Poster ID: 713TiP

A Phase 1 Study of PRT3789, a Potent and Selective Degrader of SMARCA2 in Patients With Advanced or Metastatic Solid Tumors and a *SMARCA4* Mutation

Ibiayi Dagogo-Jack,[1] Afshin Dowlati,[2] Robin Guo,[3] Mark Awad,[4] Aurelie Swalduz,[5] Emiliano Calvo,[6] Victor Moreno,[7] Alex Adjei,[8] Patricia LoRusso,[9] Salman Punekar,[10] Ticiana Leal,[11] Timothy A. Yap,[12] Antoine Italiano,[13,14] William Novotny,[15] Chris Tankersley,[15] Sarah Rowe,[15] Gina Paris,[15] William Sun,[15] Alex Spira,[16] and Benjamin Besse[17]

[1]Massachusetts General Hospital, Boston, MA, USA; [2]University Hospitals Seidman Cancer Center and Case Western Reserve University, Cleveland, OH, USA; [3]Memorial Sloan Kettering Cancer Center, Commack, NY, USA; [4]Dana-Farber Cancer Institute, Boston, MA, USA; [5]Léon Bérard Center, Lyon, France; [6]START Madrid-CIOCC, Centro Integral Oncológico Clara Campal, Madrid, Spain; [7]START Madrid-FJD, Madrid, Spain; [8]Cleveland Clinic Taussig Cancer Institute, Cleveland, OH, USA; [9]Yale Cancer Center, New Haven, CT, USA; [10]NYU Langone Health, New York, NY, USA; [11]Winship Cancer Center of Emory University, Atlanta, GA, USA; [12]Department of Investigational Cancer Therapeutics, The University of Texas MD Anderson Cancer Center, Houston, TX, USA; [13]Early Phase Trial Unit, Institut Bergonié, Bordeaux, France; [14]Faculty of Medicine, University of Bordeaux, Bordeaux, France; [15]Prelude Therapeutics Incorporated, Wilmington, DE, USA; [16]NEXT Oncology-Virginia, Fairfax, VA, USA; and [17]Gustave Roussy, Villejuif, France

Copies of this poster obtained through the QR (Quick Response) code are for personal use only and may not be reproduced without written permission of the authors

BACKGROUND

- Genes encoding subunits of the SWI/SNF chromatin remodeling complex have been observed in 20% of all human cancers[1,2]
 - The SWI/SNF complex contains 1 of 2 ATP enzymatic subunits: either SMARCA2 (also known as BRM) or SMARCA4 (also known as BRG1)
 - Because SMARCA2 and SMARCA4 function as mutually exclusive catalytic subunits of the SWI/SNF complex, cells exhibiting SMARCA4 loss rely on its paralog, SMARCA2, making SMARCA2 an attractive therapeutic target[3-5]
- *SMARCA4* mutations have been found in multiple tumor types, including thoracic sarcomas, lymphomas, and cancers of the lung, ovaries, and skin (**Table 1**)[2,6]

Table 1. *SMARCA4* Mutation by Cancer Indication[2,6]

Indication	Any *SMARCA4* mutation, %	*SMARCA4* LOF mutation, %
NSCLC	10.0	5
Esophageal	8.0	2.7
Gastric (stomach adeno)	8.3	2.3
Skin	21.0	2.0
Endometrial (Uterine corpus)	13.3	1.9
Squamous cell lung	7.7	1.9
Urinary (bladder)	9.0	1.5
Colorectal	6.0	1.3
Pancreatic	2.9	1.2
Melanoma	8.7	0.5

- In NSCLC, *SMARCA4* mutations are observed in ≈10% of cases and are associated with more aggressive and invasive disease and shorter survival[7,8]
- The most frequently co-mutated genes in *SMARCA4*-mutated NSCLC are *TP53*, *KRAS*, *KEAP1*, and *STK11*[7,8]
- *SMARCA4* mutation class is differentially associated with prognosis in patients with stage IV NSCLC; *SMARCA4* alterations can be divided into 2 clinically relevant genomic classes associated with differential protein expression as well as distinct prognostic and treatment implications[8]
 - **Class 1 mutations** include truncating mutations, fusions, and homozygous deletion
 - **Class 2 mutations** include missense mutations
- In patients with metastatic NSCLC, *SMARCA4* alterations have been associated with shorter OS, with truncating mutations, fusions, and homozygous deletion alterations being associated with the shortest survival times[8]
- Increased understanding of the relationship of *SMARCA4* in lung cancer may enable the development of new therapeutic opportunities
- Among NSCLCs treated with first-line chemoimmunotherapy, a *SMARCA4* mutation is associated with significantly worse ORR, mPFS, and mOS compared to those of first-line chemoimmunotherapy[9]
- *SMARCA4* LOF mutations include homozygous missense and hotspot mutations with known LOF, in addition to the other damaging mutations such as frameshift indels, nonsense (stop gain, stop loss), or splice site
- PRT3789 is an intravenous VHL-based SMARCA2 degrader (**Figure 1**) which induces proteosome-dependent degradation of SMARCA2 (**Figure 2**)
- Preclinical data indicate that PRT3789 is a potent and selective degrader of SMARCA2, which induces robust synthetic lethality in *SMARCA4*-deficient cancers

Figure 1. 3D Figure of SMARCA2 Bromodomain and E3 Ligase Complex (VBC/CUL2/RBX1) Formed by PRT3789 SMARCA2 Degrader

PRT3789

- Highly potent (plasma DC_{50}=21 nM)
- Highly selective for SMARCA2 over SMARCA4
 - Cellular assays >1000×
 - HiBit 40× selective
 - Selectivity confirmed in vivo
- Efficacious in H838 CDX model at well-tolerated doses
- Clean on hERG and safety 47
- Moderate CYP3A4 inhibition
- IC_{50} >10-fold expected C_{max}, with no TDI

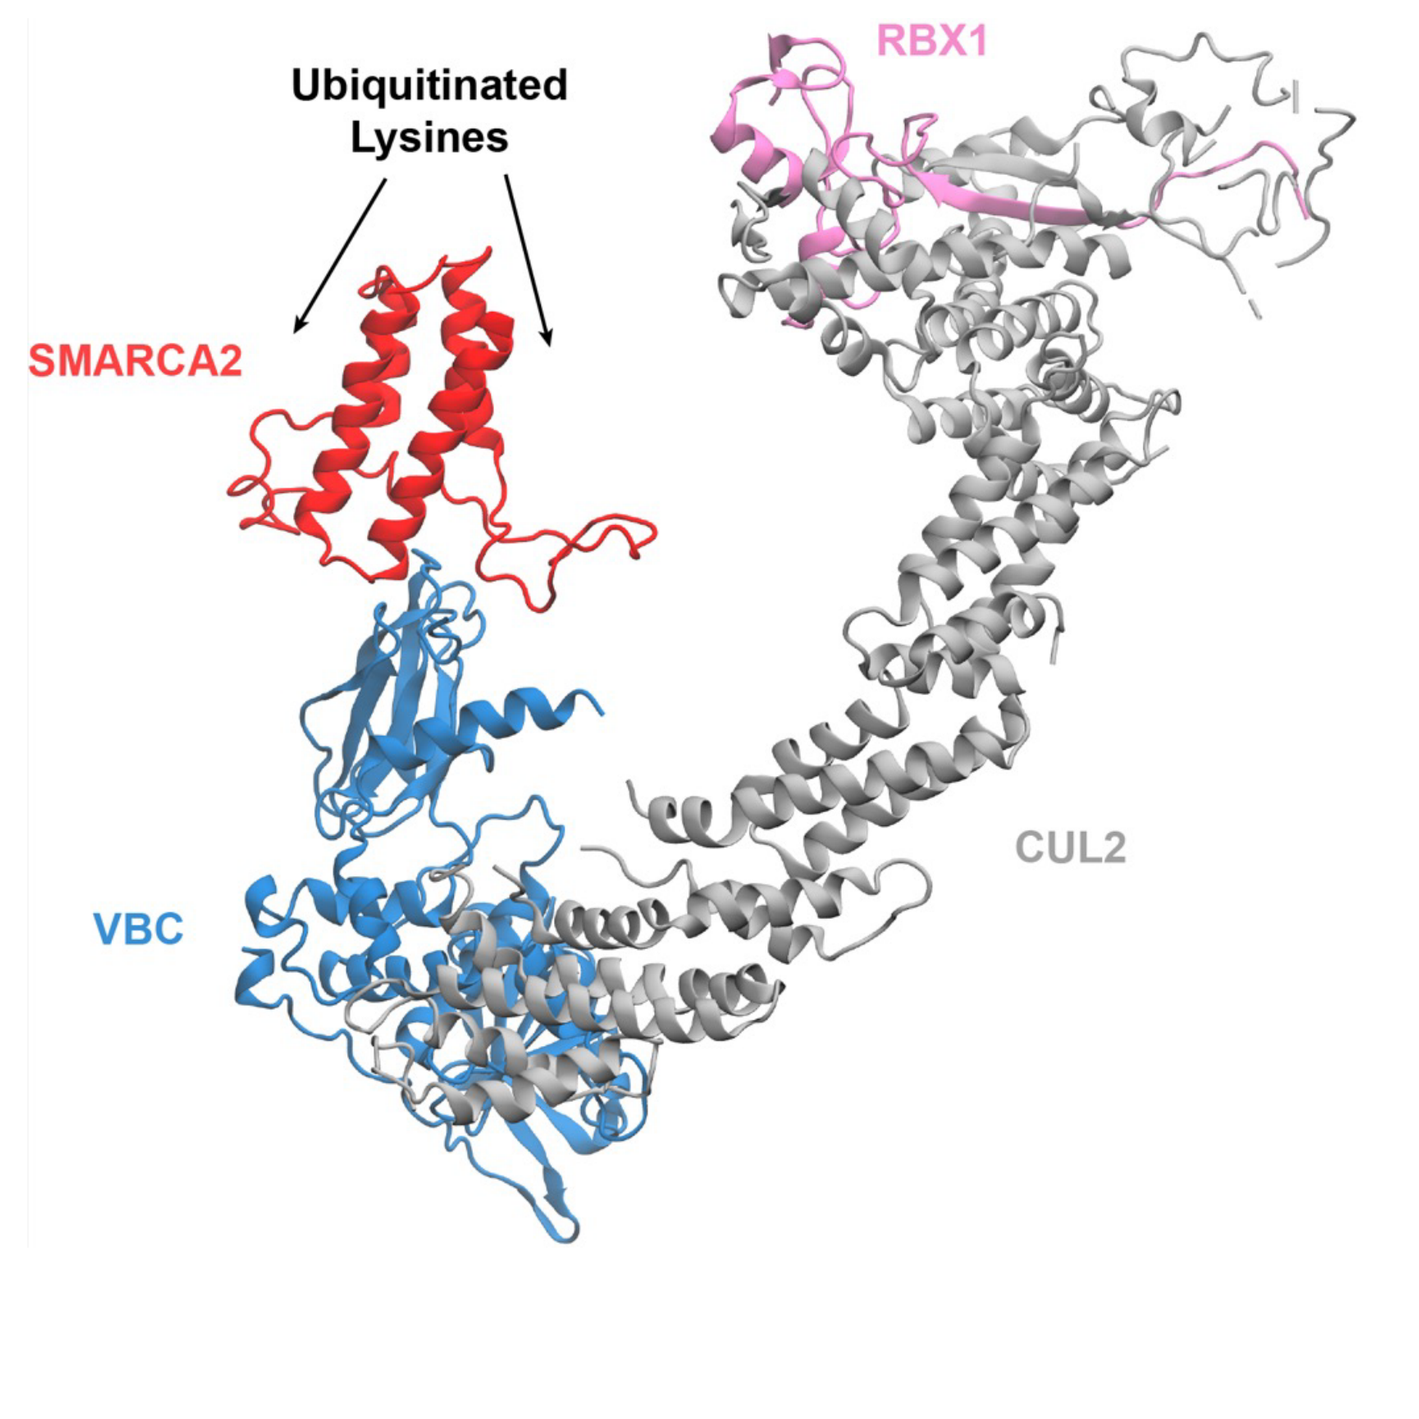
Ubiquitinated Lysines
SMARCA2
RBX1
VBC
CUL2

Figure 2. Mechanism of Action

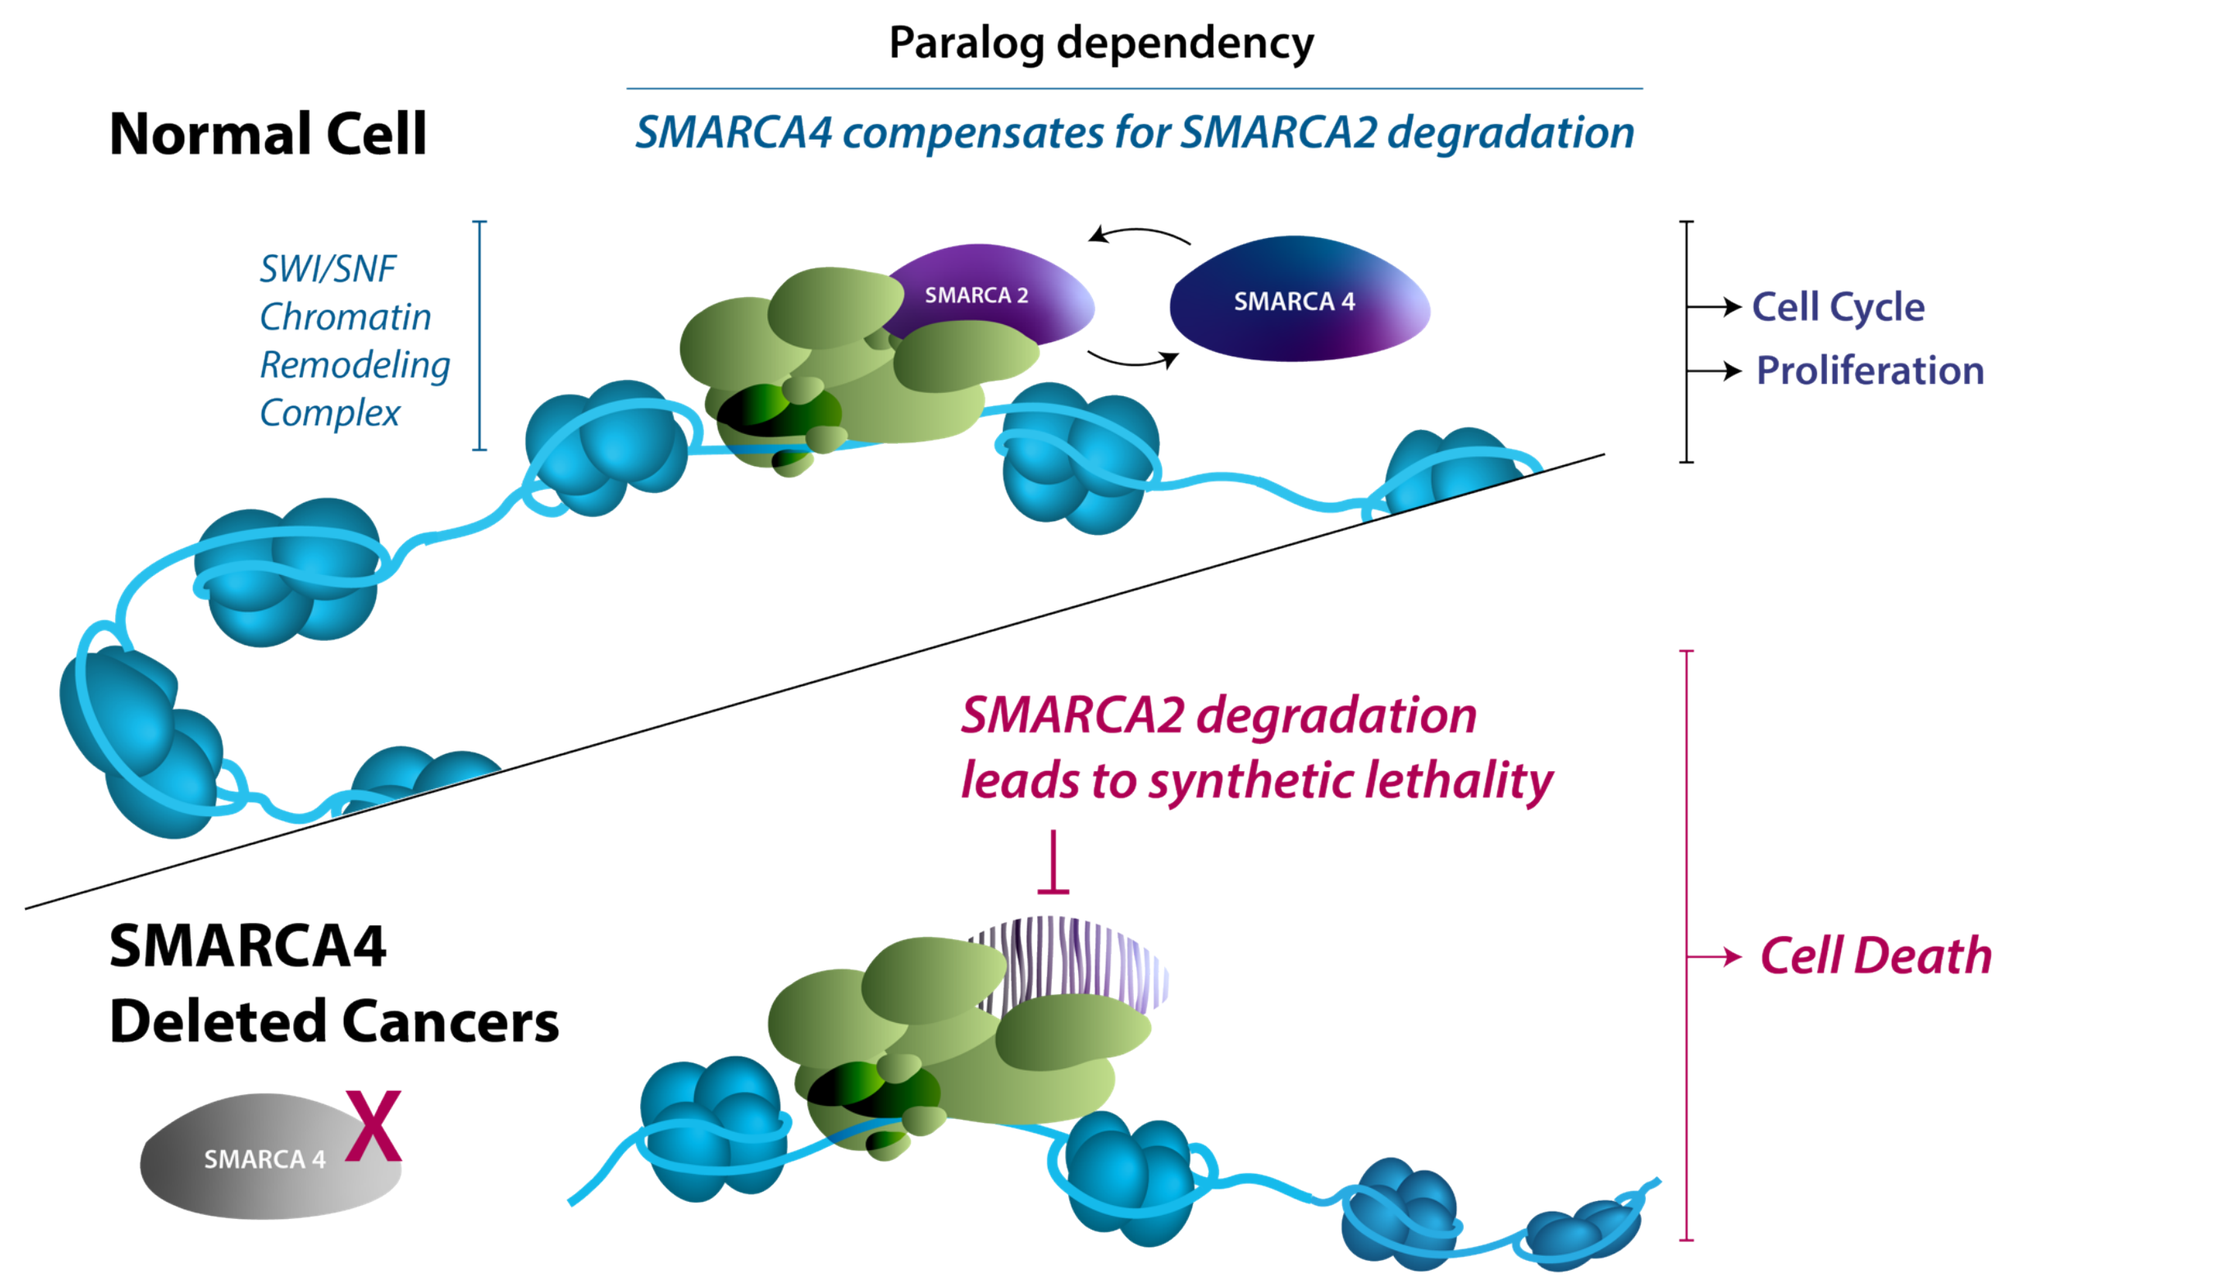
Paralog dependency
Normal Cell
SMARCA4 compensates for SMARCA2 degradation
SWI/SNF Chromatin Remodeling Complex
SMARCA 2
SMARCA 4
Cell Cycle
Proliferation
SMARCA2 degradation leads to synthetic lethality
SMARCA4 Deleted Cancers
SMARCA 4
X
Cell Death

Figure 3. PRT3789 Inhibits Proliferation of SMARCA4-Del/Knockout Cancer Cell Lines, but Not SMARCA4 WT Cancer Cell Lines in a Clonogenic Assay

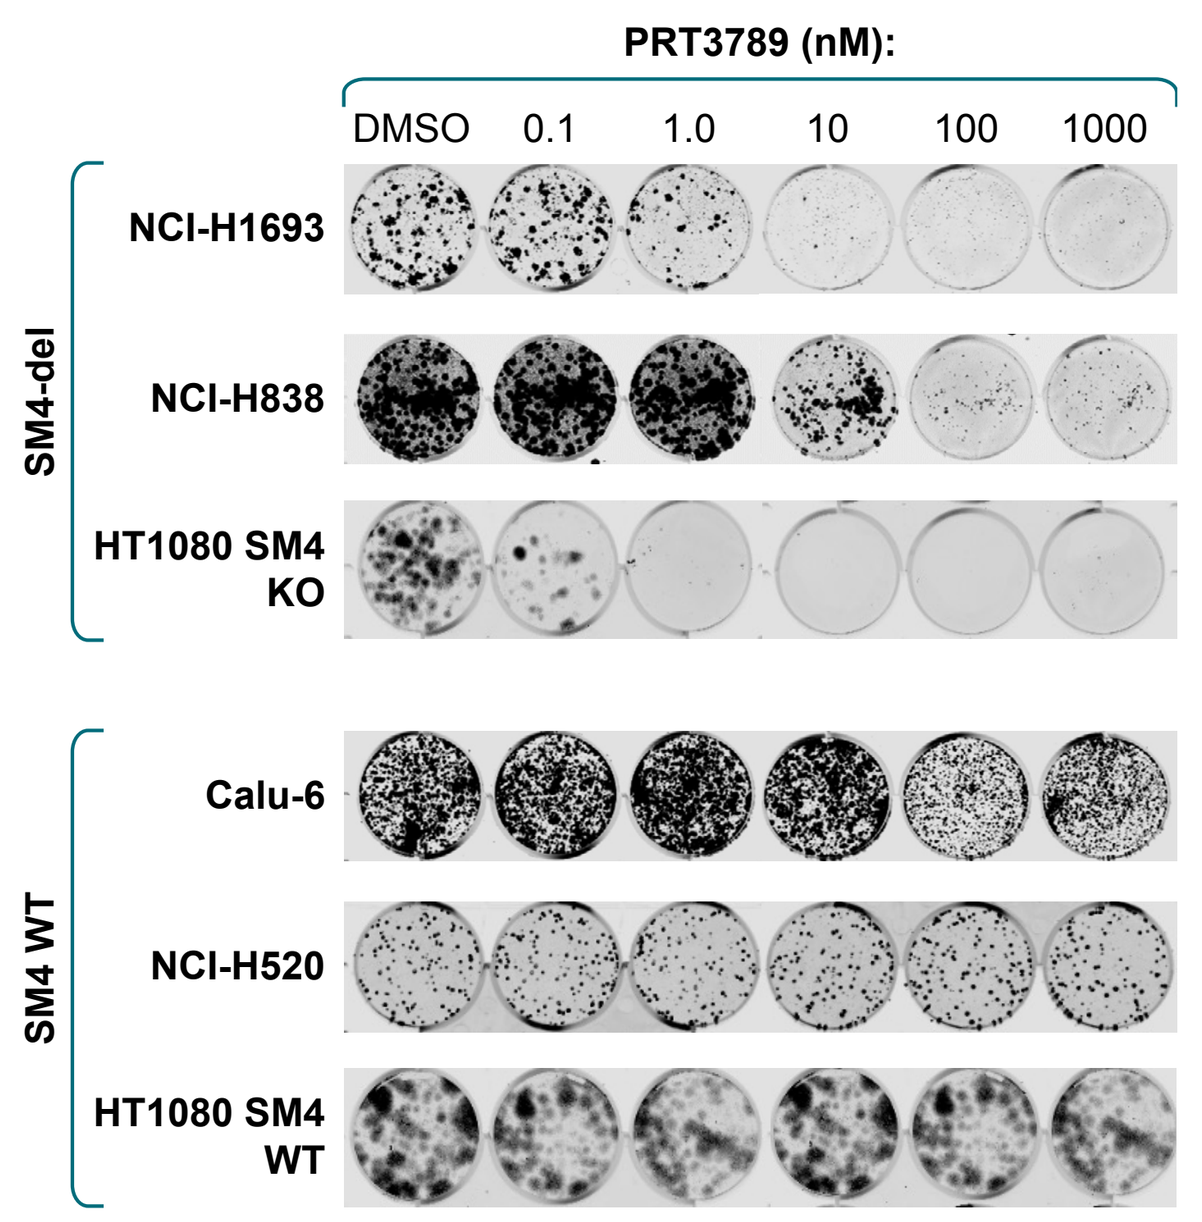
PRT3789 (nM):
DMSO
0.1
1.0
10
100
1000
SM4-del
NCI-H1693
NCI-H838
HT1080 SM4 KO
SM4 WT
Calu-6
NCI-H520
HT1080 SM4 WT

- PRT3789 selectively inhibits SMARCA4-deficient cancer cell proliferation in vitro (**Figure 3**)

Figure 4. SMARCA4-Del Cancer Cell Lines and PDX Models Are More Sensitive to PRT3789 vs WT and SMARCA2/4 Dual Loss Models

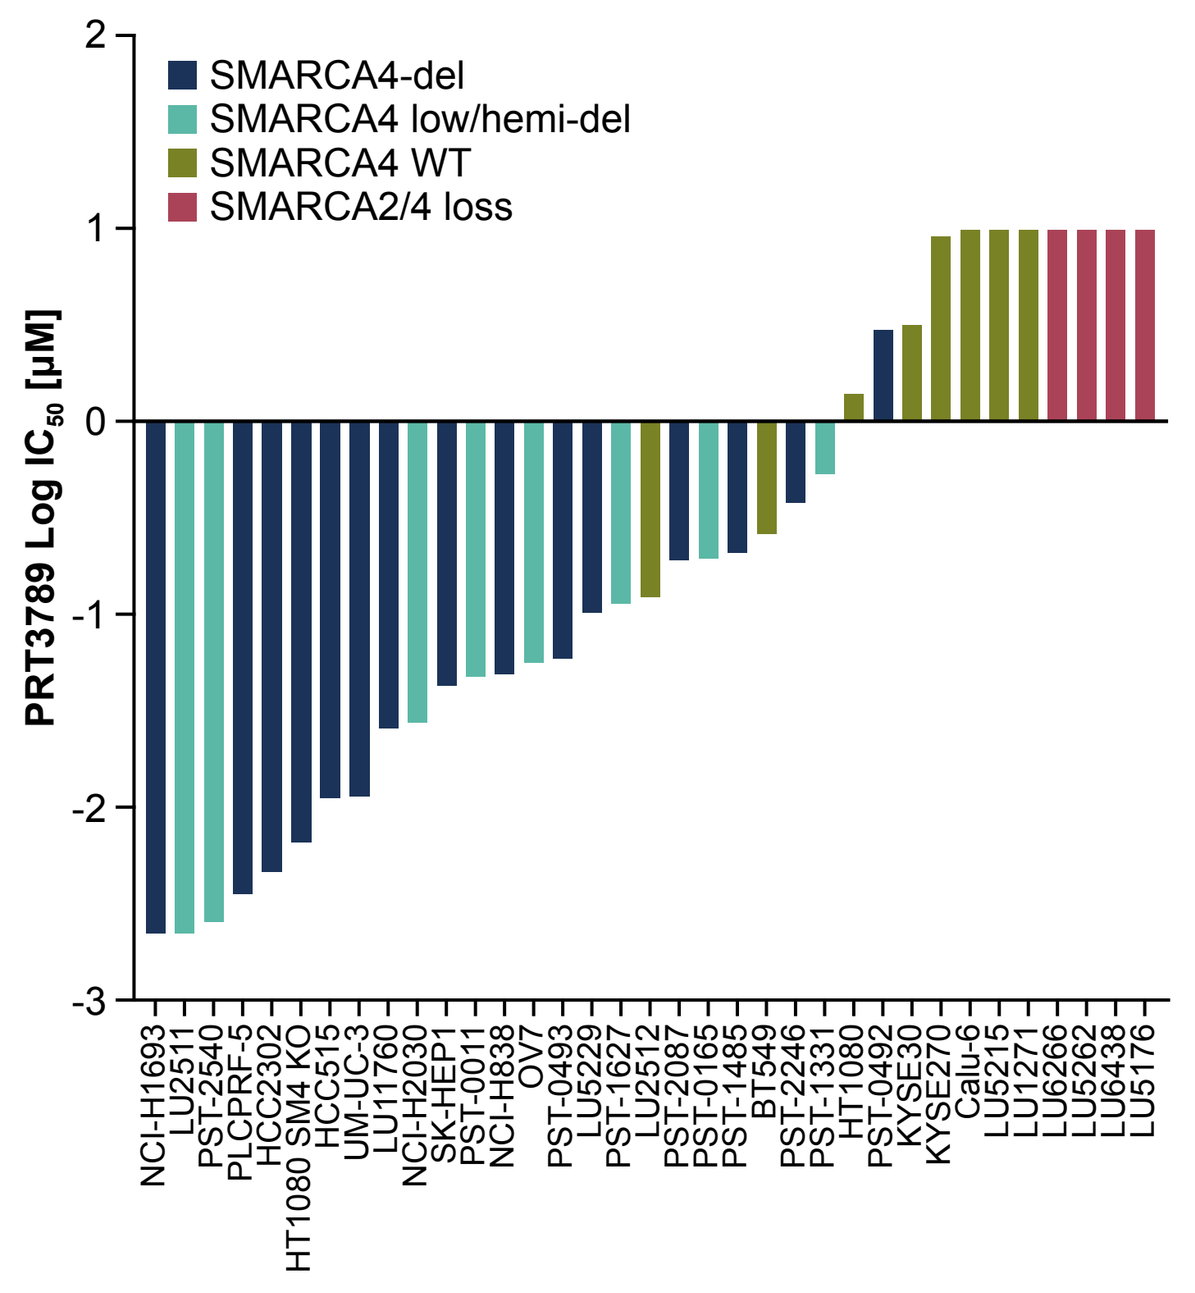
SMARCA4-del
SMARCA4 low/hemi-del
SMARCA4 WT
SMARCA2/4 loss
PRT3789 Log IC_{50} [μM]
2
1
0
-1
-2
-3
NCI-H1693
LU2511
PST-2540
PLCPRF-5
HCC2302
HT1080 SM4 KO
HCC515
UM-UC-3
LU11760
NCI-H2030
SK-HEP1
PST-0011
NCI-H838
OV7
PST-0493
LU5229
PST-1627
LU2512
PST-2087
PST-0165
PST-1485
BT549
PST-2246
PST-1331
HT1080
PST-0492
KYSE30
KYSE270
Calu-6
LU5215
LU1271
LU6266
LU5262
LU6438
LU5176

- There is none or limited response in SMARCA4 WT and SMARCA2/4 dual-loss cancer cells (**Figure 4**)

Figure 5. PRT3789 Significantly Inhibits Growth of SMARCA4-Del Lung Cancer CDX and PDX Models at Well-Tolerated Doses

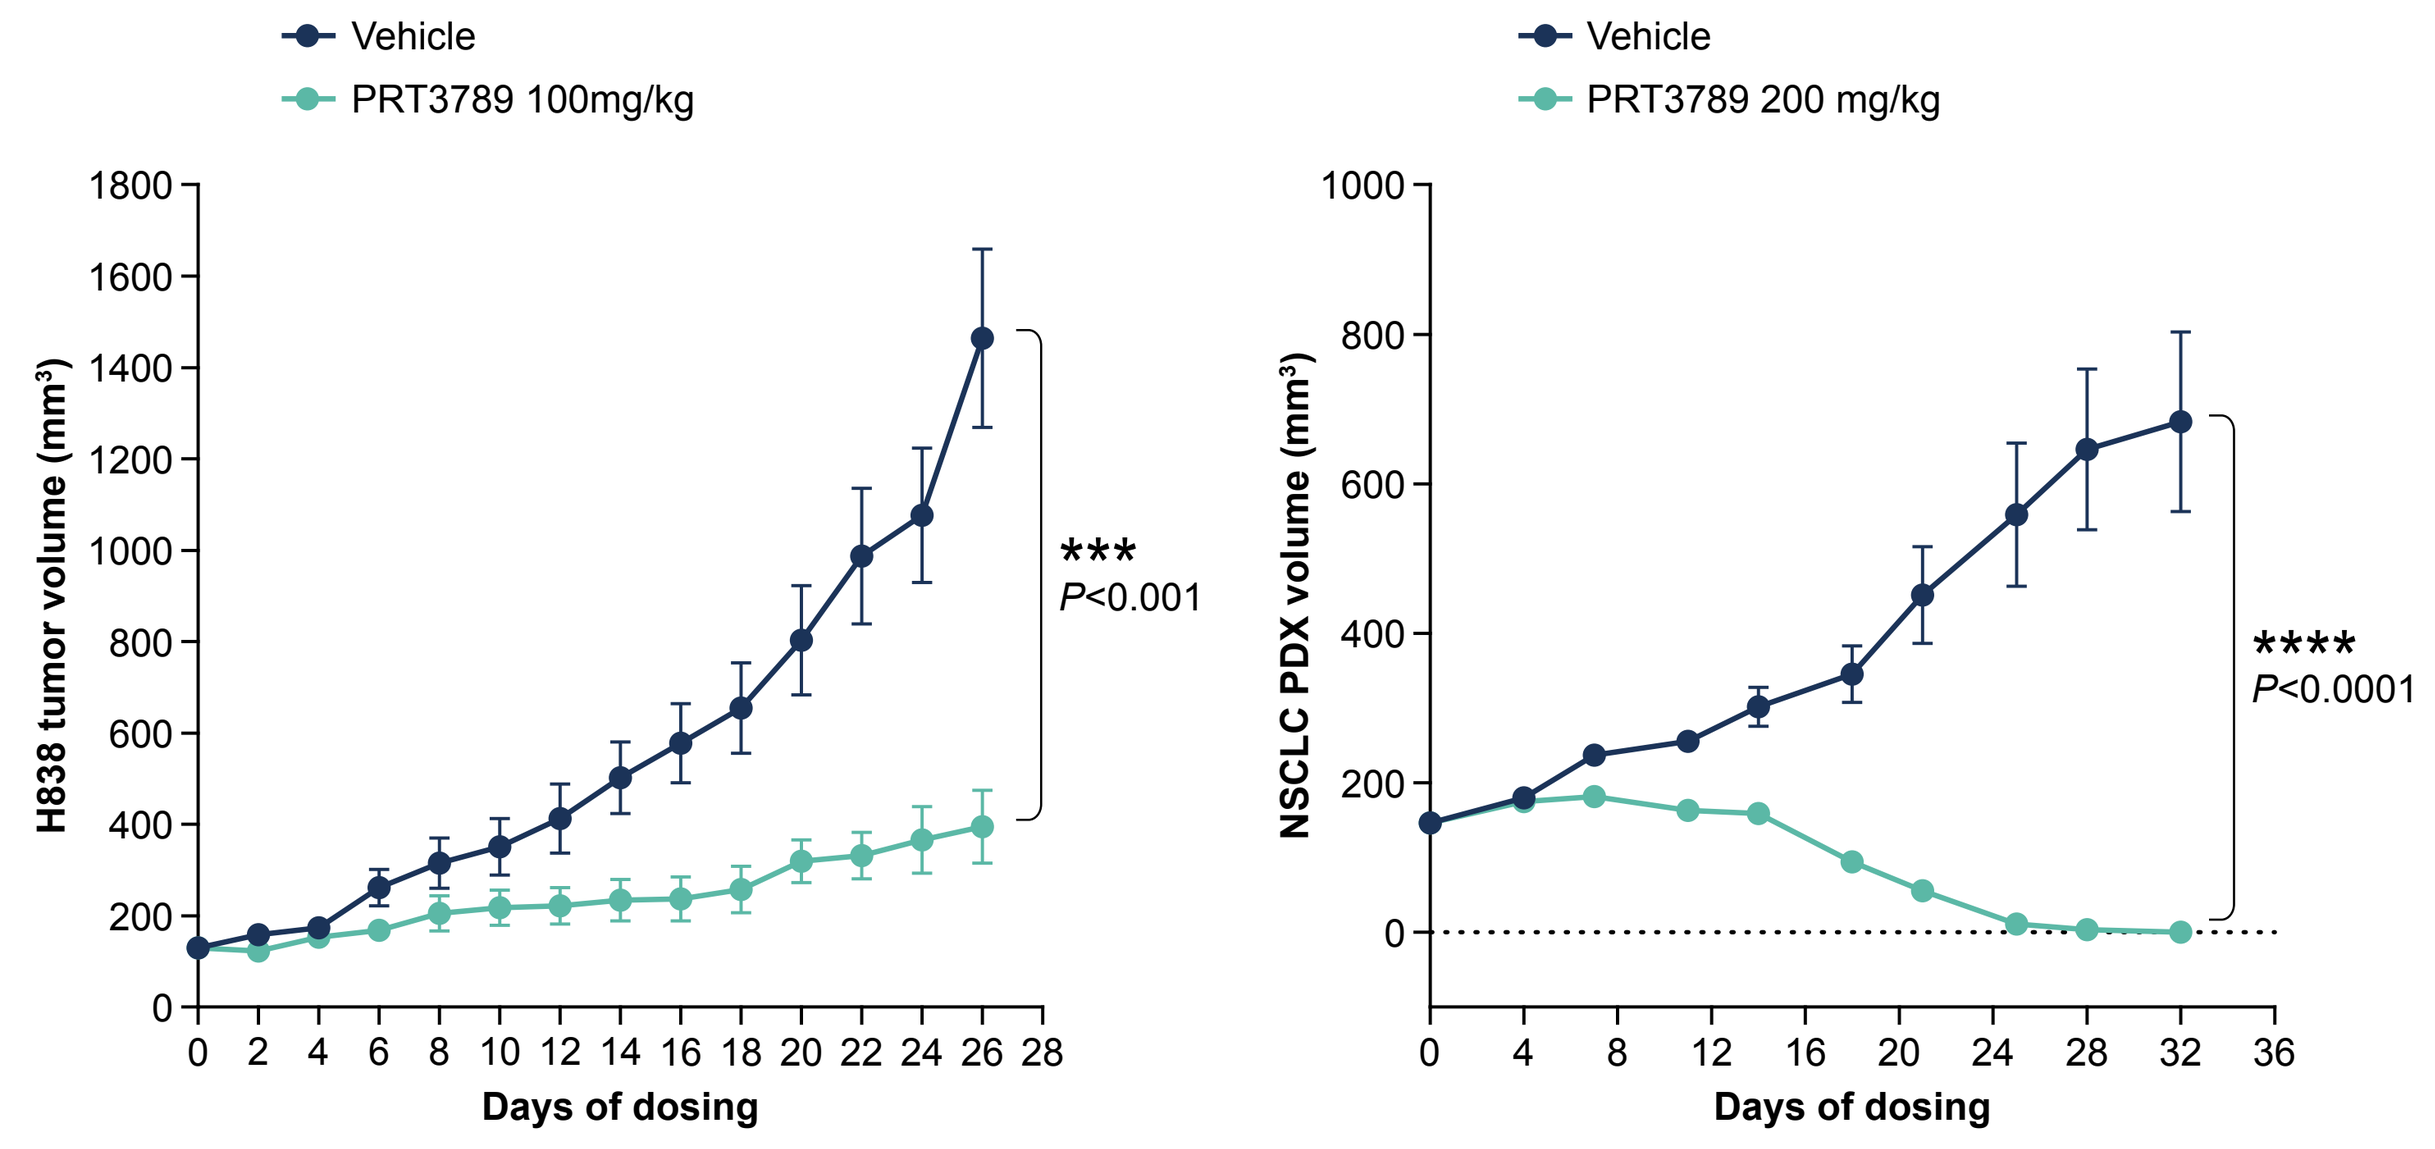
Vehicle
PRT3789 100mg/kg
H838 tumor volume (mm^3)
0
200
400
600
800
1000
1200
1400
1600
1800

P<0.001
Days of dosing
0 2 4 6 8 10 12 14 16 18 20 22 24 26 28
Vehicle
PRT3789 200 mg/kg
NSCLC PDX volume (mm^3)
0
200
400
600
800
1000

P<0.0001
Days of dosing
0 4 8 12 16 20 24 28 32 36

- PRT3789 is efficacious as a single agent at well-tolerated doses in *SMARCA4*-deficient NSCLC in preclinical models in mice (**Figure 5**)

OBJECTIVES

PRT3789-01 (NCT05639751) is an open-label, multicenter, dose-escalation, first-in-human, phase 1 study of PRT3789, a SMARCA2 degrader, for participants with select advanced or metastatic solid tumors with any SMARCA4 mutation for dose escalation cohorts and a SMARCA4 LOF mutation for backfill cohorts (**Figure 6**)

Primary Objectives

- To evaluate the safety, tolerability, and DLTs of PRT3789
- To determine the RP2D of PRT3789

Secondary Objectives

- To evaluate the efficacy (ORR, PFS, CBR, and DOR) of PRT3789 per investigator assessment per RECIST v1.1
- To evaluate the PK profile of PRT3789
- To evaluate the pharmacodynamic effect of PRT3789

Exploratory Objectives

- To evaluate additional markers of PRT3789 target engagement
- To understand baseline tumor biomarker profiles
- To explore potential role of ctDNA as surrogate endpoint for monitoring of disease response

STUDY DESIGN AND ENDPOINTS

Figure 6. PRT3789-01 Phase 1 Study Design

PRT3789 will be administered IV once weekly for 3 weeks (1 cycle)
DLTs assessed during the first 21 days of dosing in cycle 1

Dose Finding
BOIN Method

Solid tumors with any SMARCA4 mutation

Dose level 1 → Dose level 2 → Dose level 3 → Dose level 4 → Dose level 5 → Dose level 6 → Dose level 7 → Dose level 8

Possible Backfill Cohorts

Criteria to Enroll Backfill Cohort

1. All participants have cleared the DLT observation period; **AND**
2. An objective response per RECIST v1.1 has been observed; **OR**
3. The dose level is safe and biologically effective taking into account the safety, PK, and pharmacodynamic data

Up to 10 participants
(a minimum of 6 participants with NSCLC)
with a SMARCA4 LOF mutation

PATIENT ELIGIBILITY

Key Inclusion Criteria

- Patients aged ≥18 years with histologically confirmed advanced, recurrent, or metastatic solid tumor malignancy with any SMARCA4 mutation for dose escalation cohorts and a SMARCA4 LOF mutation for backfill cohorts by local testing that have either progressed on or are ineligible for SOC therapy are eligible to enroll
- *SMARCA4* mutation must be confirmed by local NGS or IHC in tumor tissue or blood using a clinically validated laboratory test
- Patients with NSCLC with driver alterations (eg, *EGFR*, *MET*, *RET*, *ALK*, *BRAF*, *KRAS*, *ROS1*, etc) are eligible after progression on approved targeted therapies
- Patients may have measurable or nonmeasurable, but evaluable, disease
- Tumor tissue sample from a core or excisional/surgical biopsy
- Adequate organ function laboratory values as defined in **Table 2**
- ECOG PS of 0 or 1

Table 2. Adequate Organ Function Laboratory Values

System	Laboratory value
Hematology[a]	
ANC	≥1.0 × 10^9/L
Platelets	≥75,000/μL
Renal	
Calculated creatinine clearance	≥50 mL/min according to the Cockcroft-Gault equation or other standard institutional method
Hepatic	
Total bilirubin	<1.5 × ULN for reference laboratory OR direct bilirubin <1.5 × ULN for participants with total bilirubin levels ≥1.5 × ULN, (≤3 × ULN if Gilbert disease)
AST and ALT	≤3.0 × ULN for reference laboratory unless there is hepatic involvement by the underlying malignancy as documented by either CT or ultrasound, in which case <5 × ULN is acceptable

[a]Patients who do not meet the criteria for hematologic function due to disease-related cytopenias and/or due to extensive bone marrow involvement may be enrolled into the study provided the ANC is ≥0.5 × 10^9/L and platelet count is ≥50,000/μL.

Key Exclusion Criteria

- Participants with solid tumors with known concomitant *SMARCA2* mutation or loss of protein expression (eg, SCCOHT-small-cell carcinoma of the ovary hypercalcemic type or thoracic sarcomatoid tumors)
- Clinically significant or uncontrolled cardiac disease, uncontrolled electrolyte disorders, uncontrolled or symptomatic CNS metastases or leptomeningeal disease
- Participant has not recovered to grade ≤1 from toxic effects of prior treatments (systemic, radiation, or surgery)
- History of another malignancy within 3 years except for adequately treated basal cell or squamous cell skin cancer, superficial bladder cancer, prostate intraepithelial neoplasm, carcinoma in situ of the cervix, or other noninvasive or indolent malignancies, or malignancies previously treated with curative intent and not on active therapy or expected to require treatment or recurrence during the study
- Concurrent treatment with strong or moderate CYP3A4 inhibitor or inducer
- Has a known history of hepatitis B or known active hepatitis C virus infection
- Pregnant or breastfeeding

ENROLLMENT, STATUS, AND REGISTRATION

- This study is registered at ClinicalTrials.gov (NCT05639751) and is active in the United States, France, Spain, and the Netherlands
- Study is enrolling well with treatment commencing at the 4th dose level, with a promising early safety profile and SMARCA2 degradation data

REFERENCES

1. Shain AH, et al. *PLoS ONE*. 2013;8:e55119.
2. Fernando TM, et al. *Nat Commun*. 2020;11(1)551.
3. Helming KC, et al. *Cancer* Cell. 2014;26:309-317.
4. Hoffman GR, et al. *Proc Natl Acad Sci U S A*. 2014;111:3128-3133.
5. Oike T, et al. *Cancer Res*. 2013;73(17):5508-5518.
6. cBioPortal. TCGA Pan Cancer Database. Accessed August 20, 2023. https://www.cbioportal.org/
7. Dagogo-Jack I, et al. *J Thorac Oncol*. 2020;15(5):766-776.
8. Schoenfeld AJ, et al. *Clin Cancer Res*. 2020;26(21):5701-5708.
9. Alessi JV, et al. *J Thorac Oncol*. 2023;18(6):731-743.

ABBREVIATIONS

ANC, absolute neutrophil count; ATP, adenosine triphosphate; BOIN, Bayesian optimal interval; BRG1, brahma-related gene-1; BRM, brahma; CBR, clinical benefit rate; CDX, cell line-derived tumors xenograft, CIT, chemoimmunotherapy; C_{max}, maximum concentration; CNS, central nervous system; CT, computed tomography; ctDNA, circulating tumor DNA; CYP3A4, cytochrome P450 3A4; DC50, half-maximal degradation concentration; DLT, dose-limiting toxicity; DOR, duration of response; ECOG PS, Eastern Cooperative Oncology Group performance status; hERG, human ether-a-go-go-related gene; HiBit, a small 11 amino acid peptide that binds with high affinity to another larger subunit called LgBiT; IC_{50}, half-maximal inhibitory concentration; ICW, intracellular water; IHC, immunohistochemistry; IV, intravenous; KEAP1, kelch-like ECH-associated protein 1; KRAS, kirsten rat sarcoma viral oncogene homolog; LOF, loss of function; mOS, median overall survival; mPFS, median progression-free survival; NGS, next-generation sequencing; NSCLC, non-small cell lung cancer; ORR, overall response rate; OS, overall survival; PD, progressive disease; PDX, patient-derived xenograft; PFS, progression-free survival; PK, pharmacokinetic; q3d, every 3 days; RECIST, Response Evaluation Criteria in Solid Tumors; RP2D, recommended phase 2 dosing; sc, subcutaneous; SCCOHT, small cell carcinoma of the ovary, hypercalcemic type; SMARCA2, SWI/SNF related, matrix associated, actin dependent regulator of chromatin, subfamily A, member 2; SMARCA4, WI/SNF related, matrix associated, actin dependent regulator of chromatin, subfamily A, member 4; SOC, standard of care; STK11, serine/threonine kinase 11STK11; SWI/SNF, switch/sucrose non-fermentable; TDI, time-dependent inhibition; TP53, tumor protein 53; VHL, von Hippel-Lindau; WT, wild type.

ACKNOWLEDGMENTS

- Funding support for PRT3789-01 was provided by Prelude Therapeutics Incorporated
- We thank the patients and their caregivers for participating in PRT3789-01; the study investigators, coordinators, and health care staff at each study site; and Kimberli Brill, BSN, for contributions to the development of the PRT3789-01 study protocol and site selection support during her tenure at Prelude Therapeutics Incorporated
- Medical writing support was provided by Laura S. Moye, PhD, ISMPP CMPP™, of Team 9 Science, funded by Prelude Therapeutics Incorporated

CORRESPONDENCE

Ibiayi Dagogo-Jack, MD
Massachusetts General Hospital
Boston, MA, USA
idagogo-jack@partners.org
Please contact **PRT3789-01IV@preludetx.com** for more information

DISCLOSURES

AD: holds a consulting or advisory role with AbbVie, Amgen, AstraZeneca, BMS, Ipsen, Merck, Seattle Genetics.
Full author disclosures are available through the Quick Response (QR) code.

Presented at the Annual European Society for Medical Oncology Congress (ESMO 2023), 20-24 October, 2023, in Madrid, Spain

FULL AUTHOR DISCLOSURES

ID-J: holds a consulting or advisory role with AstraZeneca, Bayer, Boehringer Ingelheim, BostonGene, Bristol Myers Squibb Foundation, Catalyst Pharmaceuticals, Genentech, Janssen, Novocure, Pfizer, Sanofi, Syros Pharmaceuticals, Xcovery; received honoraria from American Academic Health System, Aptitude Health, ASCO Post, Creative Educational Concepts, DAVA Oncology, Foundation Medicine, Medscape, OncLive, Research To Practice, Total Health, Triptych Health Partners; received research funding from Array BioPharma, BostonGene, Calithera Biosciences, Guardant Health, Genentech, Novartis, Tango Therapeutics, Pfizer, Vivace Therapeutics. **RG:** received research funding from Merck. **MA:** holds a consulting or advisory role with AbbVie, Achilles, ArcherDX, Ariad, AstraZeneca, Blueprint Medicines, Bristol Myers Squibb, EMD Serono, Foundation Medicine, Genentech, Gritstone, Hengrui, Maverick, Merck, Mirati Therapeutics, Nektar, Neon, NextCure, Novartis, Panvaxal/NovaRx, Syndax; received research funding from AstraZeneca, Bristol Myers Squibb, Genentech, Lilly. **ASw:** holds a consulting or advisory role with Amgen, AstraZeneca, BMS, Janssen, Lilly, Pfizer, Roche; received honoraria from Amgen, AstraZeneca, BMS, Janssen, Roche; received travel, accommodations, or paid expenses from Pfizer, Roche. **EC:** president and cofounder of INTHEOS (Investigational Therapeutics in Oncological Sciences); previous employment with HM Hospitales Group, START Madrid – CIOCC (Centro Integral Oncológico Clara Campal); holds a consulting or advisory role, is a steering/data monitoring committee member, or has been an invited speaker for Adcendo, Amunix, Anaveon, AstraZeneca, BeiGene, BMS, Chugai, CRIS Cancer Foundation, Diaccurate, Elevation Oncology, Ellipses Pharmacy, EORTC IDMC, Genmab, Janssen, Merus, MonTa, MSD, Nanobiotix, Nouscom, Novartis, OncoDNA, PharmaMar, PsiOxus, Roche/Genentech, Sanofi, Servier, Syneos Health, T-knife, TargImmune; is a member on the board of directors for PharmaMar; has an ownership interest in Ocoart Associated, START. **VM:** holds a consulting or advisory role with AstraZeneca, Basilea, Bayer, BMS, Janssen, Roche, START; received research funding from AbbVie. **AA:** holds a consulting or advisory role with Sagent Pharmaceuticals, Johnson & Johnson, Merck AG, Swiss Rockets, Zai Labs; received research funding from Vyriad; is the editor-in-chief for the International Association for the Study of Lung Cancer. **PL:** holds a consulting or advisory role or participates on a data safety monitoring board or steering committee for AbbVie, ABL Bio, Agenus, Agios, American Association for Cancer Research, American Society of Clinical Oncology, Association of American Cancer Institute, Astellas Pharmaceuticals, AstraZeneca, BAKX Therapeutics, Bayer, Black Diamond, Cancer Research United Kingdom, Case Western Reserve University, Compass BADX, Cybrexa, CytomX, EMD Serono, Five Prime, Genentech, Genmab, GSK, Halozyme, I-Mab, ImCheck, ImmunoMet, IQVIA, Kineta, Kyowa Kirin, MacroGenics, Mekanist, Mersana, Molecular Templates, National Cancer Institute, NeuroTrials, Pfizer, QED Therapeutics, Qualigen, Relay Therapeutics, Roche-Genentech, Roivant Sciences, Salarius, Scenic Biotech, Seagen, Shattuck, Silverback, SK Life, SOTIO, STCube, Stemline, Takeda, Targeted Anti-Cancer Therapies, TRIGR, Tyme, University of Arizona, University of California San Diego, University of New Mexico, Zentalis; owns stock or equity in BAXK; received research funding from AbbVie, ADC Therapeutics, ALX Oncology, Astellas Pharmaceuticals, Astex Pharmaceuticals, AstraZeneca, Bayer, Black Diamond, Boehringer Ingelheim, Calico Life Sciences, Corvus Pharmaceuticals, CytomX, Eisai Pharmaceuticals, Eli Lilly, EMD Serono, F-Star Delta Limited, Five Prime, FLX Bio, Genentech, Genmab, Incyte, Jounce, Linnaeus Therapeutics, MedImmune, Merck Sharp & Dohme, Moderna Therapeutics, NextCure, Pfizer, Ribon Therapeutics, SOTIO, Stemline, Takeda, Tesaro. **SP:** has received research funding from A2Bio, Astellas Pharmaceuticals, Constellation, Revolution Medicine, Simcere Pharmaceuticals, VITRAC. **TL:** holds a consulting or advisory role with Amgen, AstraZeneca, Daiichi Sankyo, Eisai, EMD Serono, Janssen, Jazz; received research funding from Advaxis, Pfizer. **TAY:** employment from Institute for Applied Cancer Science, University of Texas MD Anderson Cancer Center; owns stock in Seagen; holds a consulting or advisory role with AbbVie, AstraZeneca, Acrivon, Adagene, Almac, Aduro, Amphista, Artios, Athena, Atrin, Avoro, Axiom, Baptist Health Systems, Bayer, BeiGene, Blueprint Medicines, Boxer, Bristol Myers Squibb, C4 Therapeutics, Calithera, Cancer Research UK, Circle Pharma, Clovis, CUHK Committee, Cybrexa, Dark Blue Therapeutics, Diffusion, Ellipses, EMD Serono, F-Star, Genentech, Genmab, Gerson and Lehrman Group, Glenmark, GLG, Globe Life Sciences, GSK, Guidepoint, Idience, Ignyta, I-Mab, ImmuneSensor, Institut Gustave Roussy, Intellisphere, Jansen, Kyn, LRG1, MEI Pharma, Mereo, Merck, Natera, Nexys, Novocure, OHSU, OncoSec, Ono Pharma, Panangium, Pegascy, PER, Pfizer, Piper-Sandler, Pliant Therapeutics, Prolynx, Radiopharm Theranostics, Repare, resTORbio, Roche, Sanofi, Schrodinger, Seagen, Synthis Therapeutics, Terremoto Biosciences, Tessellate Bio, TD2 Theragnostics, Tome Biosciences, Varian, Versant, Vibliome, Xinthera, Zai Labs, Zentalis, ZielBio; received research funding from Acrivon, Artios, AstraZeneca, Bayer, BeiGene, BioNTech, Blueprint Medicines, BMS, Boundless bio, Clovis, Constellation, Cyteir, Eli Lilly, EMD Serono, Forbius, F-Star, GlaxoSmithKline, Genentech, Haihe, Ideaya ImmuneSensor, Ionis, Ipsen, Jounce, Karyopharm, KSQ, Kyowa, Merck, Mirati, Novartis, Pfizer, Ribon Therapeutics, Regeneron, Repare, Rubius, Sanofi, Scholar Rock, Seagen, Tesaro, Vivace, Zenith.
AI: received honoraria from, holds a consulting or advisory role with, and has received research funding from AstraZeneca, Bayer, BMS, Lilly, Merck, MSD, Novartis, Parthenon, Pharmamar, Roche. **WN:** employment, holds a leadership position, owns stock, and has received travel, accommodations, or paid expenses from Prelude Therapeutics Incorporated. **CT:** employment, owns stock, and has received travel, accommodations, or paid expenses from Prelude Therapeutics Incorporated. **SR:** employment from Prelude Therapeutics Incorporated. **KB:** current or previous employment, owns stock with Prelude Therapeutics Incorporated, Pfizer. **GP:** nothing to disclose. **WS:** received honoraria from Prelude Therapeutics Incorporated. **ASp:** holds a consulting or advisory role with Amgen, Array BioPharma, AstraZeneca/MedImmune, Bayer, Blueprint Medicines, Bristol Myers Squibb, CytomX, Daiichi Sankyo/AstraZeneca, Gritstone Bio, Gritstone Oncology, Janssen Oncology, Janssen Research & Development, Jazz, Incyte, Merck, Mersana, Mirati Therapeutics, NEXT Oncology Virginia, Novartis, Takeda; owns stock in Eli Lilly; received research funding from AbbVie, ADCT, Amgen, Arch Therapeutics, Astellas Pharmaceuticals, Astex Pharmaceuticals, AstraZeneca, Blueprint Medicines, Boehringer Ingelheim, Bristol Myers Squibb, CytomX, Daiichi Sankyo, Gritstone, Ignyta, Incyte, Janssen Oncology, Kezar, LAM Therapeutics, LOXO Oncology, MacroGenics, MedImmune, Mersana, Mirati Therapeutics, NewLink Genetics, Novartis, Plexxikon, Revolution Medicine, Roche, Rubius, Synthekine, Takeda, Trovagene.
BB: received research funding from 4D Pharmaceuticals, AbbVie, Amgen, Aptitude Health, AstraZeneca, BeiGene, Blueprint Medicines, Boehringer Ingelheim, Celgene, Cergentis, Cristal Therapeutics, Daiichi Sankyo, Eli Lilly, GSK, Janssen, Onxeo, OSE Immunotherapeutics, Pfizer, Roche-Genentech, Sanofi, Takeda, Tolero Pharmaceuticals, Chugai Pharmaceuticals, Eisai, Genzyme Corporation, Inivata, IPSEN, Turning Point Therapeutics.