PRT2527, a Novel Highly Selective Cyclin-Dependent Kinase 9 (CDK9) Inhibitor, Has Potent Antitumor Activity in Combination with BTK and BCL2 Inhibition in Various Lymphoid Malignancies

Norman Fultang[1], Ashley Schwab[1], Sophia McAneny-Droz[1], Diane Heiser[1], Peggy Scherle[1], Neha Bhagwat[1]

[1]*Prelude Therapeutics Incorporated, Wilmington, DE; contact: nfultang@preludetx.com*

5966

Prelude THERAPEUTICS

Background

- CDK9 is a master regulator of transcription that modulates transcription elongation via phosphorylation of RNA polymerase II.[1]
- Short-term inhibition of CDK9 depletes short-lived transcripts and labile proteins such as MCL1, BFL1 and MYC to promote cancer cell death.[1]
- PRT2527 is a potent, highly selective, ATP-competitive, CDK9 inhibitor currently under evaluation in a Phase I clinical trial in patients with relapsed/refractory hematologic malignancies (NCT05665530).[2]
- BTK inhibitors and BCL2 inhibitors have shown clinical efficacy in lymphoid malignancies; BTK inhibitors are approved for the treatment of diffuse large B-cell lymphoma (DLBCL), chronic lymphocytic leukemia (CLL) and mantle cell lymphoma (MCL), whereas BCL2 inhibitors are approved for the treatment of CLL.[3]
- Despite strong clinical responses to both classes of agents, resistance rapidly develops.[4,5,6]
- Here, we investigate PRT2527 in combination with BTK and BCL2 inhibitors in various preclinical models of lymphoid malignancies

Key Findings

- PRT2527 is efficacious in preclinical models of DLBCL, CLL and MCL, and combines with both BTK and BCL2 inhibition to improve depth and duration of responses.
- PRT2527 exhibits potent anti-tumor activity in therapy-resistant CLL and MCL

Results

Figure 1. Short-term treatment with PRT2527 is efficacious in DLBCL and CLL models

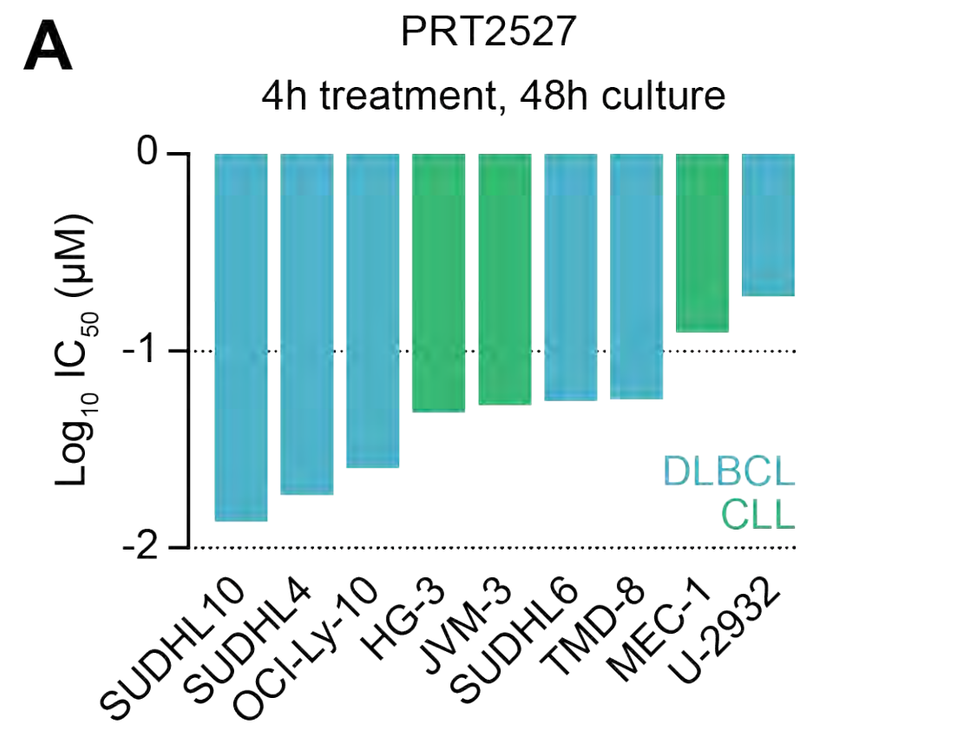
A
PRT2527
4h treatment, 48h culture
Log10 IC50 (µM)
DLBCL
CLL
SUDHL10
SUDHL4
OCI-Ly-10
HG-3
JVM-3
SUDHL6
TMD-8
MEC-1
U-2932

A) Waterfall plot of CellTiter-Glo (CTG) assay IC_{50} values assessing growth inhibition potency in DLBCL and CLL cell lines transiently treated (4h) with PRT2527. Cell viability was assessed after 48h.

B) Caspase-Glo® 3/7 assay demonstrating induction of apoptosis in DLBCL and CLL cell lines after 6 hours treatment with PRT2527.

C) Cell line-derived xenograft (CDX) DLBCL models treated with PRT2527 intravenously (BID q2h, QW) show significant inhibition of tumor growth. Data represented as mean ± SEM. N=8, ***P<0.001, ****P<0.0001 by Mann-Whitney U test

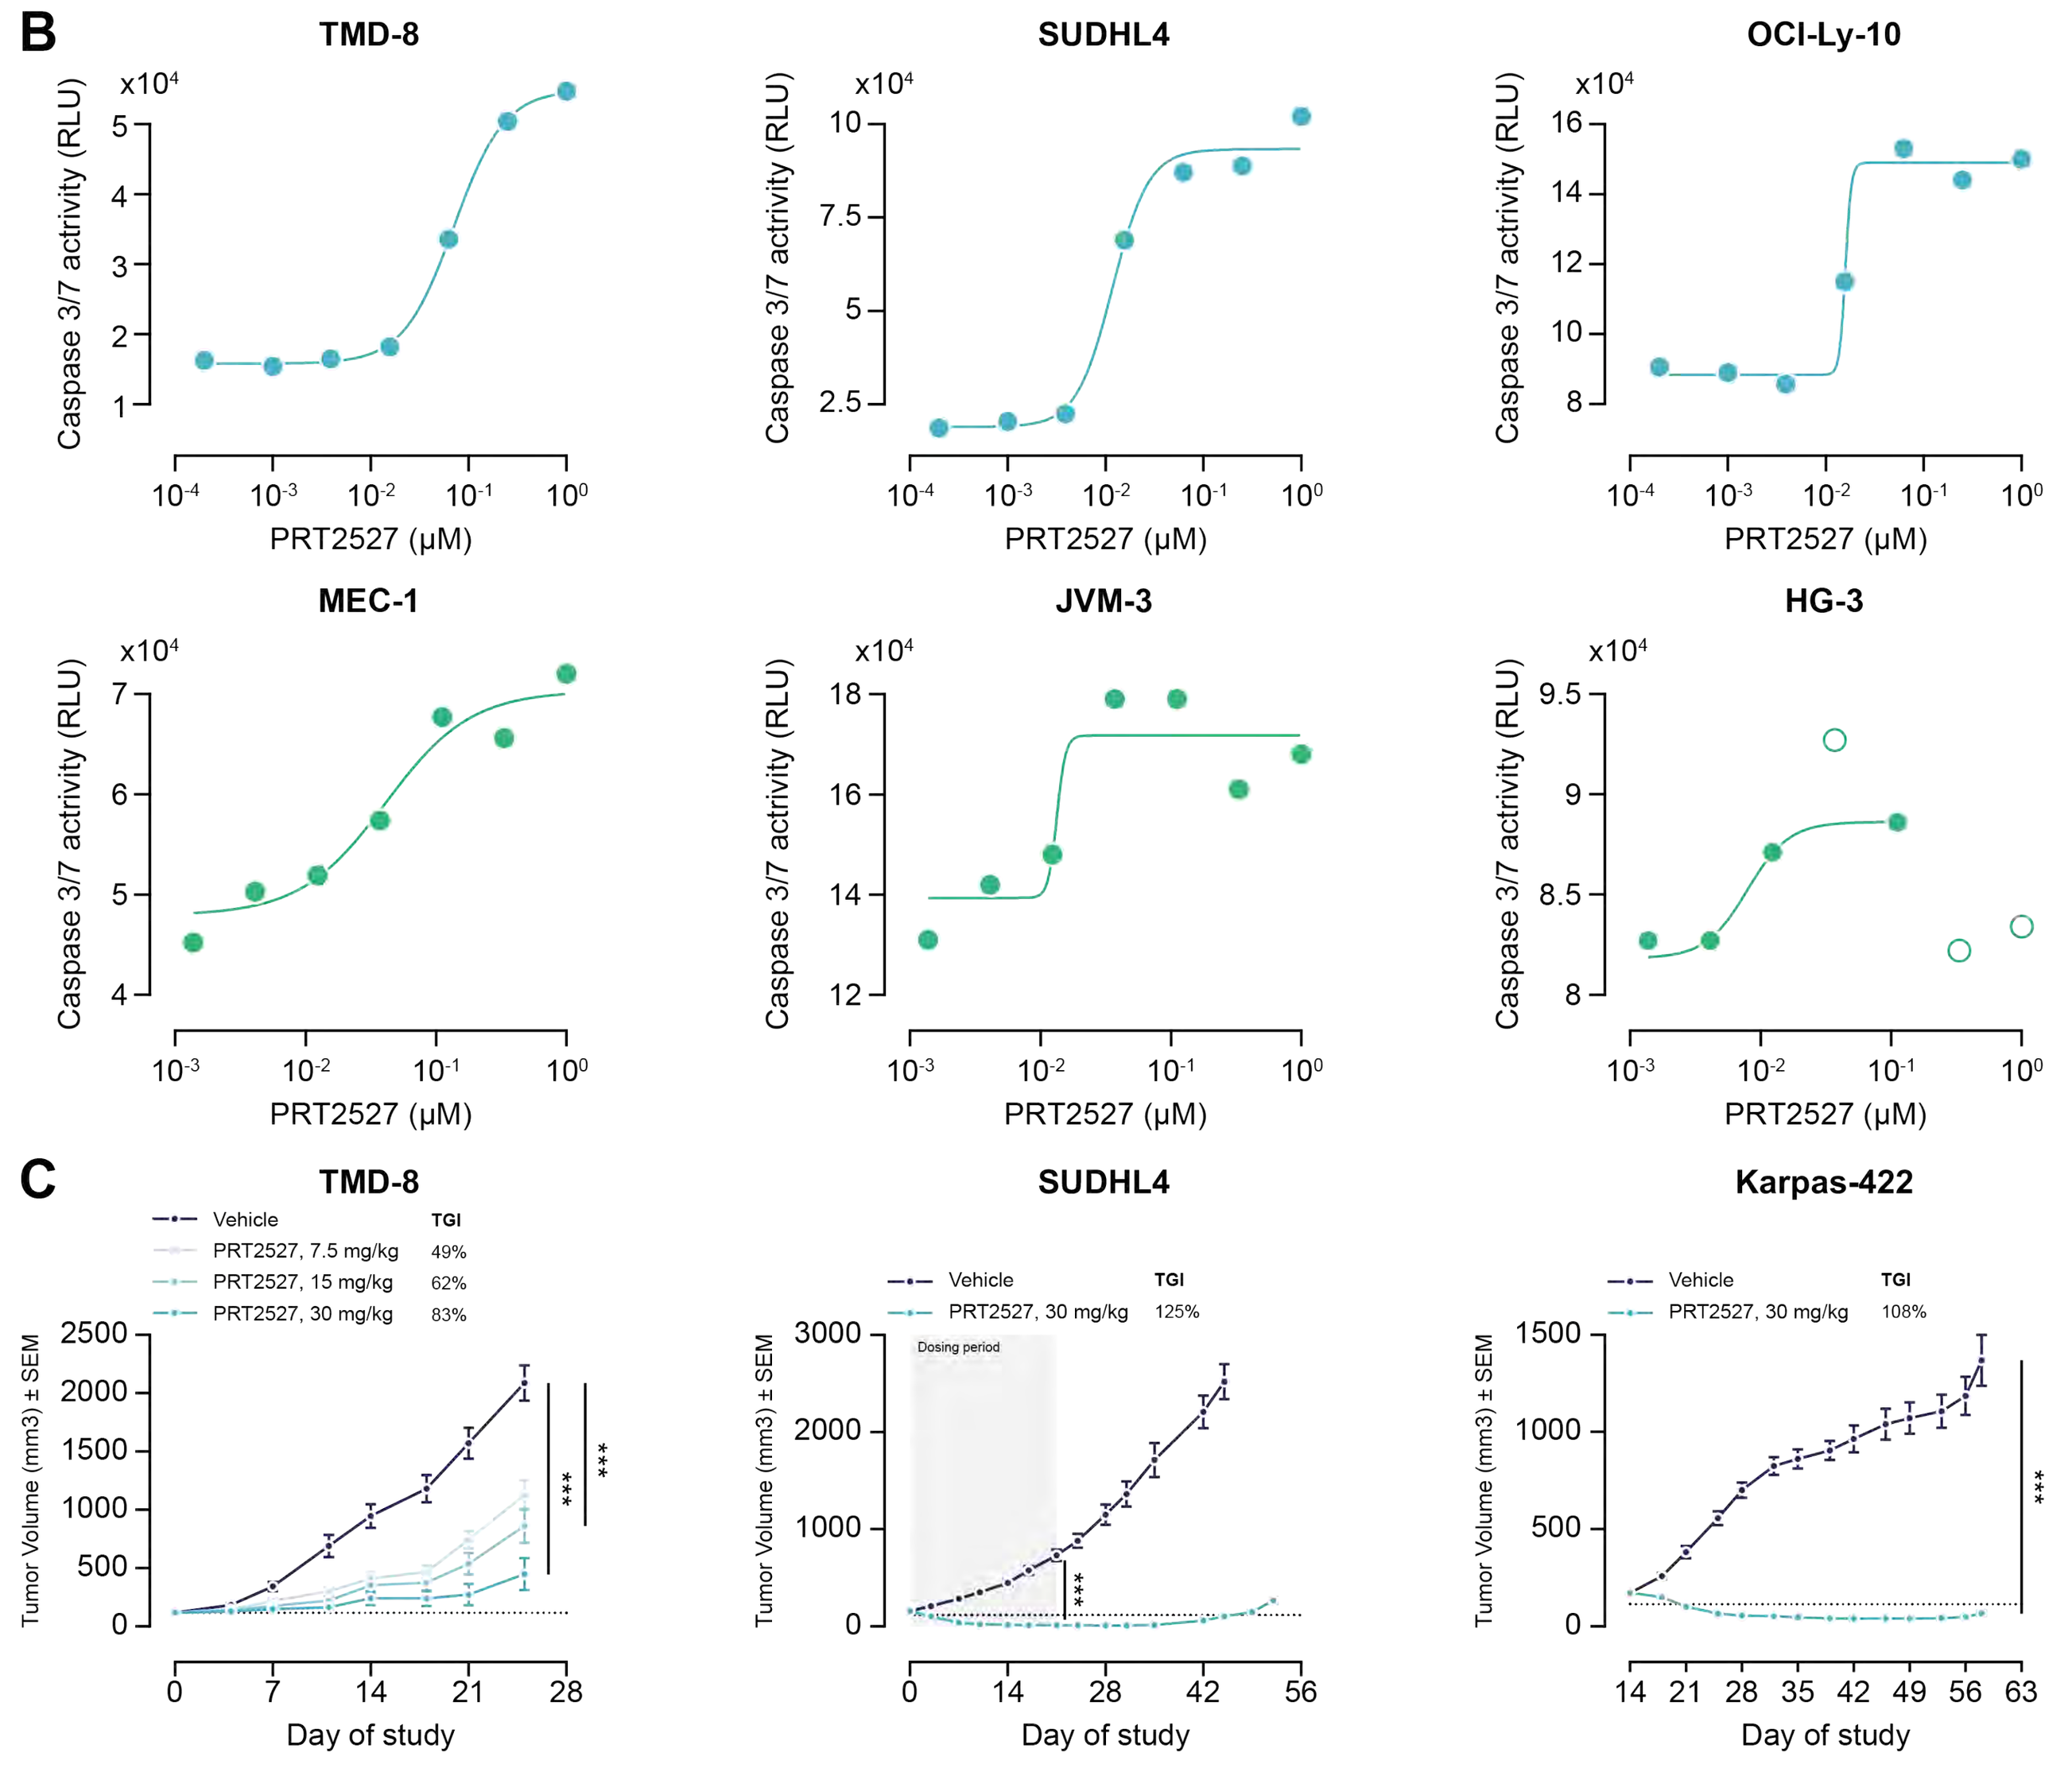
B
TMD-8
SUDHL4
OCI-Ly-10
MEC-1
JVM-3
HG-3
Caspase 3/7 activity (RLU)
PRT2527 (µM)
C
TMD-8
Vehicle
PRT2527, 7.5 mg/kg 49%
PRT2527, 15 mg/kg 62%
PRT2527, 30 mg/kg 83%
SUDHL4
Vehicle
PRT2527, 30 mg/kg 125%
Dosing period
Karpas-422
Vehicle
PRT2527, 30 mg/kg 108%
TGI
Tumor Volume (mm3) ± SEM
Day of study

Figure 2. Addition of BTK inhibitors potentiates the activity of PRT2527 *in vitro* and *in vivo*

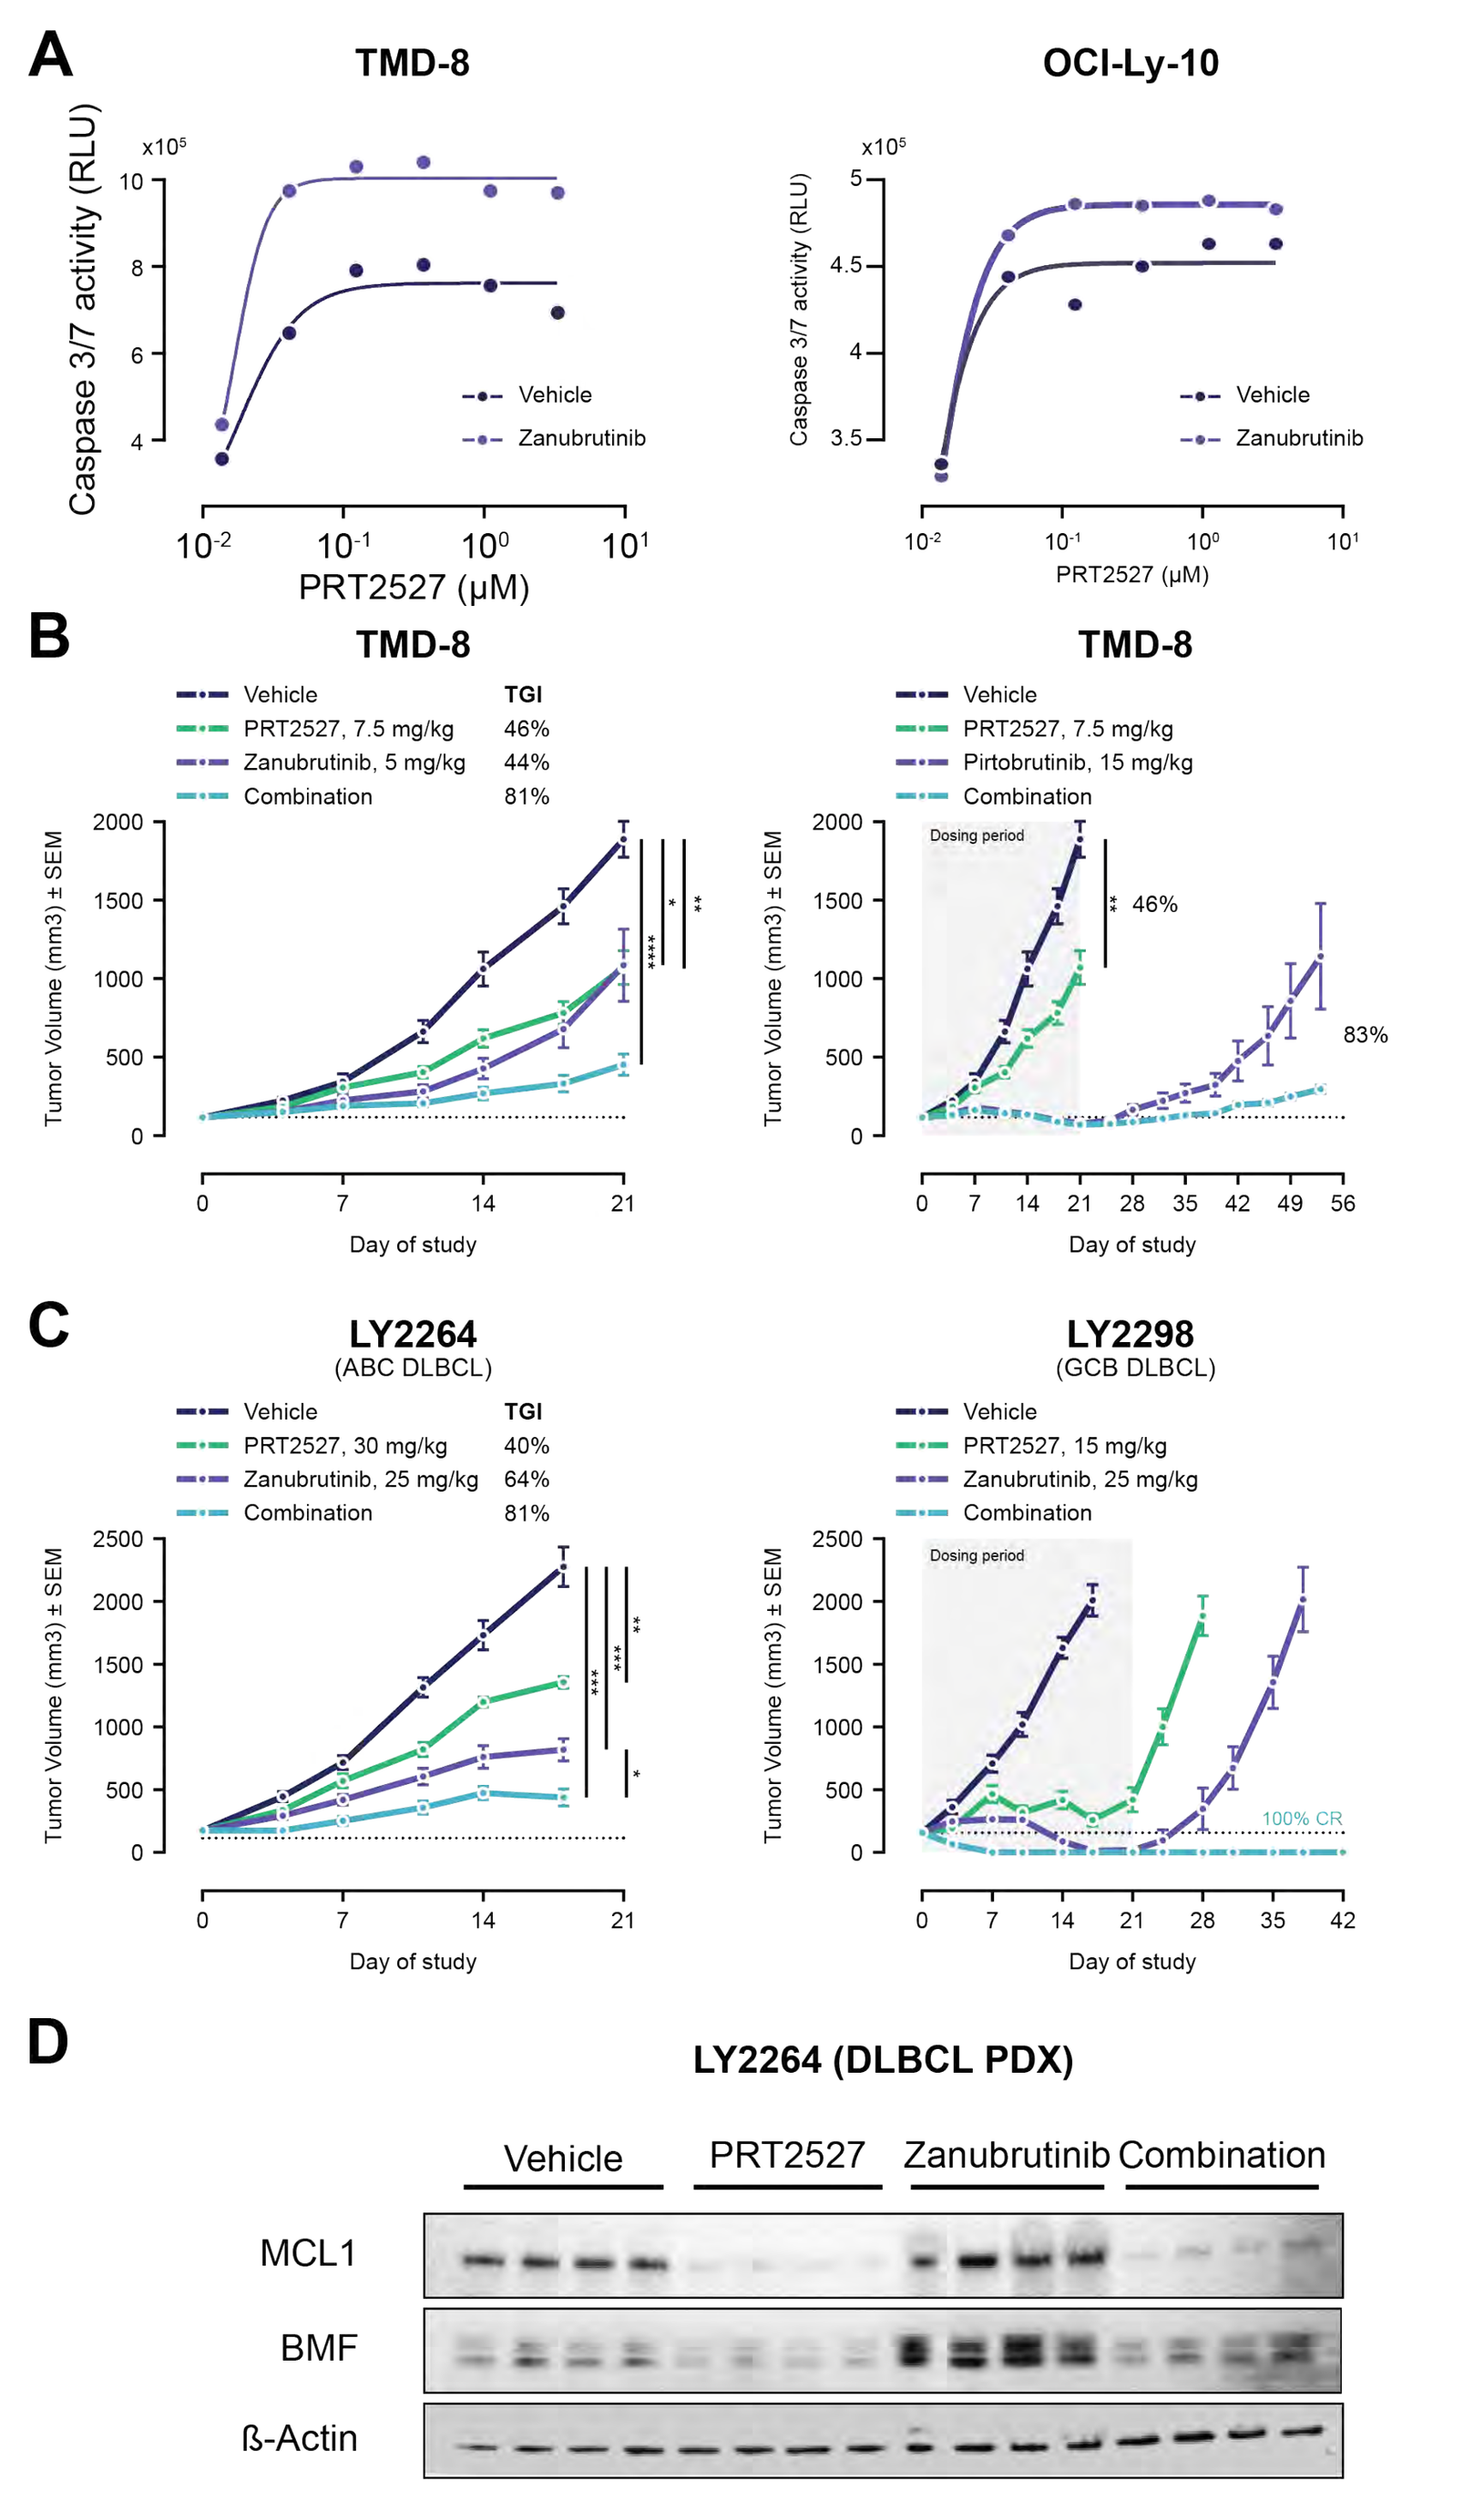
A
TMD-8
OCI-Ly-10
Vehicle
Zanubrutinib
Caspase 3/7 activity (RLU)
PRT2527 (µM)
B
TMD-8
Vehicle
PRT2527, 7.5 mg/kg 46%
Zanubrutinib, 5 mg/kg 44%
Combination 81%
TMD-8
Vehicle
PRT2527, 7.5 mg/kg
Pirtobrutinib, 15 mg/kg
Combination
Dosing period
46%
83%
C
LY2264
(ABC DLBCL)
Vehicle
PRT2527, 30 mg/kg 40%
Zanubrutinib, 25 mg/kg 64%
Combination 81%
LY2298
(GCB DLBCL)
Vehicle
PRT2527, 15 mg/kg
Zanubrutinib, 25 mg/kg
Combination
100% CR
TGI
Tumor Volume (mm3) ± SEM
Day of study
D
LY2264 (DLBCL PDX)
Vehicle
PRT2527
Zanubrutinib
Combination
MCL1
BMF
ß-Actin

A) Caspase-Glo® 3/7 assay assessing induction of apoptosis by PRT2527 in DLBCL cell lines, with and without overnight pre-treatment with Zanubrutinib (1 nM).

B) TMD-8 (DLBCL) cell line-derived xenograft (CDX) demonstrating potent inhibition of tumor growth by PRT2527 (7.5 mg/kg, BID q2h QW) administered intravenously, in monotherapy and in combination with Zanubrutinib (5 mg/kg, BID) or Pirtobrutinib (15 mg/kg, BID), administered orally.

C) Intravenously administered PRT2527 (30 mg/kg, BID q2h QW) alone and in combination with Zanubrutinib (25 mg/kg, QD), administered orally, potently inhibits tumor growth in LY2264 and LY2298 (DLBCL) patient-derived xenografts (PDX); 100% complete response (mean tumor volume = 0 mm^3) observed in combination group. Data represented as mean ± SEM. N=8, * P<0.05, **P<0.001, ***P<0.001 by Mann-Whitney U test.

D) Western blot showing decrease in MCL1 and BMF in LY2264 tumors following PRT2527 and Zanubrutinib treatment *in vivo*.

E) Western blot assessing modulation of MCL1, BFL1, BMF and Cleaved Caspase 3 in TMD8 cells following treatment with PRT2527 ± Zanubrutinib overnight pre-treatment.

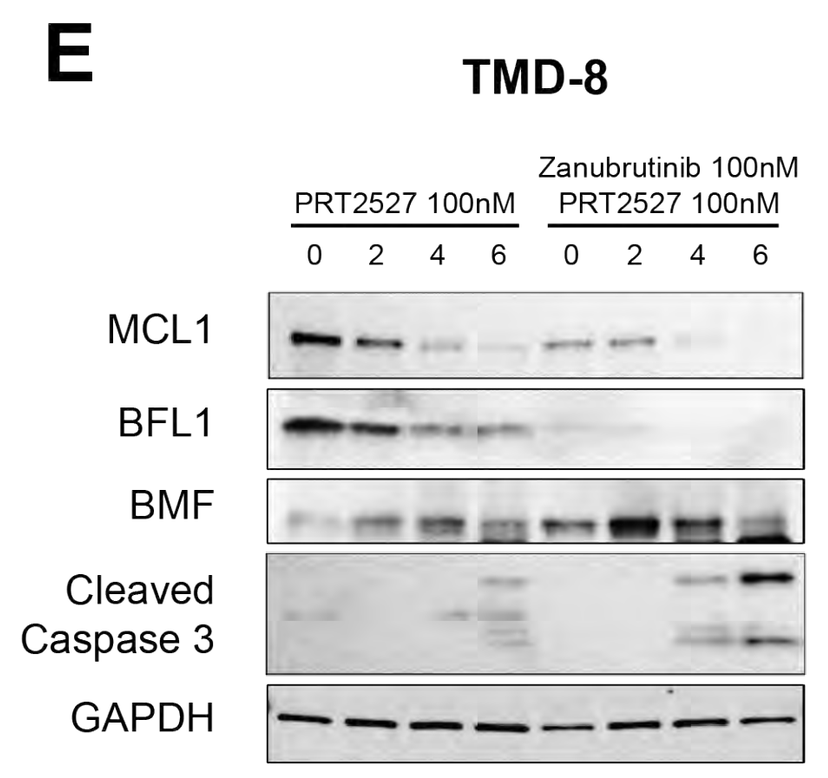
E
TMD-8
PRT2527 100nM
Zanubrutinib 100nM
PRT2527 100nM
0 2 4 6 0 2 4 6
MCL1
BFL1
BMF
Cleaved
Caspase 3
GAPDH

Figure 3. Addition of Venetoclax (BCL2 inhibitor) potentiates the activity of PRT2527 *in vitro* and *in vivo*

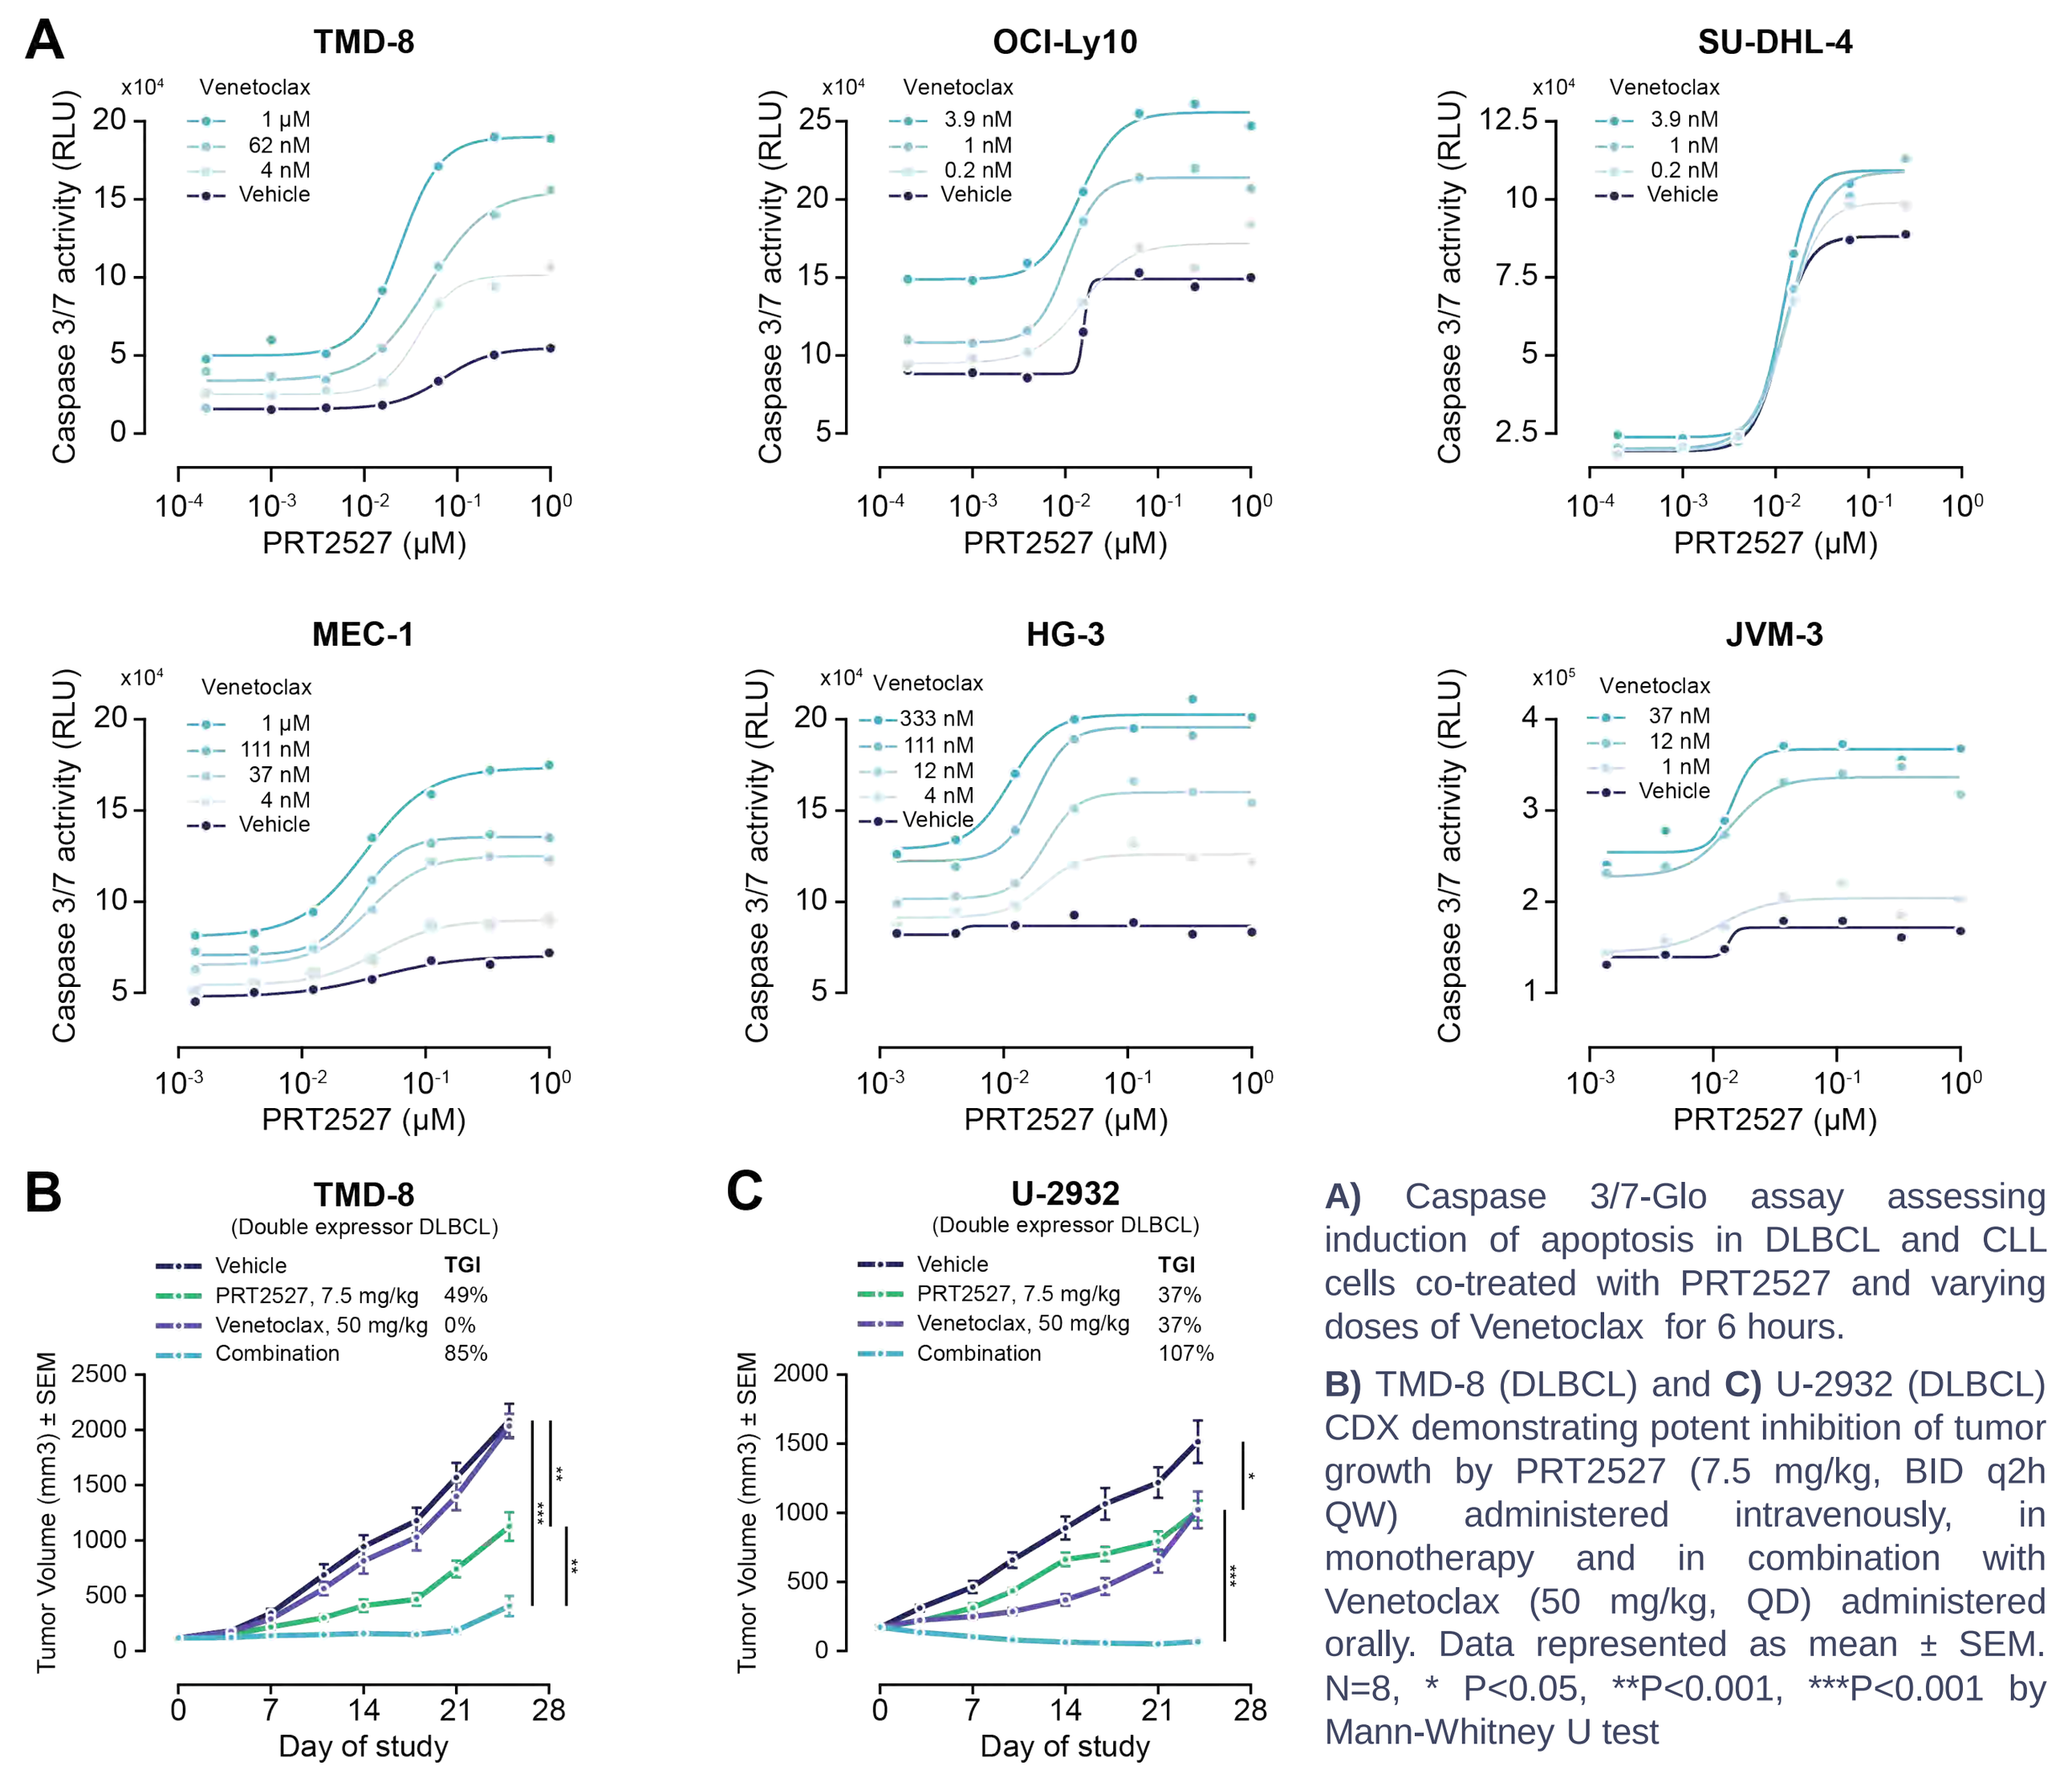
A
TMD-8
OCI-Ly10
SU-DHL-4
MEC-1
HG-3
JVM-3
Venetoclax
Vehicle
Caspase 3/7 activity (RLU)
PRT2527 (µM)
B
TMD-8
(Double expressor DLBCL)
Vehicle
PRT2527, 7.5 mg/kg 49%
Venetoclax, 50 mg/kg 0%
Combination 85%
C
U-2932
(Double expressor DLBCL)
Vehicle
PRT2527, 7.5 mg/kg 37%
Venetoclax, 50 mg/kg 37%
Combination 107%
TGI
Tumor Volume (mm3) ± SEM
Day of study

A) Caspase 3/7-Glo assay assessing induction of apoptosis in DLBCL and CLL cells co-treated with PRT2527 and varying doses of Venetoclax for 6 hours.

B) TMD-8 (DLBCL) and **C)** U-2932 (DLBCL) CDX demonstrating potent inhibition of tumor growth by PRT2527 (7.5 mg/kg, BID q2h QW) administered intravenously, in monotherapy and in combination with Venetoclax (50 mg/kg, QD) administered orally. Data represented as mean ± SEM. N=8, * P<0.05, **P<0.001, ***P<0.001 by Mann-Whitney U test

Figure 4. PRT2527 is efficacious in treatment-naïve and R/R primary CLL *ex vivo* as monotherapy and in combination with BTKi or Venetoclax

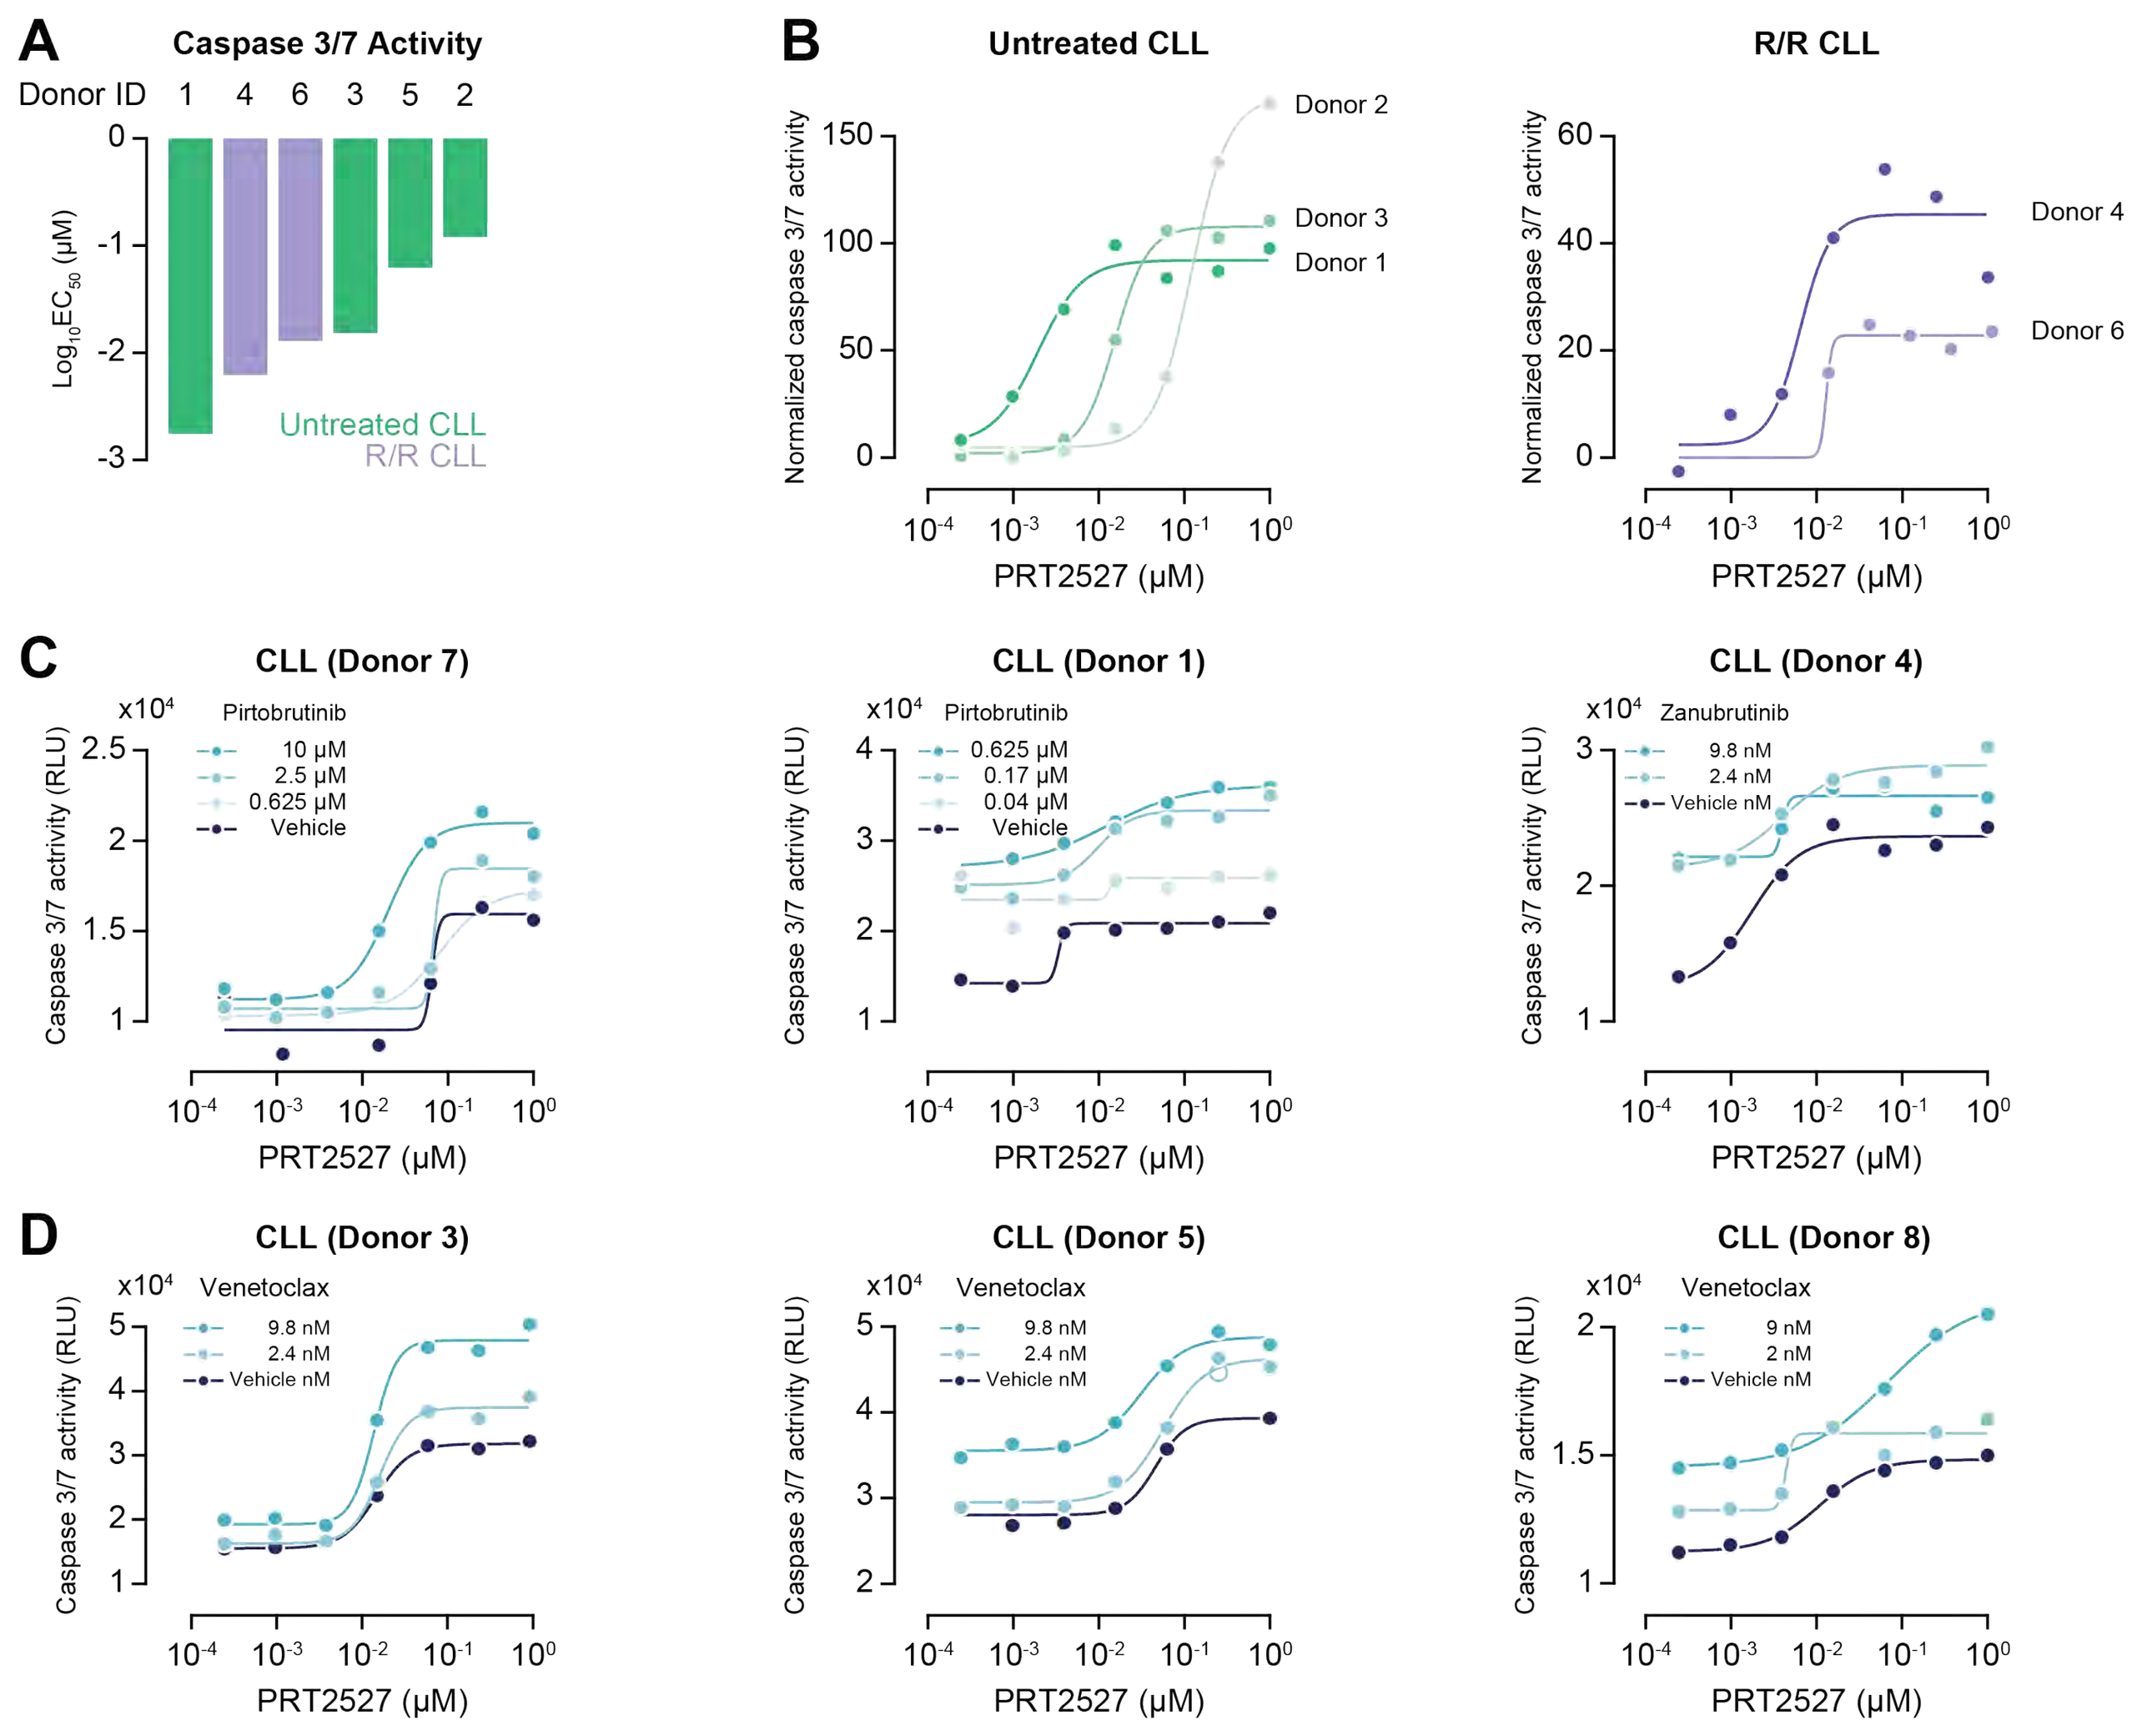
A
Caspase 3/7 Activity
Donor ID 1 4 6 3 5 2
Log10EC50 (µM)
Untreated CLL
R/R CLL
B
Untreated CLL
Donor 2
Donor 3
Donor 1
R/R CLL
Donor 4
Donor 6
Normalized caspase 3/7 activity
C
CLL (Donor 7)
Pirtobrutinib
CLL (Donor 1)
Pirtobrutinib
CLL (Donor 4)
Zanubrutinib
D
CLL (Donor 3)
Venetoclax
CLL (Donor 5)
Venetoclax
CLL (Donor 8)
Venetoclax
Caspase 3/7 activity (RLU)
PRT2527 (µM)

A, B) Caspase-Glo® 3/7 activity assay showing induction of apoptosis in PBMCs derived from patients with untreated or relapsed/refractory (R/R) CLL, following 6h treatment with PRT2527.

C) Caspase-Glo® 3/7 activity assay showing induction of apoptosis by PRT2527 in CLL PBMCs, with and without overnight pre-treatment with varying doses of BTKi.

D) Caspase-Glo® 3/7 activity assay showing induction of apoptosis in CLL PBMCs co-treated with PRT2527 and varying doses of Venetoclax for 6 hrs.

Figure 5. PRT2527 overcomes Ibrutinib resistance in Mantle Cell Lymphoma

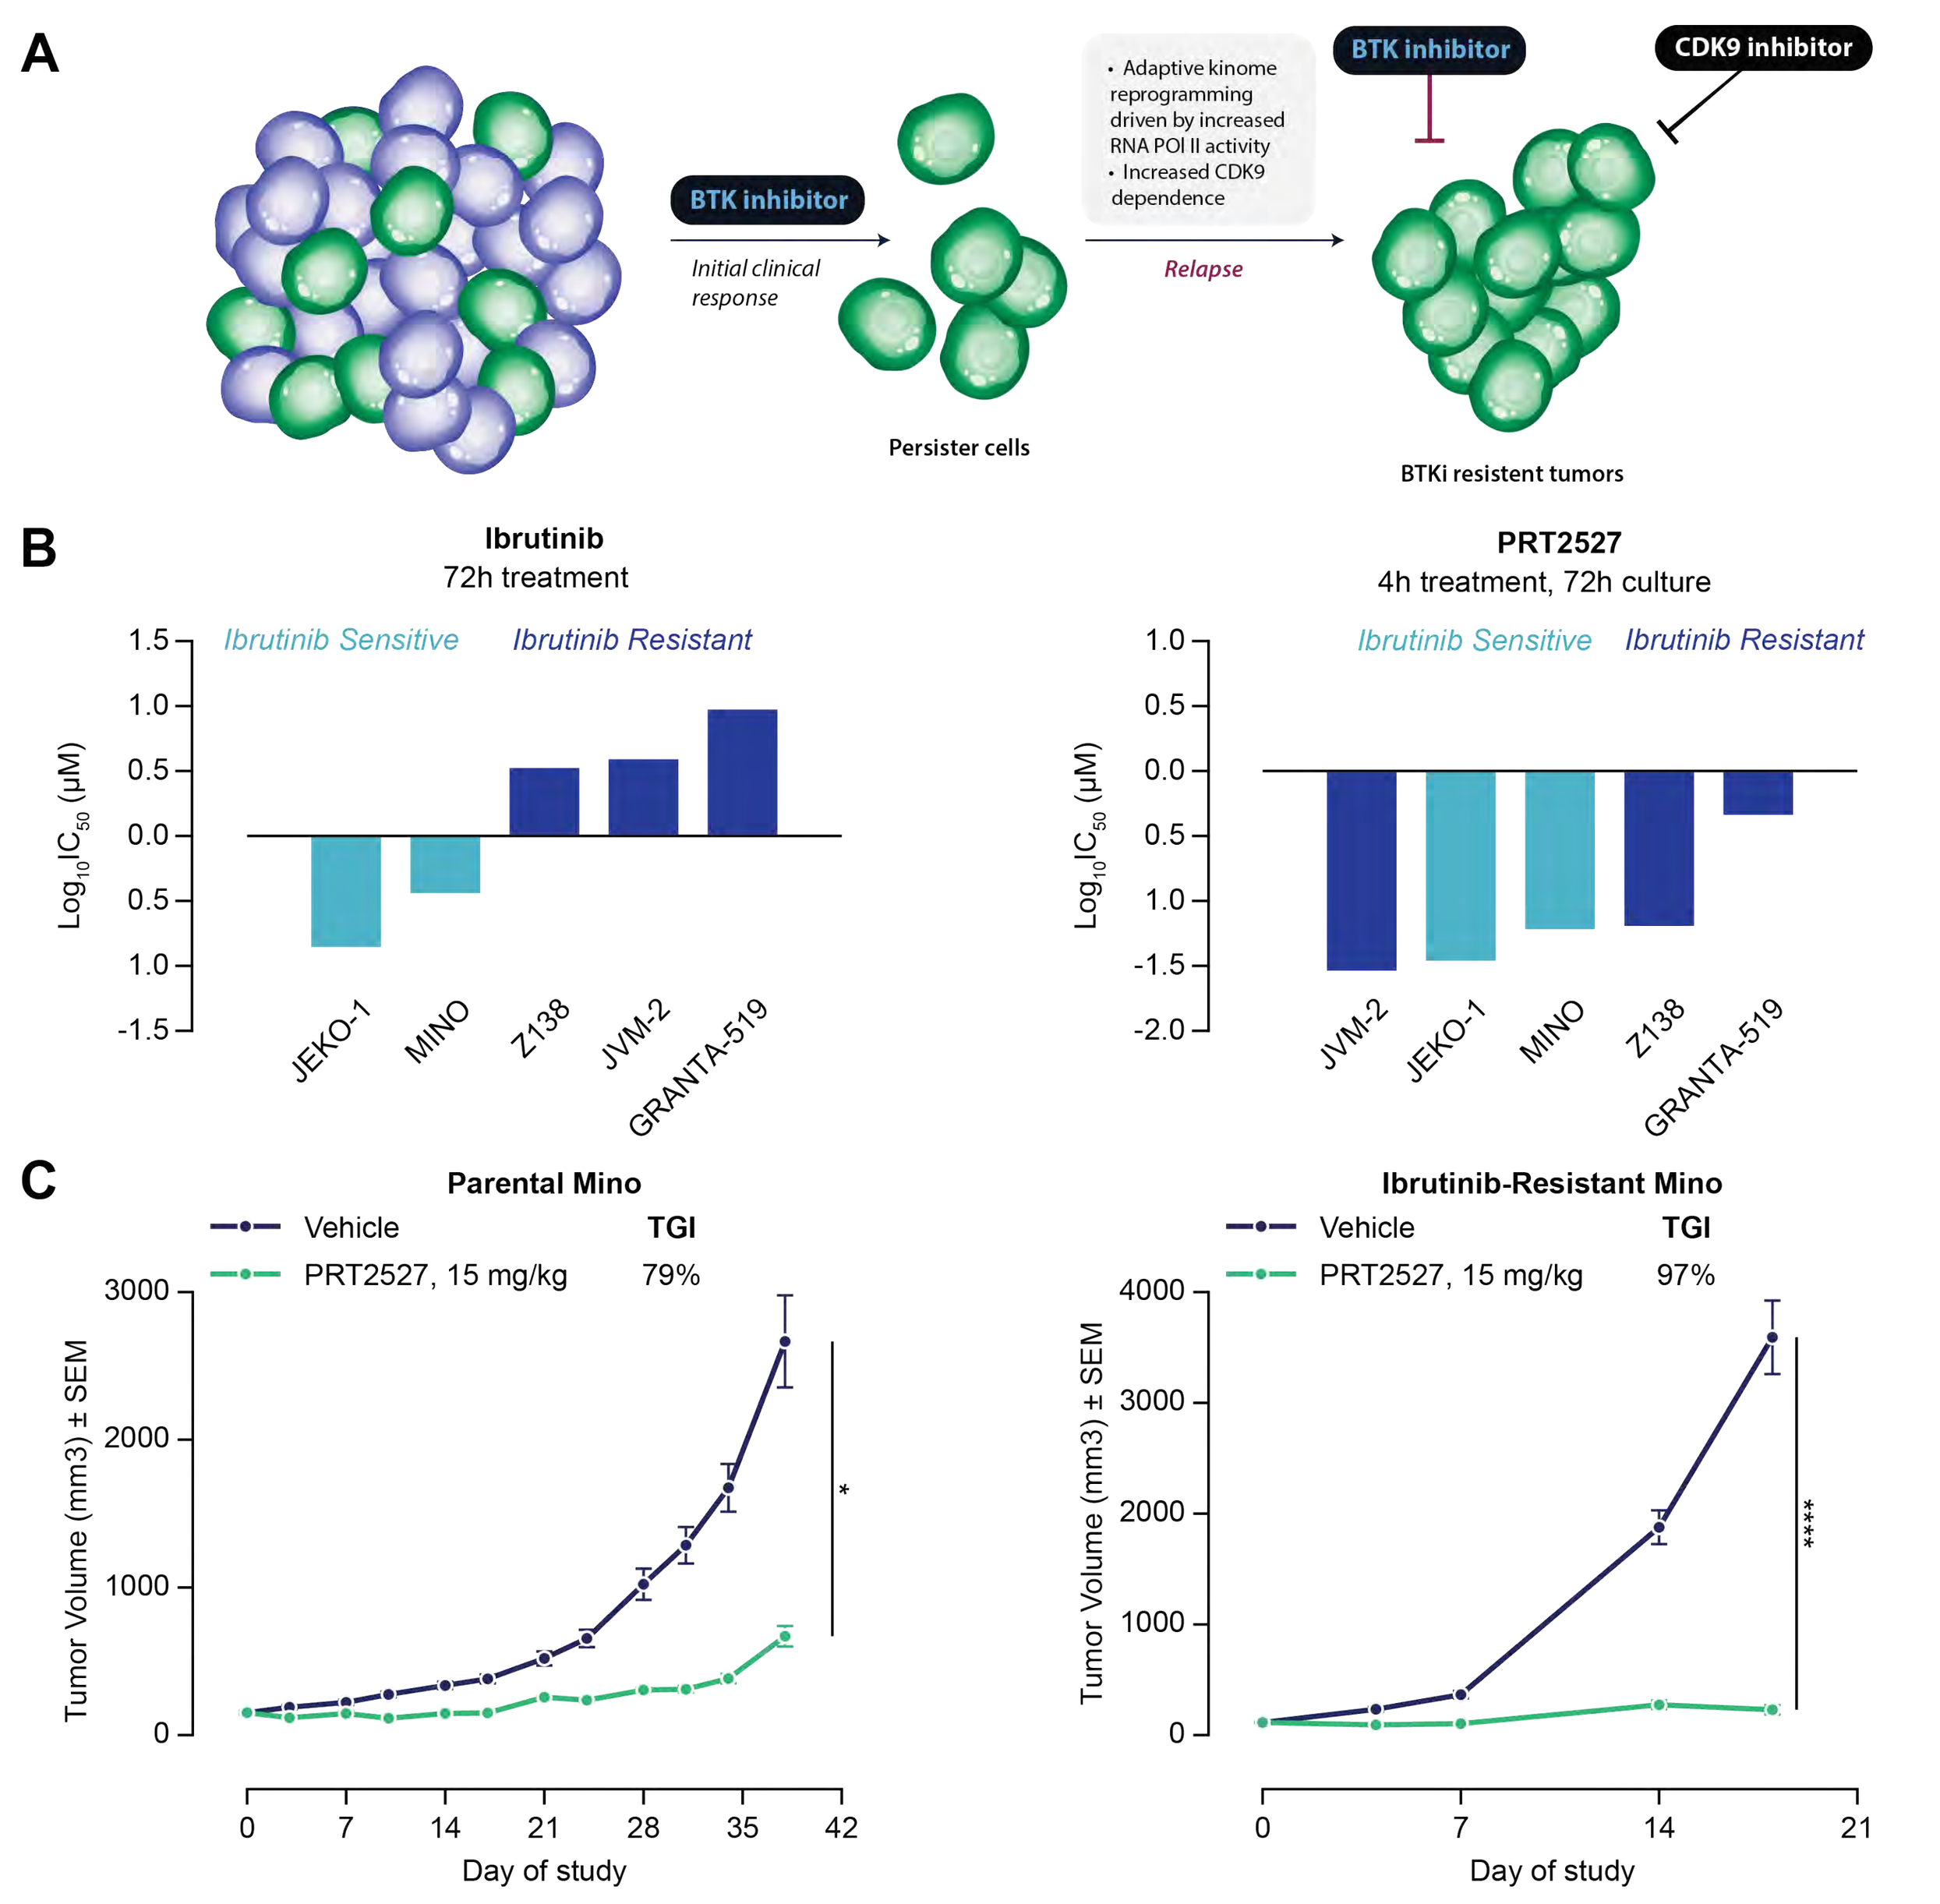
A
BTK inhibitor
Initial clinical response
Persister cells
Adaptive kinome reprogramming driven by increased RNA POl II activity
Increased CDK9 dependence
Relapse
BTK inhibitor
CDK9 inhibitor
BTKi resistent tumors
B
Ibrutinib
72h treatment
PRT2527
4h treatment, 72h culture
Ibrutinib Sensitive
Ibrutinib Resistant
Log10IC50 (µM)
JEKO-1
MINO
Z138
JVM-2
GRANTA-519
C
Parental Mino
Vehicle
PRT2527, 15 mg/kg 79%
Ibrutinib-Resistant Mino
Vehicle
PRT2527, 15 mg/kg 97%
TGI
Tumor Volume (mm3) ± SEM
Day of study

A) CDK9/RNA Pol II-mediated transcriptional reprogramming drives BTKi-resistance in MCL.[7]

B) PRT2527 shows activity in Ibrutinib sensitive and resistant MCL cell lines in vitro in a CTG assay.

C) PRT2527 (15 mg/kg, BID q2h QW i.v.) shows significant tumor growth inhibition in the MINO CDX model of Ibrutinib resistant MCL. Anti-tumor activity remains similar to the parental Mino model. Ibrutinib-resistance was induced through repeat *in vivo* passage and treatment with Ibrutinib. Data represented as mean ± SEM. N=7, * P<0.05, ****P<0.0001 by Mann-Whitney U test

Figure 6. Mechanism of Action of BTK and BCL2 Inhibitor Combination with the CDK9 Inhibitor, PRT2527 in Lymphoid Malignancies

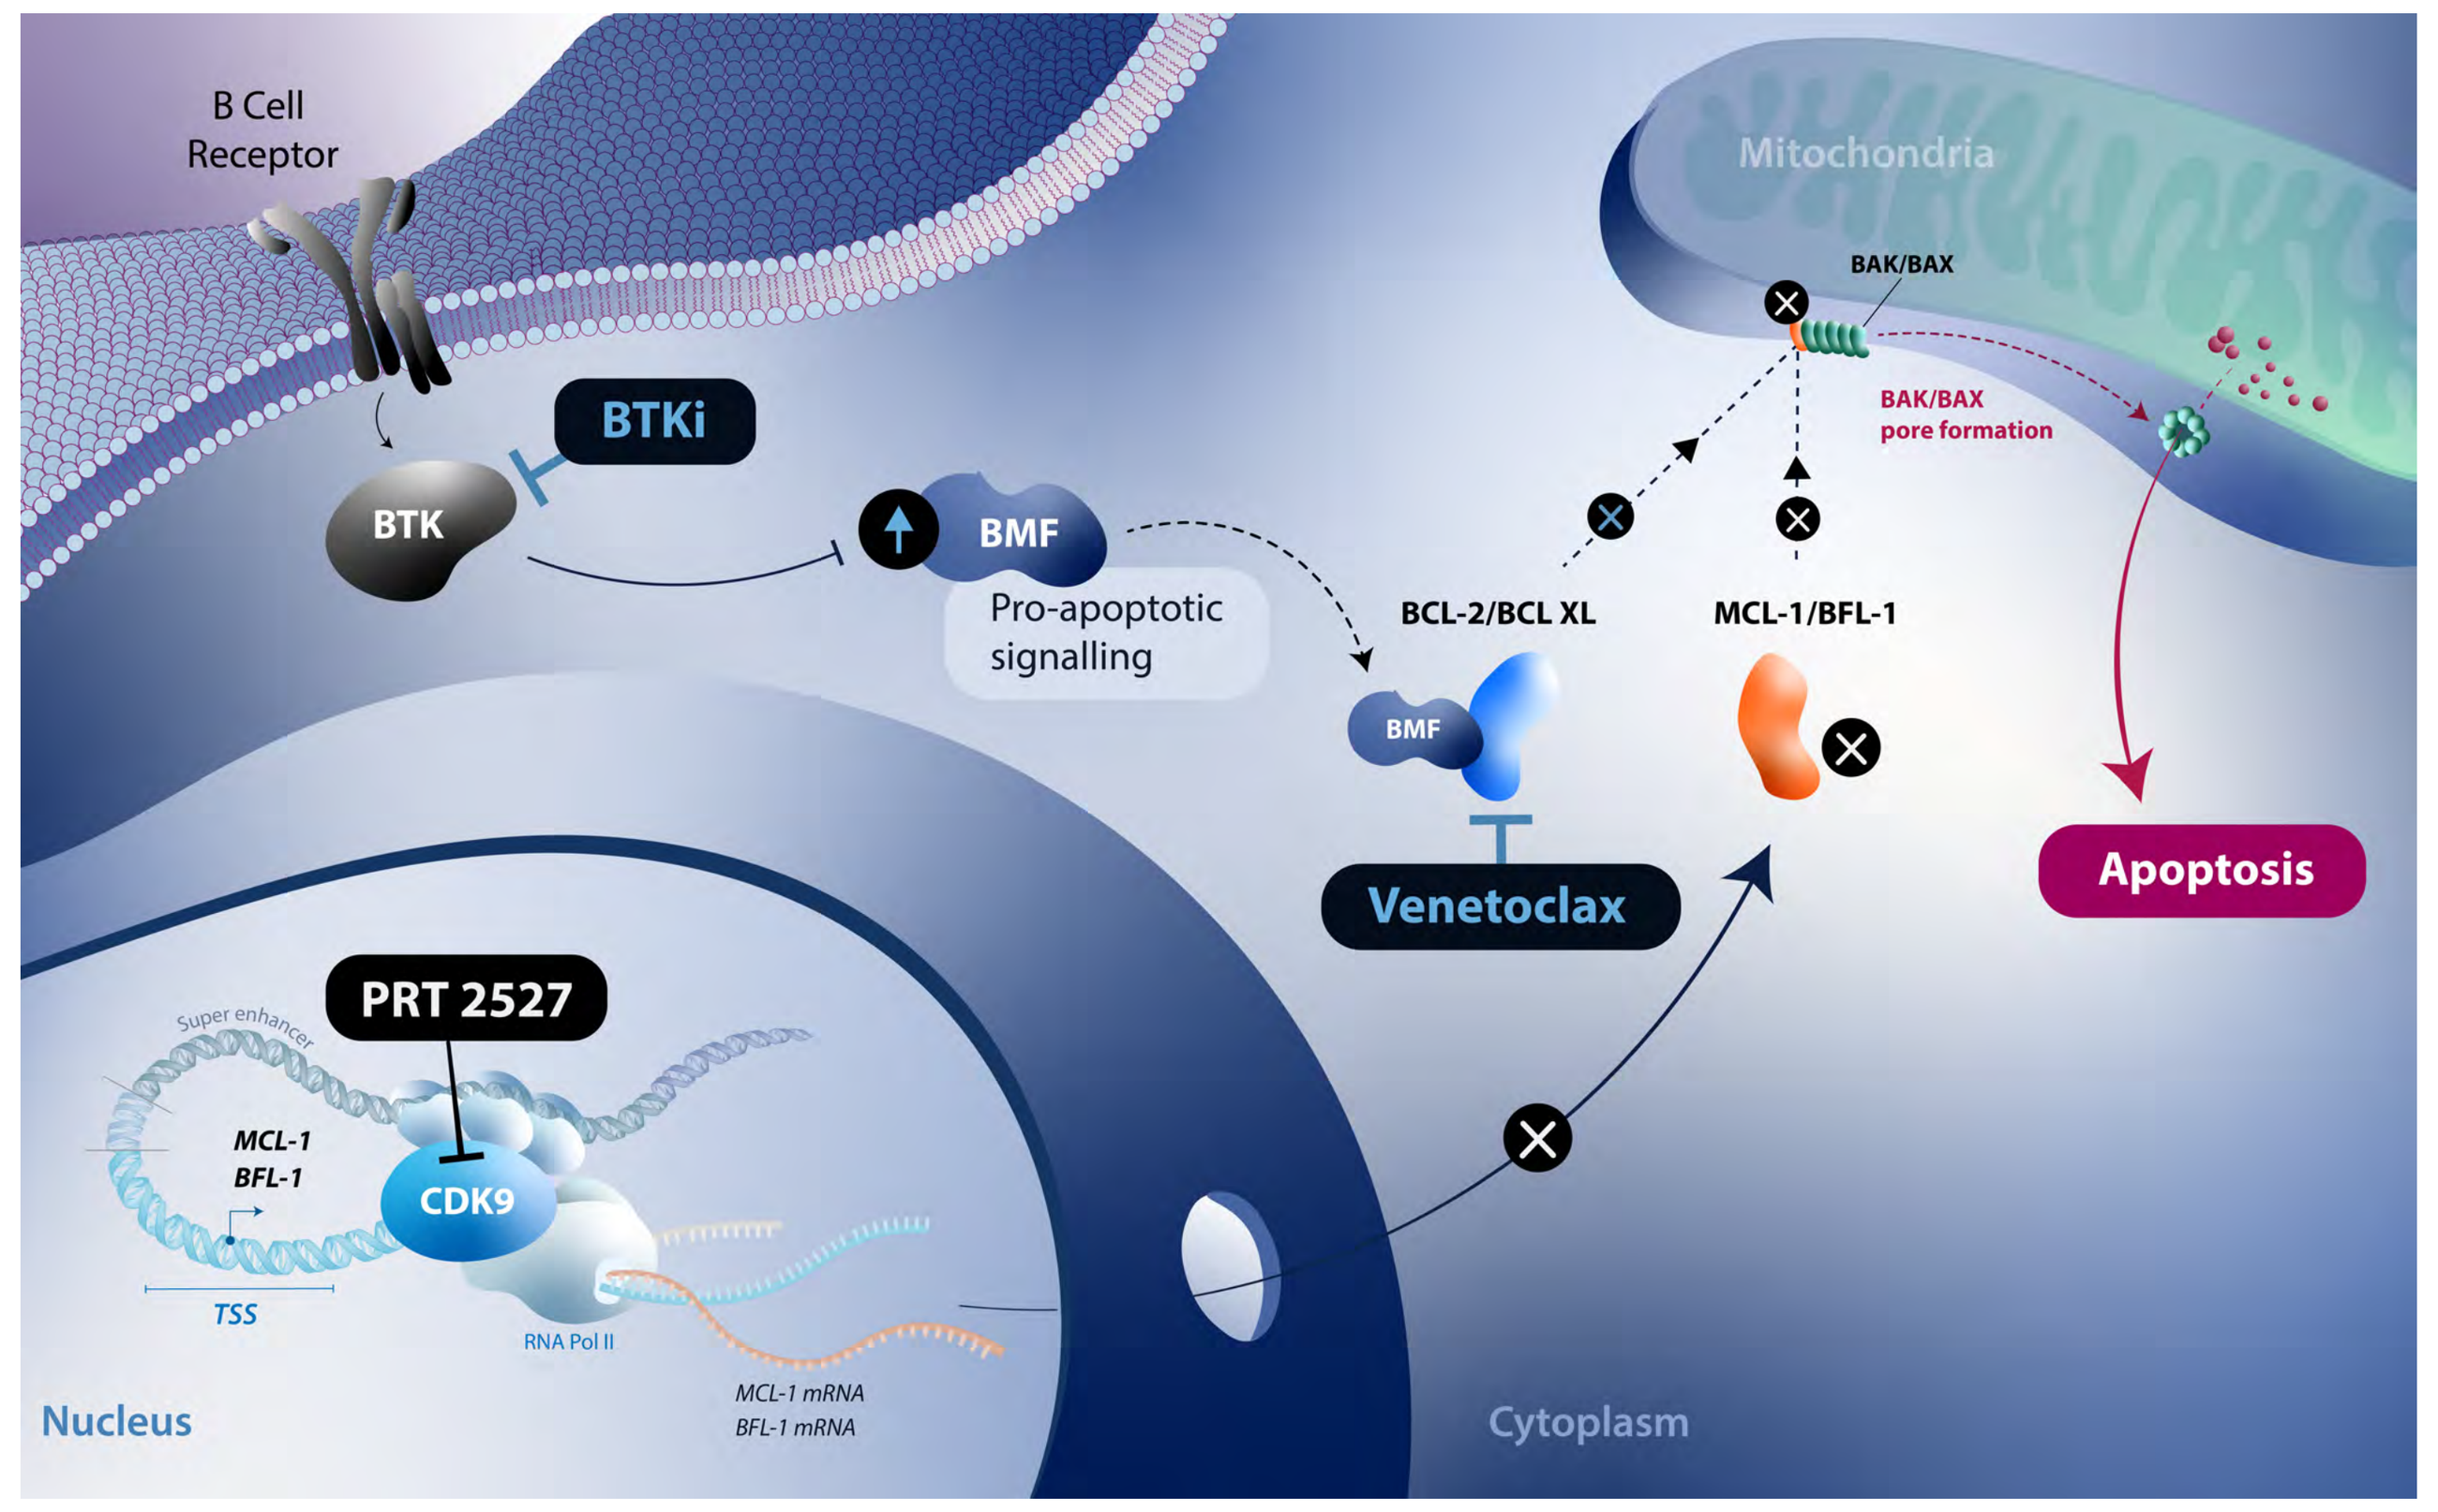
B Cell
Receptor
BTKi
BTK
BMF
Pro-apoptotic
signalling
Mitochondria
BAK/BAX
BAK/BAX
pore formation
BCL-2/BCL XL
MCL-1/BFL-1
Venetoclax
Apoptosis
PRT 2527
CDK9
MCL-1
BFL-1
TSS
RNA Pol II
MCL-1 mRNA
BFL-1 mRNA
Nucleus
Cytoplasm

- BTK inhibitors 'prime' cancer cells for apoptosis by inducing pro-apoptosis BH3 proteins like BMF. Upregulated BMF preferentially binds and inhibits BCL2 and BCL-xL, thereby increasing dependency on MCL1 and BFL1. CDK9 inhibition by PRT2527 depletes both MCL1 and BFL1, synergistically inducing cell death.
- Targeting BCL2 with inhibitors like Venetoclax often results in increased dependence of cancer cells on other pro-survival BCL2 family members such as MCL1, and is a frequent mechanism of intrinsic and acquired resistance to BCL2 inhibitors. The indirect targeting of MCL1 with the CDK9 inhibitor, PRT2527, in combination with Venetoclax can overcome resistance and result in synergistic induction of apoptosis in hematologic malignancies

Conclusions

- PRT2527 is efficacious in pre-clinical models of DLBCL, CLL and MCL
- PRT2527 combines with BCL2i and BTKi to potently induce apoptosis *in vitro* and inhibit tumor growth (including regressions) *in vivo*.
- PRT2527 potently induces apoptosis in treatment-naïve and relapsed/refractory primary CLL *ex vivo*.
- PRT2527 overcomes resistance to Ibrutinib in MCL *in vitro* and *in vivo*.
- PRT2527 is currently being evaluated in a Phase I clinical trial in patients with relapsed/refractory hematologic malignancies as a monotherapy and in combination with zanubrutinib (NCT05665530)[2].

References

1. Boiko, S., et al 2021. Blood, Am. J. Hematol., 137(21), pp.2947-2957.
2. NCT05665530 (ClinicalTrials.gov).
3. National Comprehensive Cancer Network. B-Cell Lymphomas (Version 1.2024). https://www.nccn.org/professionals/physician_gls/pdf/b-cell.pdf. Accessed February 1st, 2024.
4. Wang, H., et al 2021. Hematol. Oncol., 39(5), pp.605-615.
5. Reyes, A. et al 2023. Cancer Drug Resistance, 6(4), p.828.
6. Thompson, E.R., et al 2022. Blood Advances, 6(2), pp.503-508.
7. Zhao, X., et al 2021. Cell reports, 34(11).

Acknowledgments

This study was funded by Prelude Therapeutics. *In vivo* data provided by Crown Bioscience, Wuxi Apptec and Medicilon. Editorial Support was provided by Endosymbiont GmbH.

Disclosures

All authors are employees of Prelude Therapeutics, Inc., Wilmington, DE and may own equity in the Company.