The Brain Penetrant CDK4/6 Inhibitor, PRT3645, is Highly Effective in Combination with Other Targeted Therapies in Preclinical Models of Breast Cancer, CRC and NSCLC

Yue Zou, Kirsten Gallagher, Srijita Dhar, Andrew Buesking, Jack Carter, Anthony Reichelderfer, Jimin Park, Sarah Pawley, Ryan Holmes, Xiaowei Wu, Min Wang, Joseph Rager, Tom Emm, Stefan Ruepp, Bo Shen, Miles Cowart, Andrew Combs, Kris Vaddi, Sandy Geeganage, Peggy Scherle, Sang Hyun Lee

Prelude Therapeutics Incorporated, Wilmington, DE; contact: yzou@preludetx.com

5710

Prelude THERAPEUTICS

Background

- Deregulation of the cell cycle is a key characteristic of cancer, with the overactivity and increased expression of cyclin-dependent kinases (CDKs) frequently driving disease progression.[1]
- CDK4 and CDK6 play pivotal roles in controlling the cell's entry into the S phase, which is essential for DNA replication and cell division, making them significant factors in the onset and development of numerous cancers.[2,3]
- At present, three CDK4/CDK6 inhibitors are approved for the treatment of estrogen receptor-positive (ER+), HER2-negative breast cancer. They are being explored in other cancer indications as well.[4]

Objective

- To bolster the therapeutic potential of PRT3645 in tumor types with notable unmet needs, we investigated the synergistic effects of combining PRT3645 with established FDA-approved drugs in various preclinical models.

Key Findings

- PRT3645 demonstrated synergistic potential in vitro and led to tumor regression in vivo when combined with a clinically approved selective estrogen receptor degrader (SERD) in a breast cancer patient derived xenograft model (PDX) harboring the ESR1 Y537S mutation.
- PRT3645 was well-tolerated and demonstrated notable anti-tumor efficacy as a monotherapy. The efficacy was further enhanced when combined with a MEK1/2 inhibitor in preclinical models with RAS/RAF/MEK/ERK pathway activation driven by either a KRAS or BRAF class III mutation.
- Co-targeting CDK4/6 and CDK2 attenuated RB phosphorylation and augmented cell cycle arrest in TNBC cells harboring CCNE1 amplification and CDKN2A loss.

Results

Table 1. PRT3645 is a potent CDK4/6 inhibitor with biased selectivity for CDK4

	PRT3645
Biochemical IC_{50} (CDK4, nM)[1]	3
Biochemical IC_{50} (CDK6, nM)[2]	14
Proliferation IC_{50} (MCF7, nM)	47
Proliferation IC_{50} (U87 MG, nM)	21
Fold selectivity CDK4 vs other CDKs	**PRT3645**
CDK1	>1000
CDK2	>1000
CDK3	>500
CDK5	>1000
CDK7	>1000
CDK9	>1000

Biochemical TR-FRET assay carried out at 1 mM ATP[2] or 2 mM ATP[1] and 10-day cell proliferation IC_{50} determined by CellTiter-Glo® assay.

Figure 1. The combination of PRT3645 and an ER degrader demonstrates synergistic potential in vitro in an ESR1 mutant breast cancer cell line

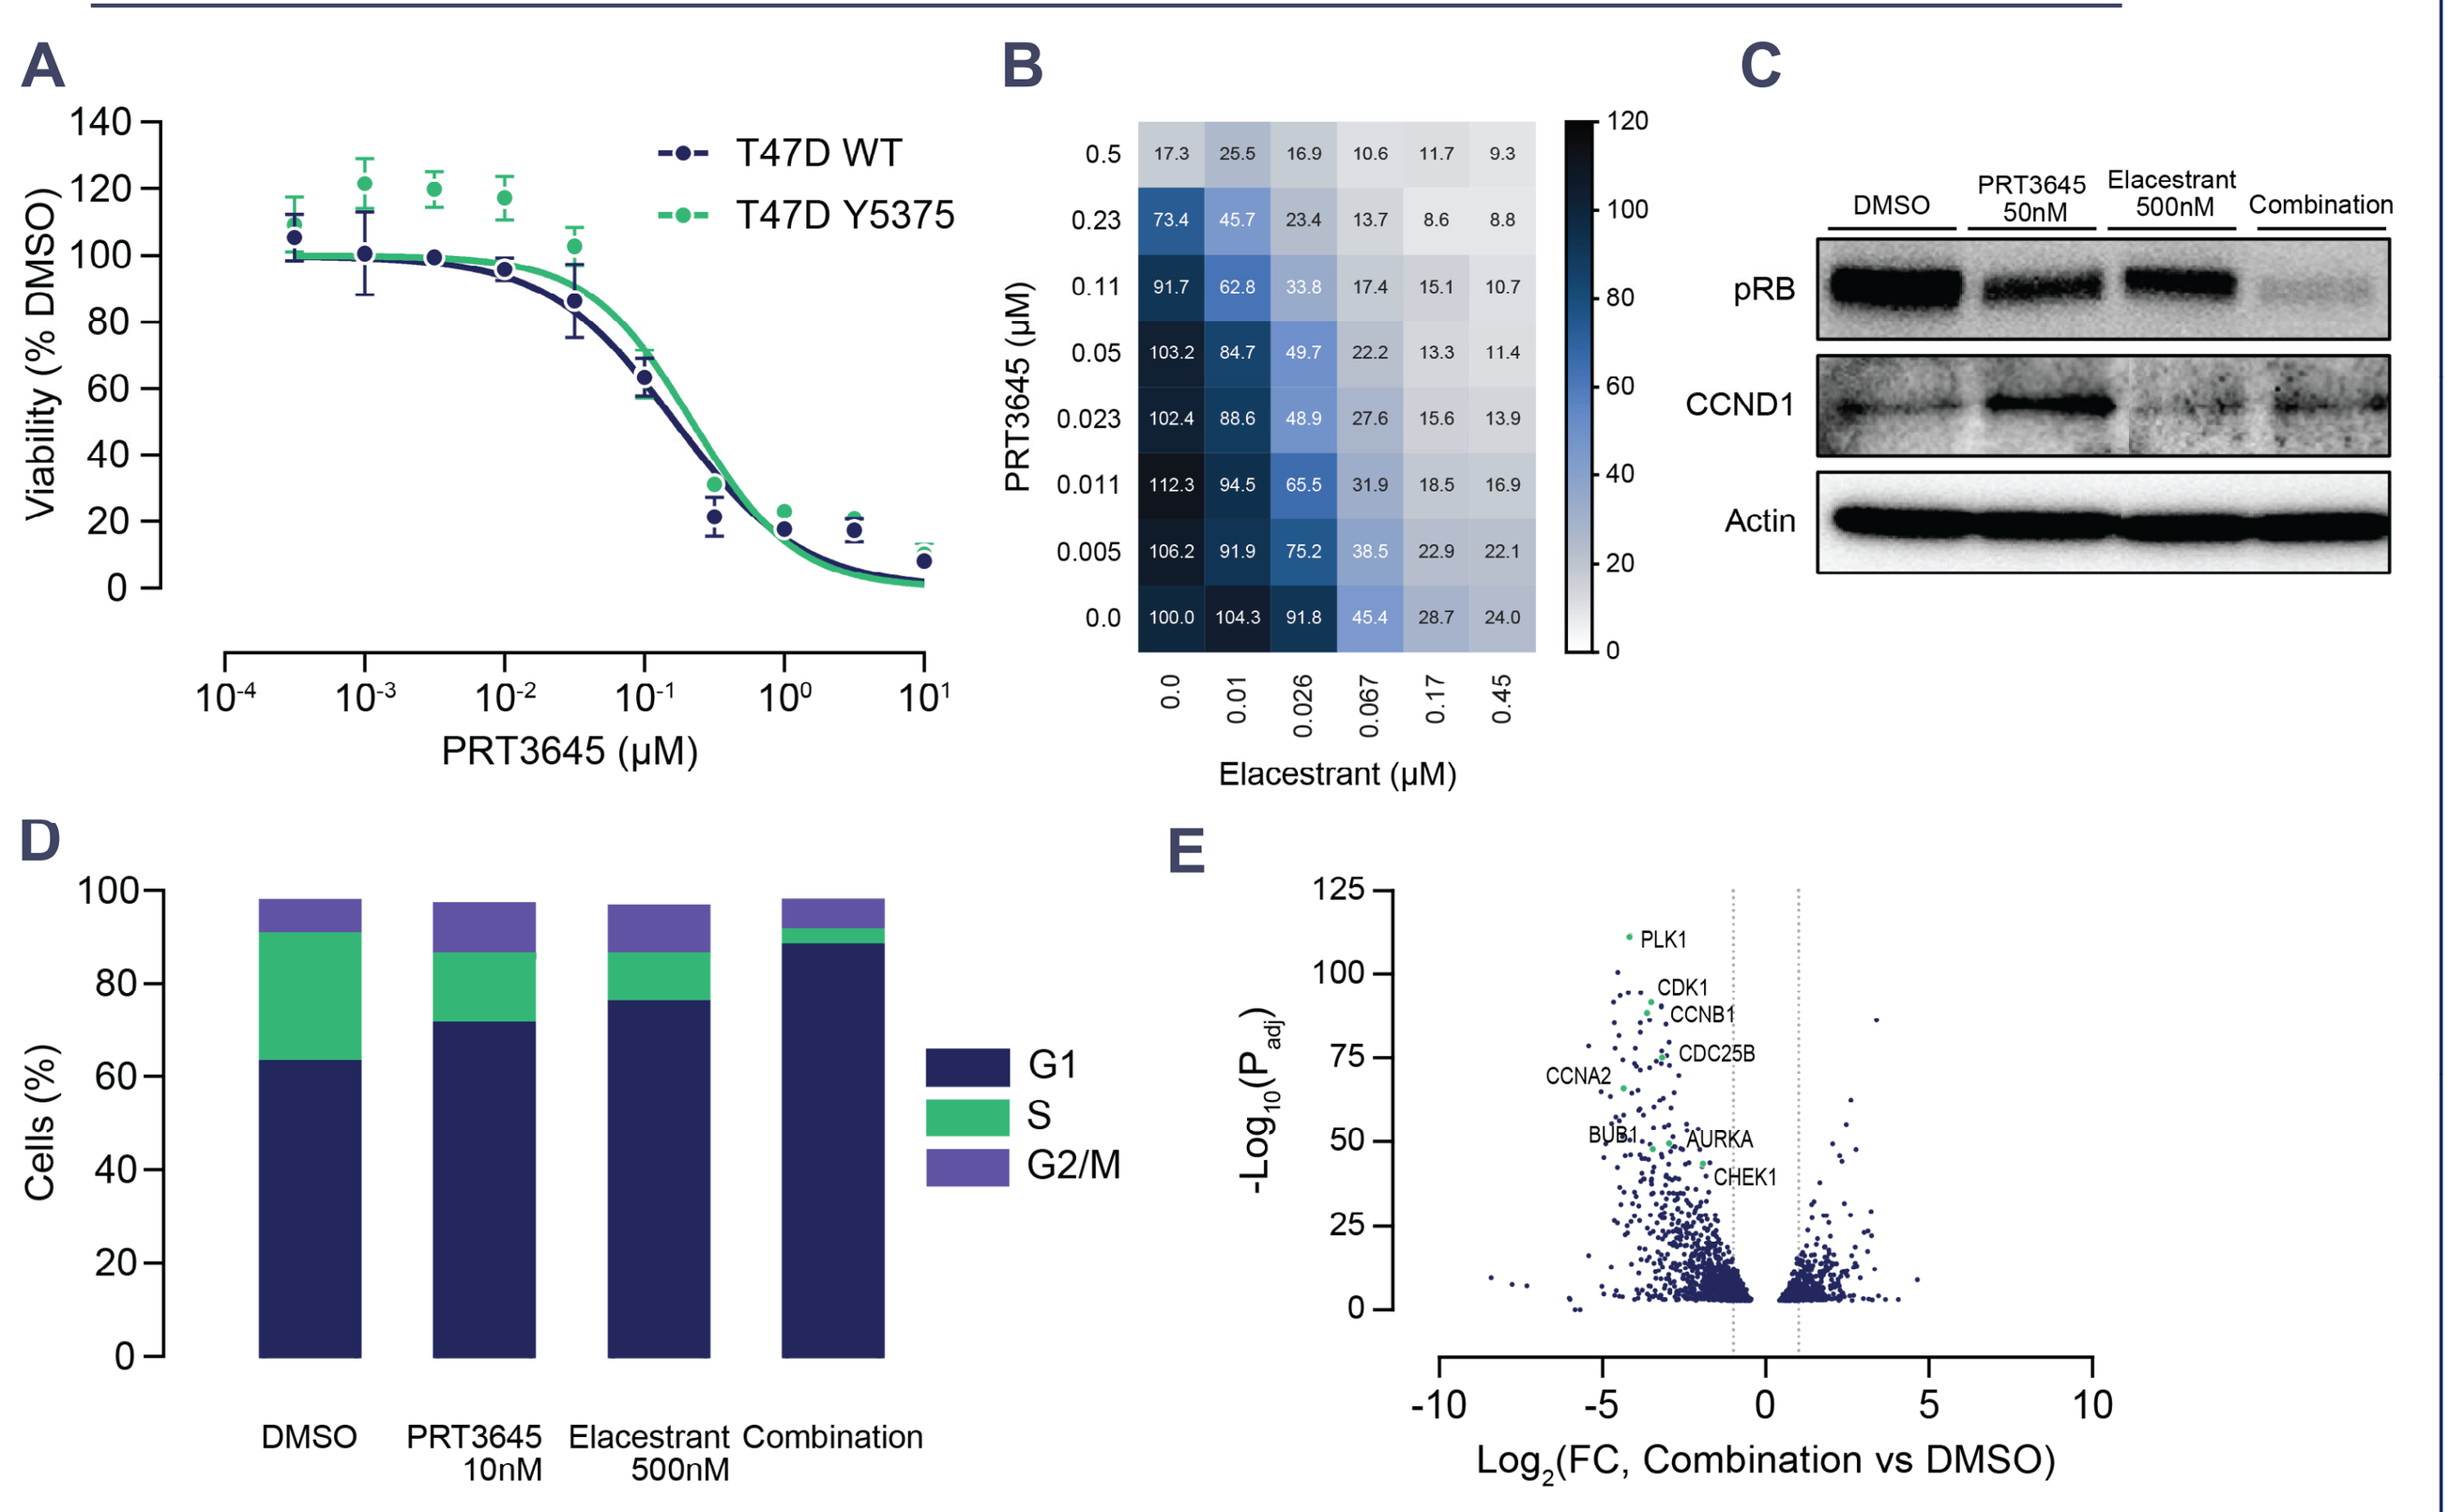
A
T47D WT
T47D Y5375
Viability (% DMSO)
PRT3645 (µM)
B
PRT3645 (µM)
Elacestrant (µM)
C
DMSO
PRT3645 50nM
Elacestrant 500nM
Combination
pRB
CCND1
Actin
D
Cells (%)
G1
S
G2/M
DMSO
PRT3645 10nM
Elacestrant 500nM
Combination
E
PLK1
CDK1
CCNB1
CDC25B
CCNA2
BUB1
AURKA
CHEK1
$-Log_{10}(P_{adj})$
Log_2(FC, Combination vs DMSO)

Combination of a CDK4/6 inhibitor and an ER degrader in breast cancer cells with ESR1 mutation in vitro A) The anti-proliferative effect of PRT3645 was assessed in both ESR1 wild type (WT) and mutant (Y537S) T47D cells using a 7-day CTG assay. **B)** The interaction landscape of the combination measured by a 7-day CTG assay in T47D (Y537S) cells. **C**) Western blots were performed to assess the levels of pRB and CCND1 in T47D (Y537S) after 24h treatment. **D)** Cell cycle progression was analyzed via flow cytometry in T47D (Y537S) after 24h treatment. **E)** Volcano plot displays Log2 (fold change combination vs DMSO) gene expression and adjusted -Log10 P adj value in T47D Y537S cells treated for 24 hours. Key cell cycle genes downregulated by PRT3645+Elacestrant combination were labeled.

Figure 2. The combination of PRT3645 and an ER degrader induces tumor regression in an ESR1 Y537S breast cancer patient derived xenograft model

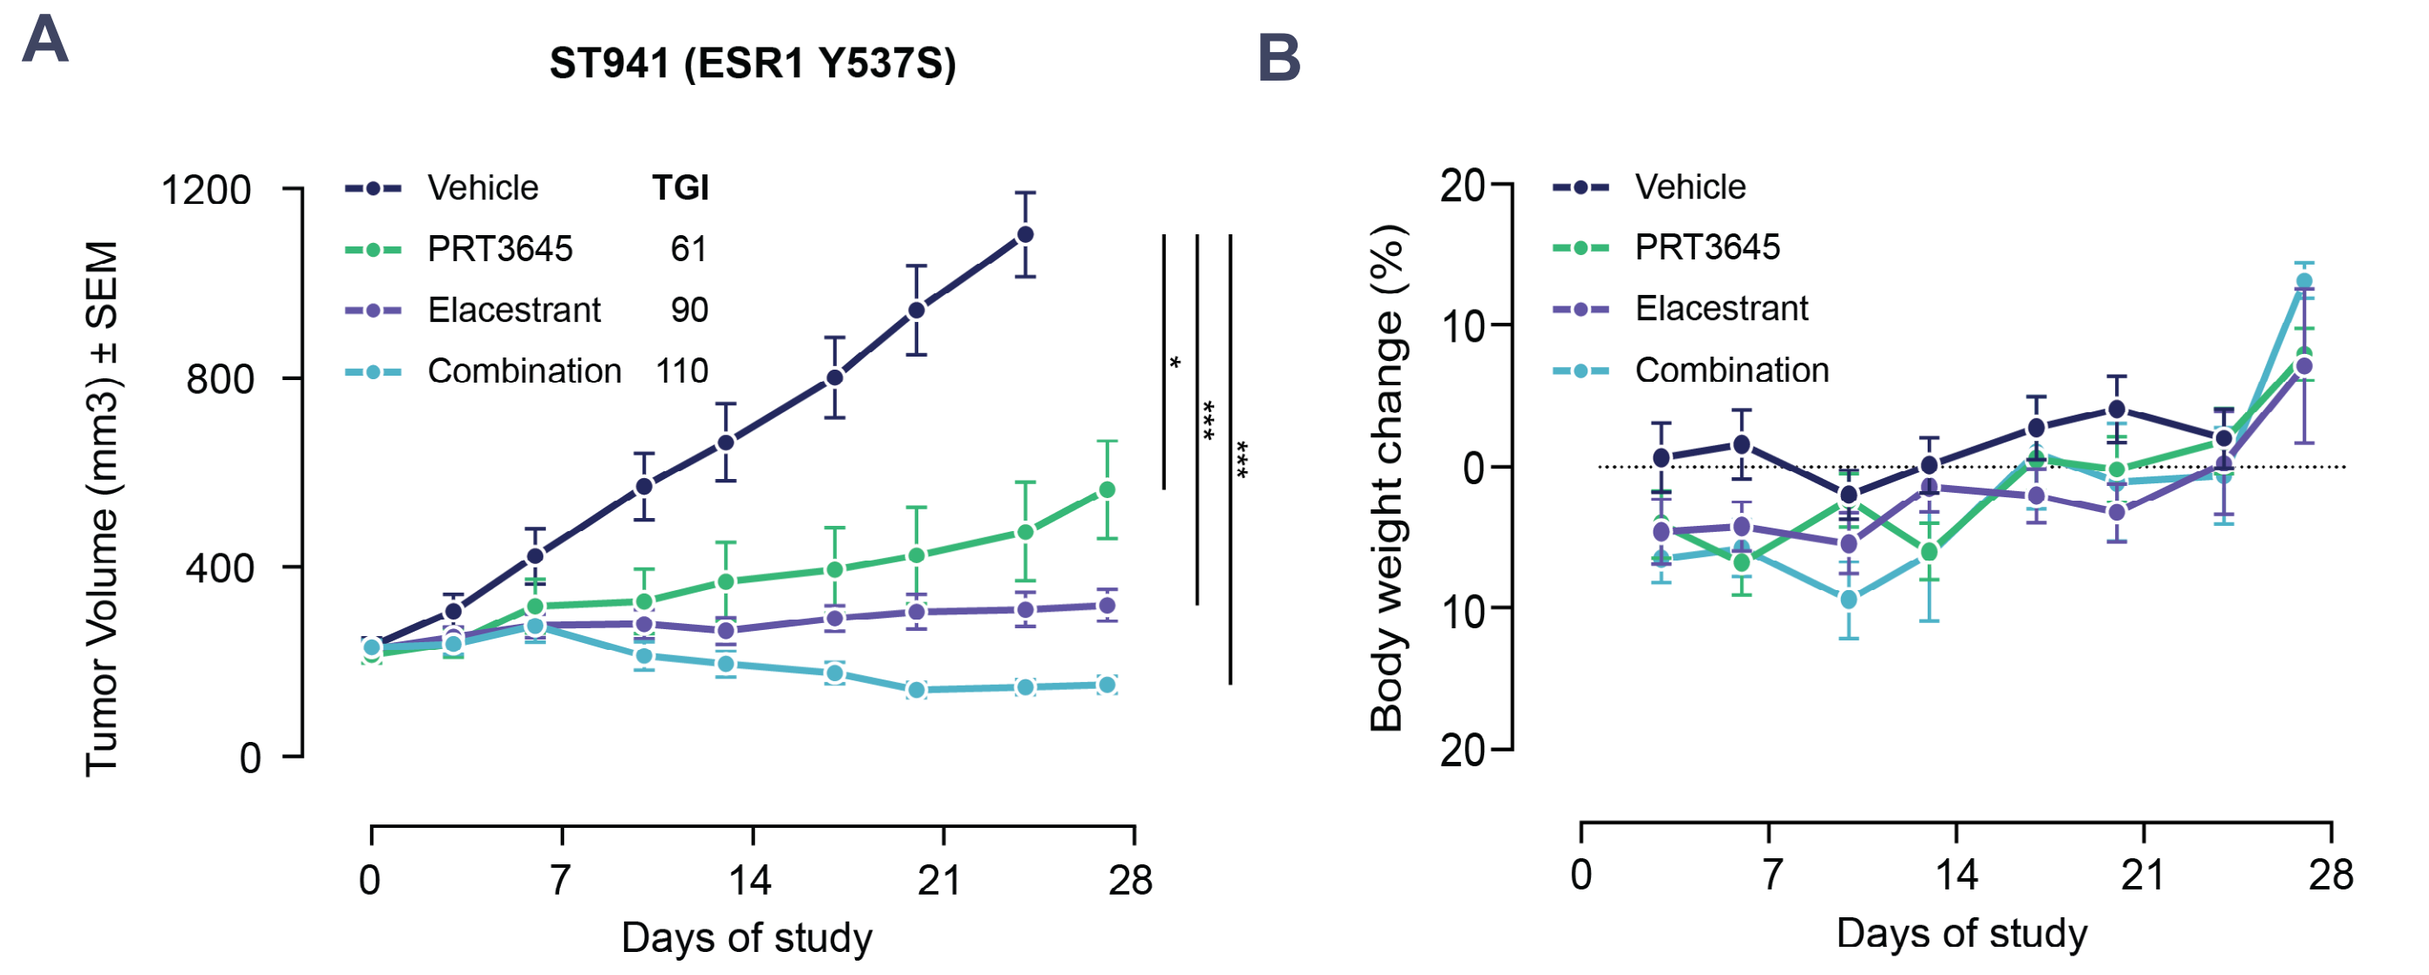
A
ST941 (ESR1 Y537S)
Vehicle
PRT3645
Elacestrant
Combination
TGI
61
90
110
Tumor Volume (mm3) ± SEM
Days of study
B
Body weight change (%)
Days of study

Combination of PRT3645 and elacestrant in vivo A) PRT3645 and elacestrant combination exhibited significant anti-tumor efficacy in the ESR1 Y537S PDX model. Elacestrant was dosed at 60mg/kg, oral daily; PRT3645 was dosed at 25mg/kg, oral daily. Drug treatments were initiated when tumors reached ~150–250 mm3 and continued through Day 27. The tumor growth inhibition ratio (TGI, %). *P<0.05, **P<0.01, ***P<0.001, versus vehicle (Mann-Whitney test). **B)** Body weight change percentage versus Day 0.

Figure 3. Combination of PRT3645 and MEK1/2 inhibitor synergistically inhibits both the cell cycle and the MAPK pathway in BRAF class III mutated colorectal (CRC) and non small cell lung cancer (NSCLC) cell lines in vitro

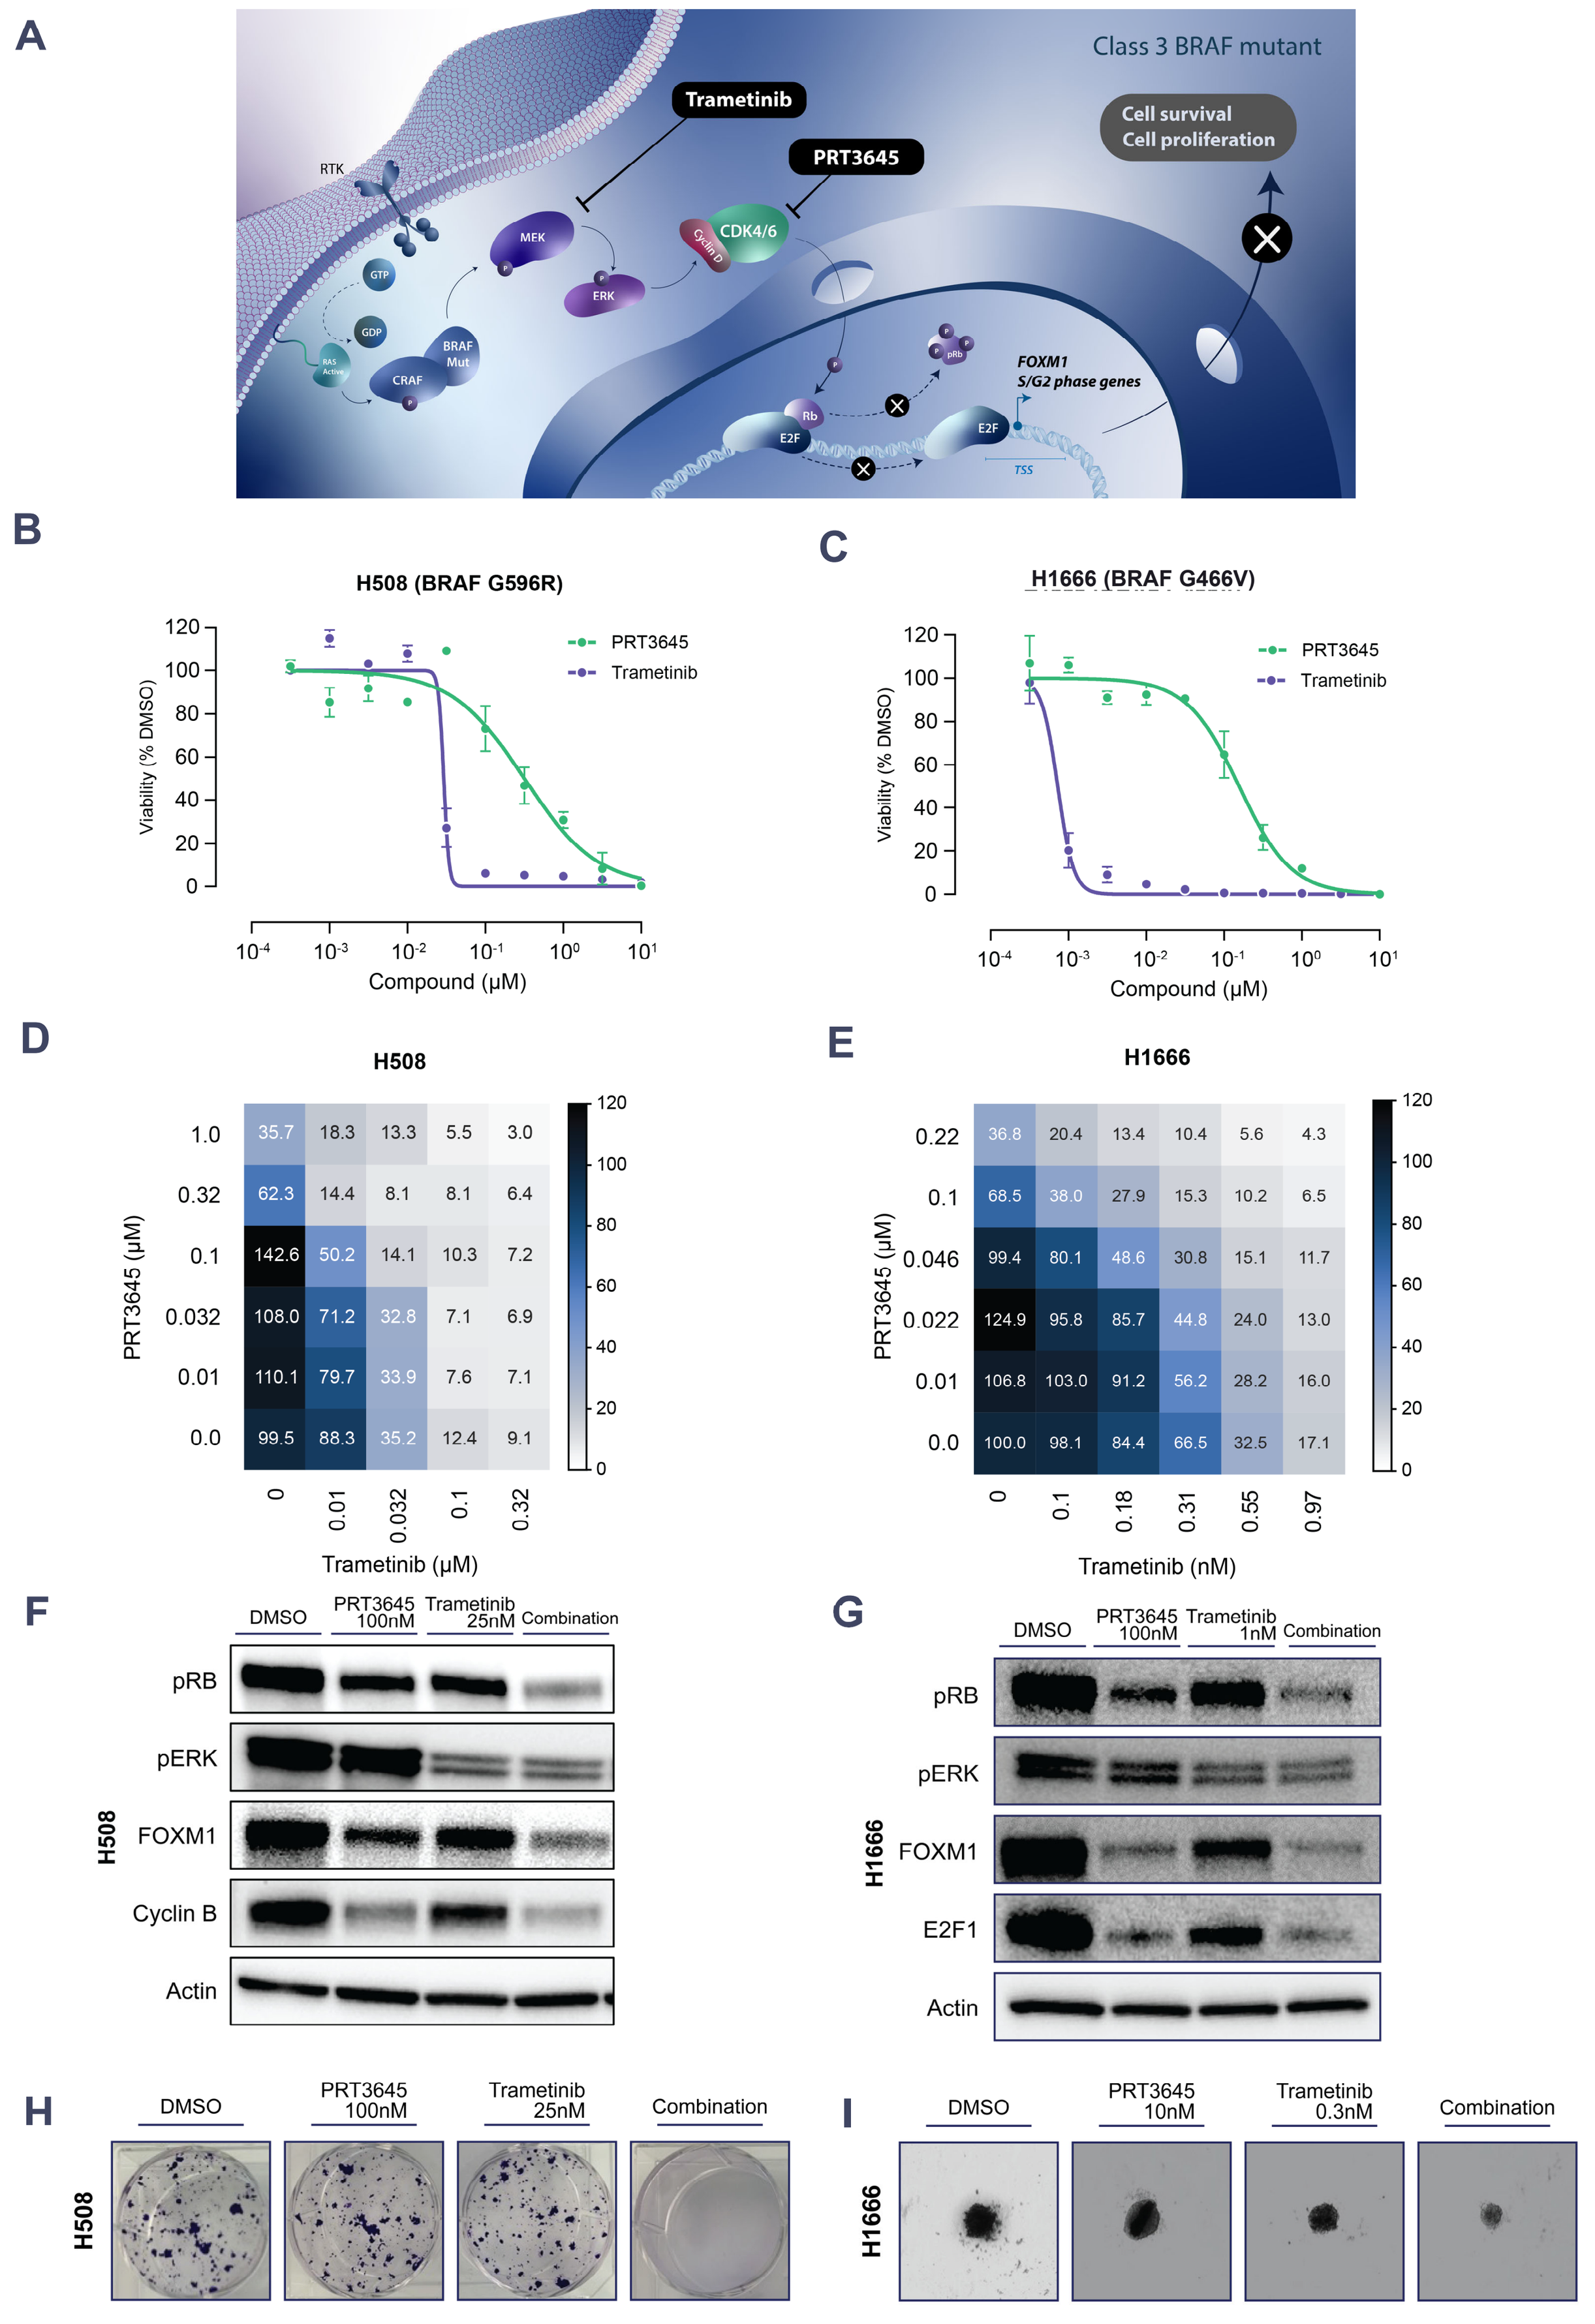
A
Class 3 BRAF mutant
Trametinib
PRT3645
Cell survival
Cell proliferation
RTK
GTP
GDP
CRAF
BRAF Mut
MEK
ERK
CDK4/6
Rb
E2F
FOXM1
S/G2 phase genes
TSS
B
H508 (BRAF G596R)
C
H1666 (BRAF G466V)
PRT3645
Trametinib
Viability (% DMSO)
Compound (µM)
D
H508
E
H1666
PRT3645 (µM)
Trametinib (µM)
Trametinib (nM)
F
DMSO
PRT3645 100nM
Trametinib 25nM
Combination
H508
pRB
pERK
FOXM1
Cyclin B
Actin
G
DMSO
PRT3645 100nM
Trametinib 1nM
Combination
H1666
pRB
pERK
FOXM1
E2F1
Actin
H
DMSO
PRT3645 100nM
Trametinib 25nM
Combination
H508
I
DMSO
PRT3645 10nM
Trametinib 0.3nM
Combination
H1666

Combination of PRT3645 and trametinib in vitro A) Proposed schematic illustrating the mechanism of action for the combination of a CDK4/6 inhibitor and a MEK inhibitor in cancers carrying the BRAF class III mutation. **B, C)** The anti-proliferative effect of PRT3645 and trametinib was assessed in both H508 and H1666 cells using the 7-day CCK8 assay. **D, E)** The interaction landscape of the combination measured by the 7-day CCK8 assay in H508 and H1666. **F, G)** Western blots were performed to evaluate alterations in gene expression related to the cell cycle and the MAPK signaling in both H508 and H1666 cells after 24h treatment. **H)** The colony formation in H508 cells and **I)** the 3D spheroid assay in H1666 cells were performed after 15 days of treatment.

Figure 4. The combination of PRT3645 and MEK1/2 inhibitor demonstrates anti-tumor efficacy in BRAF class III mutated CRC and NSCLC xenograft models

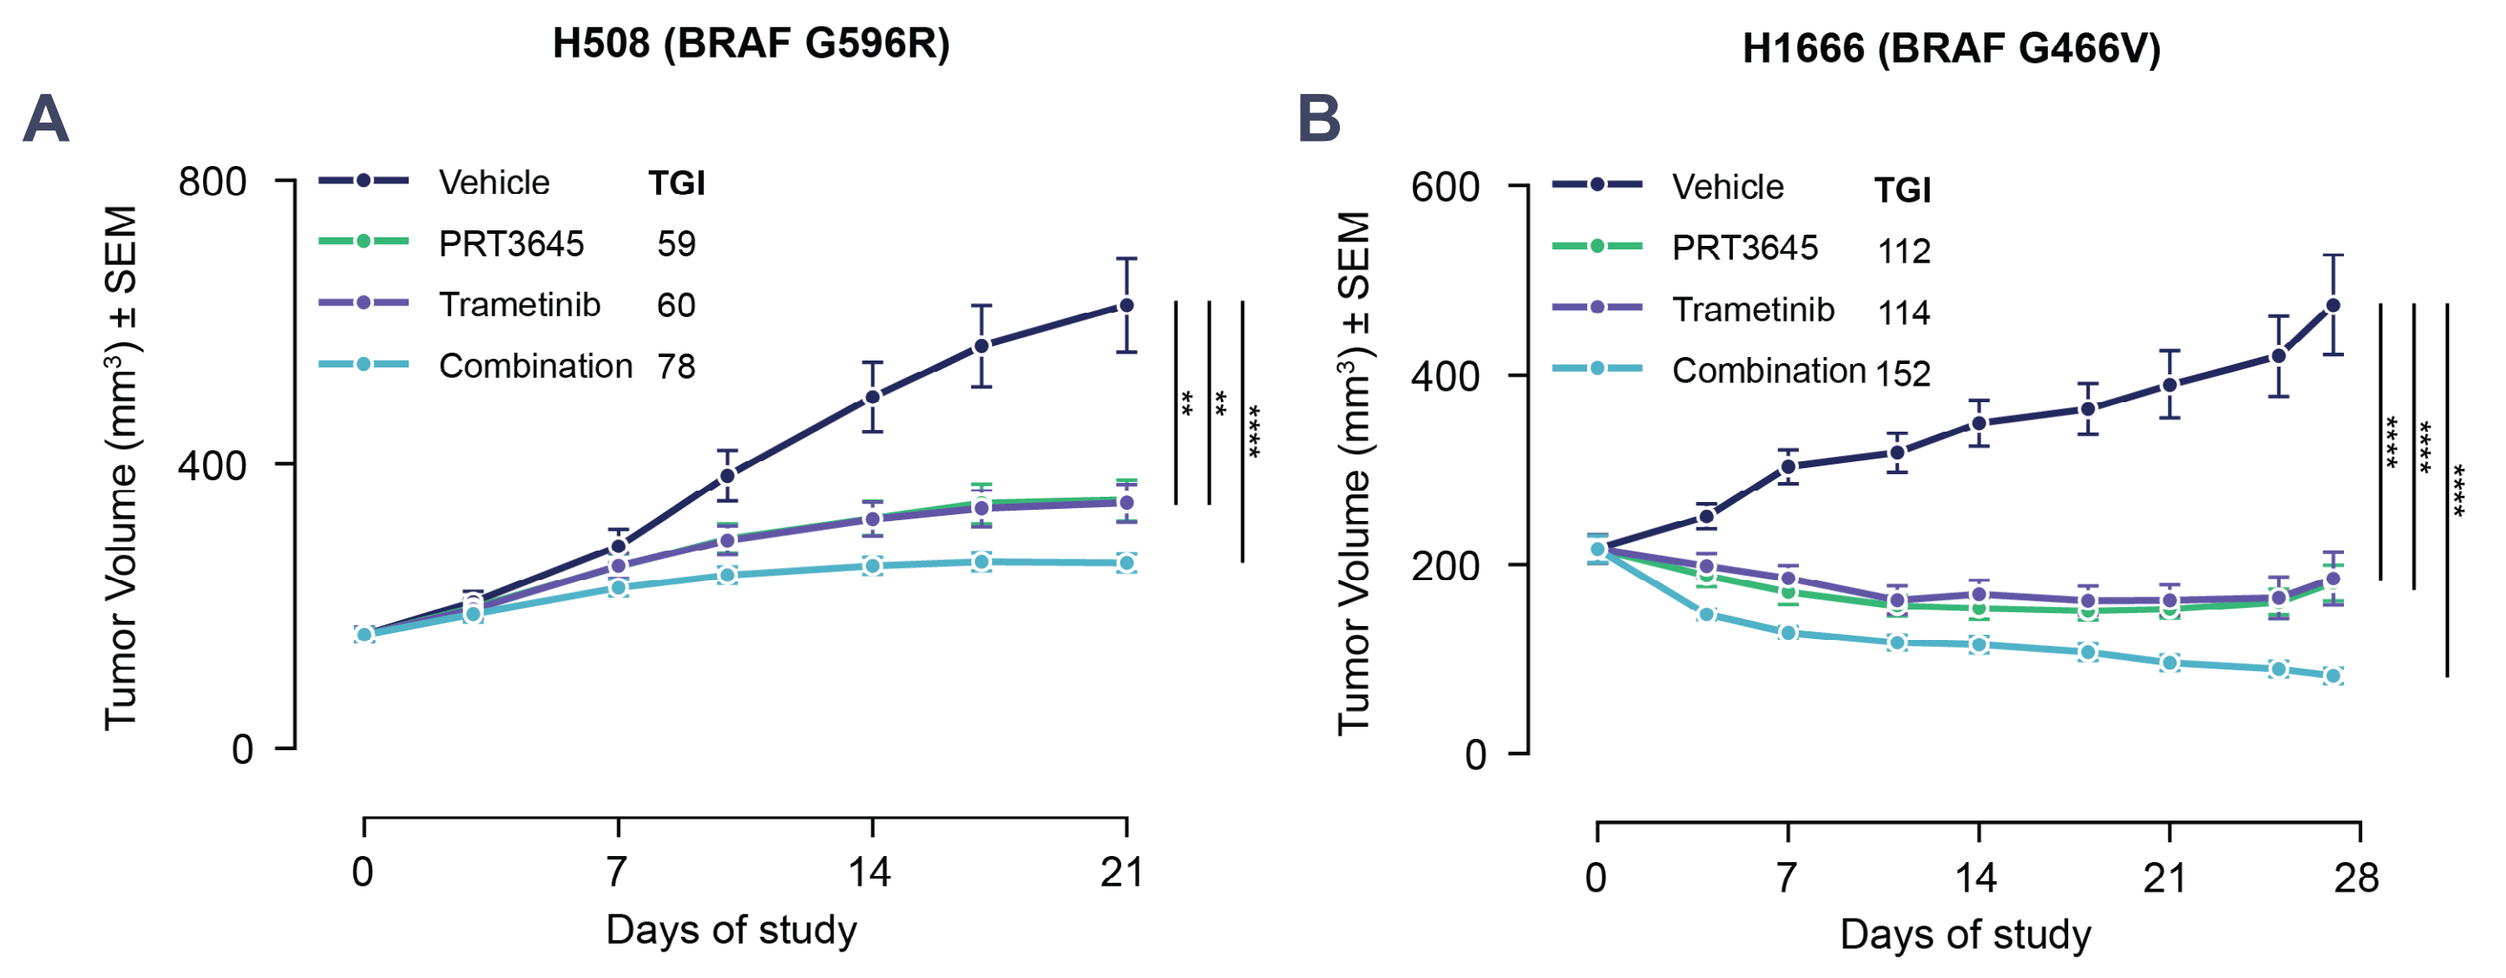
A
H508 (BRAF G596R)
Vehicle
PRT3645
Trametinib
Combination
TGI
59
60
78
B
H1666 (BRAF G466V)
Vehicle
PRT3645
Trametinib
Combination
TGI
112
114
152
Tumor Volume (mm^3) ± SEM
Days of study

Combination of PRT3645 and trametinib in vivo A) PRT3645 and trametinib combination exhibited anti-tumor efficacy in H508 xenografts. Trametinib was dosed at 0.2mg/kg, 5 days on 2 days off, oral daily. PRT3645 was dosed at 25mg/kg, oral daily. **B)** PRT3645 and trametinib combination enhanced tumor regression in H1666 xenografts. Trametinib was dosed at 0.2mg/kg, 5 days on 2 days off, oral daily. PRT3645 was dosed at 25mg/kg, oral daily. Drug treatments were initiated when tumors reached ~150–250 mm^3 and continued through Day 21 (A) Day 27 (B). The tumor growth inhibition ratio (TGI, %). **P<0.01, ***P<0.001, ****P<0.0001, versus vehicle (Mann-Whitney test).

Figure 5. Combination of PRT3645 and MEK1/2 inhibitor synergistically suppresses proliferation in KRAS G13D colorectal cancer cells by down-regulating genes involved in cell cycle and MAPK pathways

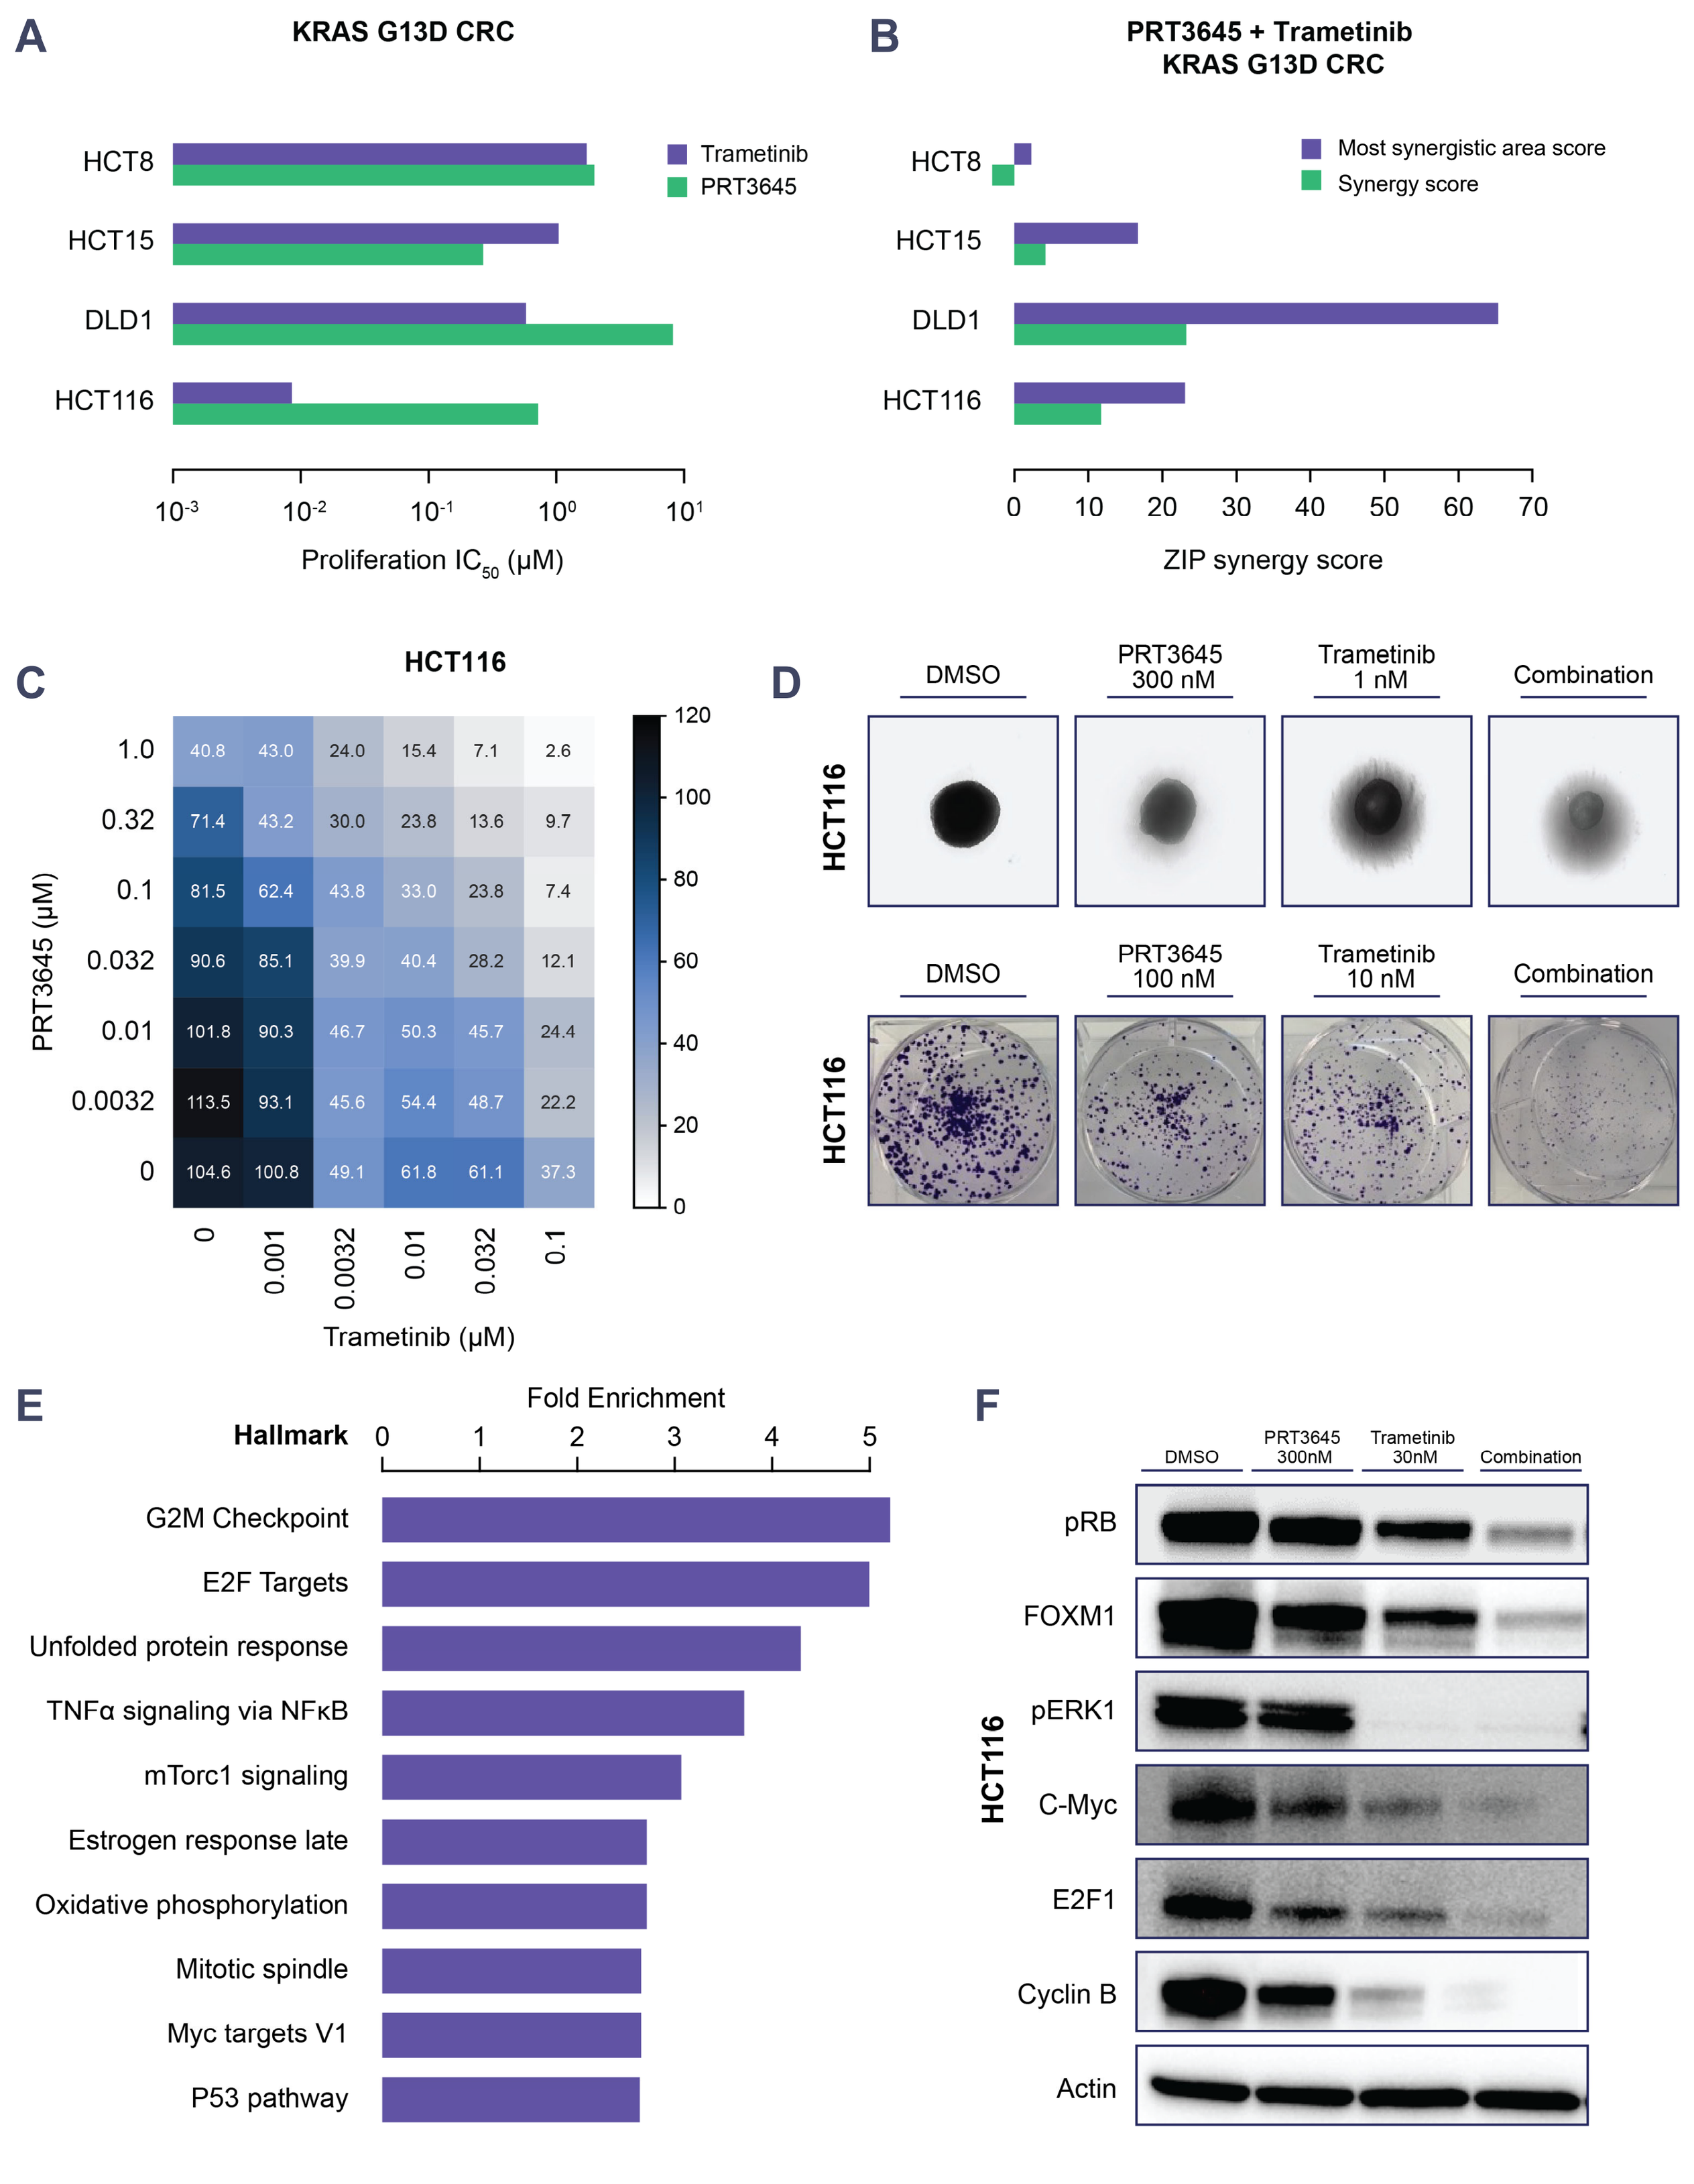
A
KRAS G13D CRC
HCT8
HCT15
DLD1
HCT116
Trametinib
PRT3645
Proliferation IC_{50} (µM)
B
PRT3645 + Trametinib
KRAS G13D CRC
Most synergistic area score
Synergy score
ZIP synergy score
C
HCT116
PRT3645 (µM)
Trametinib (µM)
D
DMSO
PRT3645 300 nM
Trametinib 1 nM
Combination
HCT116
DMSO
PRT3645 100 nM
Trametinib 10 nM
Combination
HCT116
E
Hallmark
Fold Enrichment
G2M Checkpoint
E2F Targets
Unfolded protein response
TNFα signaling via NFκB
mTorc1 signaling
Estrogen response late
Oxidative phosphorylation
Mitotic spindle
Myc targets V1
P53 pathway
F
DMSO
PRT3645 300nM
Trametinib 30nM
Combination
HCT116
pRB
FOXM1
pERK1
C-Myc
E2F1
Cyclin B
Actin

Combination of PRT3645 and trametinib in vitro A) The anti-proliferative effect of PRT3645 and trametinib was assessed in KRAS G13D cell lines by the 7-day CCK8 assay. **B)** Synergy score of PRT3645 and trametinib combination in KRAS G13D cell lines. Synergy zip scores were calculated via Synergy Finder. Zip synergy score: <-10 antagonistic; -10 to 10 additive; > 10 synergistic. **C)** The interaction landscape of the combination measured by the 7-day CCK8 assay in HCT116 cells. **D)** The 3D spheroid assay and colony formation assay were performed in HCT116 cells after 15 days treatment. **E)** Gene enrichment pathways down-regulated by PRT3645+trametinib combination in HCT116 cells after 24h treatment were analyzed in Shiny Go 0.80 with hallmark MSigDB database. **F)** Western blots were performed to evaluate alterations in gene expression related to the cell cycle and MAPK signaling in HCT116 cells after 24h treatment.

Figure 6. The combination of PRT3645 and MEK1/2 inhibitor exhibits superior efficacy to monotherapy in human colorectal HCT116 xenografts harboring KRAS G13D

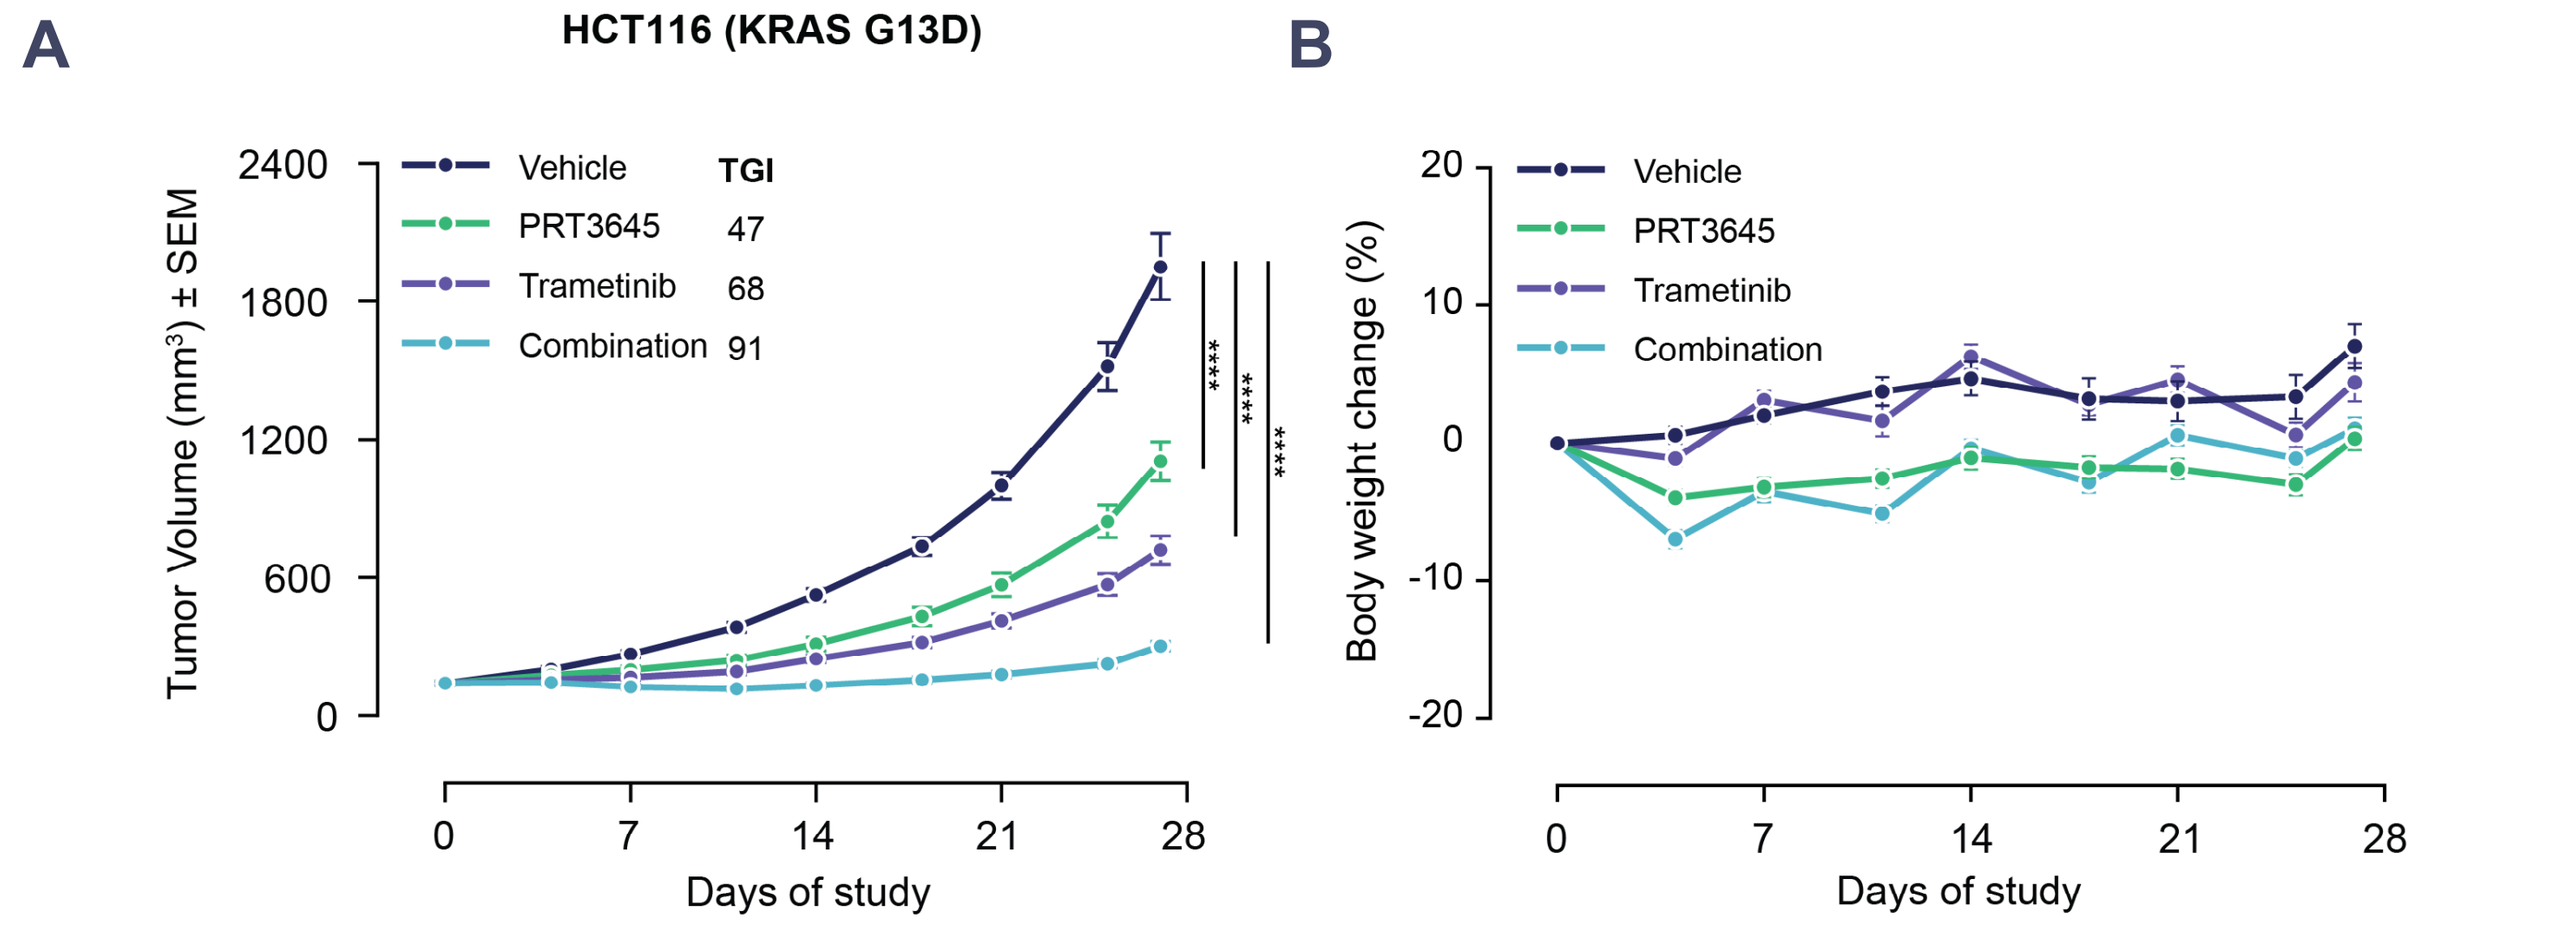
A
HCT116 (KRAS G13D)
Vehicle
PRT3645
Trametinib
Combination
TGI
47
68
91
Tumor Volume (mm^3) ± SEM
Days of study
B
Body weight change (%)
Days of study

Combination of PRT3645 and trametinib in vivo A) PRT3645 and trametinib combination exhibited anti-tumor efficacy in HCT116 xenografts. Trametinib was dosed at 0.2mg/kg, 5 days on 2 days off, oral daily. PRT3645 was dosed at 25mg/kg, oral daily. Drug treatments were initiated when tumors reached ~150–250 mm^3 and continued through Day 27. The tumor growth inhibition ratio (TGI, %). ****P<0.0001, versus vehicle (Mann-Whitney test). **B)** Body weight change percentage versus Day 0.

Figure 7. Co-targeting CDK4/6 and CDK2 attenuates RB phosphorylation and augments cell cycle arrest in TNBC HCC1806 cells harboring CCNE1 amplification and CDKN2A loss in vitro

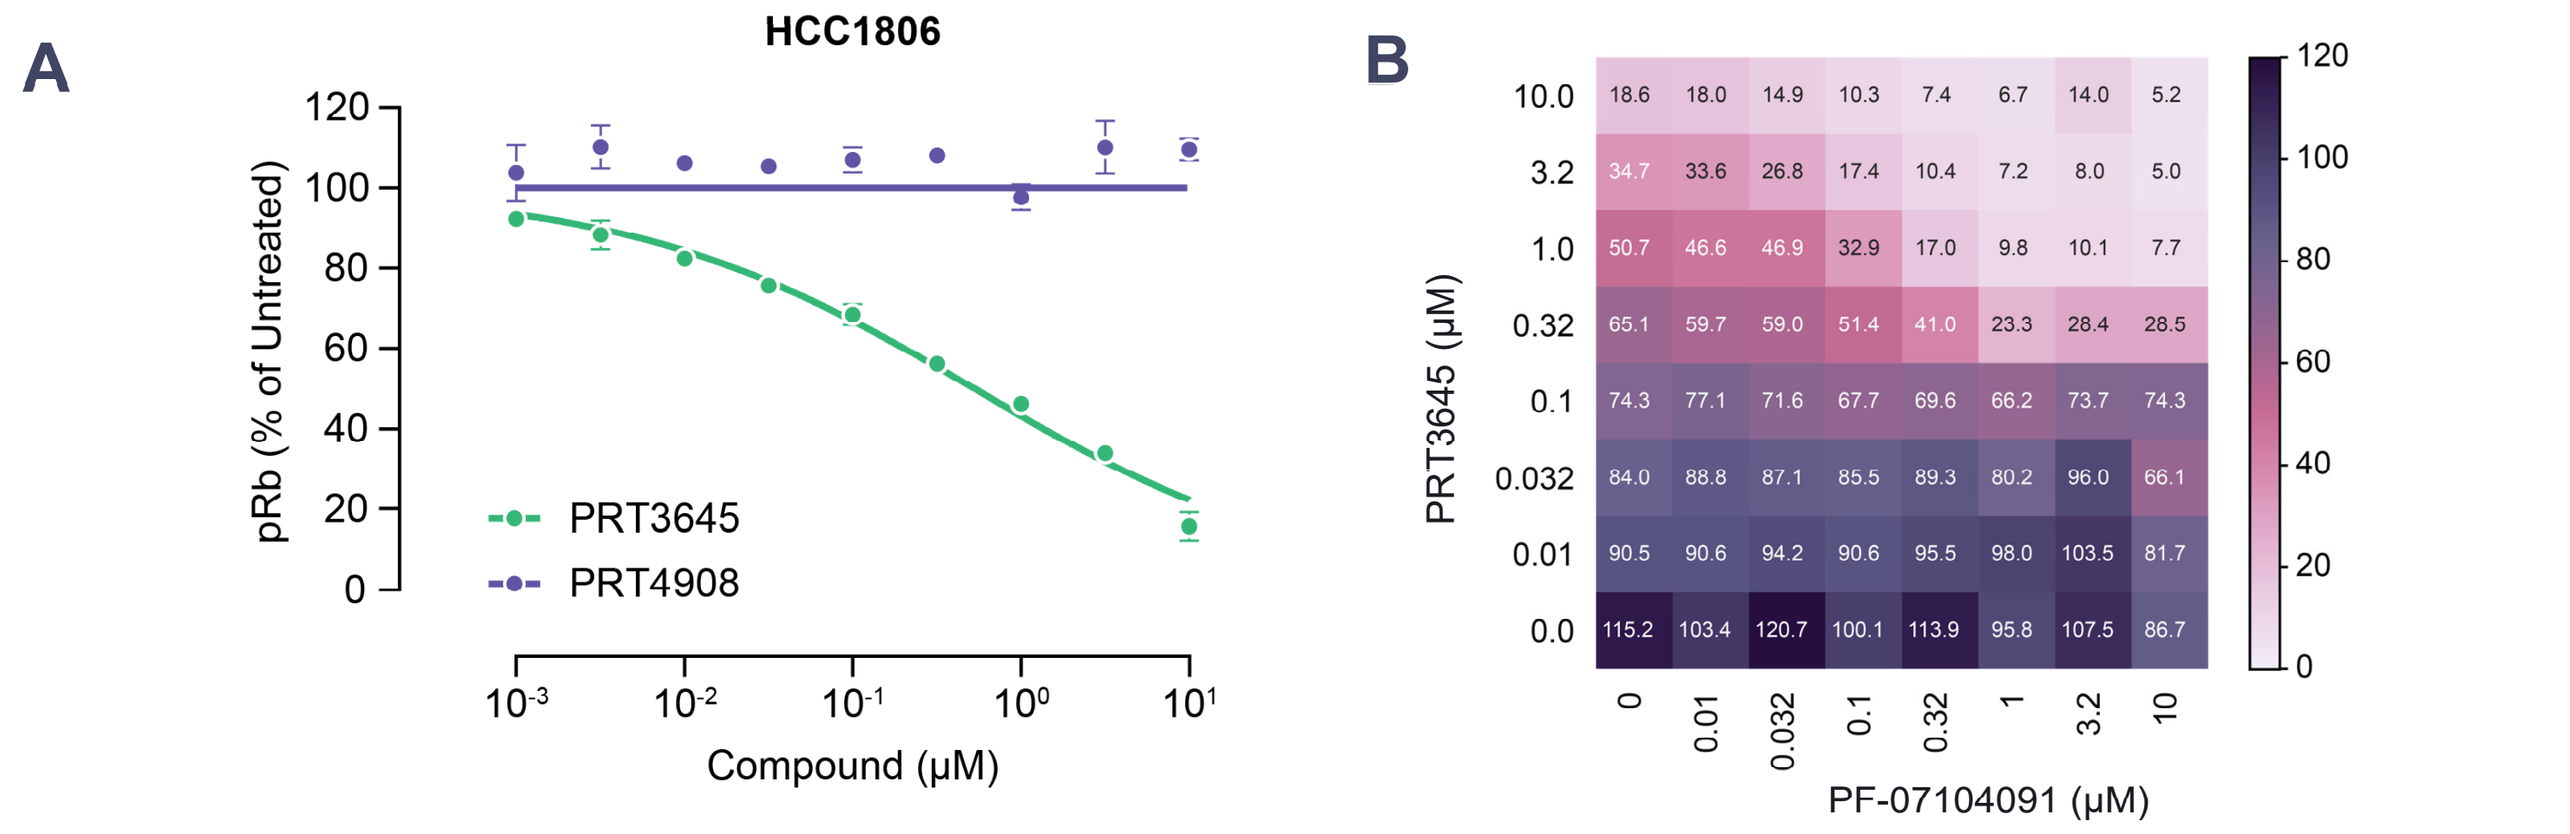
A
HCC1806
PRT3645
PRT4908
pRb (% of Untreated)
Compound (µM)
B
PRT3645 (µM)
PF-07104091 (µM)

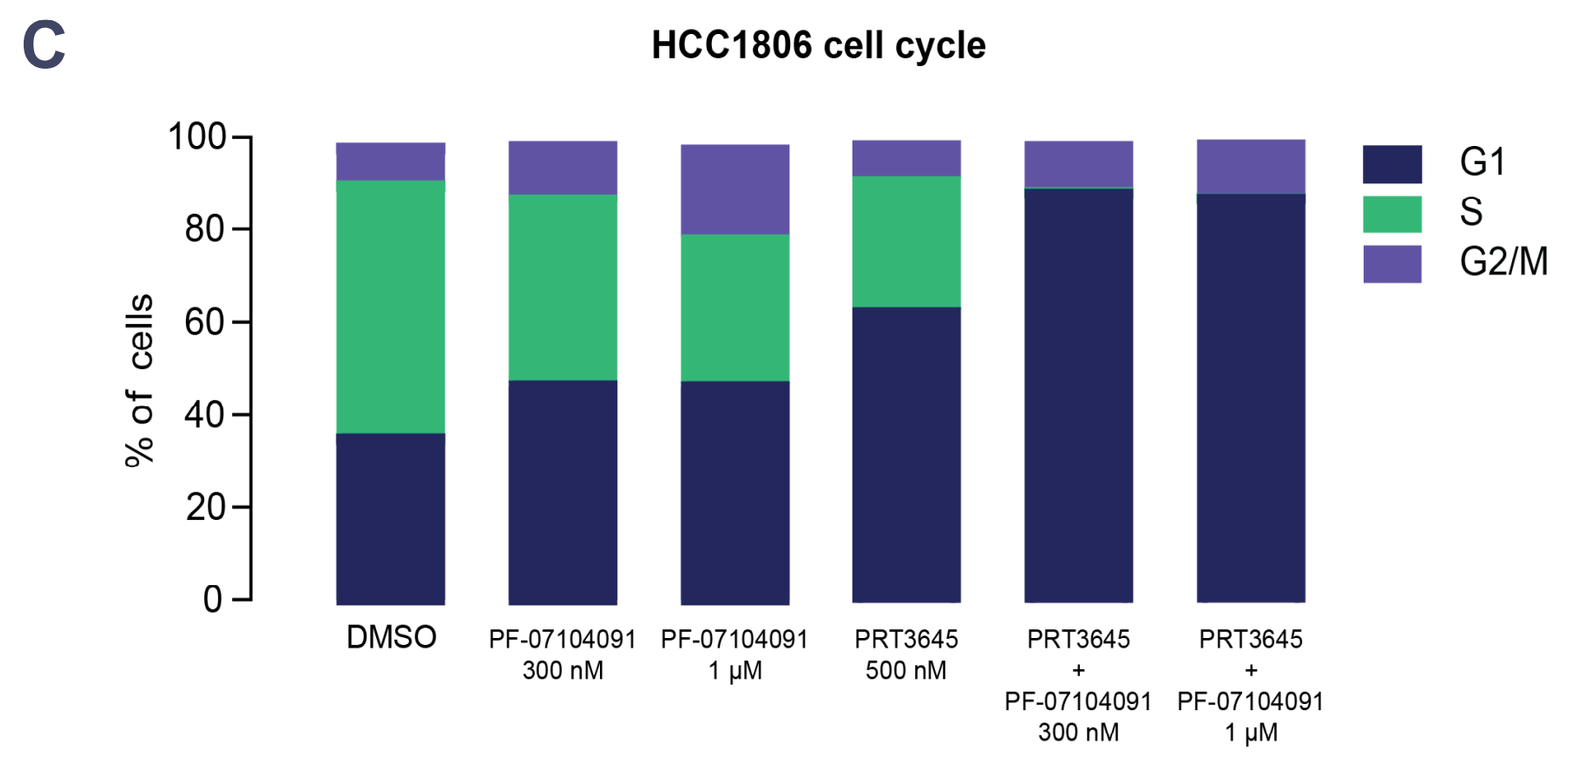
C
HCC1806 cell cycle
% of cells
G1
S
G2/M
DMSO
PF-07104091 300 nM
PF-07104091 1 µM
PRT3645 500 nM
PRT3645 + PF-07104091 300 nM
PRT3645 + PF-07104091 1 µM

Combination of PRT3645 and a CDK2 inhibitor in vitro A) In Cell Western analysis (ICW) was performed to evaluate the levels of pRB in HCC1806 cells following treatment of PRT3645 and PF-07104091 (CDK2 selective inhibitor) for 24 hours. **B)** The interaction landscape of the combination measured by pRB ICW assay after 24h treatment **C)** Cell cycle progression was analyzed via flow cytometry in HCC1806 cells after 24h treatment.

Conclusions

- PRT3645 demonstrated synergistic potential and enhanced cell cycle arrest at G1/S when combined with the ER degrader elacestrant in T47D ESR1 Y537S mutated cells in vitro and led to significant tumor regression in a breast cancer PDX model harboring the ESR1 Y537S mutation.
- PRT3645 exhibited anti-proliferative effects and demonstrated synergistic potential with trametinib in cell lines harboring BRAF class III mutations (G596R, G466V), as manifested by downregulation of genes involved in the cell cycle and the MAPK signaling pathway. PRT3645 was well-tolerated and exhibited significant anti-tumor efficacy as single agent. Moreover, the efficacy was further augmented when combined with trametinib in xenograft models of CRC and NSCLC with the BRAF class III mutation.
- The combination of PRT3645 and trametinib proved highly effective in suppressing cell survival, proliferation, and colony formation in KRAS G13D colorectal cell lines. Furthermore, the combination of PRT3645 and trametinib led to significant tumor growth inhibition in a xenograft model with the KRAS G13D mutation.
- PRT3645 attenuated RB phosphorylation and induced cell cycle G1/S arrest when combined with a CDK2 inhibitor in HCC1806, a TNBC cell line carrying both a CCNE amplification and CDKN2A deletion. This underscores the therapeutic potential of co-inhibition of CDK2 and CDK4/6.

References

1. Jiating Qi et. al. "Targeting CDK4/6 for Anticancer Therapy" Biomedicines. 2022 Mar 16.
2. Shom Goel et. al. "Targeting CDK4 and CDK6 in Cancer." Nat Rev Cancer. 2022 Mar 18
3. Silvia Lapenna et. al. "Cell cycle kinases as therapeutic targets for cancer." Nat Rev Drug Discov. 2009 July 8.
4. Usma Asghar et. al. "The history and future of targeting cyclin-dependent kinases in cancer therapy." Nat Rev Drug Discov. 2015 Jan 30.

Acknowledgments

This study was funded by Prelude Therapeutics Incorporated (Company). *In vivo* data was provided by Wuxi AppTec and XenoSTART. T47D ESR1 Y537S cell pool was provided by Synthego. RNA-Seq data was provided by Novogene. Editorial support provided by Arne Fabritius, Endosymbiont GmbH.

Disclosures

All authors are employees of the Company at the time of research and may own equity in the Company.