A Phase 1 Dose Escalation Study of Protein Arginine Methyltransferase 5 (PRMT5) Inhibitor PRT543 in Patients With Myeloid Malignancies

Manish R. Patel, MD[1]; Varun Monga, MD[2]; Shekeab Jauhari, MD[3]; Don Ambrose Stevens, MD[4]; Lucia Masarova, MD[5]; Meredith McKean, MD, MPH[6]; David Mauro, MD, PhD[7]; John Viscusi[7]; Peggy Scherle, PhD[7]; Neha Bhagwat, PhD[7]; Andrew Moore, MS[7]; William Sun, PhD[7]; Rachel Chiaverelli, PhD[7]; Eric Mintah, PhD[7]; Rajneesh Nath, MD[8]; Eytan Stein, MD[9]; Neil Palmisiano, MD[10]; Grerk Sutamtewagul, MD[2]; Srdan Verstovsek, MD, PhD[5]

[1]Sarah Cannon Research Institute, Florida Cancer Specialists & Research Institute, Sarasota, FL; [2]Department of Medicine, University of Iowa Hospitals and Clinics, Iowa City, IA; [3]Sarah Cannon Research Institute, Florida Cancer Specialists & Research Institute, Lake Mary, FL; [4]Norton Cancer Institute, Louisville, KY; [5]The University of Texas MD Anderson Cancer Center, Houston, TX; [6]Sarah Cannon Research Institute, Tennessee Oncology, Nashville, TN; [7]Research and Development, Prelude Therapeutics, Wilmington, DE; [8]Banner Health at MD Anderson Cancer Center, Gilbert, AZ; [9]Memorial Sloan Kettering Cancer Center, New York, NY; [10]Sidney Kimmel Cancer Center, Thomas Jefferson University, Philadelphia, PA

2609

Background and Objectives

- PRMT5 catalyzes symmetric arginine dimethylation of protein substrates that play important roles in the cell cycle, including histone modification, transcription, and spliceosome assembly, and is overexpressed in a variety of cancers[1]
 - Notably, specific splicing mutations confer poor prognosis in myeloid malignancies[2]
- PRT543 is a potent, selective, oral inhibitor of PRMT5[3]
- An open-label phase 1 dose escalation study of PRT543 was conducted in unselected patients with myeloid malignancies (NCT03886831)
- The key objectives of this study were to evaluate safety, pharmacokinetics, pharmacodynamics, and preliminary clinical signals and to determine the recommended expansion dose

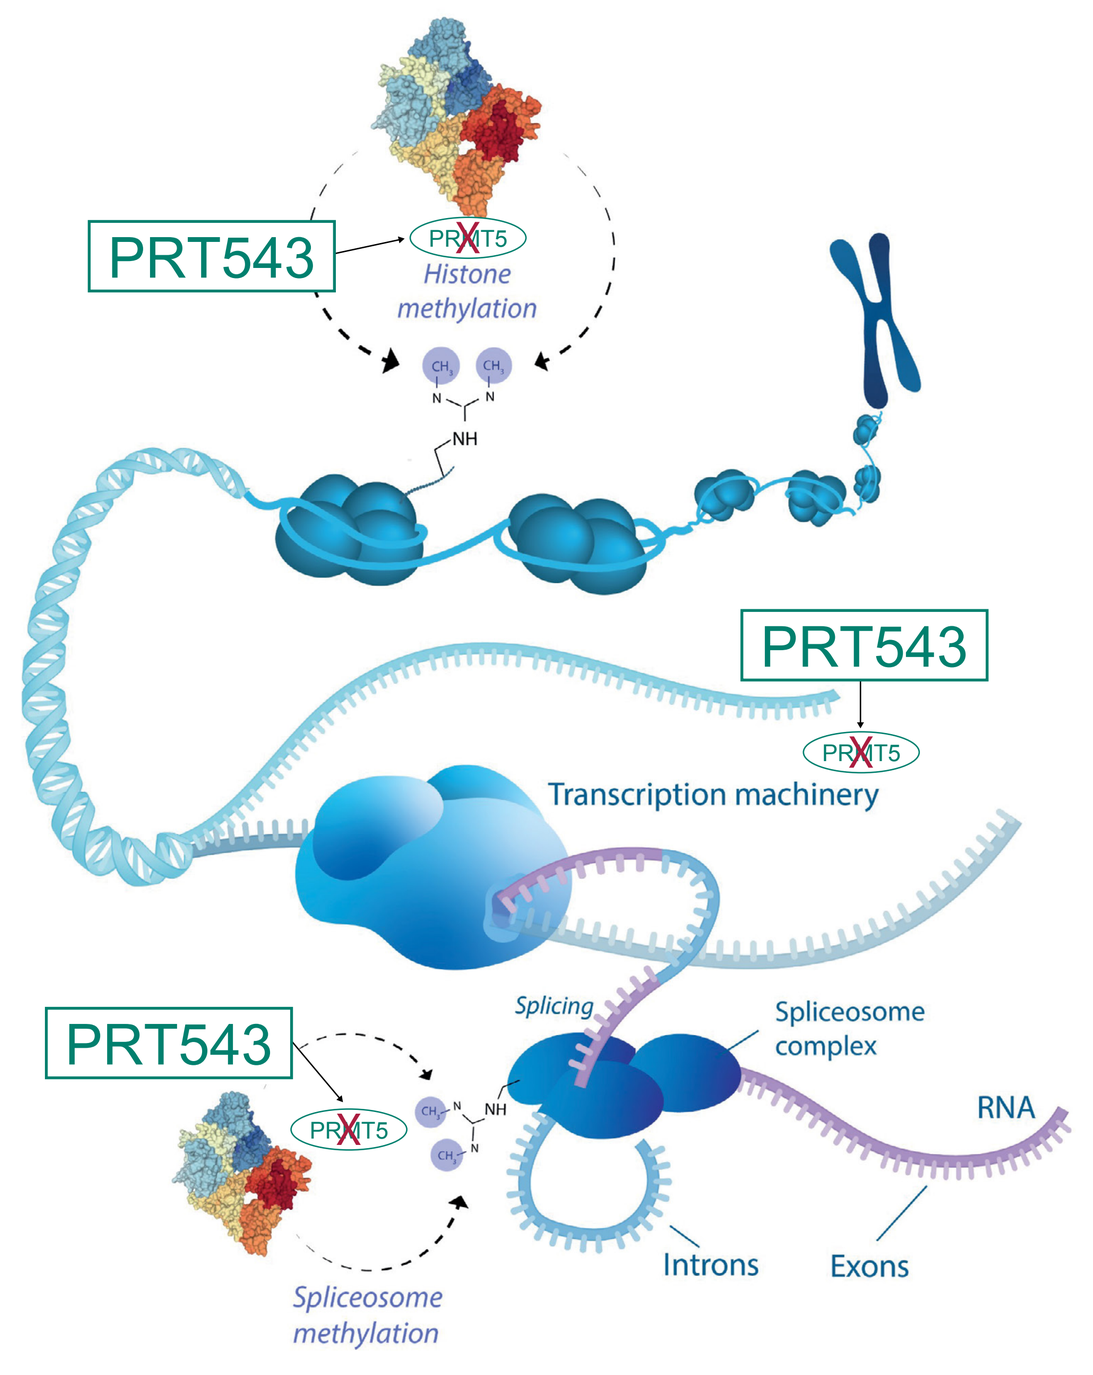
PRT543
PRMT5
Histone methylation
PRT543
PRMT5
Transcription machinery
PRT543
PRMT5
Splicing
Spliceosome complex
RNA
Introns
Exons
Spliceosome methylation

Study Design

- This was an open-label phase 1 study of 23 unselected patients with relapsed/refractory myelofibrosis (MF, n=12) or myelodysplastic syndrome (MDS, n=11) (Fig. 1)

Figure 1. Dose Escalation in Patients With Relapsed/Refractory MF or MDS[a]

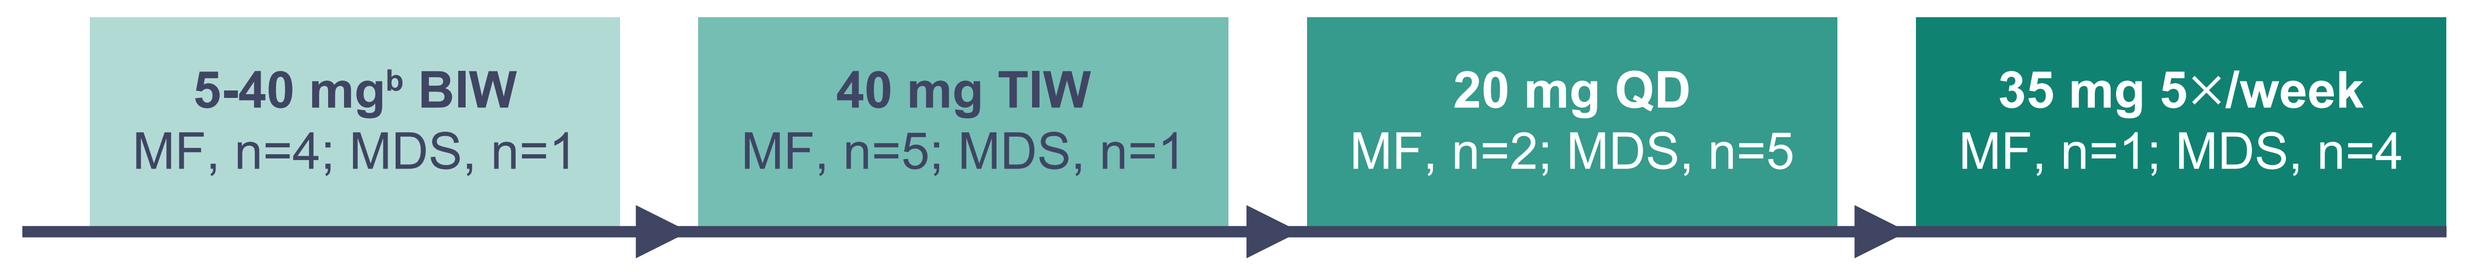
5-40 mg[b] BIW
MF, n=4; MDS, n=1
40 mg TIW
MF, n=5; MDS, n=1
20 mg QD
MF, n=2; MDS, n=5
35 mg 5×/week
MF, n=1; MDS, n=4

[a]Dosing in 28-day cycles for all patients. Accelerated 3+3 design. [b]Dosing at 5, 10, 20, and 40 mg BIW.
Data cutoff October 1, 2021.
BIW, 2 times a week; QD, once daily; TIW, 3 times a week.

Results

Table 1. Demographics

	MF (n=12)	**MDS** (n=11)
Age, years, median (range)	71 (61-78)	75 (68-84)
Female, n (%)	6 (50)	2 (18)
ECOG PS, n (%)		
0	3 (25)	0
1	9 (75)	10 (91)
2	0	1 (9)
Race, n (%)		
White or Caucasian	11 (92)	11 (100)
Other	1 (8)	0
IPSS-MF risk,[a] n (%)		
Intermediate 2	2 (20)	--
Not applicable	8 (80)	--
IPSS-MDS risk, n (%)		
High	--	3 (27)
Intermediate 1	--	4 (36)
Intermediate 2	--	2 (18)
Low	--	1 (9)
Not applicable	--	1 (9)
Platelet count (10^3/μL), median (range)	177 (33-655)	52 (14-359)
Hemoglobin (g/L), median (range)	92 (59-131)	80 (68-117)
Spleen[b] (cm below costal margin), median (range)	17 (10-20)	--
Prior lines of systemic therapy, median (range)	2 (0-6)	2 (0-6)
Prior ruxolitinib, n (%)	10 (83)	--
Splicing factor mutation–positive,[c] n (%)	5 (42)	6 (55)
JAK2 V167F mutation–positive, n (%)	10 (83)	--

[a]Data not available for 2 patients. [b]n=7. [c]Mutations were in (n=3 MF; 2 MDS), SRSF2 (n=1 MF; 3 MDS), U2AF1 (n=1 MF), and ZRSR2 (n=1 MDS). Splicing factor mutation status unevaluable for 3 MF patients.
ECOG PS, Eastern Cooperative Oncology Group performance status; IPSS, International Prognostic Scoring System.

Pharmacokinetics

- At the recommended expansion dose (35 mg 5×/week), half-life ($t_{½}$) was 10 hours, and plasma drug exposures, ie, maximum observed plasma concentration (C_{max}) and area under the curve (AUC), were 1480 nM and 15,557 nM.hr, respectively (Fig. 2)

Figure 2. Dose-dependent Increases in C_{max} and AUC[a]

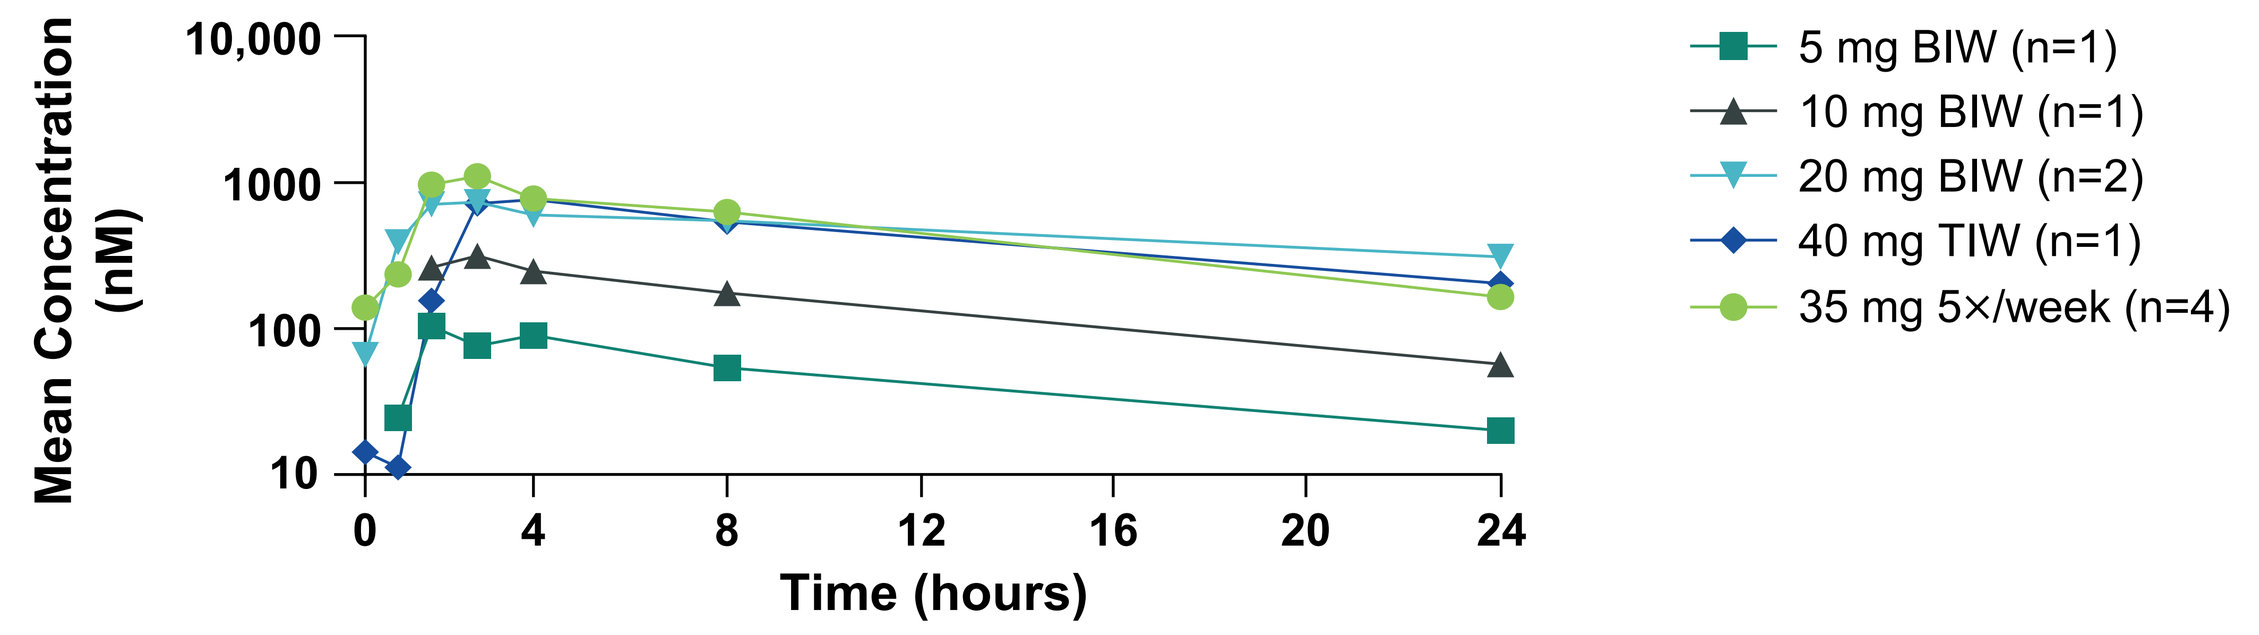
Mean Concentration (nM)
10,000
1000
100
10
0
4
8
12
16
20
24
Time (hours)
5 mg BIW (n=1)
10 mg BIW (n=1)
20 mg BIW (n=2)
40 mg TIW (n=1)
35 mg 5×/week (n=4)

	5 mg BIW	10 mg BIW	20 mg BIW	40 mg TIW	35 mg 5×/week
C_{max}, nM	114	345	808 (4)	841	1480 (5)
AUC, nM.hr	1231	3763	12,522 (18)	11,045	15,557 (37)
$t_{½}$, hours	10	10	20	N/A	10 (45)

[a]Data are from C1D25 and are considered steady state. Data are not available for patients treated with 40 mg BIW or 20 mg QD. Values in parentheses in the table are coefficient of variation (CV%). C, cycle; D, day; N/A, not applicable.

Pharmacodynamics

- PRT543 exhibited dose-dependent reduction of serum symmetric dimethylarginine (sDMA), a marker of PRMT5 target engagement, with a 58% decrease at the recommended expansion dose of 35 mg 5×/week (Fig. 3A)
- PRT543 decreased PRMT5 functional activity in peripheral blood mononuclear cells (PBMCs) at the recommended expansion dose of 35 mg 5×/week (Fig. 3B), as demonstrated by increased intron retention of transcripts known to be regulated by PRMT5

Figure 3. Confirmation of Target Engagement and Reduction in PRMT5 Functional Activity

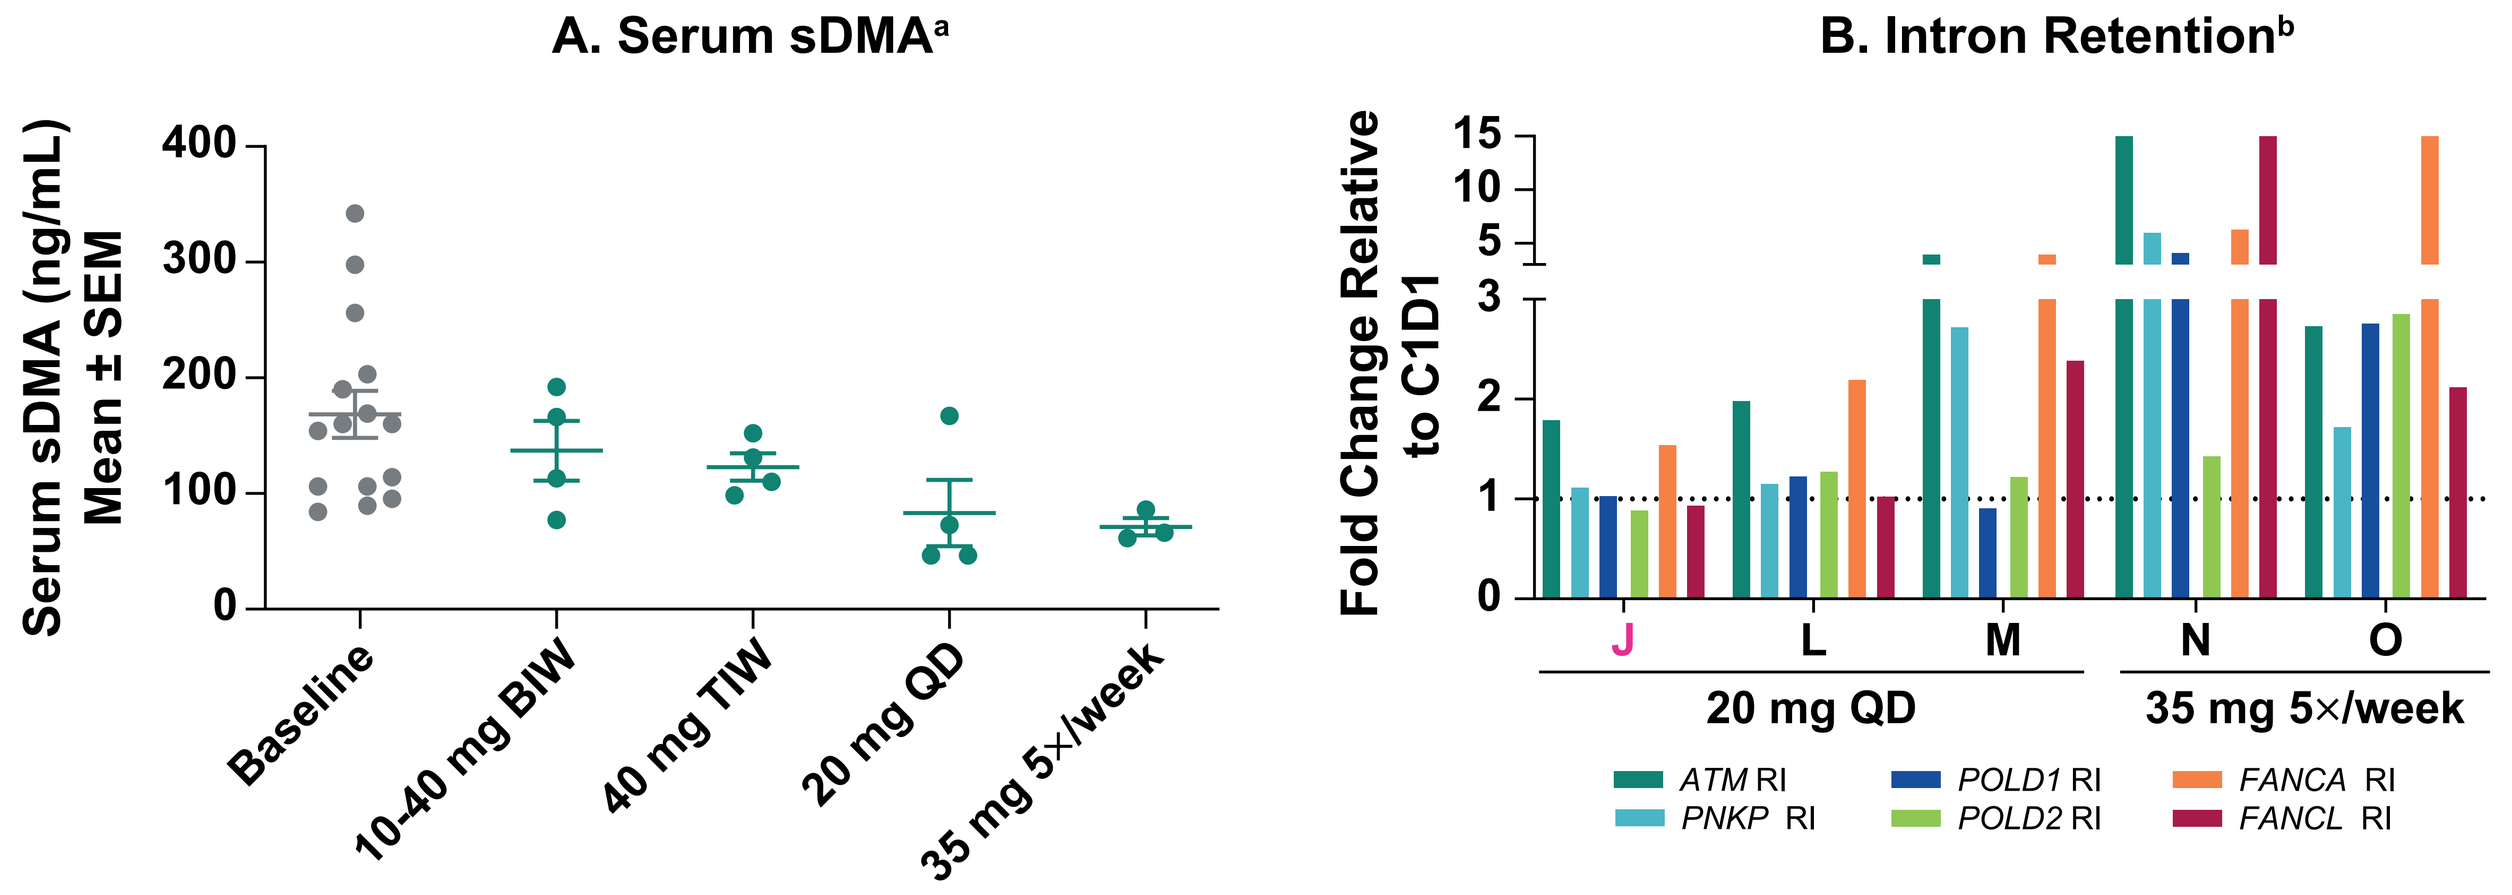
A. Serum sDMA[a]
Serum sDMA (ng/mL)
Mean ± SEM
400
300
200
100
0
Baseline
10-40 mg BIW
40 mg TIW
20 mg QD
35 mg 5×/week
B. Intron Retention[b]
Fold Change Relative to C1D1
15
10
5
3
2
1
0
J
L
M
N
O
20 mg QD
35 mg 5×/week
ATM RI
PNKP RI
POLD1 RI
POLD2 RI
FANCA RI
FANCL RI

[a]Serum sDMA was measured on C1D25 in 35 mg 5×/week and all BIW dose groups, and on C1D15 in 40 mg TIW and 20 mg QD dose groups. In BIW and TIW schedules, samples were not collected at steady state; n=15.
[b]Expression of retained introns known to be regulated by PRMT5 was evaluated in PBMCs on C1D25 for 35 mg 5×/week and on C2D1 for 20 mg QD. All are patients with MDS, except Patient J with MF. RI, retained intron.

- Reductions in inflammatory serum markers and cytokines were observed in MF patients (Fig. 4)

Figure 4. Reductions in Inflammatory Serum Markers and Cytokines in MF Patients

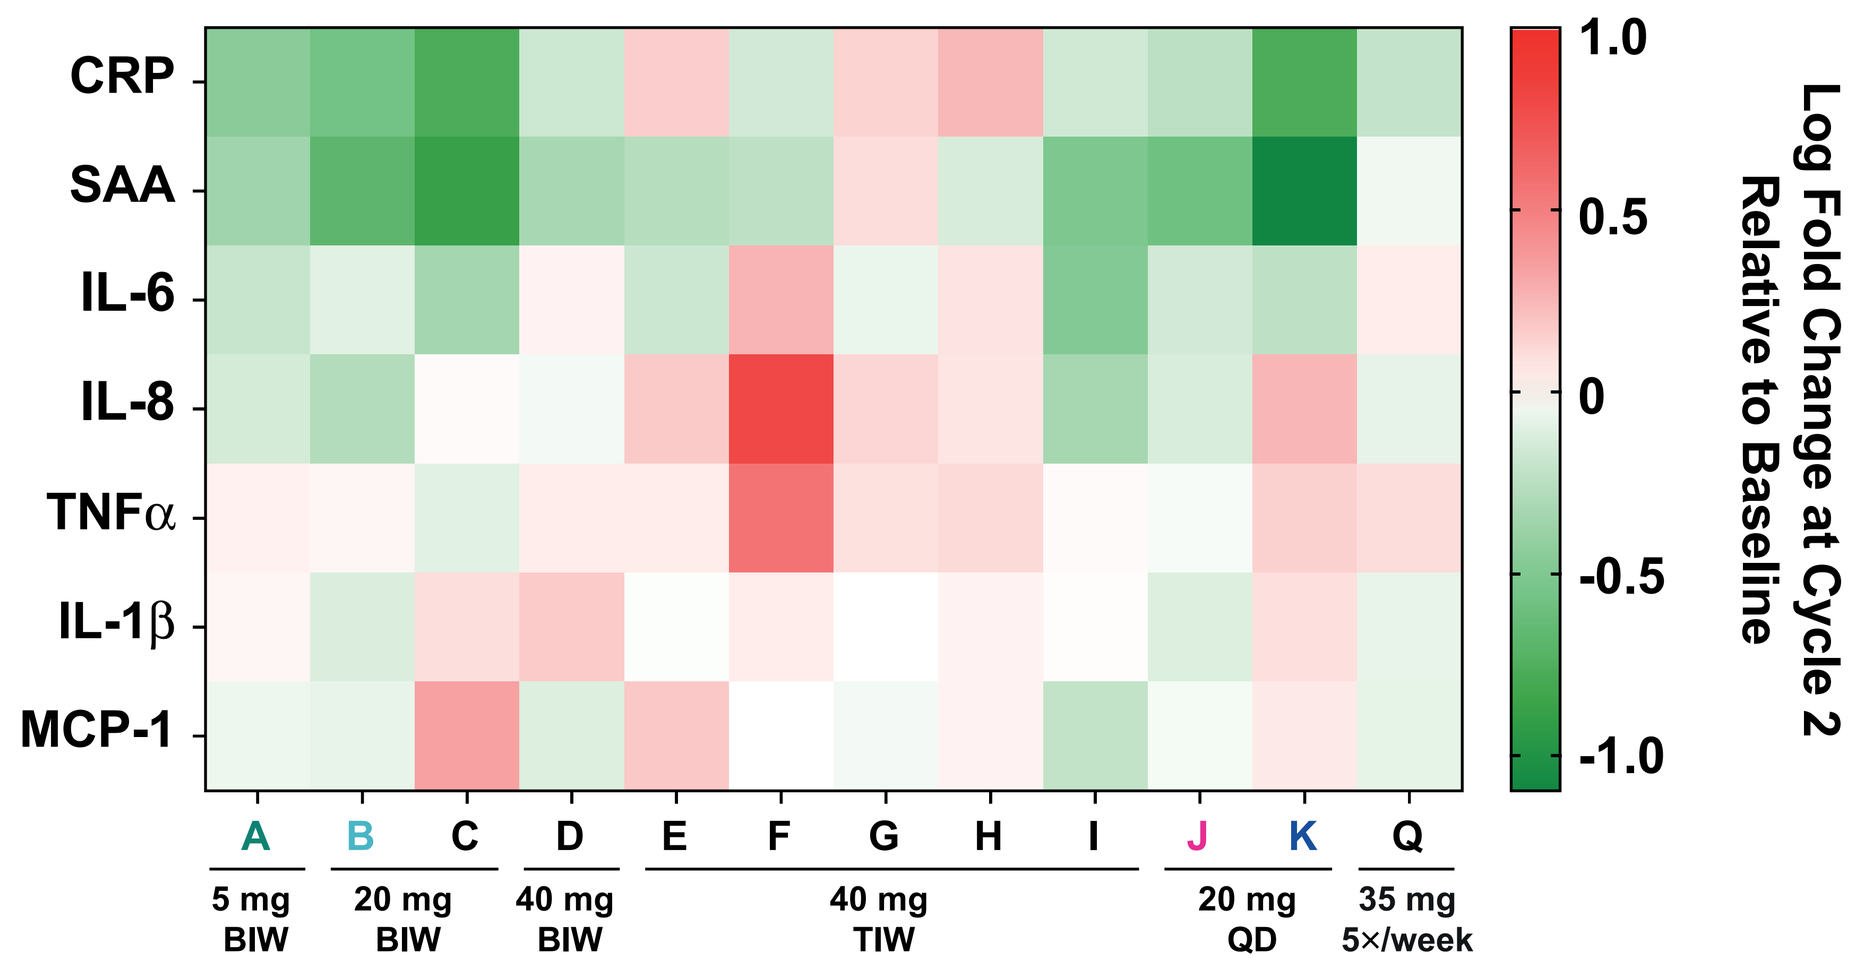
CRP
SAA
IL-6
IL-8
TNFα
IL-1β
MCP-1
A
B
C
D
E
F
G
H
I
J
K
Q
5 mg BIW
20 mg BIW
40 mg BIW
40 mg TIW
20 mg QD
35 mg 5×/week
1.0
0.5
0
-0.5
-1.0
Log Fold Change at Cycle 2 Relative to Baseline

Assessments on C2D1.
CRP, C-reactive protein, IL, interleukin; MCP, monocyte chemoattractant protein; SAA, serum amyloid A; TNF, tumor necrosis factor.

Safety

- Three of 23 patients (13%) had a single occurrence of dose-limiting thrombocytopenia (1 each at 40 mg TIW, 35 mg 5×/week, and 20 mg QD)
- Eighteen patients discontinued treatment, primarily because of disease progression or investigator decision. One patient (35 mg 5×/week) discontinued due to an adverse event (performance status decreased/weakness)
- No treatment-related deaths were reported
- Grade ≥3 treatment-related adverse events of anemia and thrombocytopenia (Table 2) were reversible with dose modification

Table 2. Most Common Treatment-Related Adverse Events of Any Grade Occurring in ≥5% of Patients

	All Patients (N=23)		Recommended Expansion Dose 35 mg 5×/week (n=5)	
Adverse Event, n (%)	**Any Grade**	**Grade ≥3**	**Any Grade**	**Grade ≥3**
Patients with events	14 (61)	6 (26)	2 (40)	1 (20)
Thrombocytopenia	5 (22)	4 (17)	1 (20)	1 (20)
Nausea	5 (22)	0	1 (20)	0
Anemia	4 (17)	4 (17)	0	0
Diarrhea	4 (17)	0	0	0
Fatigue	3 (13)	0	0	0
Bone pain	2 (9)	0	0	0

Preliminary Clinical Activity

- Early signals of clinical activity were detected in 5 of 23 patients (Table 3) across assessments that included:
 - International Working Group (IWG) response (n=3/23; Table 3);
 - investigator-reported symptom response with concomitant improvements in inflammatory serum markers (n=3/23; Fig. 5);
 - sustained increases in hemoglobin with reductions in transfusion frequency (n=3/23; Fig. 6); and
 - improvement in bone marrow reticulin fibrosis (n=1/23; Fig. 7)

Table 3. Patients With IWG Response and/or Symptom Improvement

Patient	Initial PRT543 Dose[a]	Time on Treatment (Days)[b]	Splicing Factor Mutation	Prior Ruxolitinib (Years)	Best IWG Response	Symptom Improvement[d]
A (MF)	5 mg BIW	382 PD	No	1.5	Anemia response[4]	Yes C4
B (MF)	20 mg BIW	704 PD	No	4.0	None	Yes C4
J (MF)	20 mg QD	141 Other[c]	SRSF2 P95L	1.0	None	Yes C2 + C4
K (MF)	20 mg QD	365+ SD	SF3B1 K700E	0	Anemia response[4]	Yes C3 to C13
P (MDS)	35 mg 5×/week	188+ PD	SRSF2 P95L	N/A	Hematologic improvement-erythropoietic[5]	N/A

[a]Modification of initial dose occurred in all 5 patients (dosing range, 5 mg BIW to 20 mg QD). [b]Response at data cutoff on October 1, 2021. [c]Patient underwent off-study allogeneic transplant. [d]Per investigator assessment.
PD, progressive disease; SD, stable disease; +, ongoing.

Figure 5. Concomitant Reductions in Inflammatory Serum Markers in Patients With Symptom Improvement[a]

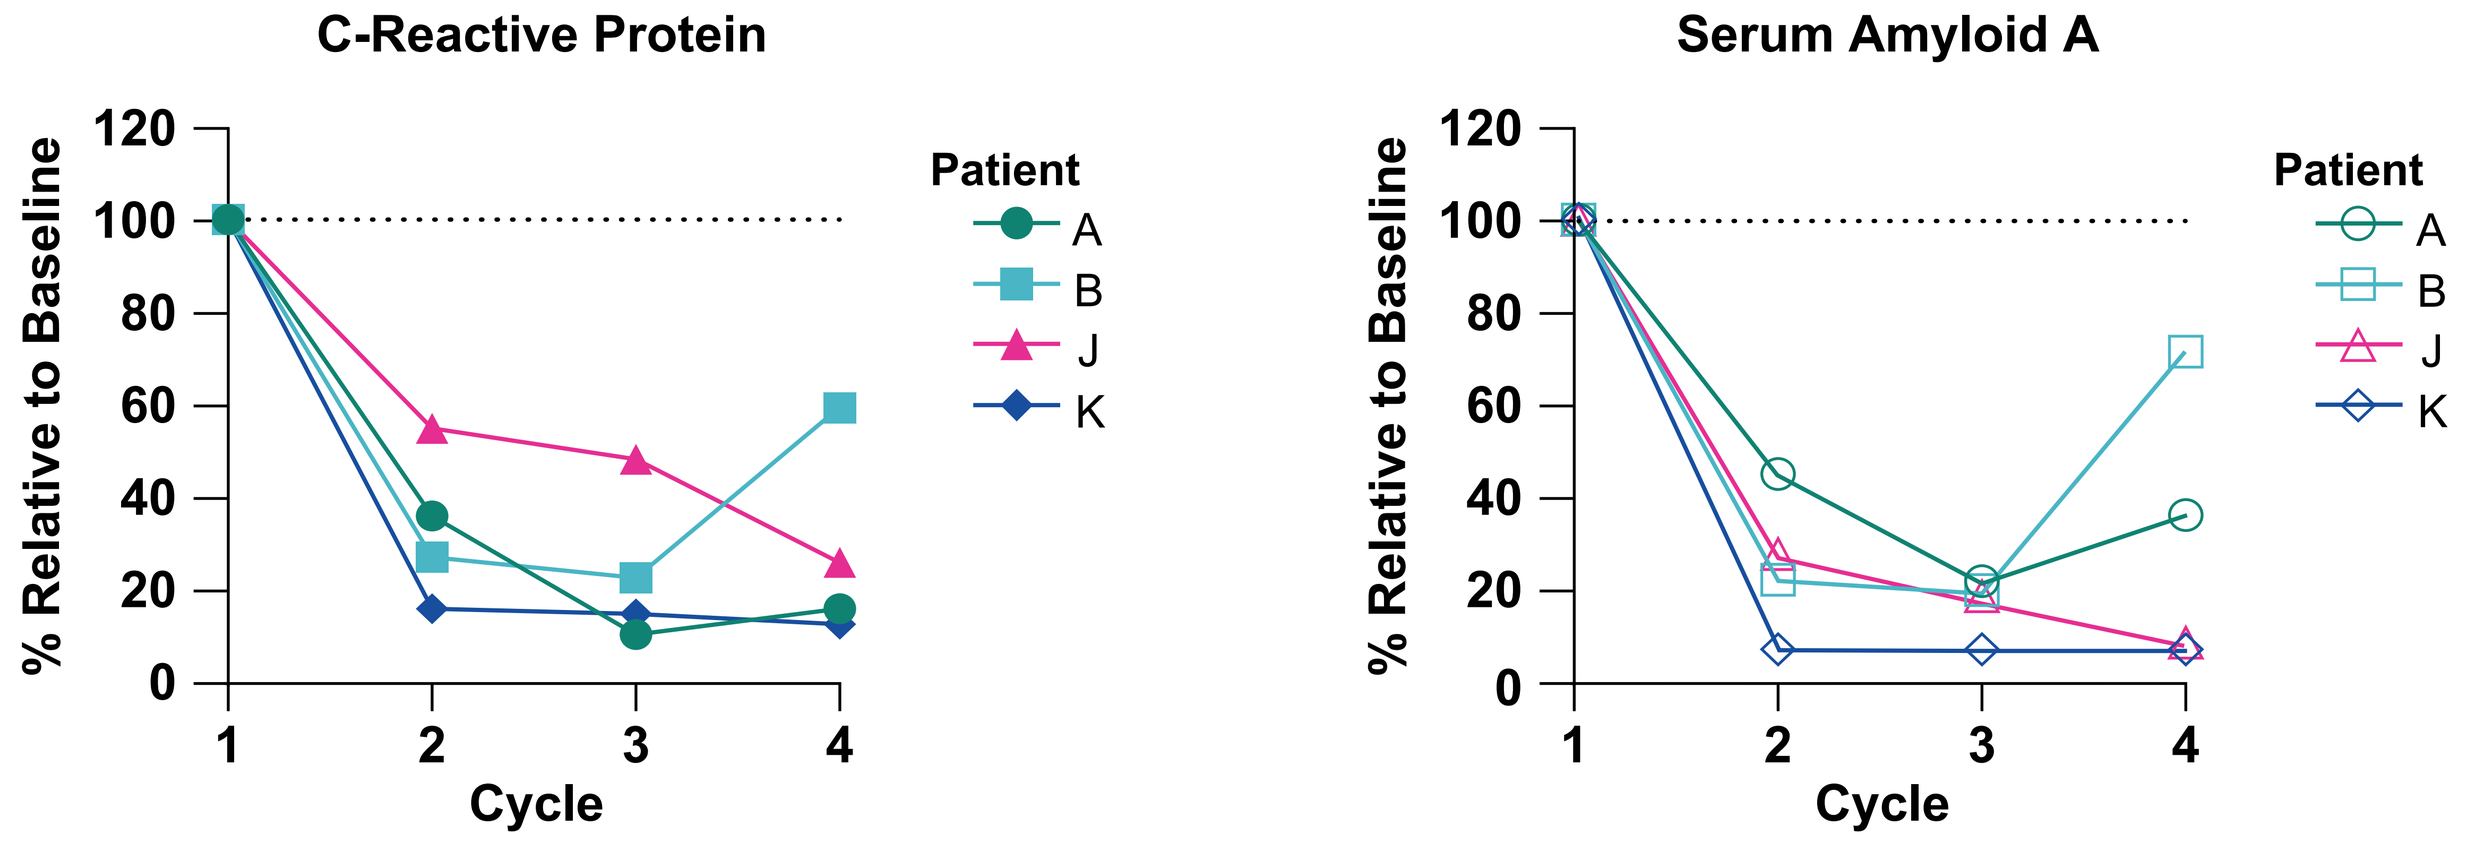
C-Reactive Protein
% Relative to Baseline
120
100
80
60
40
20
0
1
2
3
4
Cycle
Patient
A
B
J
K
Serum Amyloid A
% Relative to Baseline
Cycle
Patient
A
B
J
K

[a]Baseline assessments on C1D25.

Figure 6. Reduction in Transfusion Frequency (A) and Sustained Increase in Hemoglobin (B) in Patients A, K, and P

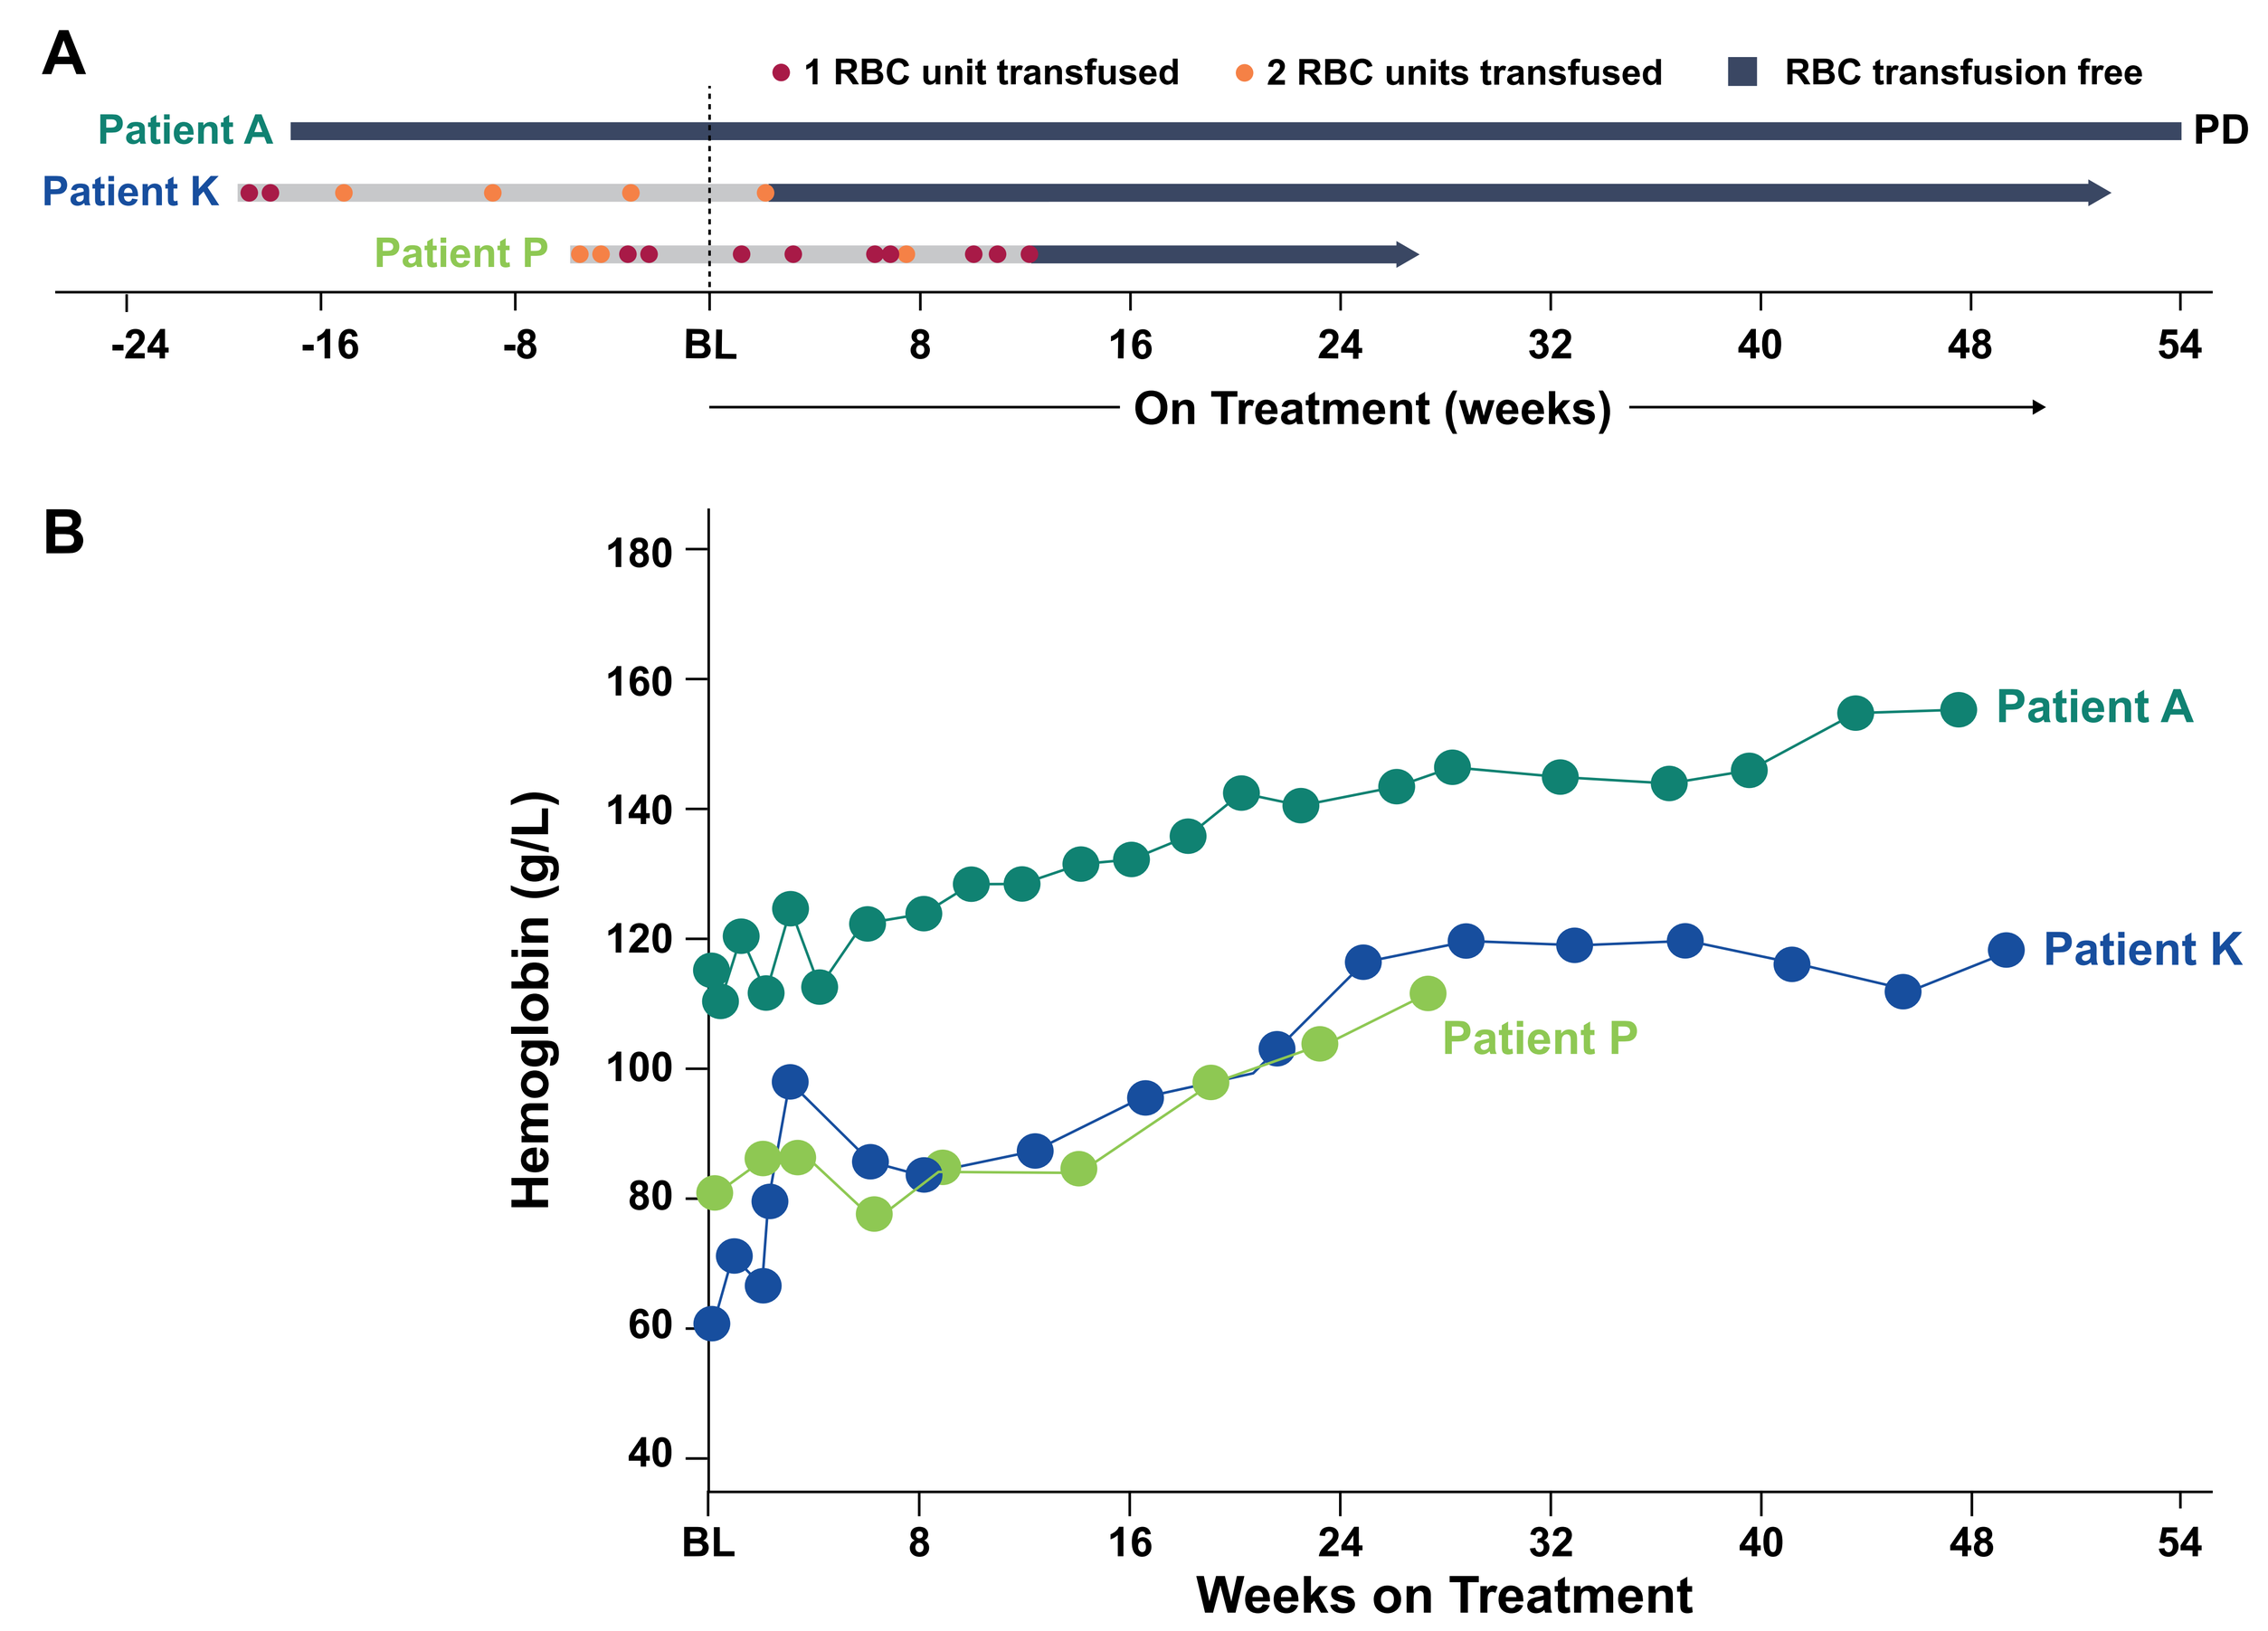
A
1 RBC unit transfused
2 RBC units transfused
RBC transfusion free
Patient A
Patient K
Patient P
PD
-24
-16
-8
BL
8
16
24
32
40
48
54
On Treatment (weeks)
B
Hemoglobin (g/L)
180
160
140
120
100
80
60
40
Patient A
Patient K
Patient P
Weeks on Treatment

RBC, red blood cell.

Figure 7. Improvement in Bone Marrow Reticulin Fibrosis (+2-3 to +1-2) in Patient B

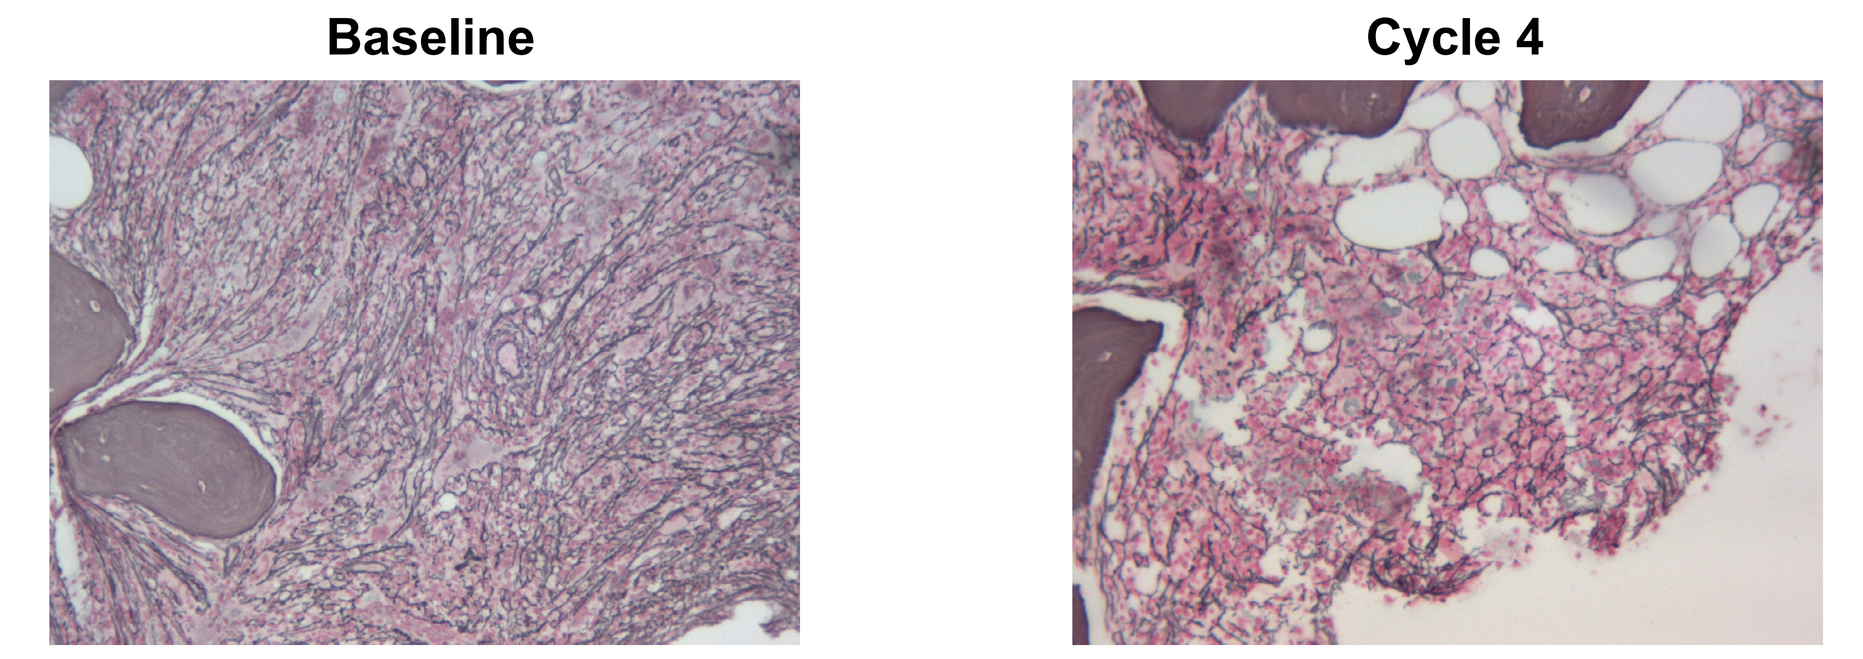
Baseline
Cycle 4

Conclusions

- PRT543 was well tolerated with a favorable safety profile
- Dose-dependent inhibition of PRMT5 target engagement and functional activity were observed
- Preliminary signals of clinical activity, including hematological responses and symptomatic improvements with concomitant reductions in serum inflammatory makers, were noted
- 35 mg 5×/week was determined to be the recommended expansion dose, and the expansion phase of the study is ongoing (NCT03886831)

References

1. Kim H, Ronai ZA. *Cell Stress*. 2020;4(8):199-215. 2. Fong JY, et al. *Cancer Cell*. 2019;36(2):194-209. 3. Bhagwat N, et al. AACR Virtual Meeting; June 22-24, 2020. Poster 2915. 4. Tefferi A, et al. *Blood*. 2013;122(8):1395-1398. 5. Cheson BD, et al. *Blood*. 2006;108(2):419-425.

Acknowledgments

We would like to thank the patients who participated in this study and their families, as well as the study teams at each participating institution. Thank you to Dr. Wilfredo Blasini for the bone marrow images.
This study was sponsored by Prelude Therapeutics. Editorial assistance was provided by Parexel and was funded by Prelude Therapeutics.

Disclosures

Research Funding: Florida Cancer Specialists; Acerta Pharma; ADC Therapeutics; Agenus Pharma; Aileron Therapeutics; Artios Pharma; AstraZeneca; Bicycle Therapeutics; BioNTech; Boehringer Ingelheim; Calithera; Celgene; Checkpoint Therapeutics; Ciclomed; Clovis; Curis; Cyteir; Daiichi Sankyo; Effector Therapeutics; Eli Lilly; EMD Serono; Evelo Biosciences; Forma Therapeutics; Genentech/Roche; Gilead; GlaxoSmithKline; H3 Biomedicine; Hengrui; Hutchinson MediPharma; Ignyta; Incyte; Jacobio; Janssen; Jounce Therapeutics; Klus Pharma; Kymab; Loxo Oncology; LSK Biopartners; Lycera; Mabspace; Macrogenics; Merck; Millennium Pharmaceuticals; Mirati Therapeutics; ModernaTX; ORIC Pharmaceuticals; Pfizer; Phoenix Molecular Designs; Placon Therapeutics; Portola Pharmaceuticals; Prelude Therapeutics; Qilu Puget Sound Biotherapeutics; Revolution Medicines; Ribon Therapeutics; Seven and Eight Biopharmaceuticals; Syndax; Synthorx; Stemline Therapeutics; Taiho; Takeda; Tesaro; TopAlliance; Vedanta; Verastem; Vigeo; Xencor

Membership on an entity's Board of Directors of advisory committees: AbbVie; Bayer; Celgene; EMD Serono; Exelixis; Genentech/Roche; Janssen; Pfizer; Pharmacyclics

Other: Alexion, AstraZeneca Rare Disease: study investigator