A Phase 2 Safety and Efficacy Study of PRT3789 in Combination With Pembrolizumab in Patients With Advanced or Metastatic Solid Tumors and a *SMARCA4* Mutation

Timothy A. Yap, MD, PhD[1]; Martin E. Gutierrez, MD[2]; Wade T. Iams, MD[3]; Victor Moreno, MD, PhD[4]; Tatiana Hernández Guerrero, MD, PhD[5]; Guzmán Alonso, MD[6]; Carlos A. Gomez-Roca, MD[7]; Sophie Postel-Vinay, MD, PhD[8]; Fabian Acker, MD[9]; Anna R. Minchom, MB BCh, FRCP, MD(res)[10]; Natasha B. Leighl, MD, MMSc, FRCPC, FASCO[11]; William F. Novotny, MD[12]; Chris Tankersley, MSc[12]; Megan Henry, BSN, RN[12]; Sunhee Ro, PhD[12]; Michael J. Chisamore, PhD[13]; John Bridgewater, MRCP, PhD, FRCP[14]

[1]The University of Texas, MD Anderson Cancer Center, Houston, TX, USA; [2]Hackensack University Medical Center, Hackensack, NJ, USA; [3]Tennessee Oncology, Nashville, TN, USA; [4]START Madrid-FJD, Hospital Universitario Fundación Jiménez Díaz, Madrid, Spain; [5]START Barcelona - HM Nou Delfos, Barcelona, Spain; [6]NEXT Barcelona, Barcelona, Spain; [7]CHU de Toulouse, Toulouse, France; [8]Gustave-Roussy Cancer Institute, Villejuif, France; [9]Goethe-Universität Hospital Frankfurt, Frankfurt, Germany; [10]The Royal Marsden, Sutton, UK; [11]Princess Margaret Cancer Centre, Toronto, ON, Canada; [12]Prelude Therapeutics Incorporated, Wilmington, DE, USA; [13]Merck & Co., Inc., Rahway, NJ, USA; [14]University College Hospital, London, UK

137a

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® or the author of this poster.

BACKGROUND

- Mutations in genes encoding subunits of the SWI/SNF chromatin remodeling complex have been observed in 20% of all human cancers[1,2]
 - The SWI/SNF complex contains 1 of 2 ATP enzymatic subunits, either SMARCA2 (also known as BRM) or SMARCA4 (also known as BRG1)[1]
 - Because SMARCA2 and SMARCA4 function as mutually exclusive catalytic subunits of the SWI/SNF complex, cells exhibiting SMARCA4 loss rely on its paralog, SMARCA2, making SMARCA2 an attractive therapeutic target[3-5]
- In NSCLC, *SMARCA4* mutations are observed in approximately 10% of cases and are associated with more aggressive and invasive disease and shorter survival[6-8]
- PRT3789 is a potent and selective, intravenous, VHL-based SMARCA2 degrader (**Figure 1**) which induces proteosome-dependent degradation of SMARCA2 and robust synthetic lethality in *SMARCA4*-deficient cancers

Figure 1. Characterization of PRT3789

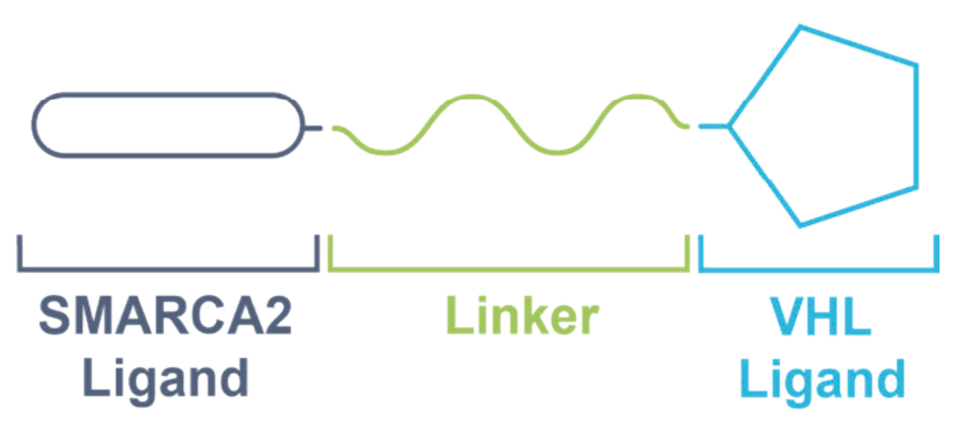

SMARCA2
Ligand
Linker
VHL
Ligand

PRT3789, a VHL-recruiting SMARCA2 degrader IV drug candidate

- Heterobifunctional SMARCA2 degrader
- Highly potent and selective for SMARCA2 vs SMARCA4
- Efficacious in *SMARCA4*-mutated CDX and PDX in vivo models at well-tolerated doses

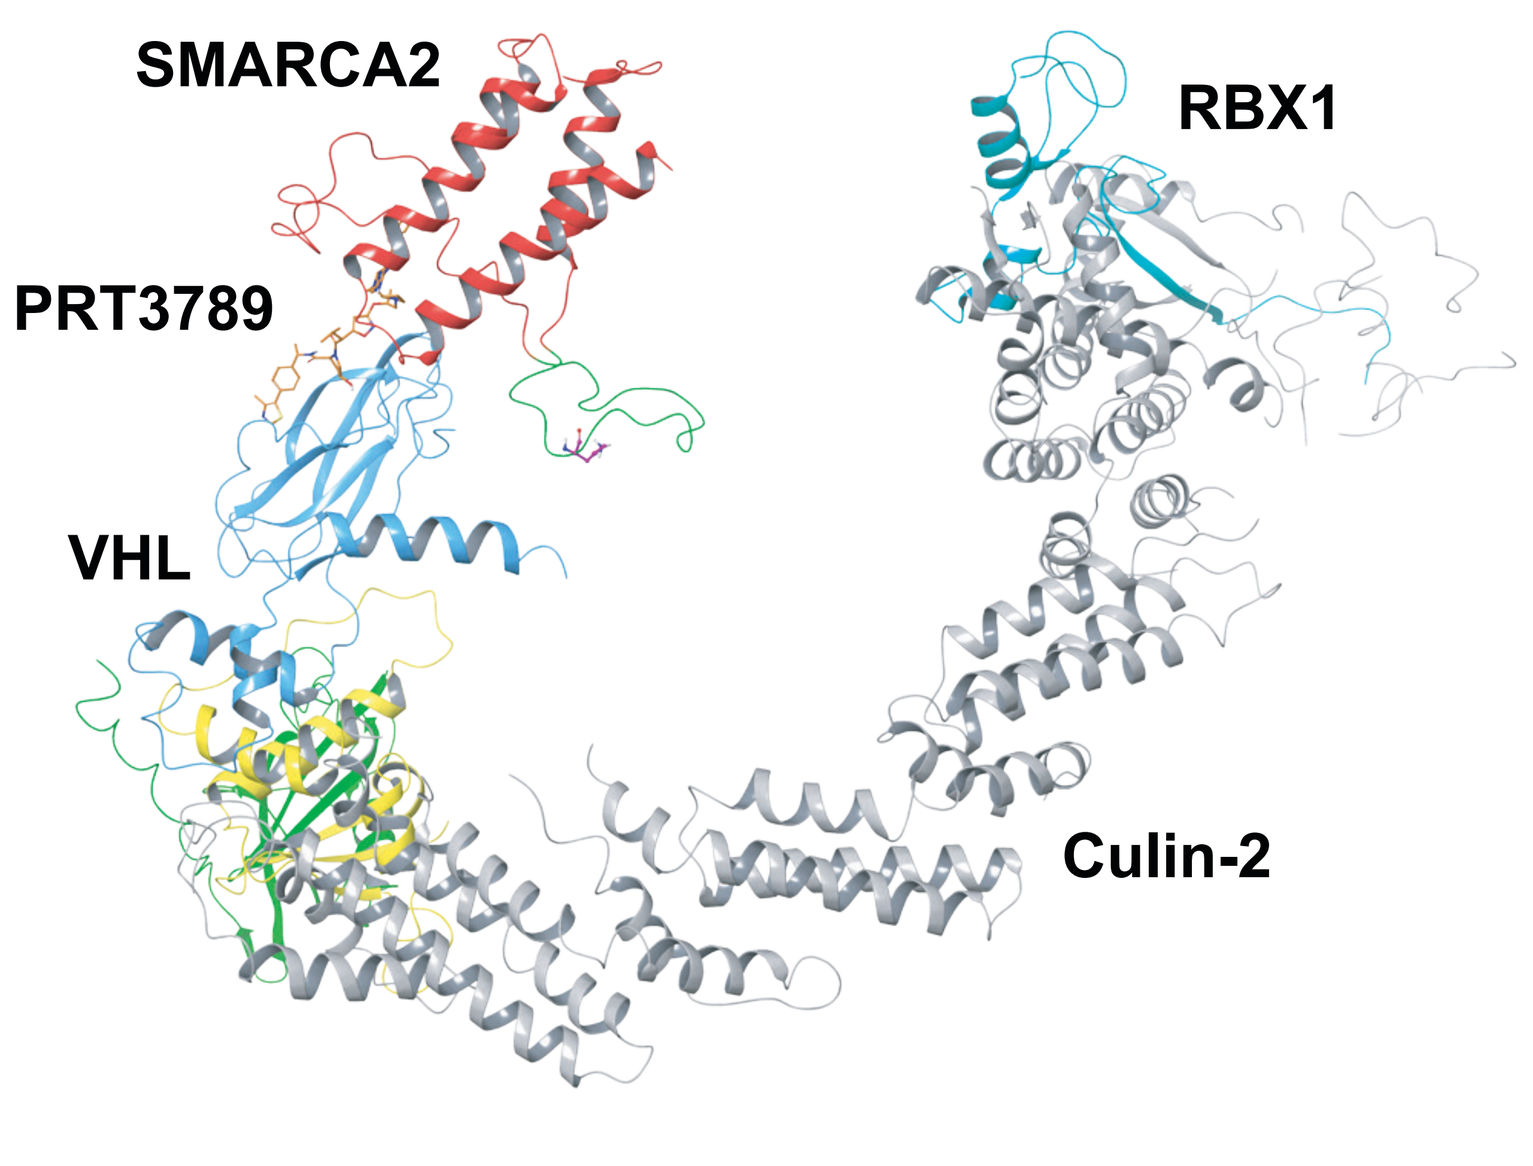

SMARCA2
RBX1
PRT3789
VHL
Culin-2

3-Dimensional structure of SMARCA2 bromodomain and E3 ligase complex (VBC/CUL2/RBX1) formed by PRT3789 SMARCA2 degrader

- PRT3789 at doses up to 500 mg IV once weekly was well tolerated and demonstrated encouraging clinical activity in patients with NSCLC and upper gastric or esophageal cancers, with LOF *SMARCA4* mutations (PRT3789-01; NCT05639751)[9]
- Although ICIs have shown remarkable clinical activity across a broad range of tumor types, various resistance mechanisms contribute to limited effectiveness in some patients. These resistance mechanisms include altered antigen presentation, reduced T-cell infiltration, and altered immune signaling pathways[8]
- PRT3789 was shown to increase antigen processing and presentation of unique MHC class I peptides and increase T-cell activity and IFN-γ production in *SMARCA4*-mutated cancer cells
- PRT3789 in combination with pembrolizumab may re-sensitize patients with cancers resistant to subsequent anti–PD-L1 therapy (**Figure 2**)

Figure 2. Scientific Rationale for Combination Treatment of PRT3789 and Anti-PD1/PD-L1 Therapy

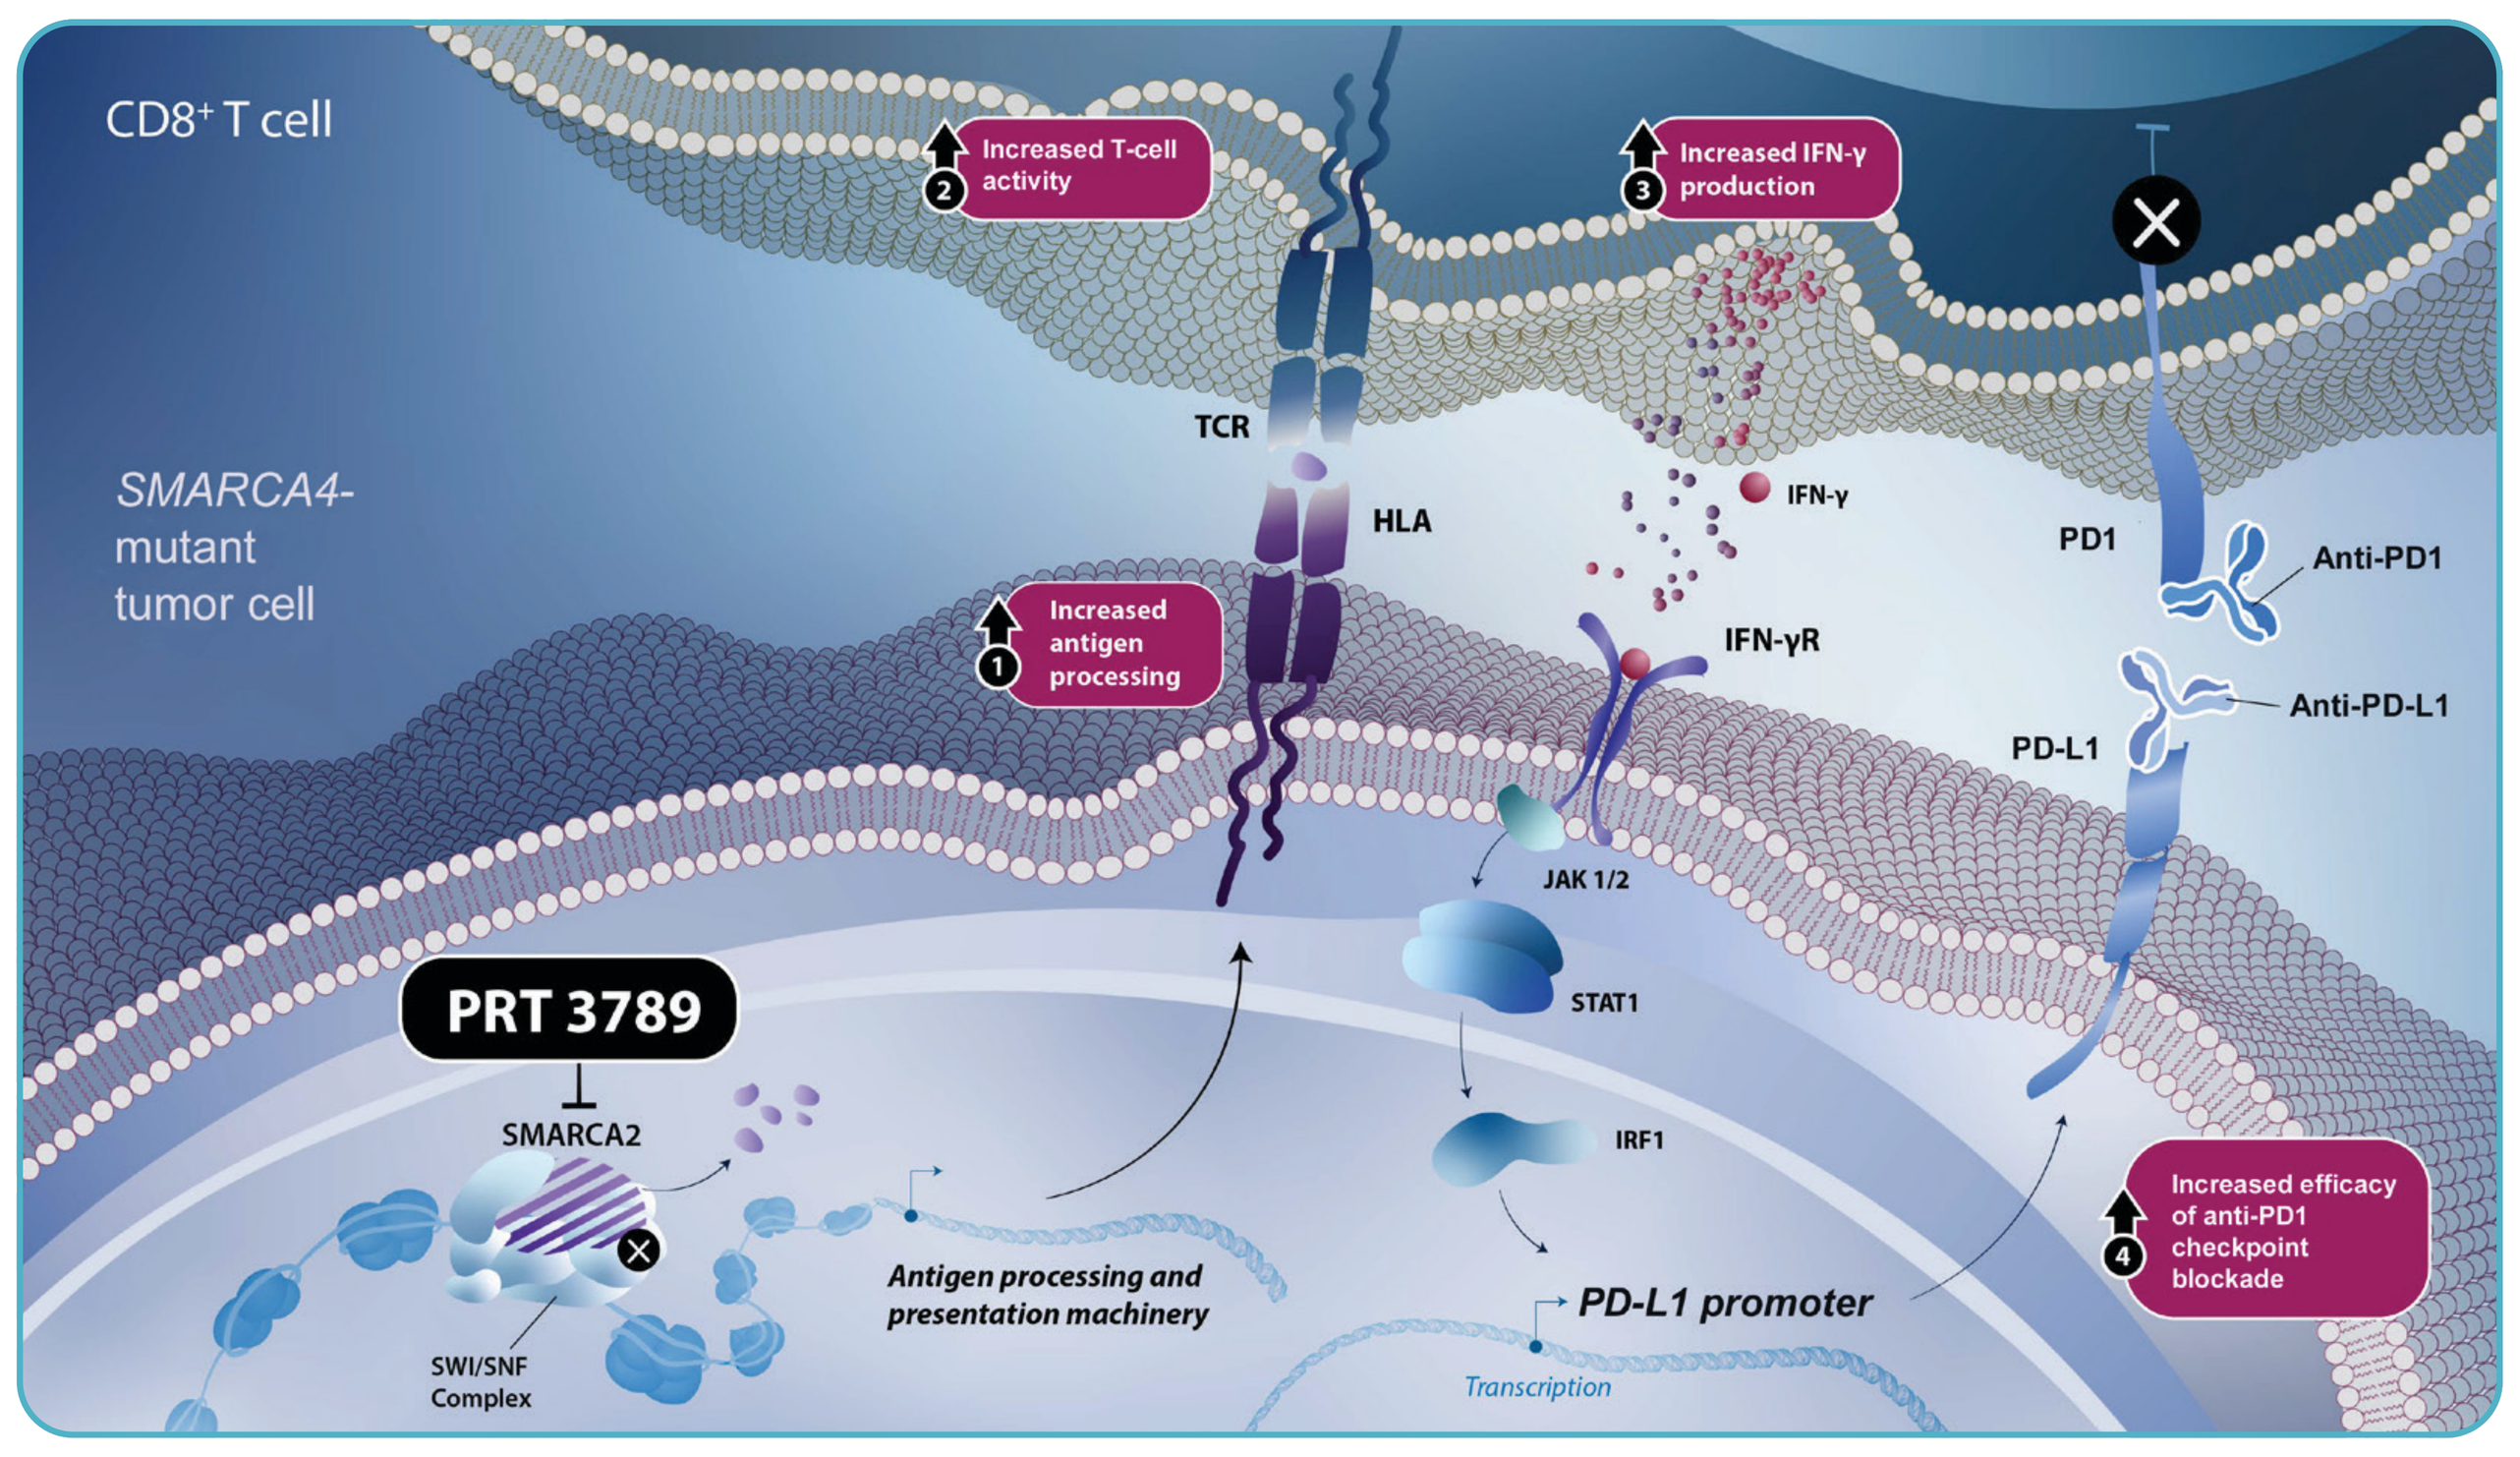

CD8+ T cell
2 Increased T-cell activity
3 Increased IFN-γ production
SMARCA4-mutant tumor cell
TCR
HLA
IFN-γ
IFN-γR
PD1
Anti-PD1
PD-L1
Anti-PD-L1
1 Increased antigen processing
JAK 1/2
STAT1
IRF1
PRT 3789
SMARCA2
SWI/SNF Complex
Antigen processing and presentation machinery
PD-L1 promoter
Transcription
4 Increased efficacy of anti-PD1 checkpoint blockade

- PRT3789 increases antigen processing and presentation machinery in *SMARCA4*-deficient NSCLC cells
- PRT3789 combined with pembrolizumab promotes T-cell activity and *SMARCA4*-deficient NSCLC cell killing
- SMARCA2 degradation promotes the effect of anti-PD1 therapy in a *SMARCA4*-deficient cancer syngeneic mouse model in vivo

METHODS

KEY OBJECTIVES

- PRT3789-02 (NCT06682806) is a phase 2, open-label, multicenter study of PRT3789 in combination with pembrolizumab in patients with advanced, recurrent, or metastatic solid tumors with a *SMARCA4* mutation (**Figure 3**)

Primary objectives

- **Part 1:** Evaluate the safety and tolerability of PRT3789 in combination with pembrolizumab in patients with advanced, recurrent, or metastatic solid tumors, with any *SMARCA4* mutation
- **Part 2:** Evaluate the efficacy of PRT3789 in combination with pembrolizumab in patients with advanced or metastatic NSCLC or upper GI/esophageal cancer, with a LOF *SMARCA4* mutation

Secondary objectives

- Evaluate the efficacy (ORR, DOR, CBR, PFS, and OS), safety and tolerability, and PK profile of PRT3789 in combination with pembrolizumab

STUDY DESIGN AND ENDPOINTS

Figure 3. PRT3789-02 Study Design

Safety Run-In

Patient Population

Any advanced, recurrent, metastatic solid tumor and any *SMARCA4* mutation

6 to 10 Patients
PRT3789 376 mg IV weekly + pembrolizumab 200 mg once every 3 weeks

6 to 10 Patients
PRT3789 283 mg IV weekly + pembrolizumab 200 mg once every 3 weeks

Stop Study

Main Study

Patient Population

Advanced, recurrent, metastatic NSCLC or upper GI/esophageal cancer and LOF *SMARCA4* mutation

Up to 40 Patients
PRT3789 376 mg IV weekly + pembrolizumab 200 mg once every 3 weeks

Up to 40 Patients
PRT3789 283 mg IV weekly + pembrolizumab 200 mg once every 3 weeks

PATIENT ELIGIBILITY

Key inclusion criteria

- **Part 1 safety run-in:** Patients aged ≥18 years with histologically or cytologically confirmed advanced, recurrent, or metastatic solid tumor malignancy with any *SMARCA4* mutation by local NGS testing or SMARCA protein expression loss per IHC. Patients must have measurable or nonmeasurable but evaluable disease
- **Part 2 main study:** Patients aged ≥18 years with histologically or cytologically confirmed advanced, recurrent, or metastatic NSCLC or upper GI/esophageal cancer with a LOF *SMARCA4* mutation by local NGS testing or SMARCA protein expression loss per IHC. Patients must have measurable disease
- Patients must meet 1 of the following criteria
 - Primary resistance to prior anti–PD1/PD-L1, defined by disease that did not respond at all and instead progressed (eg, best response of progressive disease per RECIST 1.1)
 - Acquired resistance to prior anti–PD1/PD-L1, defined as patients who received anti–PD1/PD-L1 therapy and had a partial or complete response, then progressed within 6 months
 - Received prior standard-of-care therapy but did not receive prior anti–PD1/PD-L1 therapy because the tumor PD-L1 expression was negative
 - Prior anti–PD1/PD-L1 therapy was discontinued for reasons other than disease progression, and subsequent progression >6 months following prior anti–PD1/PD-L1 therapy
- Patients must either progress on standard-of-care therapy or be ineligible for standard-of-care therapy. Patients with driver mutation in oncogenes (eg, *EGFR*, *MET*, *RET*, *ALK*, *BRAF*, *KRAS*, *ROS1*, *HER2*) are eligible after progression on approved targeted therapies, as applicable
- Adequate organ function

Key exclusion criteria

- Participants with solid tumors with known concomitant *SMARCA2* mutation or loss of protein expression (eg, SCCOHT small-cell carcinoma of the ovary hypercalcemic type or thoracic sarcomatoid tumors)
- Clinically significant or uncontrolled cardiac disease, uncontrolled electrolyte disorders, uncontrolled or symptomatic CNS metastases or leptomeningeal disease and/or carcinomatous meningitis
- History of or current noninfectious pneumonitis or interstitial lung disease
- Active autoimmune disease or diagnosis of immunodeficiency disease/disorder
- Concurrent treatment with strong or moderate CYP3A4 inhibitor or inducer
- Patients who received prior treatment with an agent directed to a stimulatory or co-inhibitory T-cell receptor (eg, CTLA-4, OX40, CD137) other than an anti–PD1, anti–PD-L1, or anti–PD-L2 agent

ENROLLMENT AND STATUS

This study is currently active with approximately 20 sites in North America, Europe, and Asia Pacific

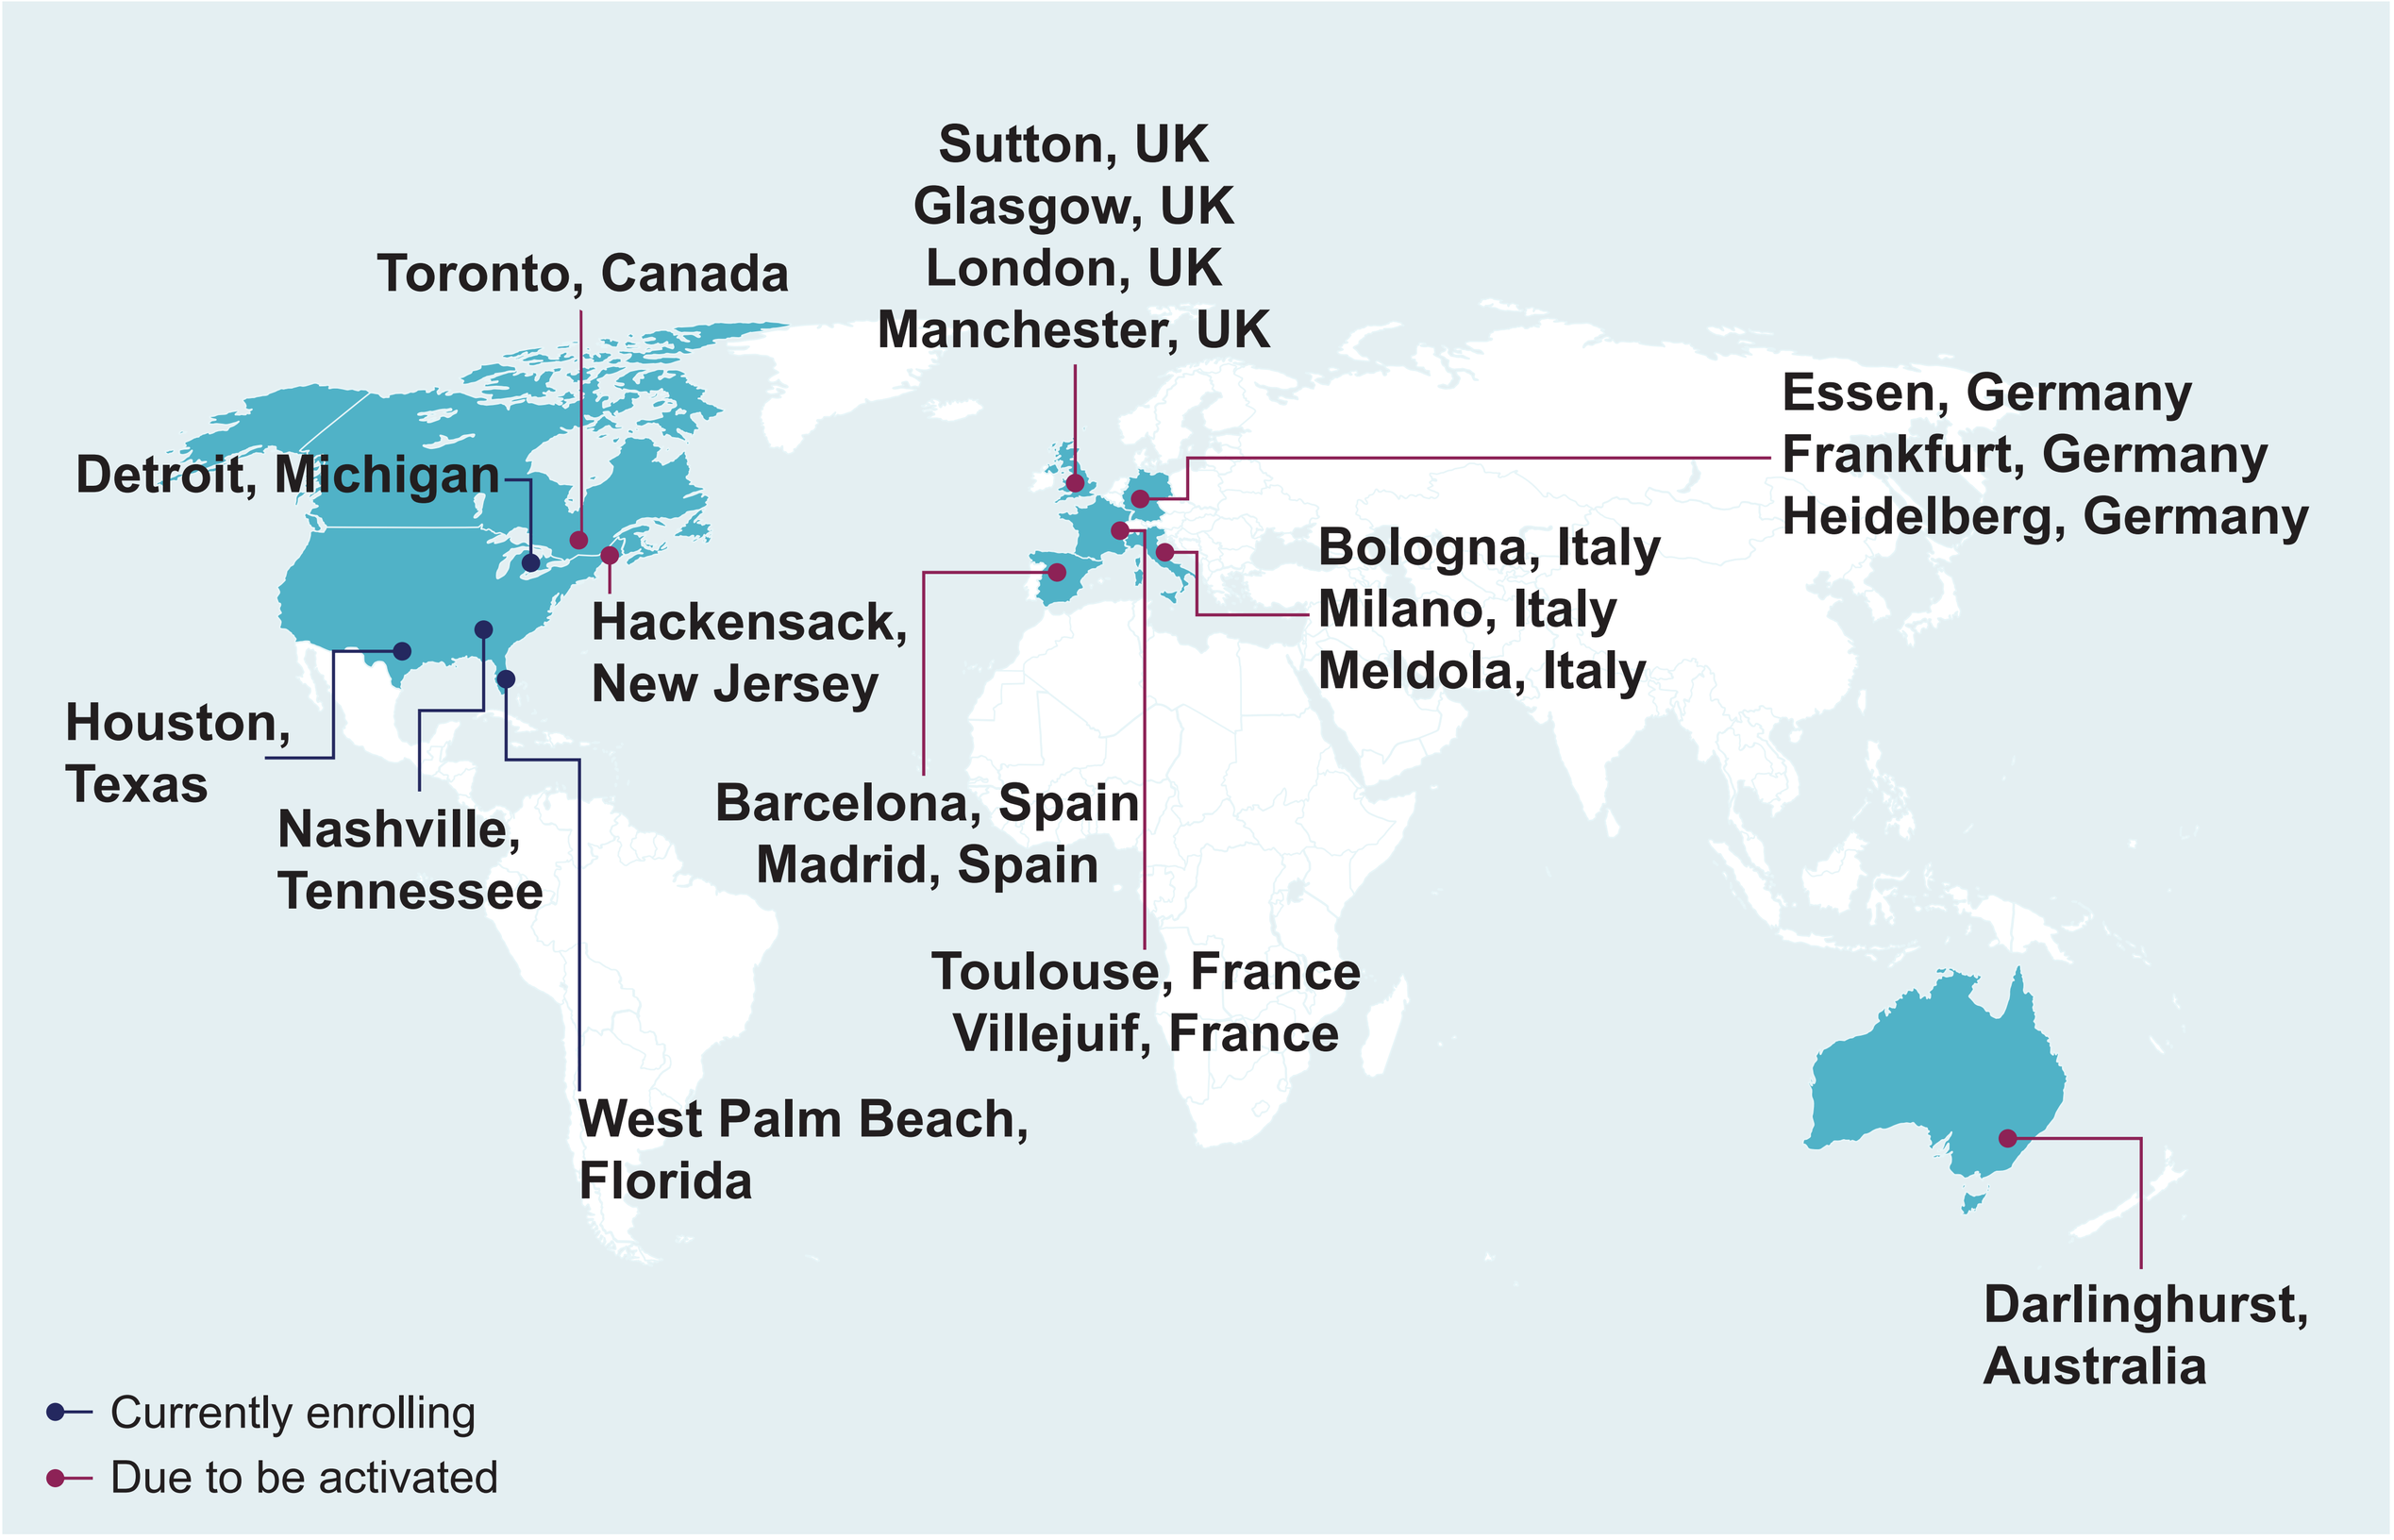

Sutton, UK
Glasgow, UK
London, UK
Manchester, UK
Toronto, Canada
Essen, Germany
Frankfurt, Germany
Heidelberg, Germany
Detroit, Michigan
Bologna, Italy
Milano, Italy
Meldola, Italy
Hackensack, New Jersey
Houston, Texas
Nashville, Tennessee
Barcelona, Spain
Madrid, Spain
Toulouse, France
Villejuif, France
West Palm Beach, Florida
Darlinghurst, Australia
Currently enrolling
Due to be activated

REGISTRATION

This study is registered at ClinicalTrials.gov (NCT06682806)

ABBREVIATIONS

ALP, alkaline phosphate; ALT, alanine aminotransferase; ANC, absolute neutrophil count; aPTT, activated partial thromboplastin time; AST, aspartate aminotransferase; ATP, adenosine triphosphate; BRG1, Brahma-related gene 1; BRM, Brahma; CBR, clinical benefit rate; CD137, tumor necrosis factor receptor superfamily member 9; CDX, cell-derived xenograft; CNS, central nervous system; CrCl, creatinine clearance; ctDNA, circulating tumor DNA; CTLA-4, cytotoxic t-lymphocyte-associated protein 4; CUL2, Cullin 2; CYP3A4, cytochrome P450 3A4; DLT, dose-limiting toxicity; DOR, duration of response; ECOG PS, Eastern Cooperative Oncology Group performance status; EGFR, epidermal growth factor receptor; GI, gastrointestinal; ICI, immune checkpoint inhibitor; IFN-γ, interferon gamma; IFN-γR, IFN-γ receptor; IHC, immunohistochemistry; INR, international normalized ratio; IV, intravenous; LOF, loss of function; MHC, major histocompatibility complex; NGS, next-generation sequencing; NSCLC, non-small cell lung cancer; ORR, overall response rate; OS, overall survival; OX40, tumor necrosis factor receptor superfamily member 4; PD, pharmacodynamics; PD1, programmed cell death protein 1; PD-L1, programmed death-ligand 1; PDX, patient-derived xenograft; PFS, progression-free survival; PK, pharmacokinetics; PT, prothrombin time; RBX1, ring-box 1; RECIST, response evaluation criteria in solid tumors; SCCOHT, small-cell carcinoma of the ovary hypercalcemic type; SMARCA, SWI/SNF related matrix associated actin dependent regulator of chromatin subfamily a member; SWI/SNF, switch/sucrose non-fermentable; ULN, upper limit of normal; VBC, VHL binding complex; VHL, Von Hippel-Lindau.

REFERENCES

1. Shain AH, Pollack JR. *PLoS ONE*. 2013;8(1):e55119.
2. Fernando TM, et al. *Nat Commun*. 2020;11(1):5551.
3. Helming KC, et al. *Cancer Cell*. 2014;26(3):309-317.
4. Hoffman GR, et al. *Proc Natl Acad Sci U S A*. 2014;111(8):3128-3133.
5. Oike T, et al. *Cancer Res*. 2013;73(17):5508-5518.
6. Dagogo-Jack I, et al. *J Thorac Oncol*. 2020;15(5):766-776.
7. Schoenfeld AJ, et al. *Clin Cancer Res*. 2020;26(21):5701-5708.
8. Alessi JV, et al. *J Thorac Oncol*. 2023;18(6):731-743.
9. Yap TA, et al. JSMO 2025. Abstract 101356.

CORRESPONDENCE

Timothy A. Yap, MD, PhD
The University of Texas MD Anderson Cancer Center Houston, TX, USA
TYap@mdanderson.org

Please contact **PRT3789-02@preludetx.com** for more information

ACKNOWLEDGMENTS

- We thank the patients and their caregivers for participating in PRT3789-02, and the study investigators, coordinators, and healthcare staff at each study site
- This study is in collaboration with Merck Sharp & Dohme LLC, a subsidiary of Merck & Co., Inc., Rahway, NJ, USA
- Funding support for PRT3789-02 was provided by Prelude Therapeutics Incorporated
- Medical writing support was provided by Miriam Cohen, PhD, ISMPP CMPP of Team 9 Science, funded by Prelude Therapeutics Incorporated

Presented at the 2025 American Society of Clinical Oncology Annual Meeting (ASCO), May 30 - June 3, 2025; Chicago IL