A Phase 1, Open-Label, Multicenter, Dose Escalation Study of PRT2527, a CDK9 Inhibitor in Adult Patients With Advanced Solid Tumors

Jason T. Henry,[1] Douglas Orr,[2] Atrayee Basu-Mallick,[3] Alex Spira,[4] Christine Lihou,[5] William Sun,[5] Neelesh Sharma,[5] Peggy Scherle,[5] Rachel Chiaverelli,[5] Manish R. Patel[6]

[1]Sarah Cannon Research Institute at HealthONE, Denver, CO; [2]Mary Crowley Cancer Research, Dallas, TX; [3]Thomas Jefferson University Hospital, Philadelphia, PA; [4]Virginia Cancer Specialists Research Institute/NEXT Oncology Virginia, Fairfax, VA; [5]Prelude Therapeutics Incorporated, Wilmington, DE; [6]Florida Cancer Specialists/Sarah Cannon Research Institute, Sarasota, FL
Contact: Jason.Henry2@sarahcannon.com

Prelude THERAPEUTICS

CT173

Background

- Cyclin-dependent kinase 9 (*CDK9*) is a master regulator of transcription that controls paused RNA polymerase II (RNAP2) release through phosphorylation of its carboxy-terminal domain, resulting in transcription elongation (Figure 1).[1,2]
- Selective CDK9 inhibition may be a promising approach to treat transcription-addicted cancers that are dependent on oncogenic drivers with a short half-life, such as the oncogenes myelocytomatosis (*MYC*), myeloblastosis (*MYB*), and myeloid leukemia cell differentiation protein (*MCL-1*).[2]
- PRT2527 is a potent and selective inhibitor of CDK9 with a biochemical half-maximal inhibitory concentration (IC_{50}) of 0.98 nM at 1 mM adenosine triphosphate that inhibits the enzymatic activity of human CDK9/CyclinT1 complex.[1] PRT2527 is highly selective among CDK family members and across 177 kinases.[1]
- Intermittent intravenous (IV) administration of PRT2527 demonstrated strong efficacy in preclinical models of solid tumors and hematological malignancies as monotherapy and in combination with other anti-cancer therapies.[1]

Figure 1. CDK9 Mechanism of Action

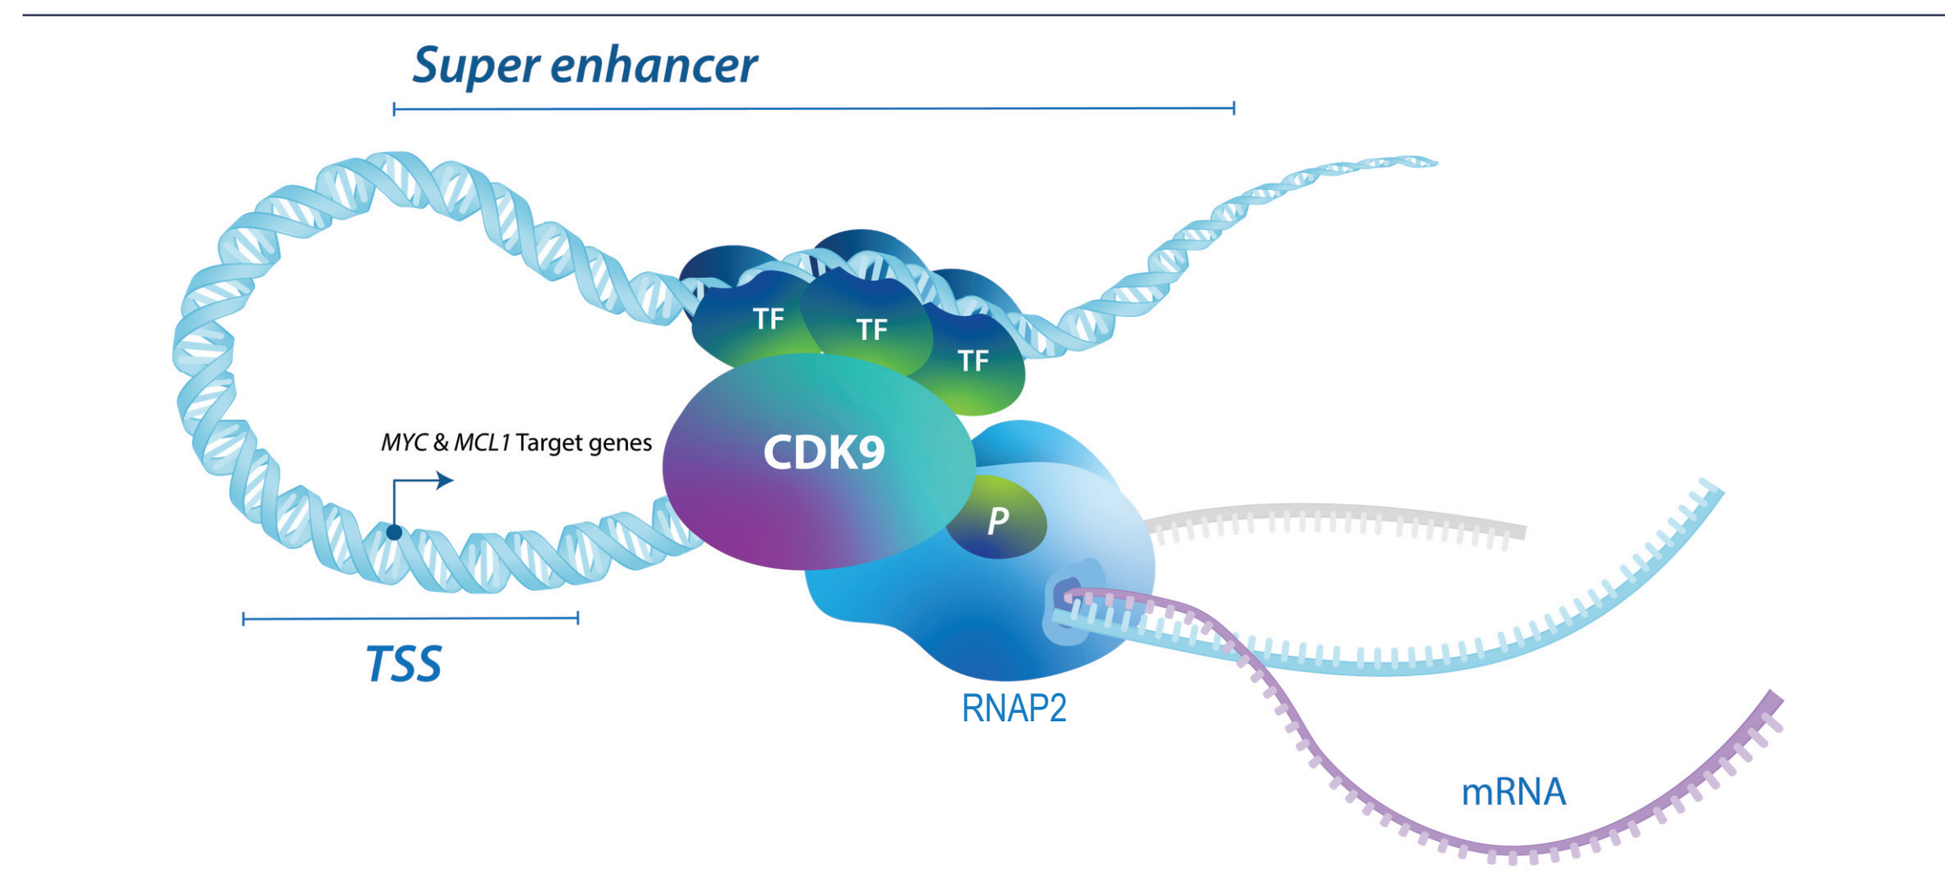
Super enhancer
TF
TF
TF
CDK9
MYC & MCL1 Target genes
P
TSS
RNAP2
mRNA

Figure reproduced with permission from Zhang Y, et al.[1]
mRNA, messenger ribonucleic acid; P, phosphate; TF, transcription factor; TSS, transcription start site.

Objectives

- To identify the dose(s) to further evaluate in a dose-confirmation cohort and evaluate the pharmacokinetic/pharmacodynamic profile, safety, and preliminary efficacy of PRT2527 in patients with advanced solid tumors.

Key Findings

- In this phase 1, open-label, dose-escalation study, the CDK9 inhibitor PRT2527 demonstrated acceptable safety and tolerability in patients with advanced solid tumors.

Methods

- This is an open-label, multicenter, phase 1 dose-escalation study of PRT2527 in patients with select advanced solid tumors (NCT05159518; Figure 2).

Figure 2. Study Design

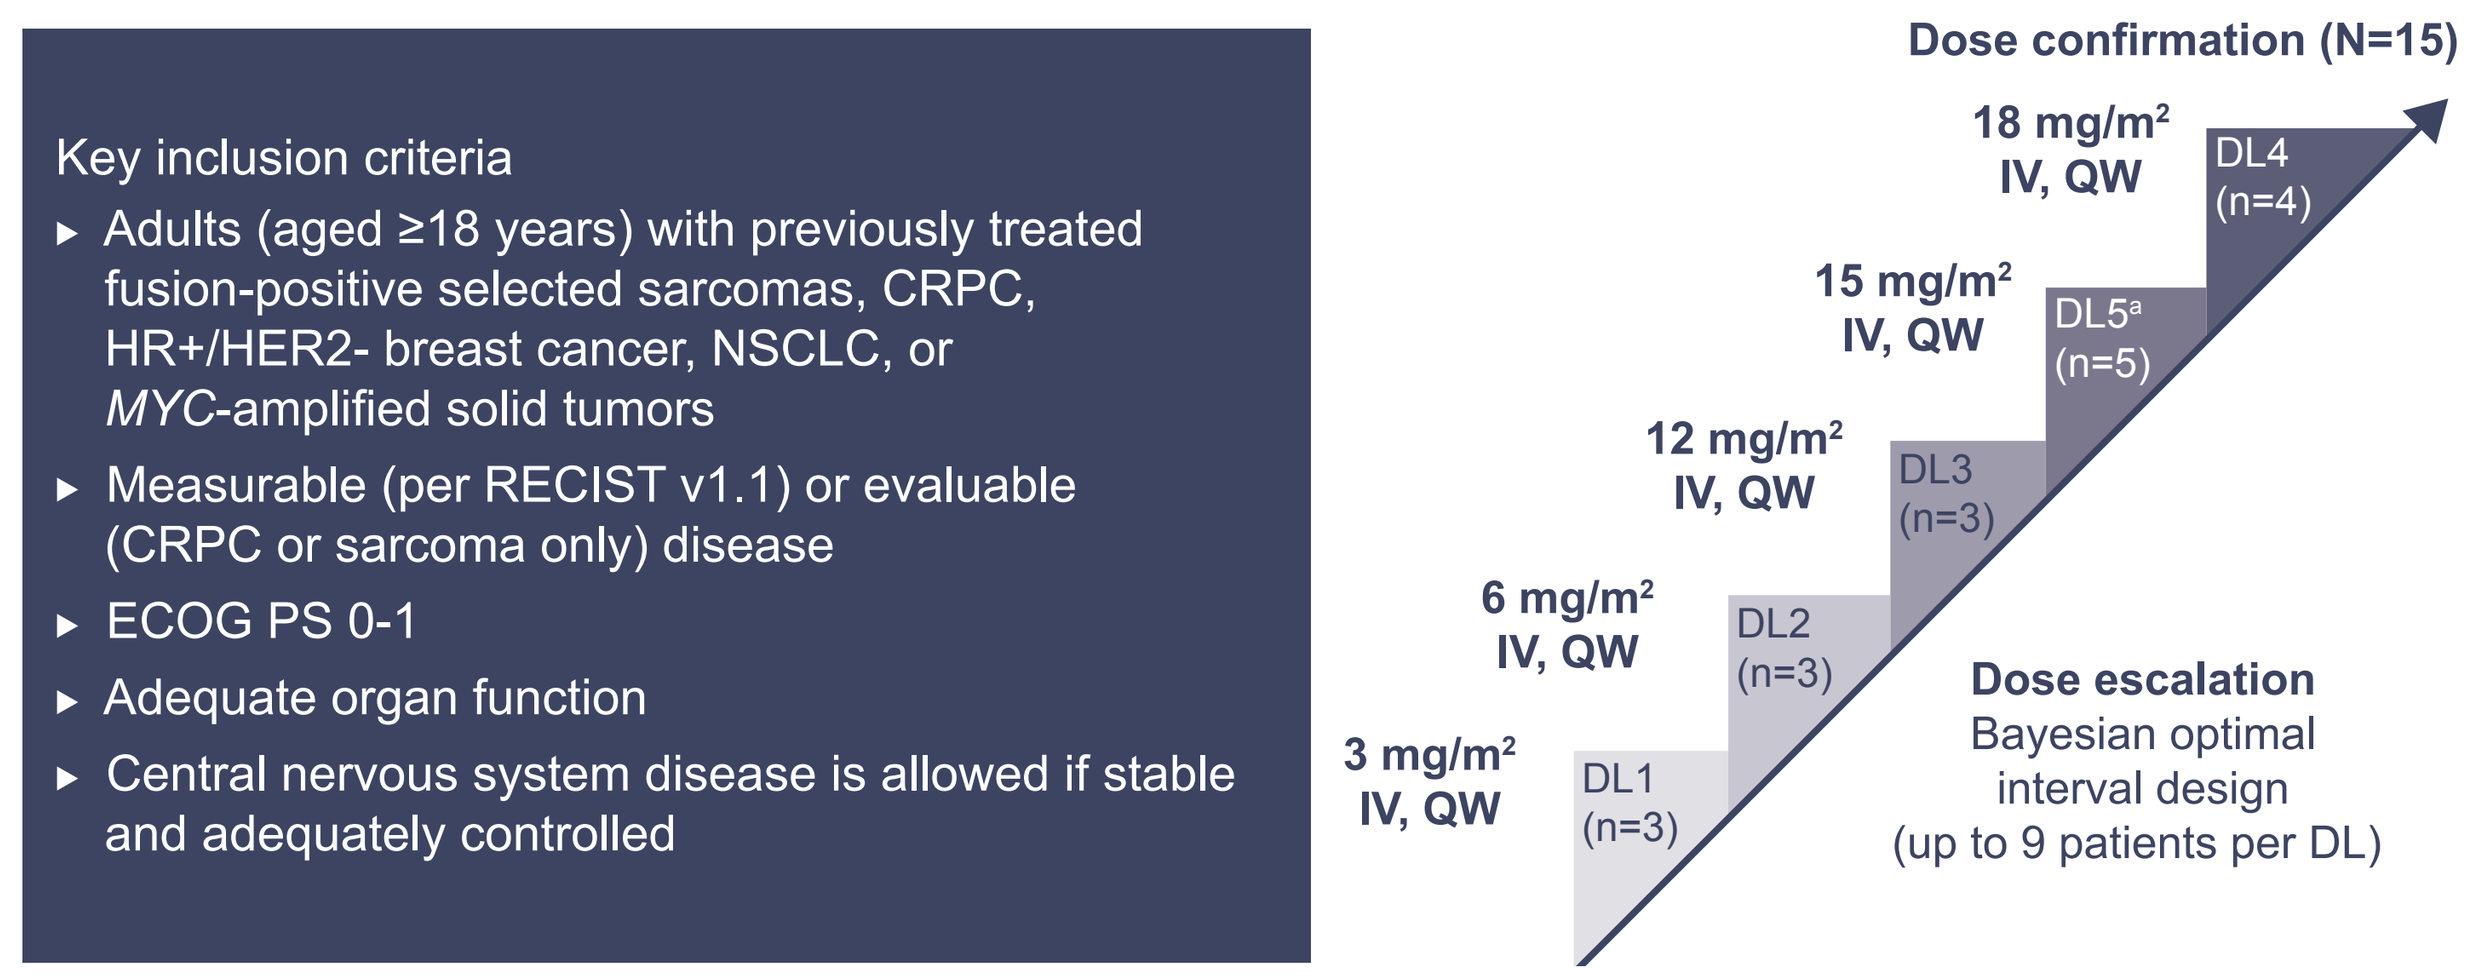
Key inclusion criteria
Adults (aged ≥18 years) with previously treated fusion-positive selected sarcomas, CRPC, HR+/HER2- breast cancer, NSCLC, or MYC-amplified solid tumors
Measurable (per RECIST v1.1) or evaluable (CRPC or sarcoma only) disease
ECOG PS 0-1
Adequate organ function
Central nervous system disease is allowed if stable and adequately controlled
Dose confirmation (N=15)
18 mg/m² IV, QW
DL4 (n=4)
15 mg/m² IV, QW
DL5ᵃ (n=5)
12 mg/m² IV, QW
DL3 (n=3)
6 mg/m² IV, QW
DL2 (n=3)
3 mg/m² IV, QW
DL1 (n=3)
Dose escalation
Bayesian optimal interval design
(up to 9 patients per DL)

[a]DL5 (15 mg/m² QW) was evaluated as an intermediate DL following safety review of DL4 (18 mg/m² QW). PRT2527 was administered intravenously once weekly in a 3-week cycle until disease progression or unacceptable toxicity. CRPC, castrate-resistant prostate cancer; DL, dose level; ECOG PS, Eastern Cooperative Oncology Group performance status; HR+/HER2-, hormone receptor positive, human epidermal growth factor receptor 2 negative; NSCLC, non-small cell lung cancer; QW, once weekly; RECIST, Response Evaluation Criteria in Solid Tumors.

Study Endpoints

- The primary endpoint was to identify dose-limiting toxicities (DLTs), maximum tolerated dose, and the dose(s) of PRT2527 to further evaluate in a dose-confirmation cohort.
- Secondary endpoints included evaluation of the pharmacokinetic profile, safety, and efficacy of PRT2527.
- Exploratory endpoints included the pharmacodynamic assessment of the expression of CDK9 transcriptional targets (*MYC*, *MCL-1*) in peripheral blood mononuclear cells (PBMCs) that may be associated with response to CDK9 inhibition. Phosphorylation of RNAP2 (p-RNAP2) was assessed by capillary electrophoresis and monitoring changes in absolute monocyte and neutrophil counts using clinical complete blood count.

Statistical Analysis

- The safety and efficacy analysis population included all patients who received at least 1 dose of PRT2527.
- All data are reported as descriptive analyses.

Results

Patients

- Patient baseline characteristics are presented in Table 1 and patient disposition is presented in Figure 3.
- At the time of data cut-off (February 13, 2023; median treatment duration of 4.1 [range, 1.0-40.1] weeks), 83.3% of patients had discontinued PRT2527. The primary reasons for discontinuation were progressive disease (PD; n=10, 66.7%), adverse events (AEs; n=3, 20.0%), physician's decision (n=1, 6.7%), and withdrawal by patient (n=1, 6.7%).

Table 1. Patient Baseline Characteristics

Characteristic	PRT2527 3 mg/m² QW (n=3)	PRT2527 6 mg/m² QW (n=3)	PRT2527 12 mg/m² QW (n=3)	PRT2527 15 mg/m² QW (n=5)	PRT2527 18 mg/m² QW (n=4)	Total (N=18)
Age, median (range), years	64.0 (59.0-64.0)	64.0 (57.0-70.0)	72.0 (71.0-73.0)	31.0 (23.0-67.0)	70.5 (37.0-86.0)	64.0 (23.0-86.0)
Female, n (%)	1 (33.3)	3 (100.0)	0	3 (60.0)	1 (25.0)	8 (44.4)
ECOG PS at baseline, n (%)						
0	1 (33.3)	2 (66.7)	0	3 (60.0)	0	6 (33.3)
1	2 (66.7)	1 (33.3)	3 (100.0)	2 (40.0)	4 (100.0)	12 (66.7)
Race, n (%)						
White or Caucasian	2 (66.7)	3 (100.0)	3 (100.0)	4 (80.0)	3 (75.0)	15 (83.3)
Black or African American	0	0	0	0	1 (25.0)	1 (5.6)
Asian	1 (33.3)	0	0	0	0	1 (5.6)
Other	0	0	0	1 (20.0)	0	1 (5.6)
Prior lines of systemic therapy, median (range)	5.0 (4.0-8.0)	4.0 (1.0-5.0)	7.0 (3.0-8.0)	4.0 (2.0-5.0)	5.5 (1.0-8.0)	4.5 (1.0-8.0)
Primary cancer diagnosis, n (%)						
Breast	1 (33.3)	1 (33.3)	0	1 (20.0)	1 (25.0)	4 (22.2)
Prostate	2 (66.7)	0	3 (100.0)	0	2 (50.0)	7 (38.9)
Sarcoma	0	2 (66.7)	0	4 (80.0)	1 (25.0)	7 (38.9)

Figure 3. Patient Disposition

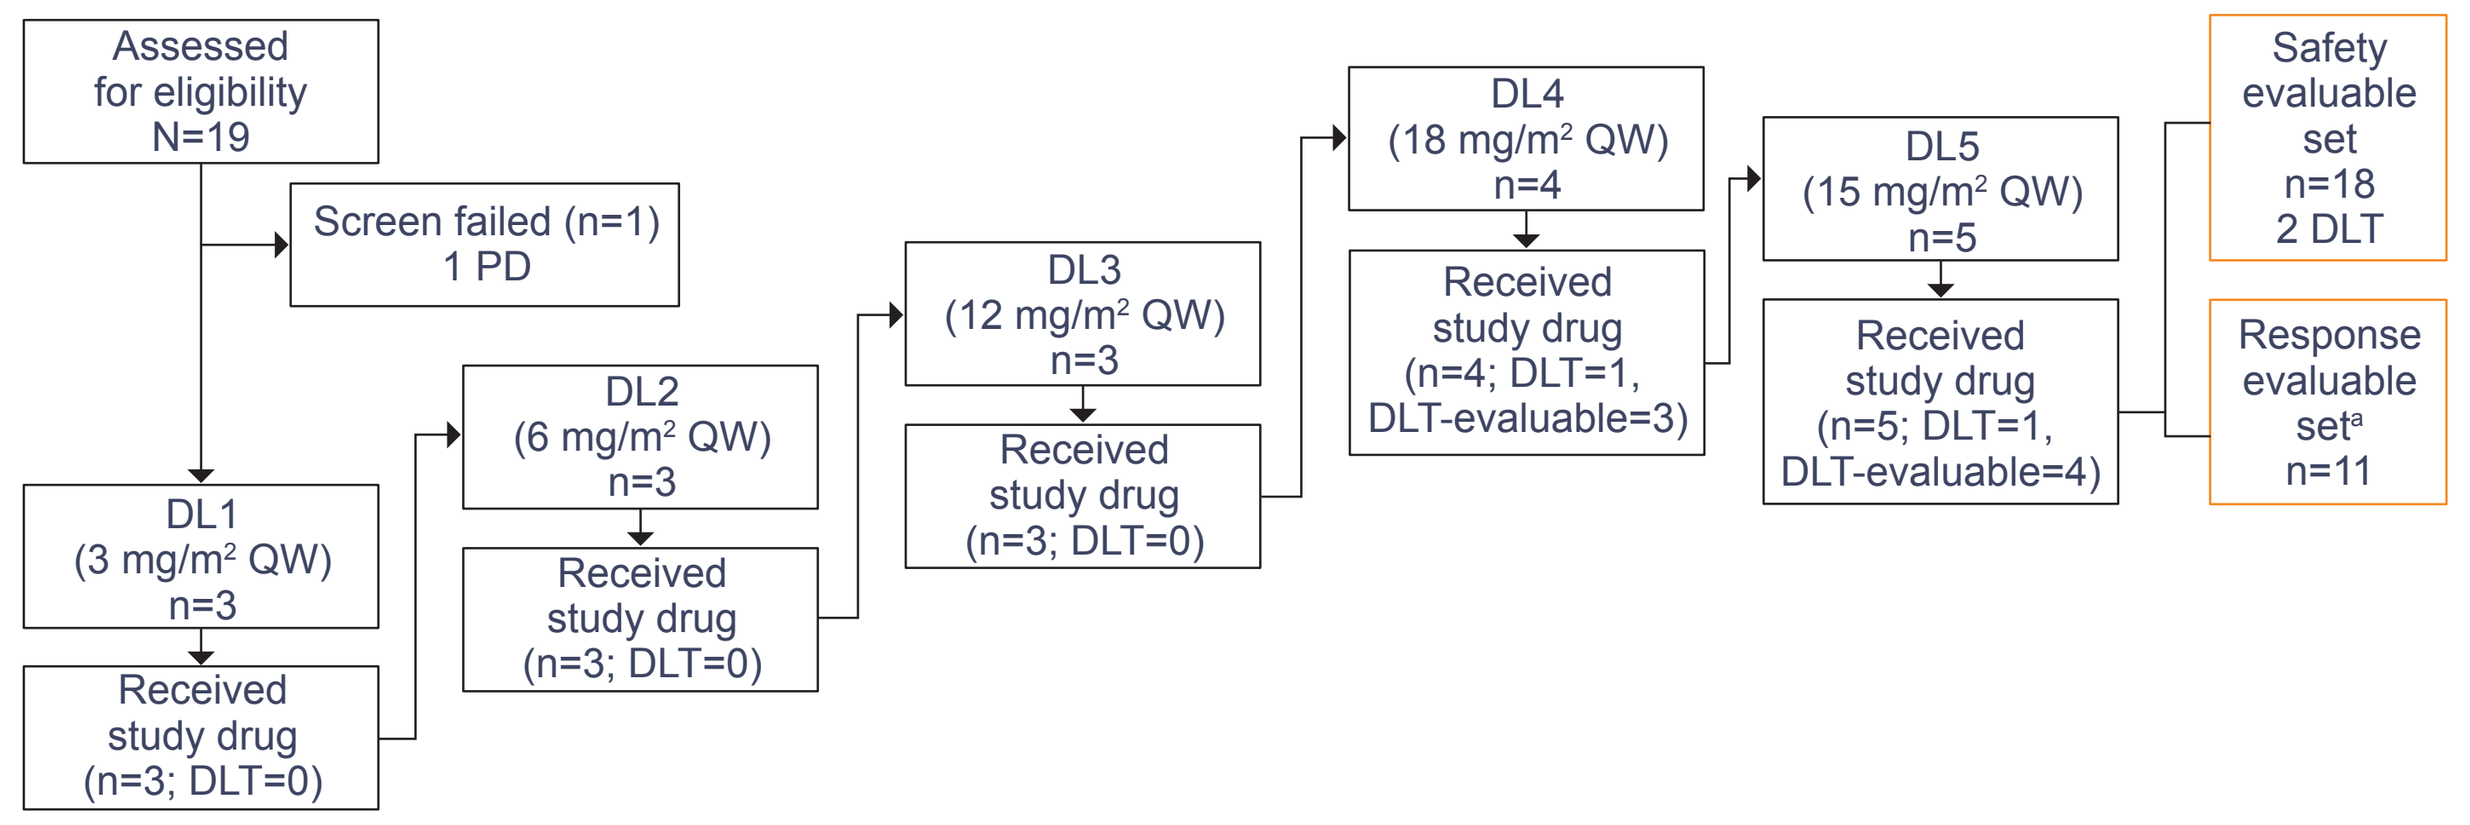
Assessed for eligibility N=19
Screen failed (n=1) 1 PD
DL1 (3 mg/m² QW) n=3
Received study drug (n=3; DLT=0)
DL2 (6 mg/m² QW) n=3
Received study drug (n=3; DLT=0)
DL3 (12 mg/m² QW) n=3
Received study drug (n=3; DLT=0)
DL4 (18 mg/m² QW) n=4
Received study drug (n=4; DLT=1, DLT-evaluable=3)
DL5 (15 mg/m² QW) n=5
Received study drug (n=5; DLT=1, DLT-evaluable=4)
Safety evaluable set n=18 2 DLT
Response evaluable setᵃ n=11

[a]Patients with at least 1 overall response assessment by Investigator(s) per RECIST v1.1.

Safety

- The most common treatment-related AEs (TRAEs) observed in the study were typically grade 1/2 in severity; grade 3/4 events were infrequent and independent of dose, except for neutropenia (Table 2).
 - The most common treatment-emergent AEs (TEAEs) were nausea (38.9%), vomiting (33.3%), fatigue (27.8%), and diarrhea (22.2%; Table 3).
 - Two patients (11.1%) had grade 4 neutropenia as a DLT; 1 patient each in the 15 mg/m² QW and 18 mg/m² QW dose cohorts.
 - No grade 3/4 gastrointestinal events or hepatotoxicity was observed.
- No treatment-related deaths or treatment-related serious AEs were observed. One grade 5 AE unrelated to study treatment was observed (sepsis as a result of aspiration pneumonia, which occurred in the context of dysphagia but without neutropenia).

Table 2. Safety Summary

Events, n (%)	PRT2527 3 mg/m² QW (n=3)	PRT2527 6 mg/m² QW (n=3)	PRT2527 12 mg/m² QW (n=3)	PRT2527 15 mg/m² QW (n=5)	PRT2527 18 mg/m² QW (n=4)	Total (N=18)
TEAEs						
Any grade	3 (100.0)	3 (100.0)	3 (100.0)	5 (100.0)	4 (100.0)	18 (100.0)
Grade ≥3	1 (33.3)	0	1 (33.3)	2 (40.0)	3 (75.0)	7 (38.9)
Serious	0	1 (33.3)	0	1 (20.0)	2 (50.0)	4 (22.2)
Grade 5 (fatal)	0	0	0	0	1 (25.0)	1 (5.6)
Leading to study drug dose reduction	0	0	0	1 (20.0)	2 (50.0)	3 (16.7)
Leading to study drug interruption	0	0	1 (33.3)	1 (20.0)	2 (50.0)	4 (22.2)
Leading to study drug withdrawal	0	0	0	1 (20.0)	2 (50.0)	3 (16.7)
Leading to a DLT	0	0	0	1 (20.0)	1 (25.0)	2 (11.1)
Treatment-related TEAEs						
Any grade	2 (66.7)	2 (66.7)	3 (100.0)	4 (80.0)	3 (75.0)	14 (77.8)
Grade 3-4	0	0	0	1 (20.0)	1 (25.0)	2 (11.1)
Serious	0	0	0	0	0	0
Grade 5 (fatal)	0	0	0	0	0	0
Leading to study drug dose reduction	0	0	0	1 (20.0)	2 (50.0)	3 (16.7)
Leading to study drug interruption	0	0	1 (33.3)	1 (20.0)	1 (25.0)	3 (16.7)
Leading to study drug withdrawal	0	0	0	0	0	0
Leading to a DLT	0	0	0	1 (20.0)	1 (25.0)	2 (11.1)

Table 3. Most Common TEAEs (in ≥10% of the Total Population)

Events, n (%)	PRT2527 3 mg/m² QW (n=3)	PRT2527 6 mg/m² QW (n=3)	PRT2527 12 mg/m² QW (n=3)	PRT2527 15 mg/m² QW (n=5)	PRT2527 18 mg/m² QW (n=4)	Total (N=18)		
	Any grade	Any grade	Any grade	Any grade	Any grade	Grade 1-2	Grade 3-4	Any grade
Any TEAE	3 (100.0)	3 (100.0)	3 (100.0)	5 (100.0)	4 (100.0)	11 (61.1)	7 (38.9)	18 (100.0)
Nausea	1 (33.3)	3 (100.0)	0	2 (40.0)	1 (25.0)	7 (38.9)	0	7 (38.9)
Vomiting	0	2 (66.7)	1 (33.3)	2 (40.0)	1 (25.0)	6 (33.3)	0	6 (33.3)
Fatigue	0	1 (33.3)	1 (33.3)	1 (20.0)	2 (50.0)	5 (27.8)	0	5 (27.8)
Diarrhea	2 (66.7)	1 (33.3)	0	1 (20.0)	0	4 (22.2)	0	4 (22.2)
Decreased appetite	1 (33.3)	0	2 (66.7)	0	0	3 (16.7)	0	3 (16.7)
Neutropenia	0	0	0	1 (20.0)	2 (50.0)	1 (5.6)	2 (11.1)	3 (16.7)
Lymphopenia	0	0	0	2 (40.0)	1 (25.0)	2 (11.1)	1 (5.6)	3 (16.7)
Leukopenia	0	0	0	2 (40.0)	1 (25.0)	2 (11.1)	1 (5.6)	3 (16.7)
Thrombocytopenia	1 (33.3)	0	0	2 (40.0)	0	2 (11.1)	1 (5.6)	3 (16.7)
Anemia	1 (33.3)	0	0	1 (20.0)	0	0	2 (11.1)	2 (11.1)
Sepsis	0	0	0	1 (20.0)	1 (25.0)	0	2 (11.1)	2 (11.1)
Urinary tract infection	0	1 (33.3)	0	0	1 (25.0)	1 (5.6)	1 (5.6)	2 (11.1)

Efficacy

- Of the 11 patients with at least 1 overall response assessment by the Investigator(s), no objective responses (per RECIST v1.1) were observed: 5 (27.8%) patients had stable disease (SD) as the best response, and 1 (5.6%) patient achieved clinical benefit response (defined as a complete response [CR], partial response [PR], or SD for a duration of more than 24 weeks; Figure 4).
- With a median study follow-up of 2.1 (range, 0.5-9.2) months, median progression-free survival was 1.3 (95% confidence interval, 1.0-2.3) months.

Figure 4. Duration of PRT2527 Treatment With Clinical Outcome

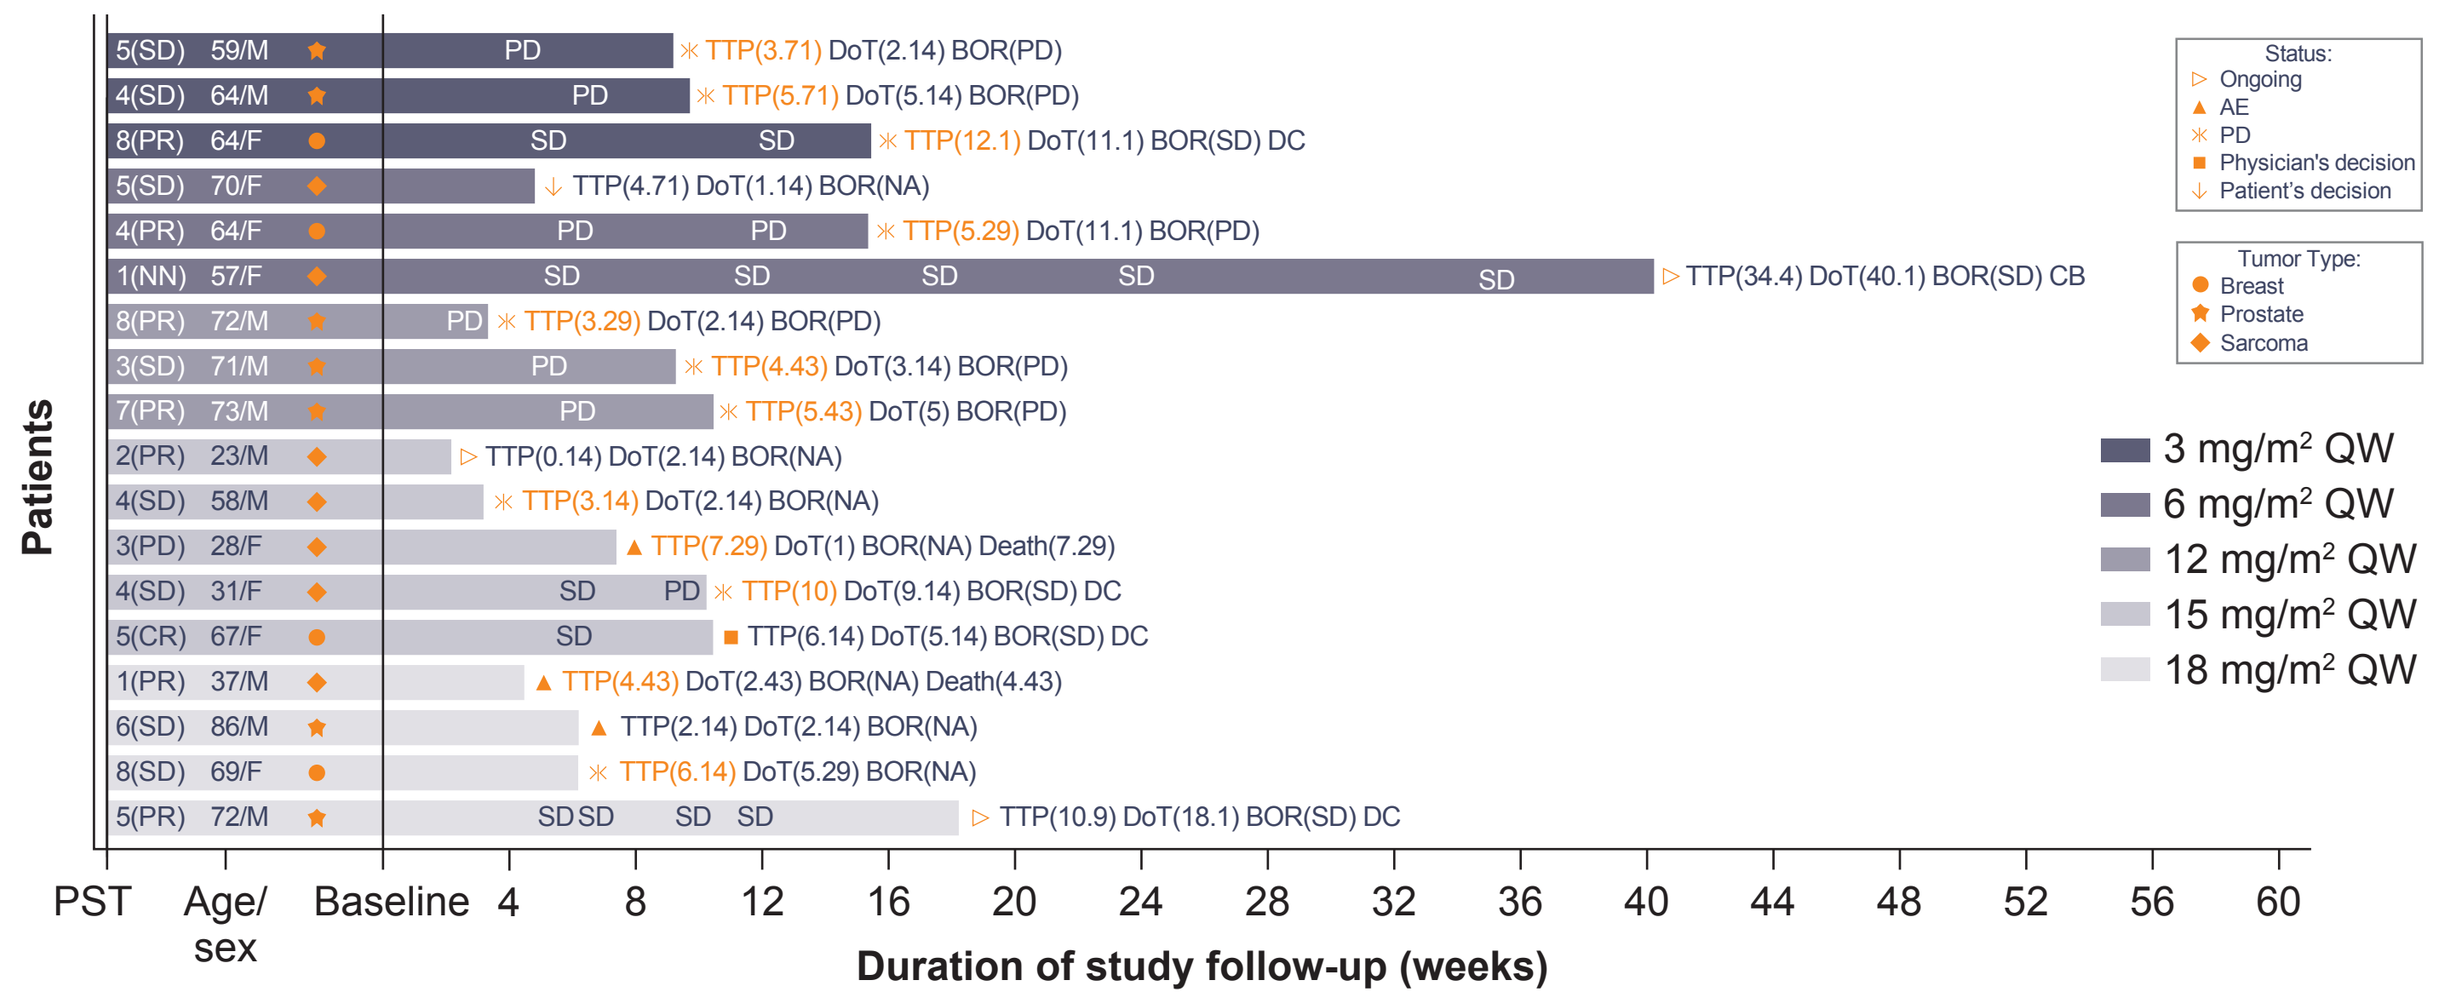
5(SD) 59/M PD TTP(3.71) DoT(2.14) BOR(PD)
4(SD) 64/M PD TTP(5.71) DoT(5.14) BOR(PD)
8(PR) 64/F SD SD TTP(12.1) DoT(11.1) BOR(SD) DC
5(SD) 70/F TTP(4.71) DoT(1.14) BOR(NA)
4(PR) 64/F PD PD TTP(5.29) DoT(11.1) BOR(PD)
1(NN) 57/F SD SD SD SD SD TTP(34.4) DoT(40.1) BOR(SD) CB
8(PR) 72/M PD TTP(3.29) DoT(2.14) BOR(PD)
3(SD) 71/M PD TTP(4.43) DoT(3.14) BOR(PD)
7(PR) 73/M PD TTP(5.43) DoT(5) BOR(PD)
2(PR) 23/M TTP(0.14) DoT(2.14) BOR(NA)
4(SD) 58/M TTP(3.14) DoT(2.14) BOR(NA)
3(PD) 28/F TTP(7.29) DoT(1) BOR(NA) Death(7.29)
4(SD) 31/F SD PD TTP(10) DoT(9.14) BOR(SD) DC
5(CR) 67/F SD TTP(6.14) DoT(5.14) BOR(SD) DC
1(PR) 37/M TTP(4.43) DoT(2.43) BOR(NA) Death(4.43)
6(SD) 86/M TTP(2.14) DoT(2.14) BOR(NA)
8(SD) 69/F TTP(6.14) DoT(5.29) BOR(NA)
5(PR) 72/M SD SD SD SD TTP(10.9) DoT(18.1) BOR(SD) DC
Status: Ongoing AE PD Physician's decision Patient's decision
Tumor Type: Breast Prostate Sarcoma
3 mg/m² QW
6 mg/m² QW
12 mg/m² QW
15 mg/m² QW
18 mg/m² QW
Patients
PST Age/sex Baseline 4 8 12 16 20 24 28 32 36 40 44 48 52 56 60
Duration of study follow-up (weeks)

The text on the bars after baseline (SD/PD) indicate overall response assessed by the Investigator per the criteria corresponding to the underlying tumor type. The text (numbers) at the end of each row indicates TTP (orange indicates real event time, and dark blue indicates censored time), DoT, and BOR assessed by the Investigator. The DoT was defined as the time interval from Day 1 to the date of end of treatment or later of the last clinical visit date and death date for those who did not reach the end of therapy. BOR, best overall response; CB, clinical benefit; DC, disease control; DoT, duration of treatment; NA, not applicable; NN, non-CR/non-PD; PST, prior systemic therapy; TTP, time to progression.

Pharmacokinetics

- PRT2527 demonstrated dose-dependent increases in peak plasma concentrations (C_{max}) and area under the curve (AUC) (Figure 5 and Table 4).
- PRT2527 did not accumulate in systemic circulation.
- The mean half-life of PRT2527 was approximately 3.8 to 4.7 hours.

Figure 5. Change in Plasma PRT2527 Concentration During Week 1 (A) and Week 3 (B) Post Administration

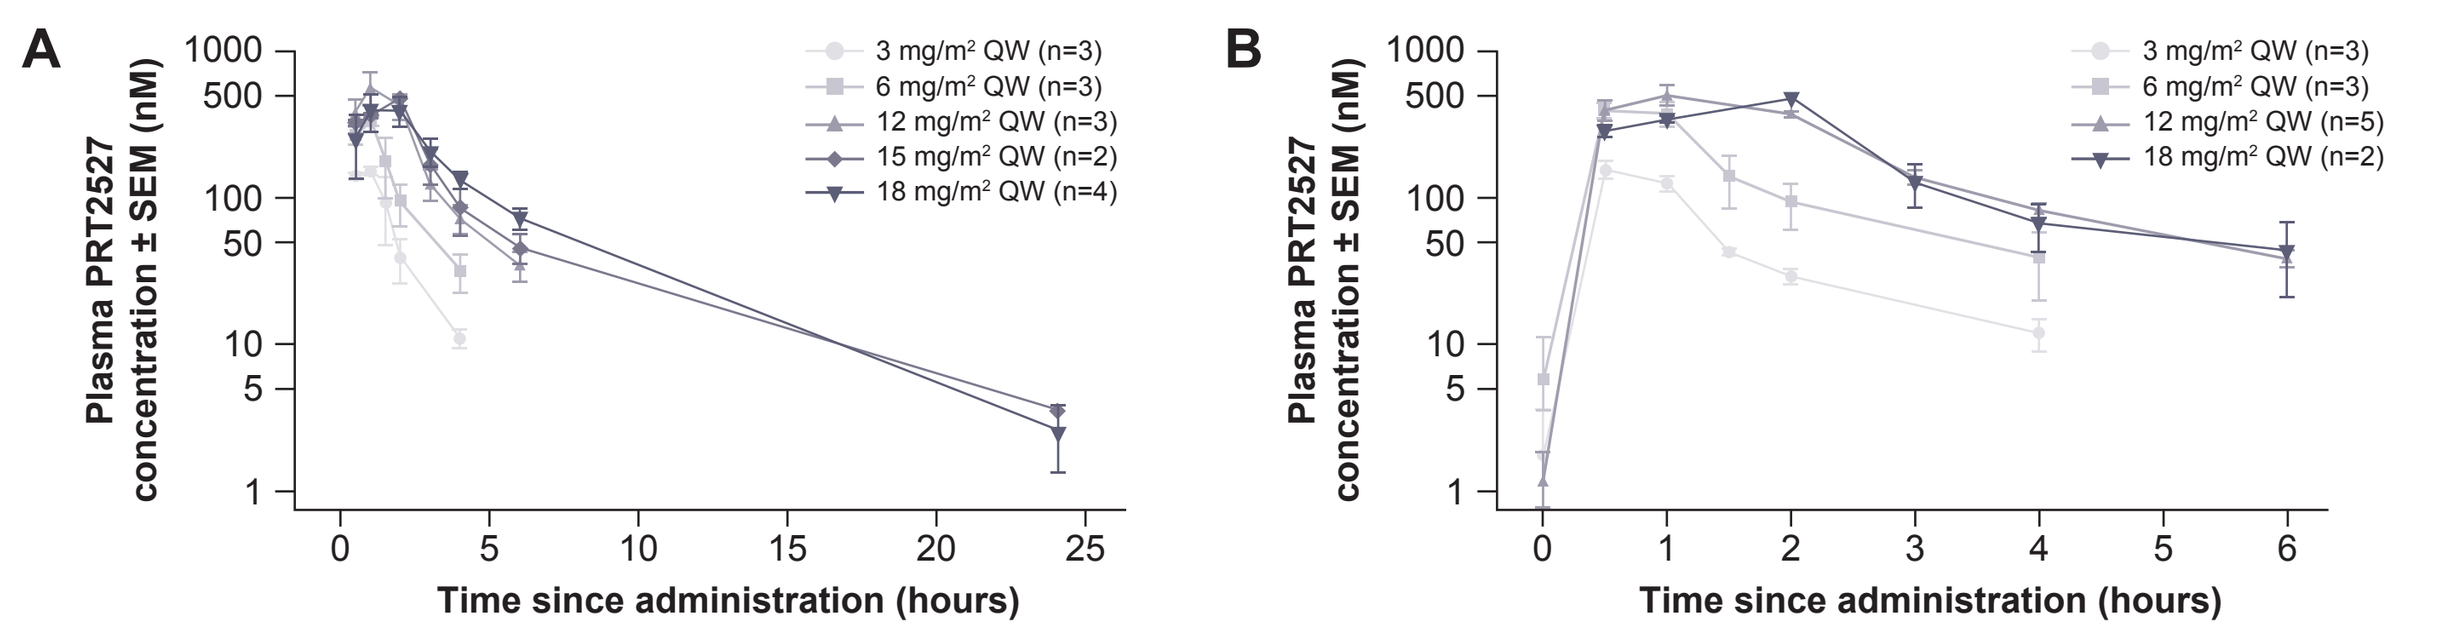
A
Plasma PRT2527 concentration ± SEM (nM)
1000 500 100 50 10 5 1
0 5 10 15 20 25
Time since administration (hours)
3 mg/m² QW (n=3)
6 mg/m² QW (n=3)
12 mg/m² QW (n=3)
15 mg/m² QW (n=2)
18 mg/m² QW (n=4)
B
Plasma PRT2527 concentration ± SEM (nM)
0 1 2 3 4 5 6
Time since administration (hours)
3 mg/m² QW (n=3)
6 mg/m² QW (n=3)
12 mg/m² QW (n=5)
18 mg/m² QW (n=2)

SEM, standard error of the mean.

Table 4. PK Profile During Week 1 and Week 3 Post Administration

Dose	Week	$AUC_{(0-t)}$, nM*hour	AUC_{inf}, nM*hour	C_{max}, nM	T_{max}, hours	Half-life, hours
3 mg/m²	1	321.7 (297.0-469.9)	NR	168.5 (153.2-184.4)	1.0 (1.0-1.5)	NR
QW (n=3)	3	220.1 (187.6-234.6)	NR	161.2 (128.9-195.4)	0.5 (0.5-1.0)	NR
6 mg/m²	1	700.8 (595.2-806.3)	NR	332.6 (298.5-387.7)	1.0 (1.0-1.5)	NR
QW (n=3)	3	623.4 (478.6-768.1)	NR	410.6 (365.4-455.7)	0.75 (0.5-1.0)	NR
12 mg/m²	1	1345.9 (1307.2-2234.0)	NR	616.2 (407.1-807.4)	1.0 (1.0-2.0)	NR
QW (n=3)	3[a]	1201.2 (1146.0-1500.2)	NR	434.7 (380.7-726.5)	1.0 (1.0-2.0)	NR
15 mg/m²	1	1708.8 (1475.3-1942.4)	1733.4 (1503.7-1963.0)	483.4 (466.4-500.3)	2.0 (2.0-2.0)	4.7 (4.1-5.3)
QW (n=3)	3	ND	ND	ND	ND	ND
18 mg/m²	1	1936.9 (1555.9-2400.3)	2306.9 (1609.8-2419.9)	522.4 (152.0-572.0)	1.5 (1.0-4.0)	3.8 (3.4-4.5)
QW (n=3)	3[b]	1059.7 (1049.6-1069.8)	NR	425.2 (360.2-490.2)	1.5 (1.0-2.0)	NR

Data are expressed as mean (range). [a]n=5; [b]n=2. $AUC_{(0-t)}$, AUC from dosing up to the last measurable concentration; AUC_{inf}, AUC from dosing extrapolated to infinity; ND, no data; NR, not reported; T_{max}, time to C_{max}.

Pharmacodynamics

- A dose-dependent downregulation of *MYC* and *MCL-1* mRNA expression in PBMCs was observed (Figure 6A-B).
 - A maximum inhibition of CDK9 transcriptional targets was observed at 2 to 4 hours post-dose.
- Generally, a dose-dependent inhibition of p-RNAP2 protein was observed in PBMCs (Figure 6C).

Figure 6. PRT2527-Associated Inhibition of CDK9 Transcriptional Targets *MYC* (A), *MCL-1* (B), and p-RNAP2 (C) in PBMCs

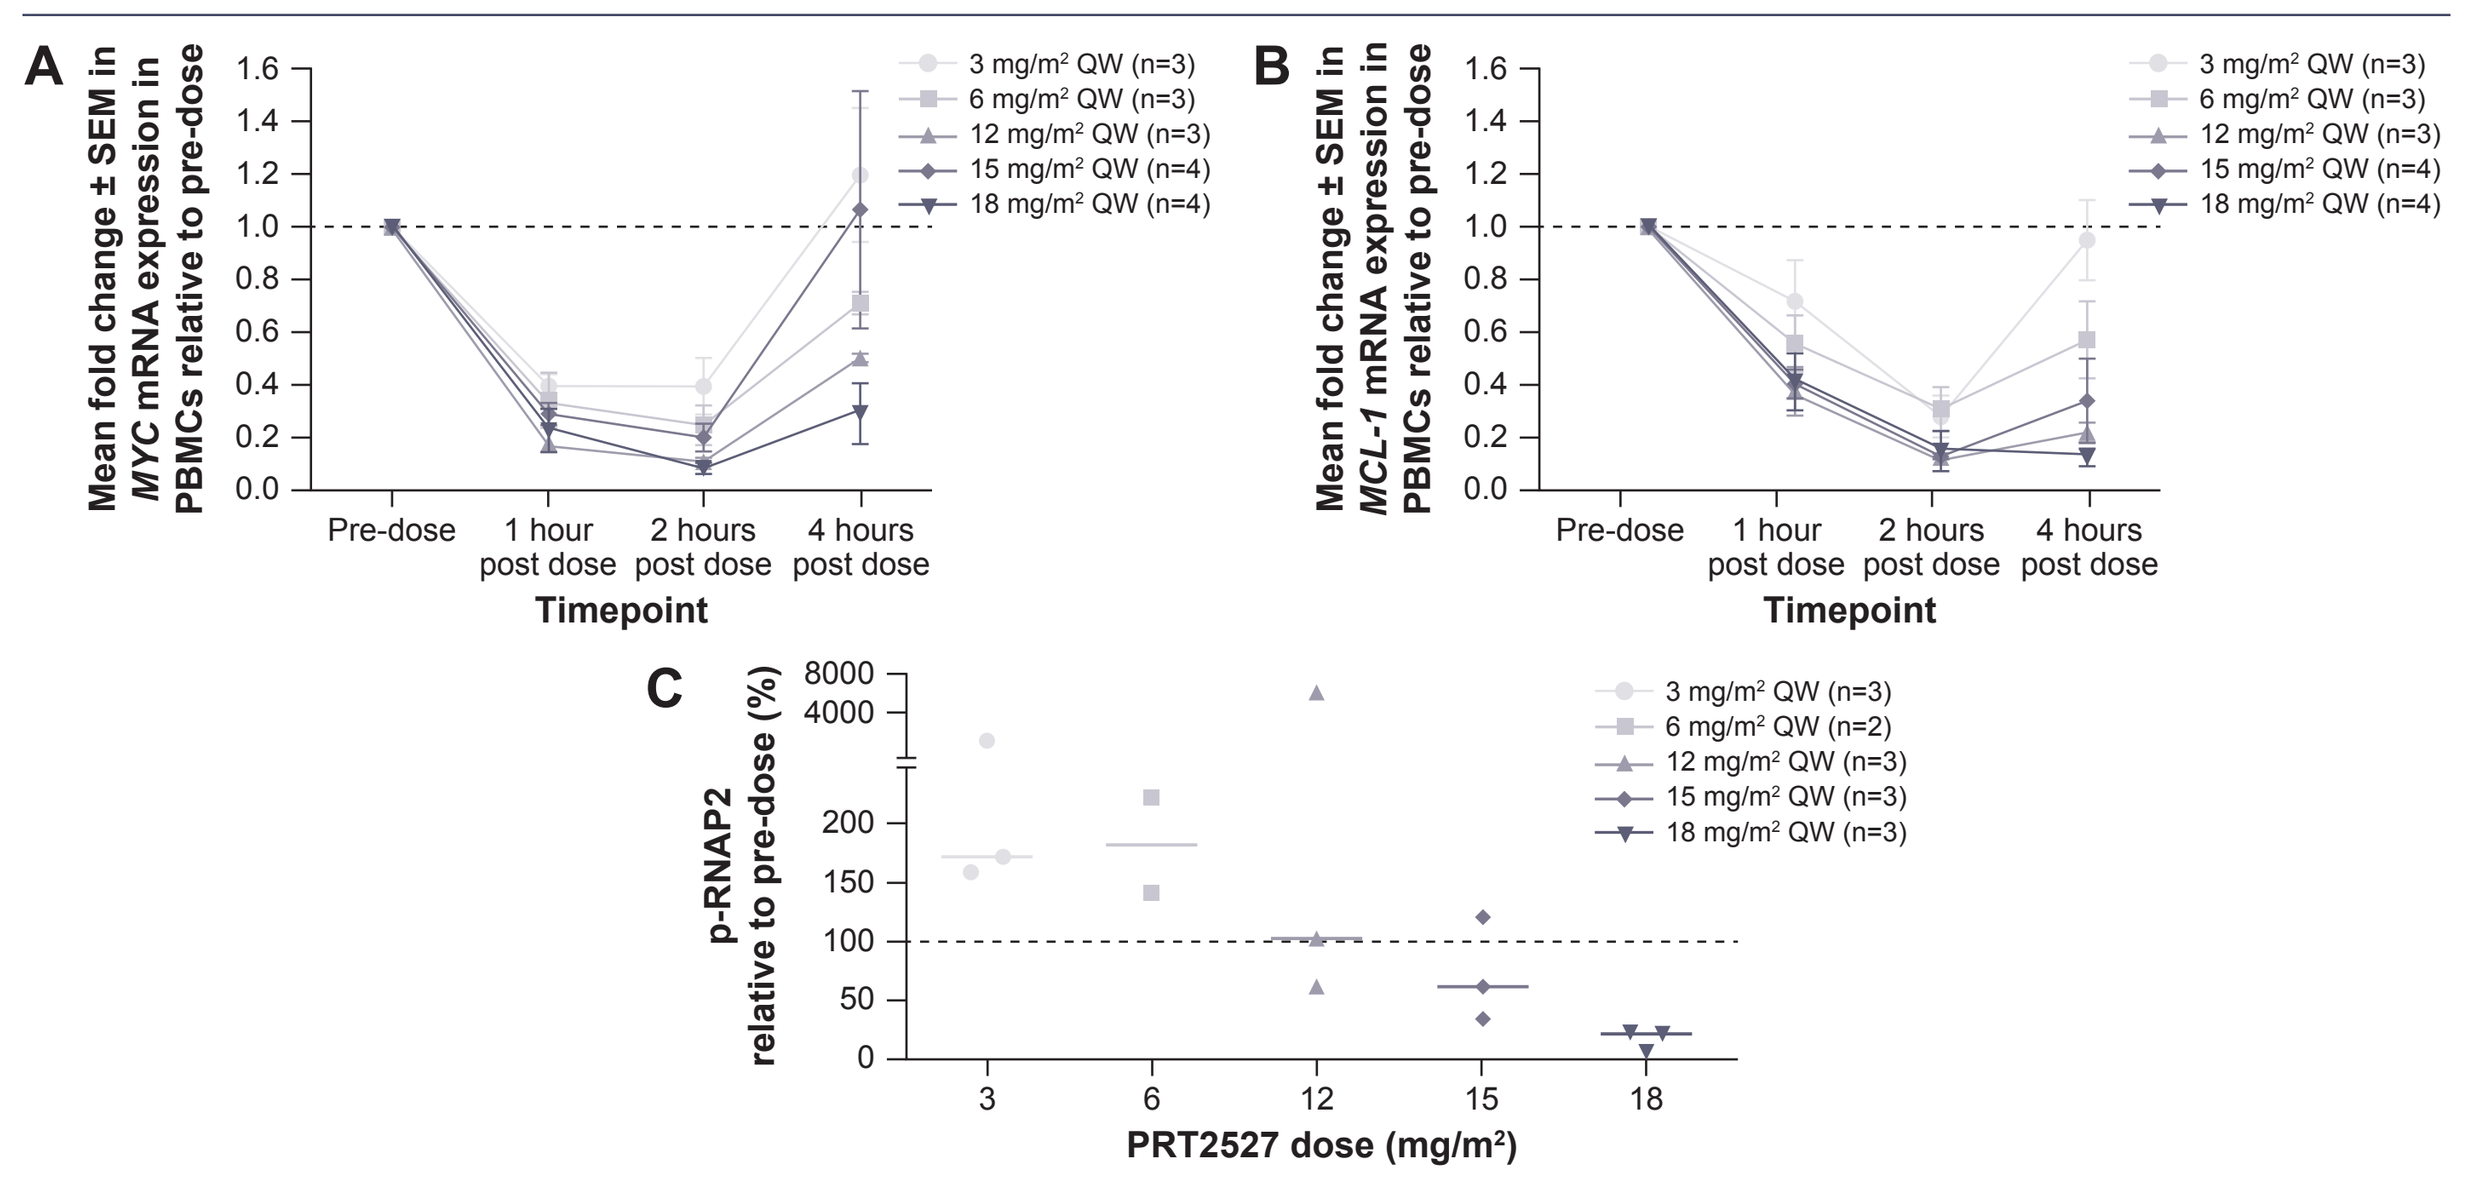
A
Mean fold change ± SEM in MYC mRNA expression in PBMCs relative to pre-dose
1.6 1.4 1.2 1.0 0.8 0.6 0.4 0.2 0.0
Pre-dose
1 hour post dose
2 hours post dose
4 hours post dose
Timepoint
3 mg/m² QW (n=3)
6 mg/m² QW (n=3)
12 mg/m² QW (n=3)
15 mg/m² QW (n=4)
18 mg/m² QW (n=4)
B
Mean fold change ± SEM in MCL-1 mRNA expression in PBMCs relative to pre-dose
Timepoint
C
p-RNAP2 relative to pre-dose (%)
8000 4000 200 150 100 50 0
3 6 12 15 18
PRT2527 dose (mg/m²)
3 mg/m² QW (n=3)
6 mg/m² QW (n=2)
12 mg/m² QW (n=3)
15 mg/m² QW (n=3)
18 mg/m² QW (n=3)

All panels: the dotted horizontal line represents the pre-dose measurement. Panel C: individual patient data are shown.

Conclusions

- In adults with advanced solid tumors, PRT2527 demonstrated favorable tolerability with manageable neutropenia and absence of significant gastrointestinal events or hepatotoxicity.
- The short half-life of PRT2527 enables acute CDK9 inhibition over a defined period, making it suitable for weekly administration without inducing significant toxicity.
- The observed dose-dependent downregulation of CDK9 transcriptional targets – *MYC* and *MCL-1* mRNA expression in PBMCs isolated from patients treated with PRT2527 – was consistent with the degree of target engagement required for preclinical efficacy.
- The 15 m/mg² QW dose of PRT2527 was selected for further evaluation in a dose-confirmation cohort.
- The overall safety profile observed in this study supports further development of PRT2527 in combination with other targeted therapies, including in hematologic malignancies (NCT05665530).

References
[1]Zhang Y, et al. Presented at the AACR-NRI-EORTC Virtual Conference on Molecular Targets and Cancer Therapeutics, October 7-10, 2021. Virtual. Available at https://preludetx.com/wp-content/uploads/2021/10/PRT2527-EORTC-2021.pdf.
[2]Mandal R, et al. *Cancers (Basel)*. 2021;13:2181.

Acknowledgments
This study was funded by Prelude Therapeutics. Medical writing and editorial support was provided by Russell Craddock, PhD, of Parexel, Uxbridge, UK, and was funded by Prelude Therapeutics.

Copies obtained through the Quick Response (QR) Code are for personal use only and may not be reproduced without permission from the author. Author disclosures can also be viewed through the QR Code.