Selective and orally bioavailable SMARCA2 targeted degraders induce synthetic lethality in SMARCA4-deficient solid tumor

Koichi Ito, Artem Shvartsbart, Joseph Rager, Anjana Agarwal, Michael Hulse, Komali Vykuntam, Min Wang, Justin Kurian, Miles Cowart, Joy Cote, Monisha Sivakumar, Jack Carter, Jessica Burtell, Alex Grego, Andrew Moore, Neha Bhagwat, Stefan Ruepp, Tom Emm, Liang Lu, Philip Pitis, Corey Basch, Klare Bersch, Song Mei, Raul Leal, John Rose, Danielle Roth, Chaoyi Xu, Ganfeng Cao, Kris Vaddi, Sandy Geeganage, Bruce Ruggeri, Andrew Combs, Peggy Scherle

Prelude Therapeutics Incorporated, Wilmington, DE; contact: kito@preludetx.com

6277

Background

- SMARCA2 (BRM) and SMARCA4 (BRG1) are the two mutually exclusive catalytic core subunits of SWI/SNF complexes that play an important role in controlling gene expression by remodeling chromatin [1].
- SMARCA4 has been shown to be mutated in multiple cancers, including up to 12% of non-small cell lung cancer [2].
- The SMARCA4-deficient cancer cells are highly dependent on the paralog gene SMARCA2 for their survival [3,4,5].
- Our selective SMARCA2 degrader PRT3789 demonstrates synthetic lethality in pre-clinical models [4] (Phase I trial underway: NCT05639751).
- We have identified novel, orally bioavailable SMARCA2 degrader series that induce strong synthetic lethality in SMARCA4-deficient cancer cells.

Introduction

Synthetic lethal relationship between targeting SMARCA2 and SMARCA4-deficiency in cancer

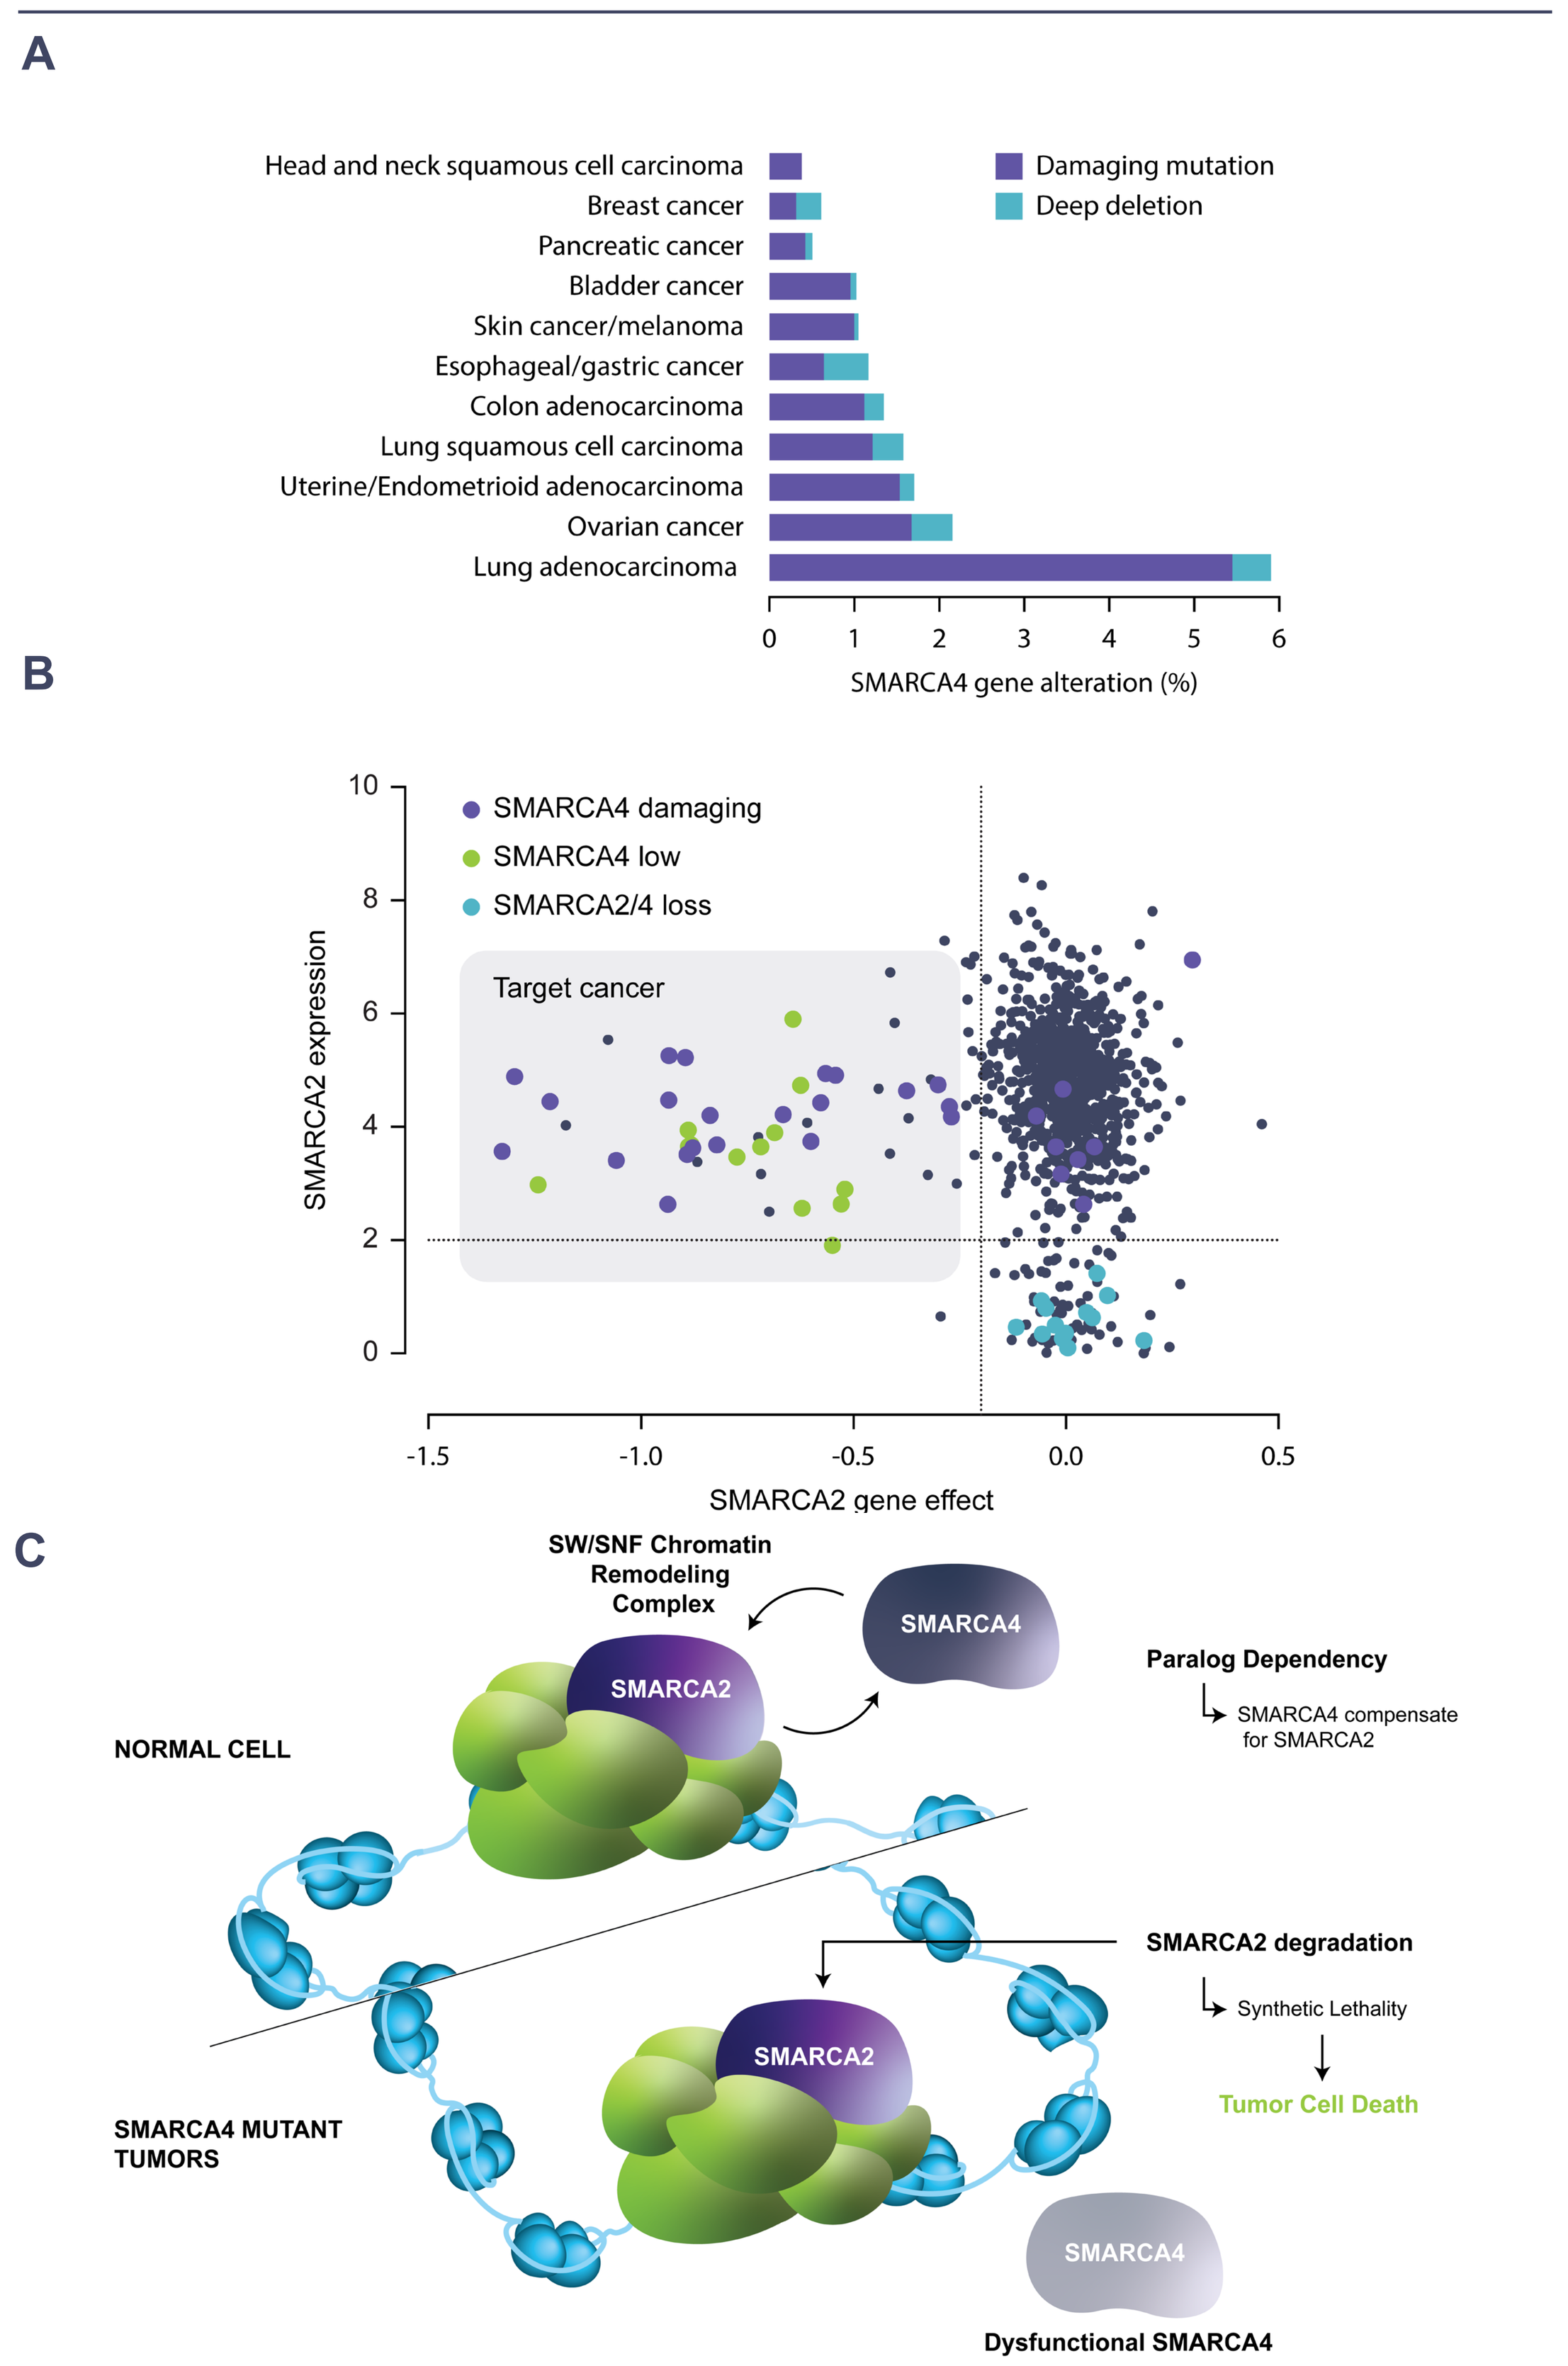
A
Head and neck squamous cell carcinoma
Breast cancer
Pancreatic cancer
Bladder cancer
Skin cancer/melanoma
Esophageal/gastric cancer
Colon adenocarcinoma
Lung squamous cell carcinoma
Uterine/Endometrioid adenocarcinoma
Ovarian cancer
Lung adenocarcinoma
Damaging mutation
Deep deletion
SMARCA4 gene alteration (%)
B
SMARCA4 damaging
SMARCA4 low
SMARCA2/4 loss
Target cancer
SMARCA2 expression
SMARCA2 gene effect
C
SW/SNF Chromatin Remodeling Complex
SMARCA4
SMARCA2
Paralog Dependency
SMARCA4 compensate for SMARCA2
NORMAL CELL
SMARCA2 degradation
Synthetic Lethality
Tumor Cell Death
SMARCA4 MUTANT TUMORS
Dysfunctional SMARCA4

A) Percentage of SMARCA4 mutations in different types of cancer. Datasets are from mixed studies (2021). **B)** Cell lines with SMARCA4 damaging mutation or low expression show high SMARCA2 gene dependency scores [3, 5], suggesting the synthetic lethal relationship of targeting SMARCA2 and SMARCA4-deficiency. **C)** Model of SMARCA2 degradation induced synthetic lethality in SMARCA4 deficient cancers.

Background

To identify and characterize orally bioavailable, highly potent and selective SMARCA2 protein degraders using *in vitro* and *in vivo* models of SMARCA4 deficient cancers.

Key Findings

- Identified novel orally bioavailable SMARCA2 degraders.
- Demonstrated superior degradation selectivity for SMARCA2
- Oral administration of SMARCA2 degraders demonstrated significant anti-tumor activity in SMARCA4-deficient NSCLC models in mice

Figure 1. SMARCA2 bromodomain and E3 ligase complex

A

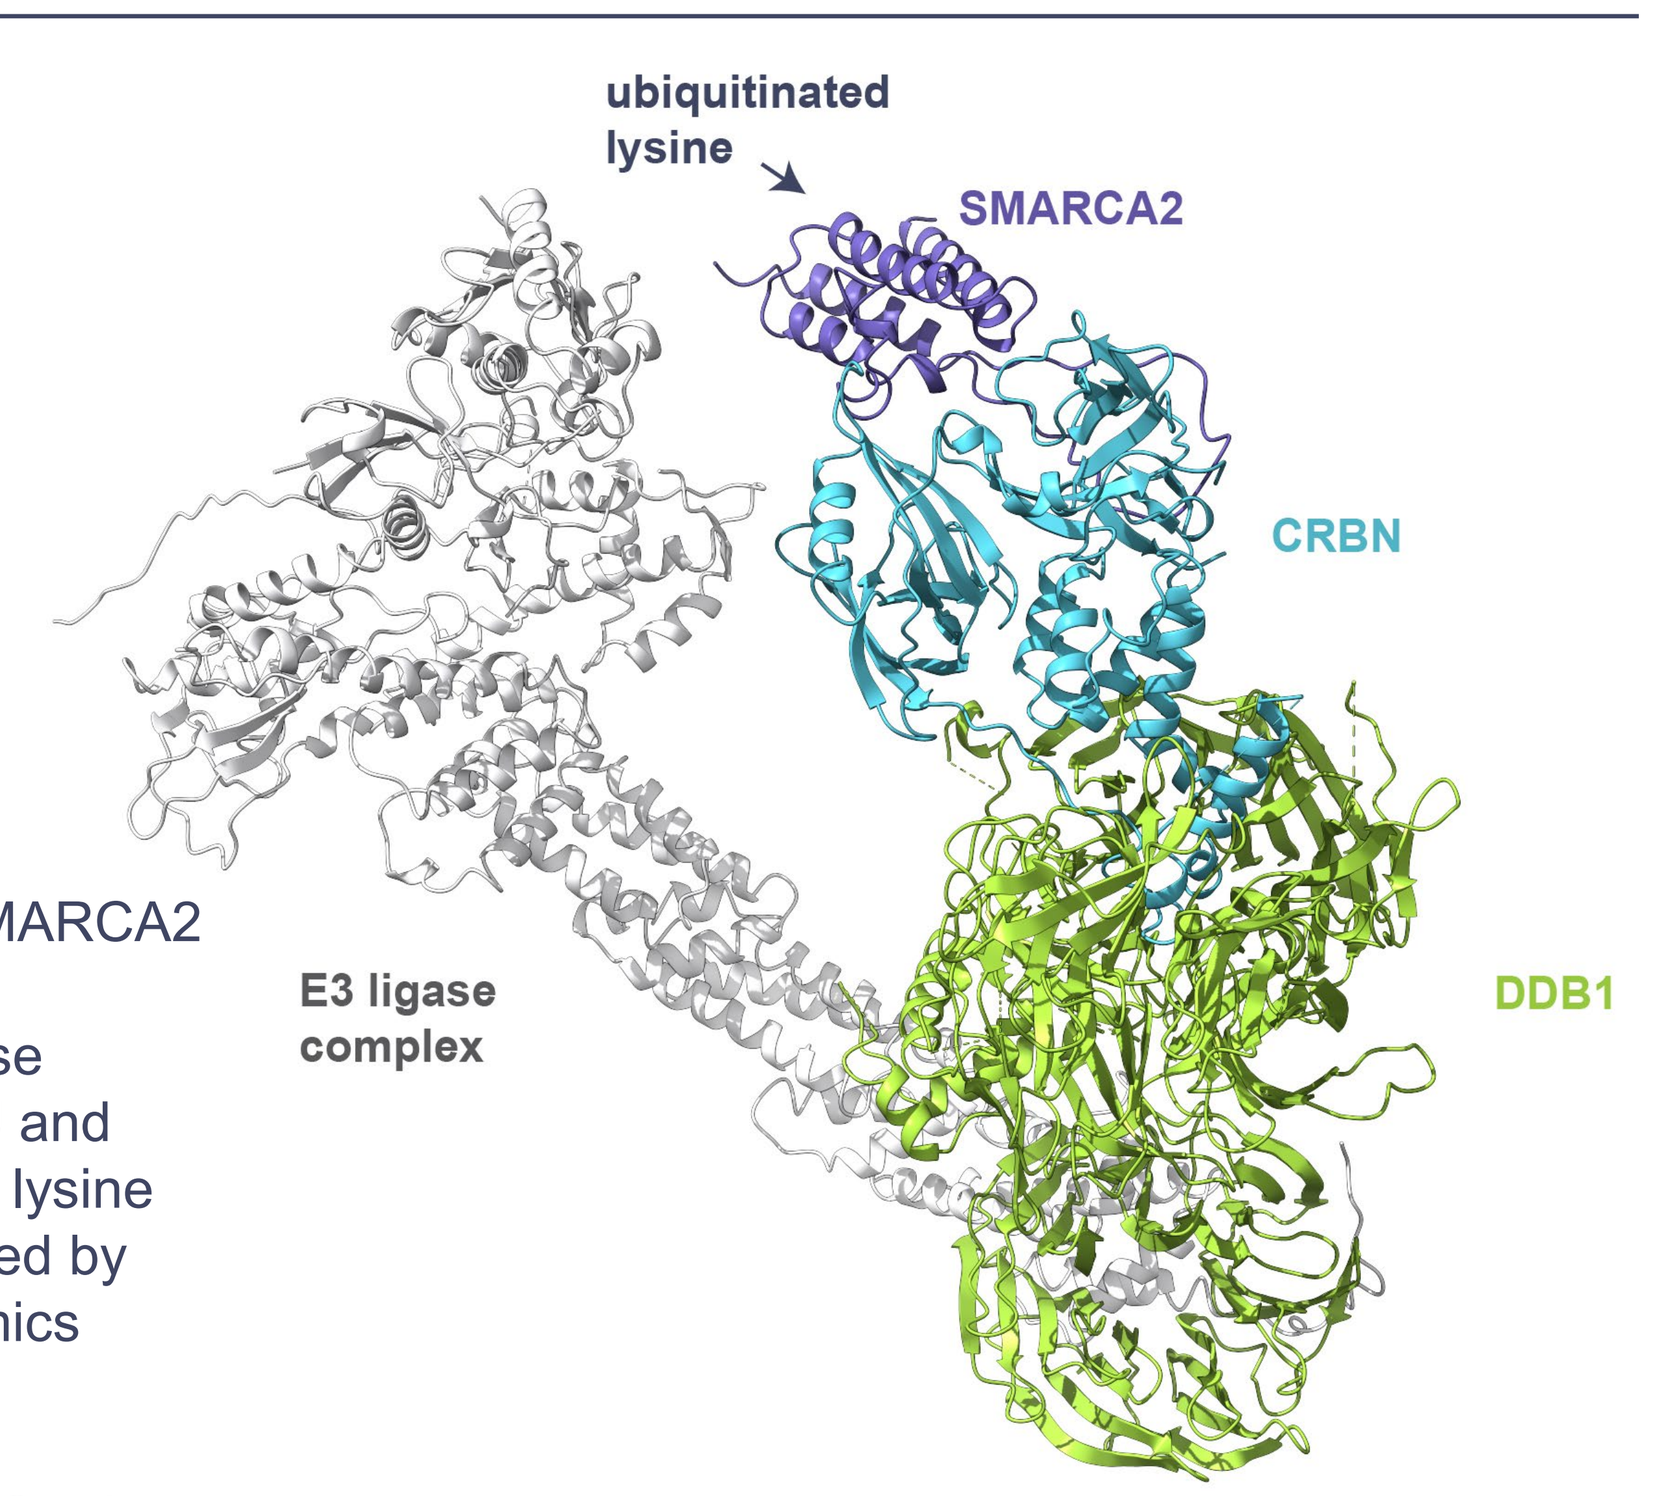
ubiquitinated lysine
SMARCA2
CRBN
E3 ligase complex
DDB1

A) 3D structure of SMARCA2 bromodomain and CRBN/DDB1 E3 ligase complex (PDB: 6TTU and 6BNB). Ubiquitinated lysine residues were detected by Ubiquitin MS proteomics study.

Figure 2. SMARCA2 oral degraders show excellent potency and selectivity for SMARCA2 in human cancer cells

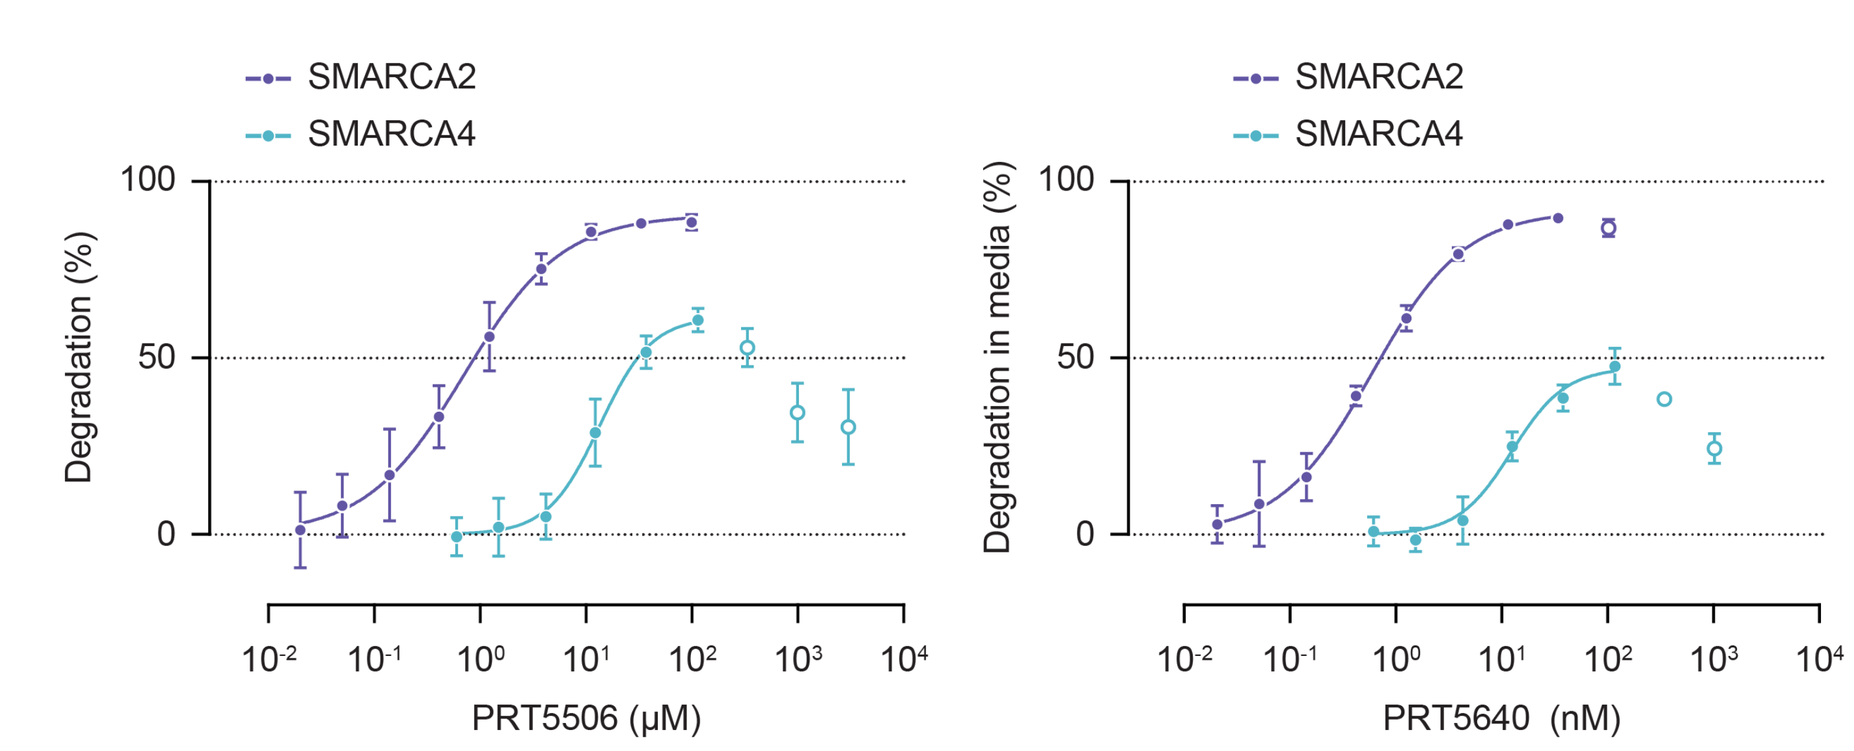
A
SMARCA2
SMARCA4
Degradation (%)
PRT5506 (μM)
Degradation in media (%)
PRT5640 (nM)

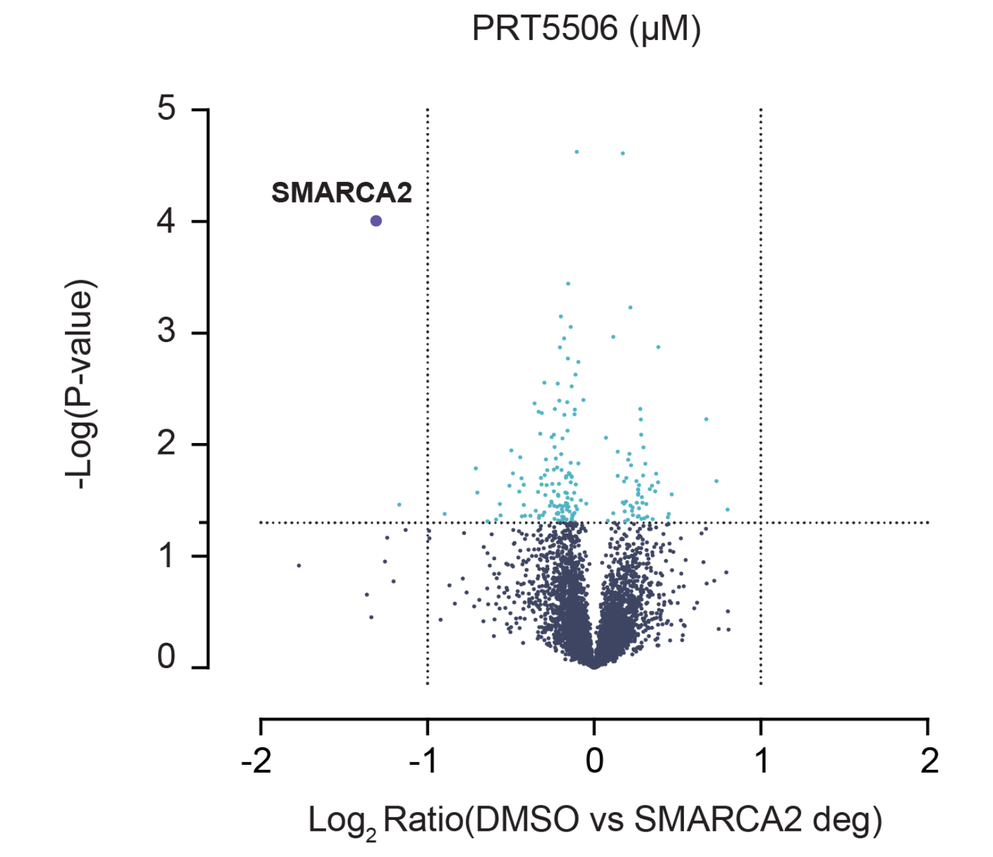
B
SMARCA2
-Log(P-value)
Log2 Ratio(DMSO vs SMARCA2 deg)

A) Our SMARCA2 oral degrader leads PRT5506 and PRT5640 demonstrate potent and selective degradation of SMARCA2 over SMARCA4, analyzed in a Hela HiBiT cell-based assay. **B**) Global proteomics reveal excellent selectivity for SMARCA2 degradation in human lung cancer cell line.

Table 1. Profile of Oral SMARCA2 Lead Degraders

Assay	PRT5506	PRT5640
HeLa SMARCA2 HiBiT DC_{50} (nM) / D_{max} (%)	1.0/91	0.33/91
HeLa SMARCA4 HiBiT DC_{50} (nM) / D_{max} (%)	54/62	98/47
Selectivity SMARCA4/SMARCA2 (HiBiT)	54	300
Selectivity Cell Proliferation (SM4-del vs. WT)	> 1000	> 1000

Figure 3. SMARCA2 oral degraders demonstrate synthetic lethality in SMARCA4-deficient human cancer cells *in vitro*

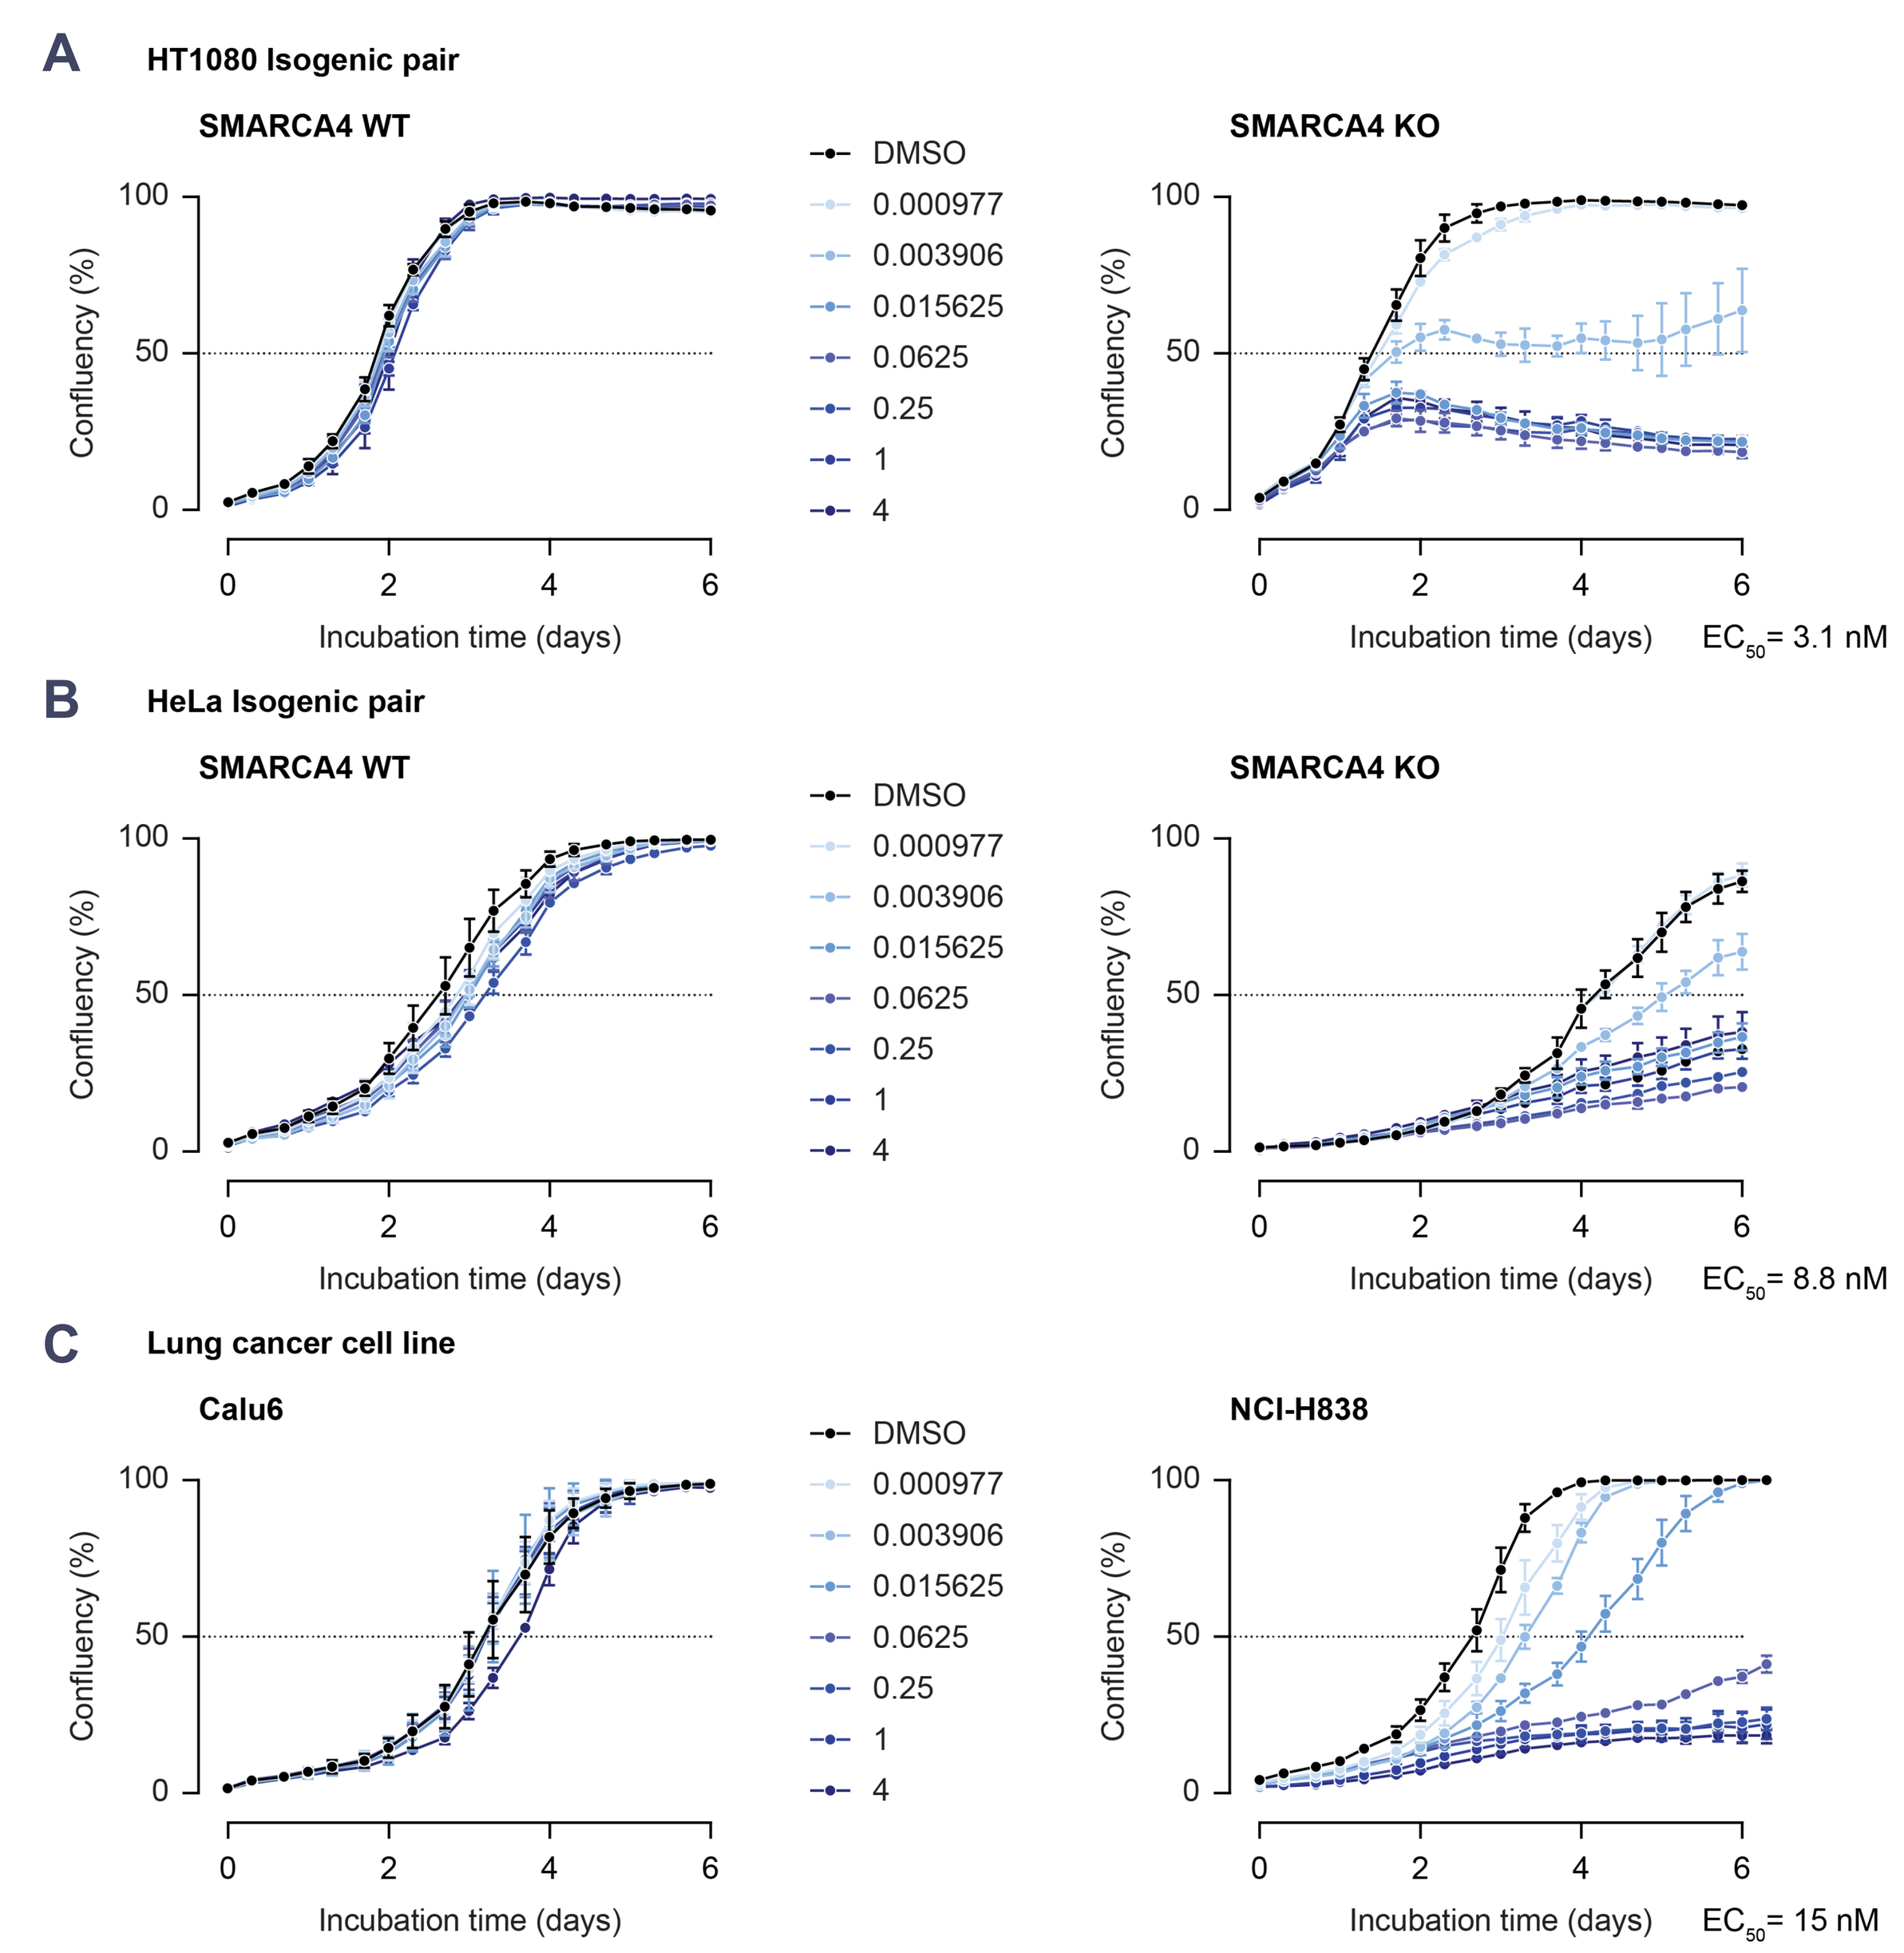
A HT1080 Isogenic pair
SMARCA4 WT
SMARCA4 KO
DMSO
0.000977
0.003906
0.015625
0.0625
0.25
1
4
Confluency (%)
Incubation time (days)
EC50= 3.1 nM
B HeLa Isogenic pair
EC50= 8.8 nM
C Lung cancer cell line
Calu6
NCI-H838
EC50= 15 nM

A) HT1080 SMARCA4 WT/KO isogenic, **B)** Hela SMARCA4 WT/KO isogenic, or **C)** Calu6 SMARCA4 WT and NCI-H838 SMARCA4-deficient human lung cancer cell lines were treated with PRT5640. The cell viability was monitored using the IncuCyte S3 system. EC_{50} values were calculated at the time of DMSO control cells become >95% confluent.

Figure 4. SMARCA2 degradation kinetics and *in vivo* PK parameters predict *in vivo* SMARCA2 and SMARCA4 protein pharmacodynamics

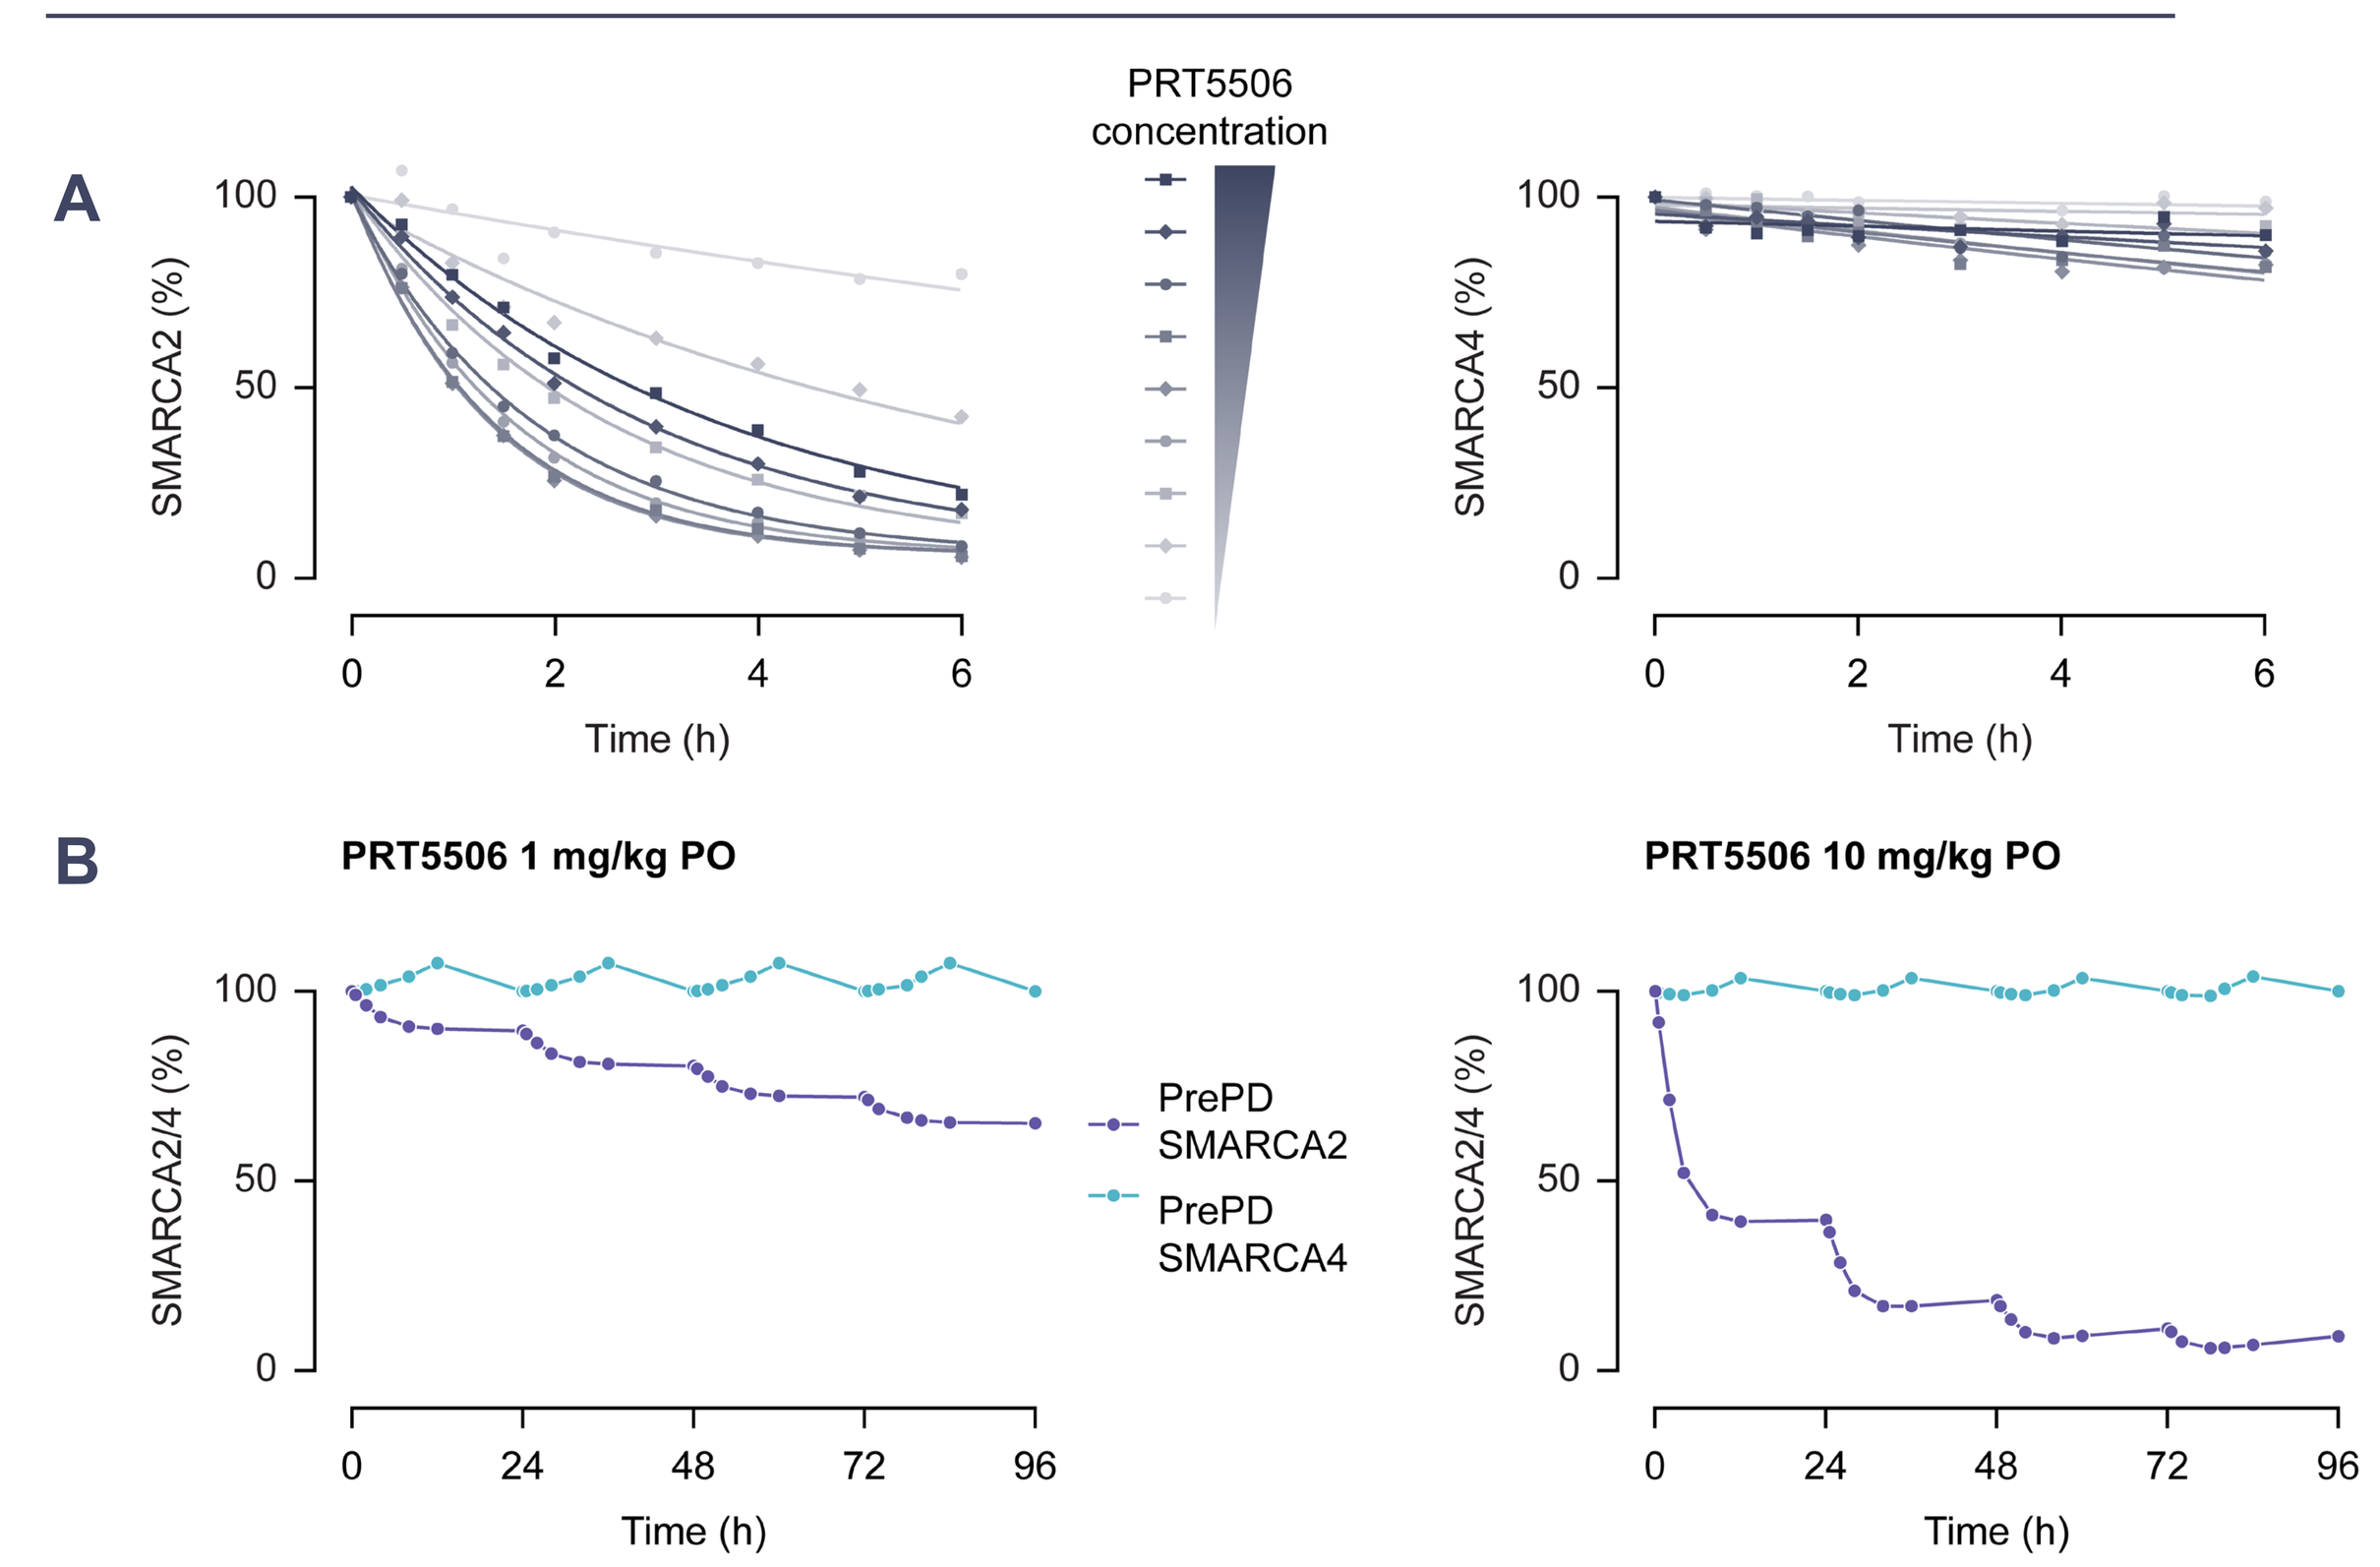
A
PRT5506 concentration
SMARCA2 (%)
SMARCA4 (%)
Time (h)
B
PRT5506 1 mg/kg PO
PRT5506 10 mg/kg PO
SMARCA2/4 (%)
PrePD SMARCA2
PrePD SMARCA4
Time (h)

A) SMARCA2 and SMARCA4 degradation kinetics of PRT5506 was analyzed in Hela HiBiT cells. **B)** Based on the degradation kinetics, selectivity parameters and *in vivo* mouse PK data, changes of *in vivo* PD levels (SMARCA2 and SMARCA4) at 1 mg/kg or 10 mg/kg oral daily dosing were estimated.

Figure 5. Oral administration of SMARCA2 degraders demonstrate significant anti-tumor activity in SMARCA4-deficient cancer xenograft models

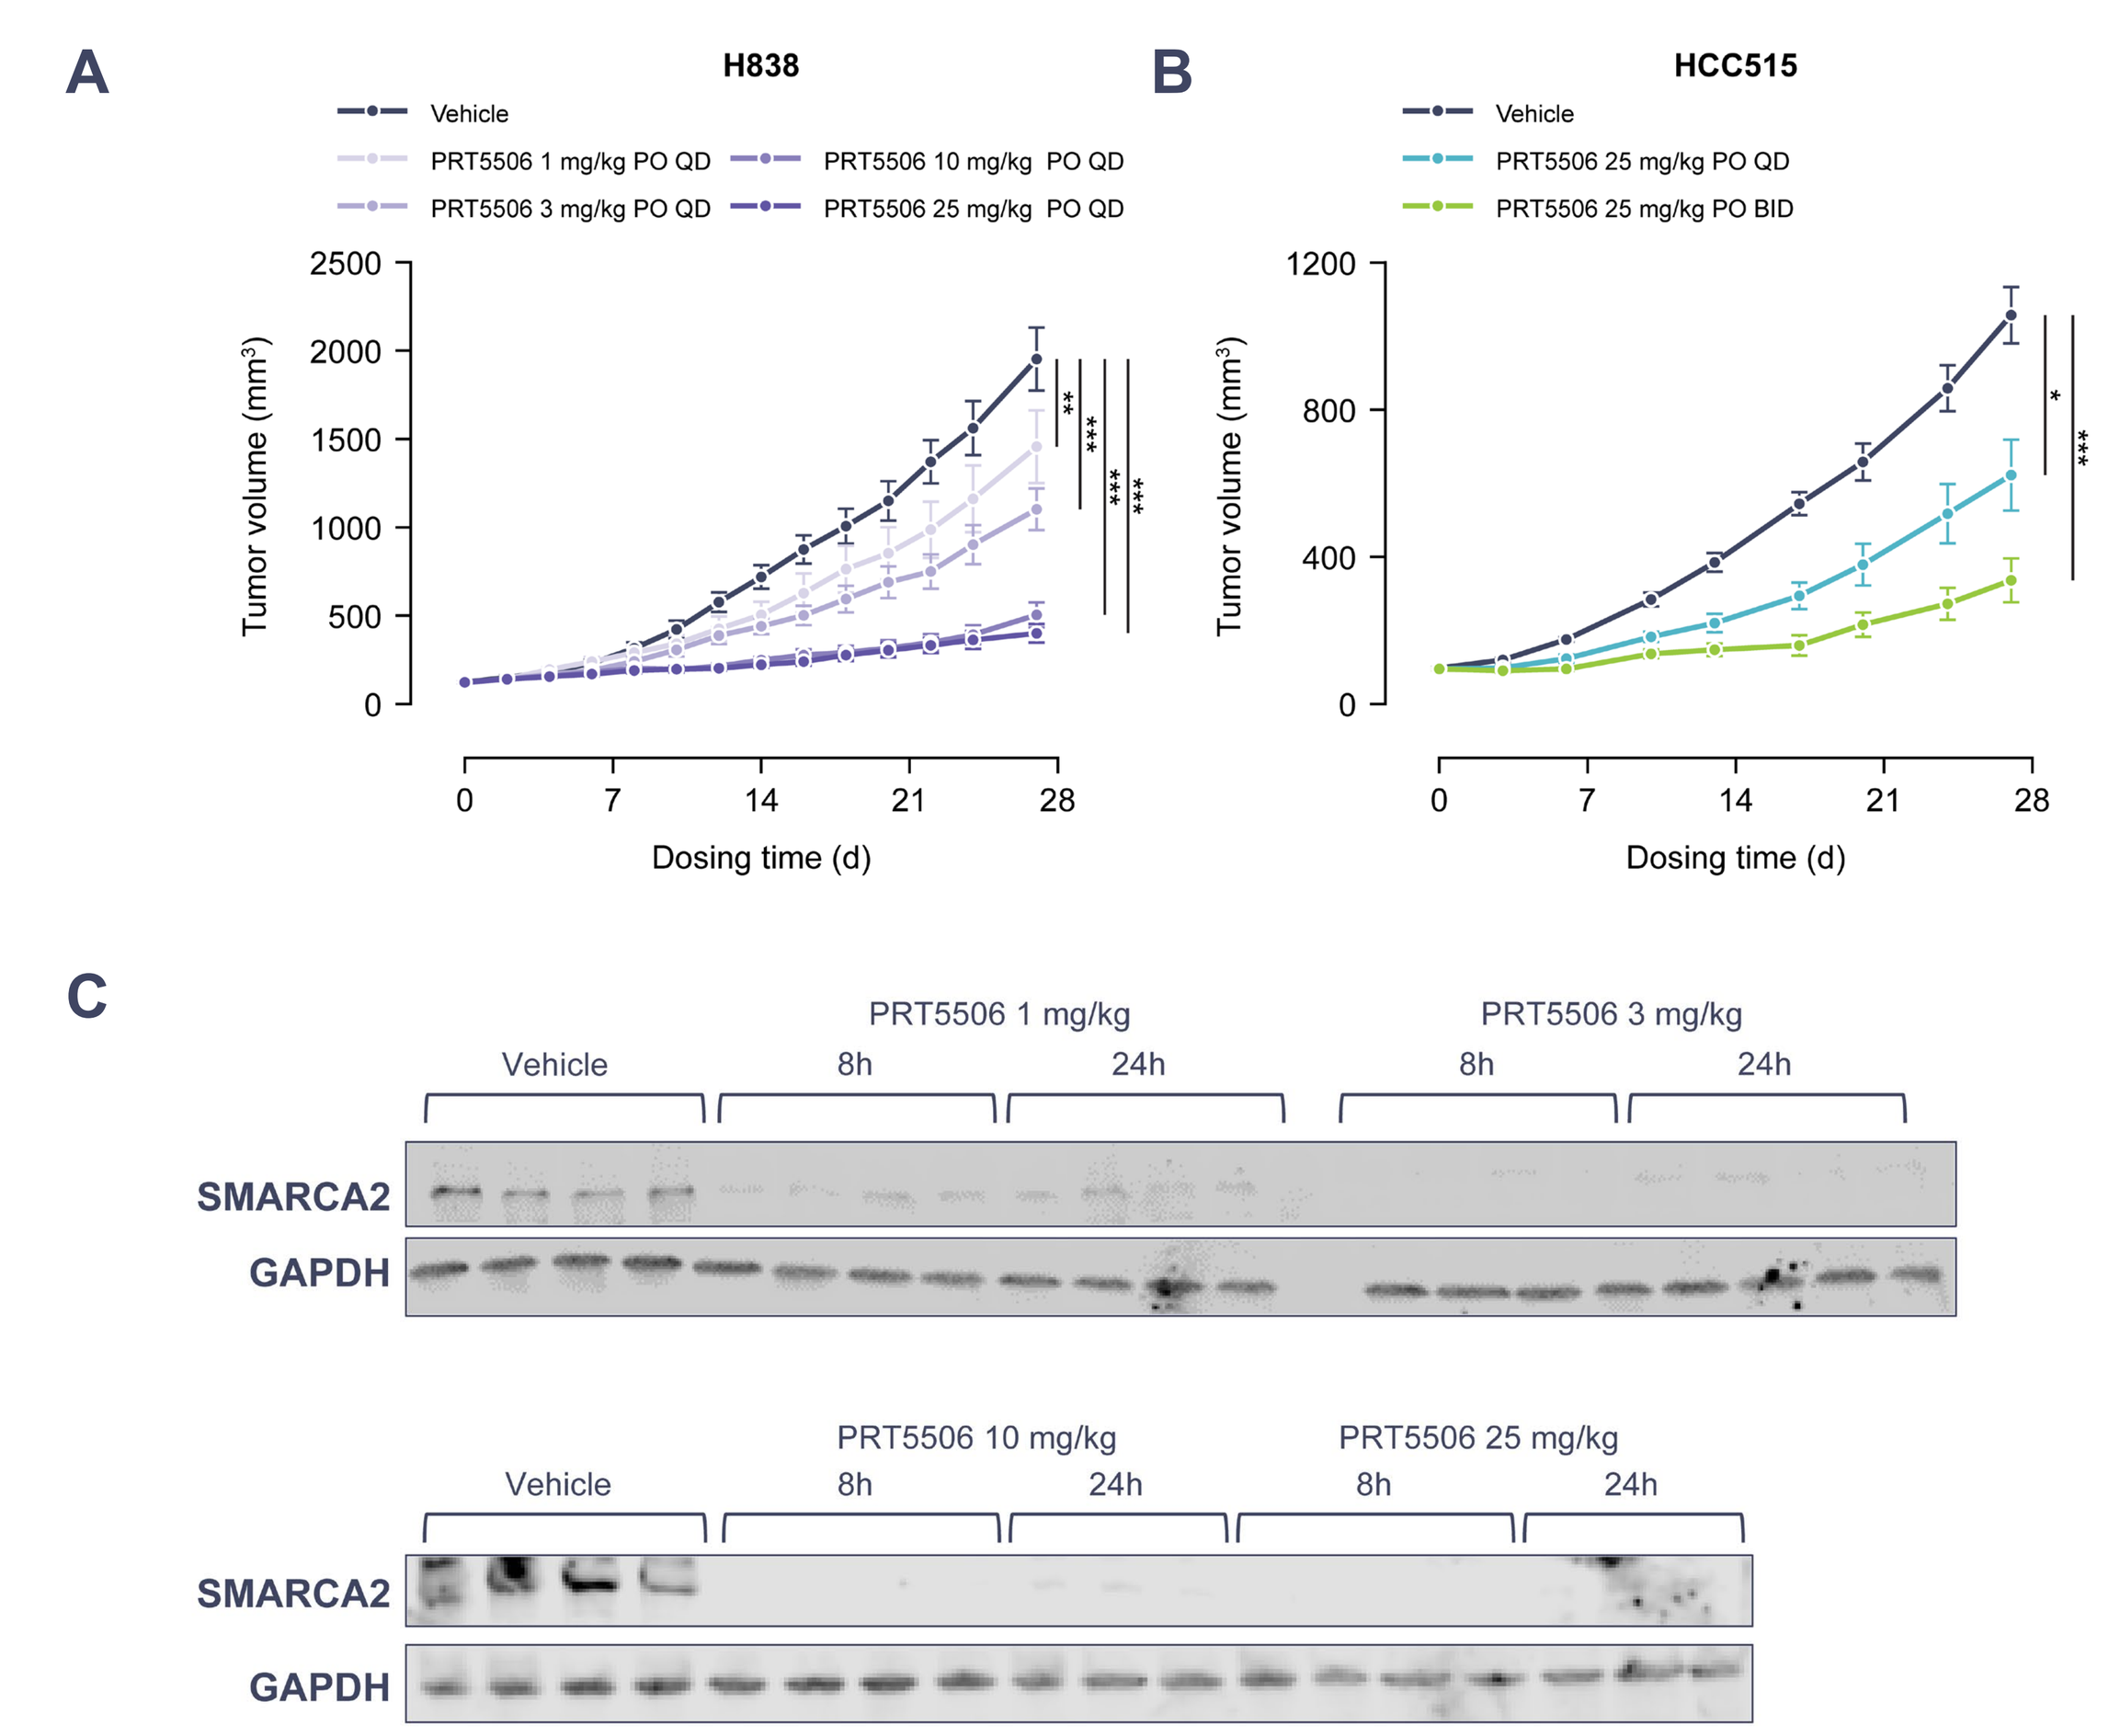
A
H838
Vehicle
PRT5506 1 mg/kg PO QD
PRT5506 3 mg/kg PO QD
PRT5506 10 mg/kg PO QD
PRT5506 25 mg/kg PO QD
Tumor volume (mm3)
Dosing time (d)
B
HCC515
PRT5506 25 mg/kg PO QD
PRT5506 25 mg/kg PO BID
C
Vehicle
PRT5506 1 mg/kg
PRT5506 3 mg/kg
PRT5506 10 mg/kg
PRT5506 25 mg/kg
8h
24h
SMARCA2
GAPDH

A,B) Oral administration of PRT5506 demonstrates significant tumor growth inhibition in SMARCA4-deficient NCI-H838 and HCC515 CDX tumors in a dose-dependent manner. *P<0.05 **P<0.01 ***P<0.001, versus vehicle (two-tailed Mann-Whitney test) **C)** Tumor PD (SMARCA2 protein) analyzed in samples from efficacy studies by Western blot were shown. Robust reduction of SMARCA2 protein in tumor tissues were detected in samples treated at efficacious doses. PO: per oral. QD: daily dosing.

Conclusions

- We have identified the first orally bioavailable bivalent molecules that selectively degrade SMARCA2 over SMARCA4 by >100 fold
- Our SMARCA2 oral degrader leads show excellent potency and selectivity for SMARCA2 degradation tested *in vitro* and *in vivo*
- Our SMARCA2 oral degrader leads show strong anti-proliferation activity in SMARCA4 KO/deficient cells, while sparing SMARCA4 WT cells, demonstrating synthetic lethality in SMARAC4-deficiency cancers
- *In vivo* SMARCA2 protein levels can be predicted by *in vitro* degradation kinetics and *in vivo* PK parameters
- Oral administration of our SMARCA2 degrader leads demonstrate robust anti-tumor activity in SMARCA4-deficient lung cancer models *in vivo*
- SMARCA2 protein levels were >90% reduced in the treated tumor at efficacious doses

References

1. Fernando, T.M. et al. Functional characterization of SMARCA4 variants identified by targeted exome-sequencing of 131,668 cancer patients. Nat Commun 11, 5551 (2020). https://doi.org/10.1038/s41467-020-19402-8
2. Ito K et al. Abstract 1139: Potent SMARCA2 targeted degraders induce genetic synthetic lethality in SMARCA4 deleted cancer. Cancer Res 1 July 2021; 81 (13_Supplement): 1139.
3. Hulse M et al. Abstract 3263: Preclinical characterization of PRT3789, a potent and selective SMARCA2 targeted degrader. Can Res 2022, 82 (12_Supplement): 3263
4. Tsherniak A., et al. Defining a Cancer Dependency Map. Cell, 2017 Jul 27; 170(3)

Acknowledgments

This study was funded by Prelude Therapeutics, Inc. Data provided by CrownBio Sciences and HD Biosciences (Wuxi). Editorial support was provided by Arne Fabritius, Endosymbiont GmbH and was funded by Prelude Therapeutics, Inc.

Disclosures

Authors are or were employees of Prelude Therapeutics, Inc at the time of research, and may own equity in the Company.