Preclinical Characterization of PRT543, a Potent and Selective Inhibitor of Protein Arginine Methyltransferase 5 (PRMT5), With Broad Antitumor Activity in *In vitro* and *In vivo* Models

Poster #: 2915

Neha Bhagwat[1], Yang Zhang[1], Hong Lin[1], Min Wang[1], Dave Rominger[1], Tom Emm[1], Divya Chugani-Mahtani[1], Dimitrios Angelis[1], Rupa Shetty[1], Raul Leal[1], William Gowen-MacDonald[1], Alexander Grego[1], Juan Luengo[1], Taghi Manshouri[2], Friederike Pastore[3], Ross L. Levine[3], Srdan Verstovsek[2], Bruce Ruggeri[1], Peggy Scherle[1], Kris Vaddi[1].
[1]Prelude Therapeutics, Wilmington, DE; [2]The University of Texas MD Anderson Cancer Center, Houston, TX; [3]Memorial Sloan-Kettering Cancer Center, New York, NY.
Contact: nbhagwat@preludetx.com

INTRODUCTION

PRMT5

Protein arginine methyltransferase 5

The major Type II PRMT that catalyzes the formation of symmetrical dimethyl arginine (SDMA) on cellular proteins

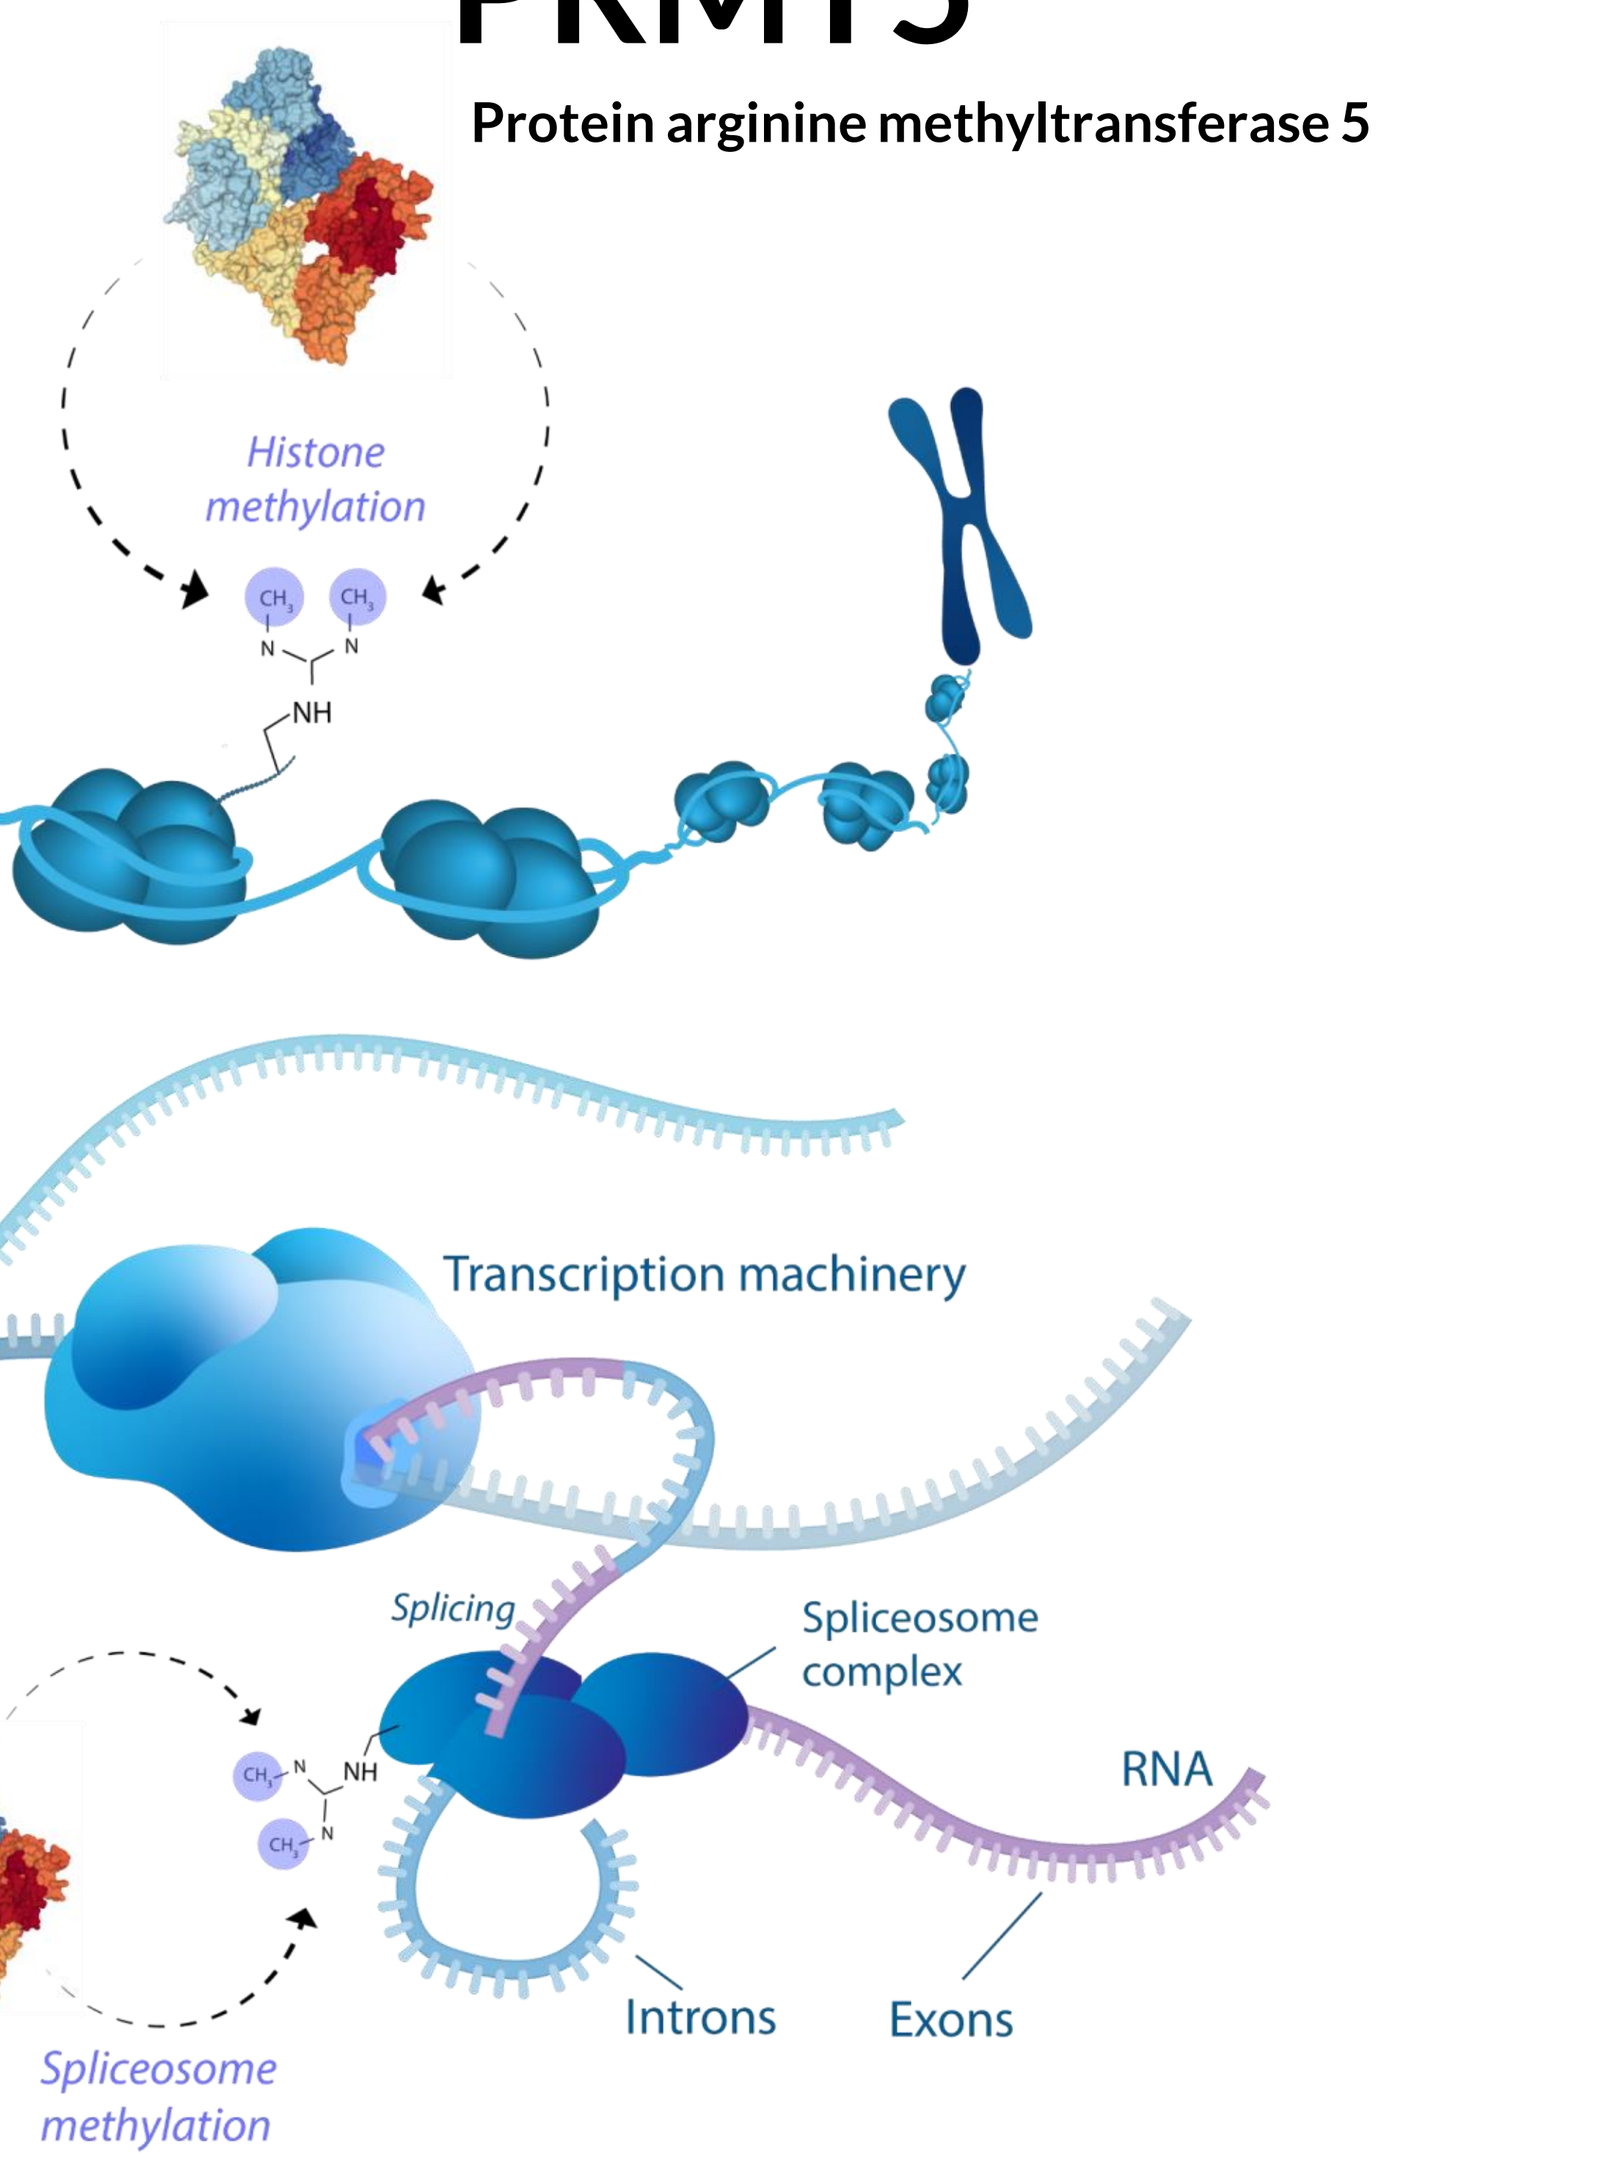
Histone methylation
Transcription machinery
Splicing
Spliceosome complex
RNA
Introns
Exons
Spliceosome methylation

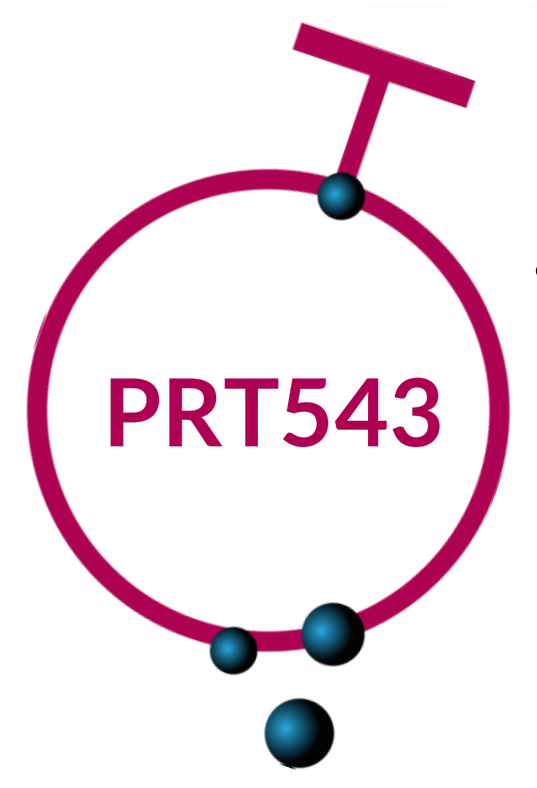
PRT543

- **Major substrates include:**
 - **Histones,** leading to repression of tumor suppressor genes at specific loci.
 - **Transcription factors** such as p53, E2F-1, NF-κB, thereby regulating gene expression on a global level
 - **Spliceosome complex,** thereby regulating pre-mRNA processing of key target genes such as MYC
- PRMT5 is overexpressed in a wide variety of cancers and arginine methylation is emerging as an important oncogenic and resistance mechanism

PRT543 is a potent and selective PRMT5 inhibitor currently under evaluation in a Phase I clinical trial in patients with advanced solid tumors and hematologic malignancies (NCT03886831).

RESULTS

PRT543 IS A POTENT AND SELECTIVE PRMT5 INHIBITOR

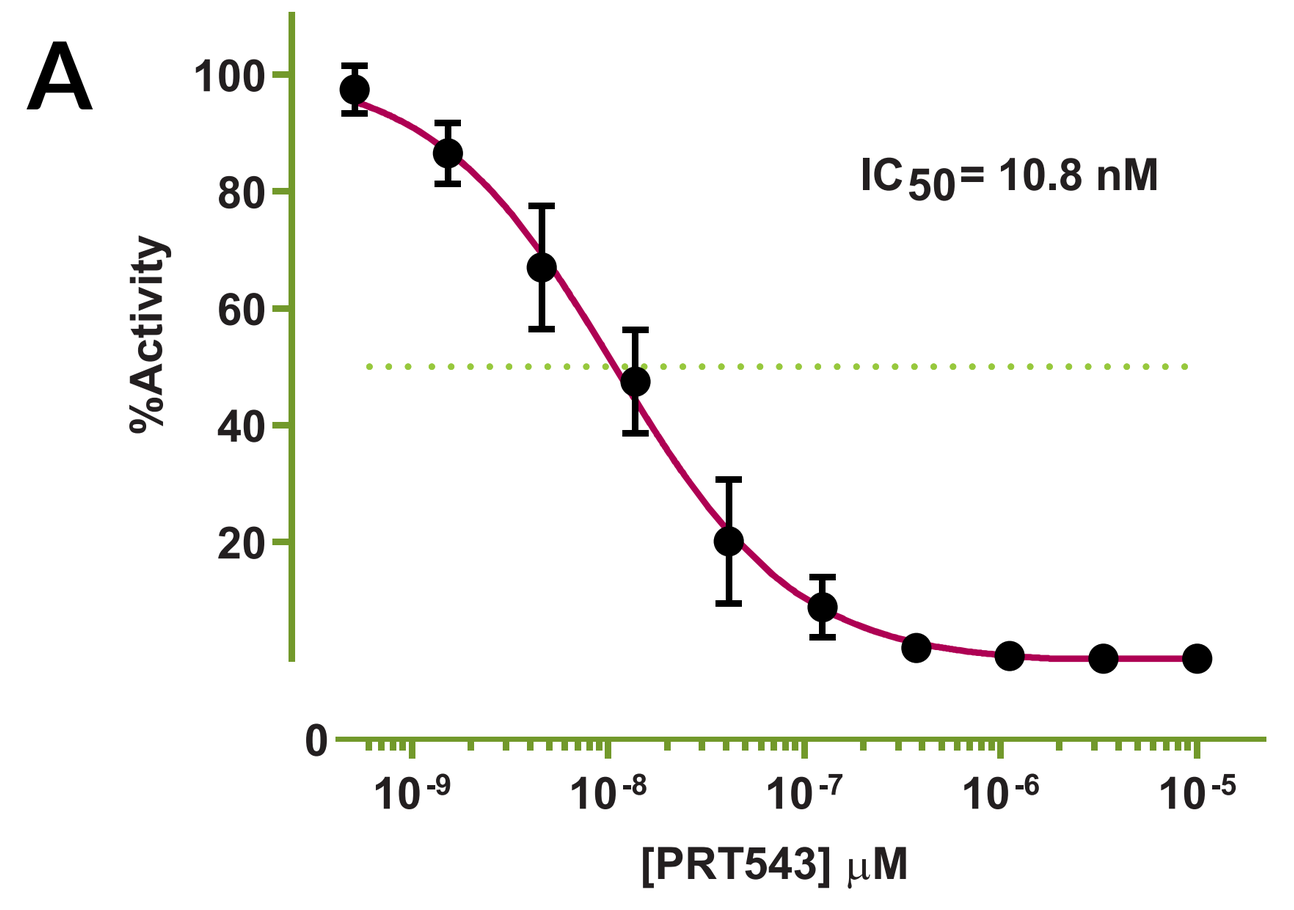
A
%Activity
IC_{50} = 10.8 nM
[PRT543] µM

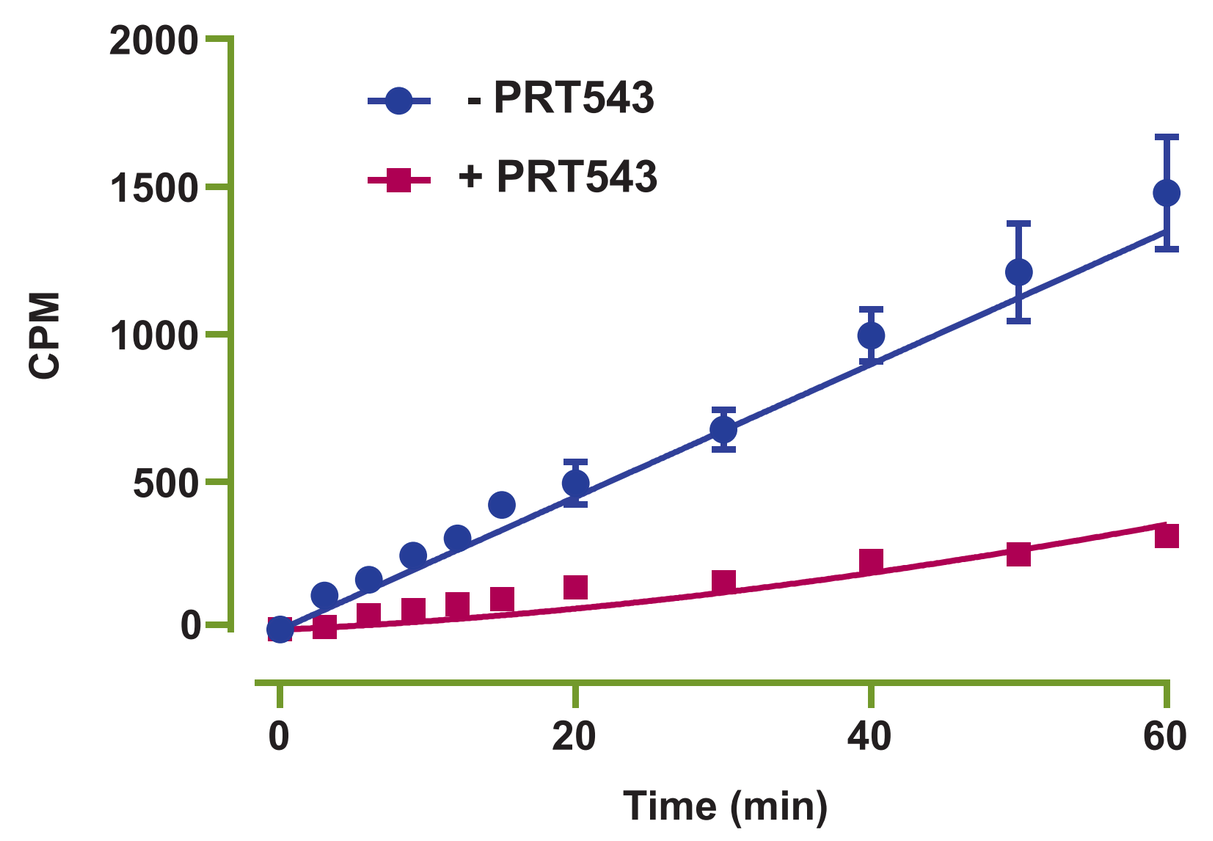
B
CPM
- PRT543
+ PRT543
Time (min)

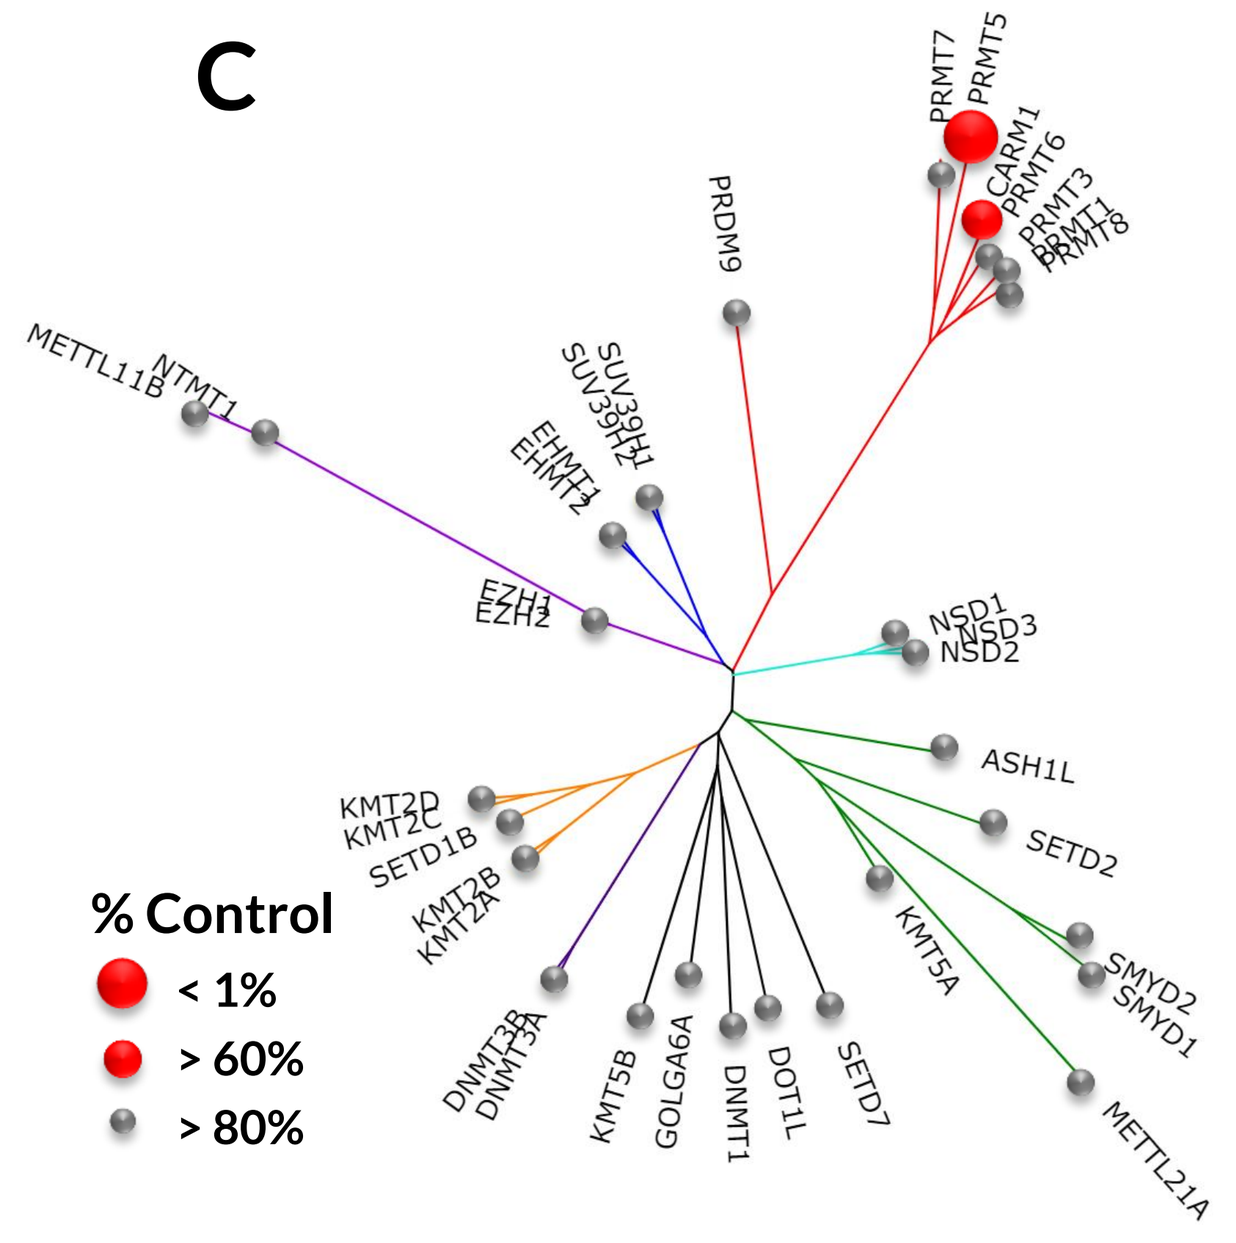
C
% Control
< 1%
> 60%
> 80%
PRMT7
PRMT5
CARM1
PRMT6
PRMT3
PRMT1
PRMT8
PRDM9
SUV39H1
SUV39H2
EHMT1
EHMT2
EZH1
EZH2
METTL11B
NTMT1
NSD1
NSD3
NSD2
ASH1L
SETD2
SMYD2
SMYD1
METTL21A
KMT5A
SETD7
DOT1L
DNMT1
GOLGA6A
KMT5B
DNMT3B
DNMT3A
KMT2B
KMT2A
SETD1B
KMT2D
KMT2C

Figure 1: A. Concentration-dependent inhibition of PRMT5/MEP50 enzymatic activity by PRT543 in a scintillation proximity based radiometric assay. Data represent mean ± SD. n = 11. **B**. Jump dilution assay showing PRMT5/MEP50 enzymatic progress curve in the absence and presence of PRT543. PRT543 is determined to be a reversible PRMT5 inhibitor with a slow off-rate and long residence time (n = 2). **C.** Biochemical selectivity of PRT543 against 37 human methyltransferases. Percent control represents % enzymatic activity remaining in the presence of 10 µM PRT543 relative to DMSO control

PRT543 POTENTLY INHIBITS CELLULAR PROLIFERATION AND SDMA IN MULTIPLE CELL LINES

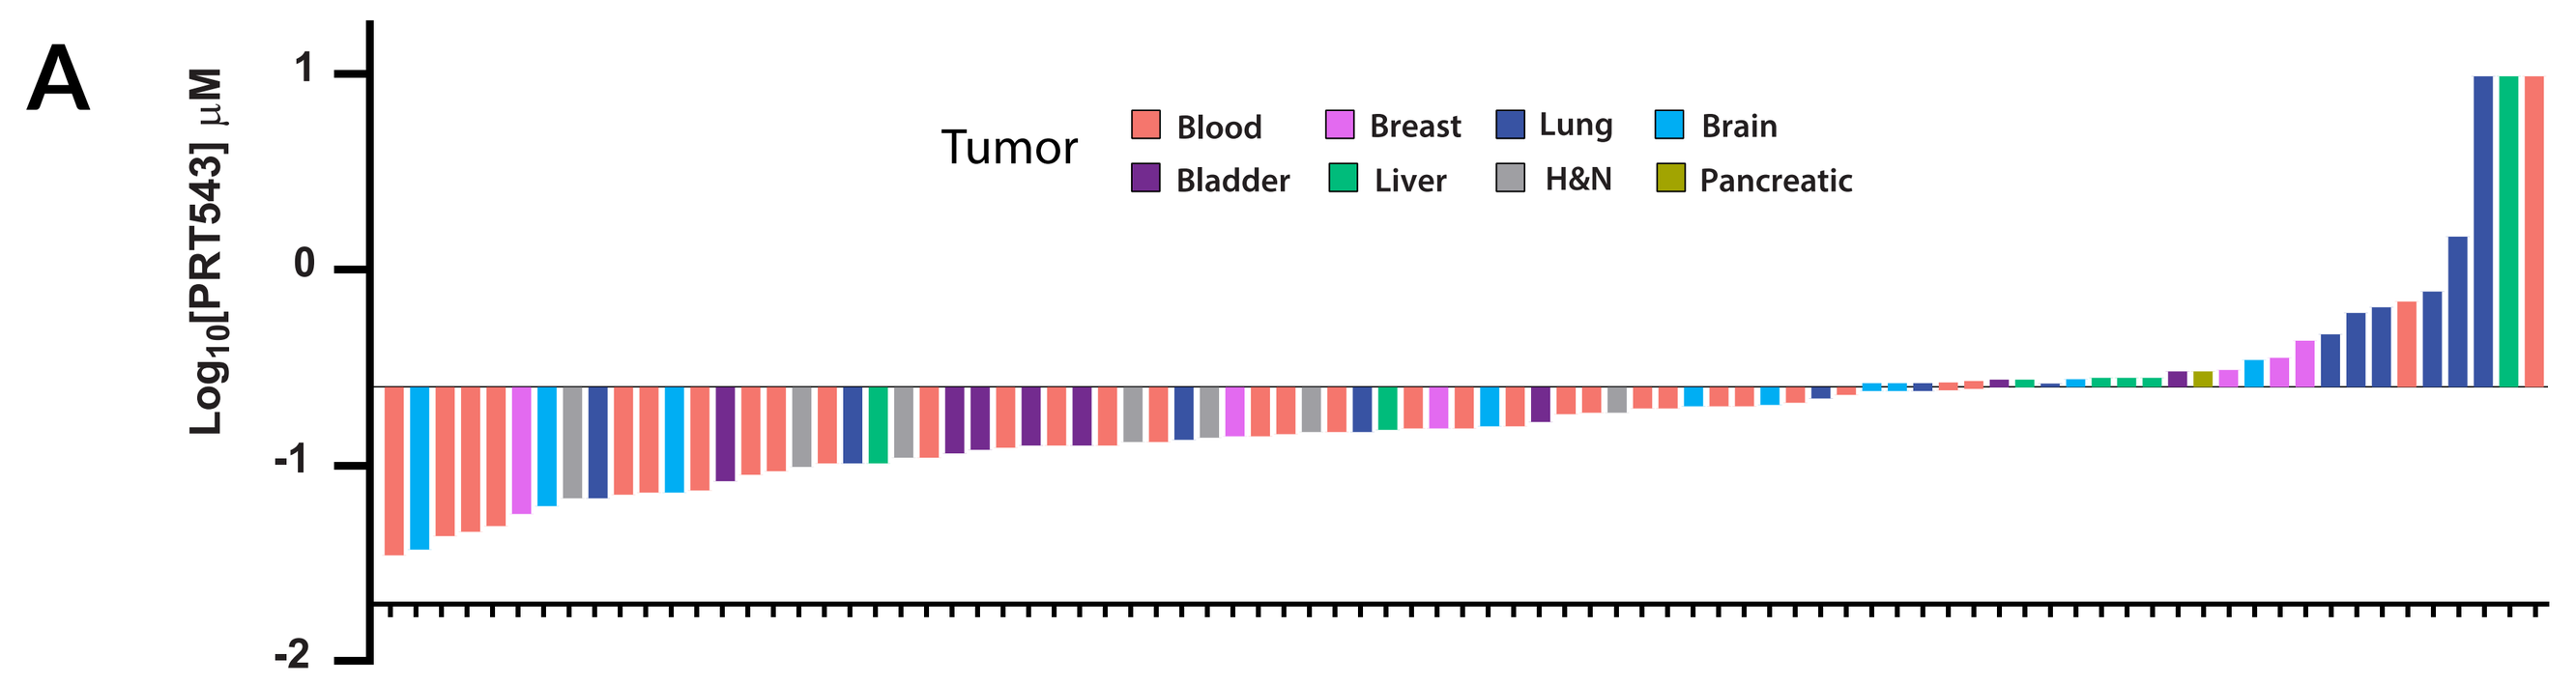
A
Log_{10}[PRT543] µM
Tumor
Blood
Bladder
Breast
Liver
Lung
H&N
Brain
Pancreatic

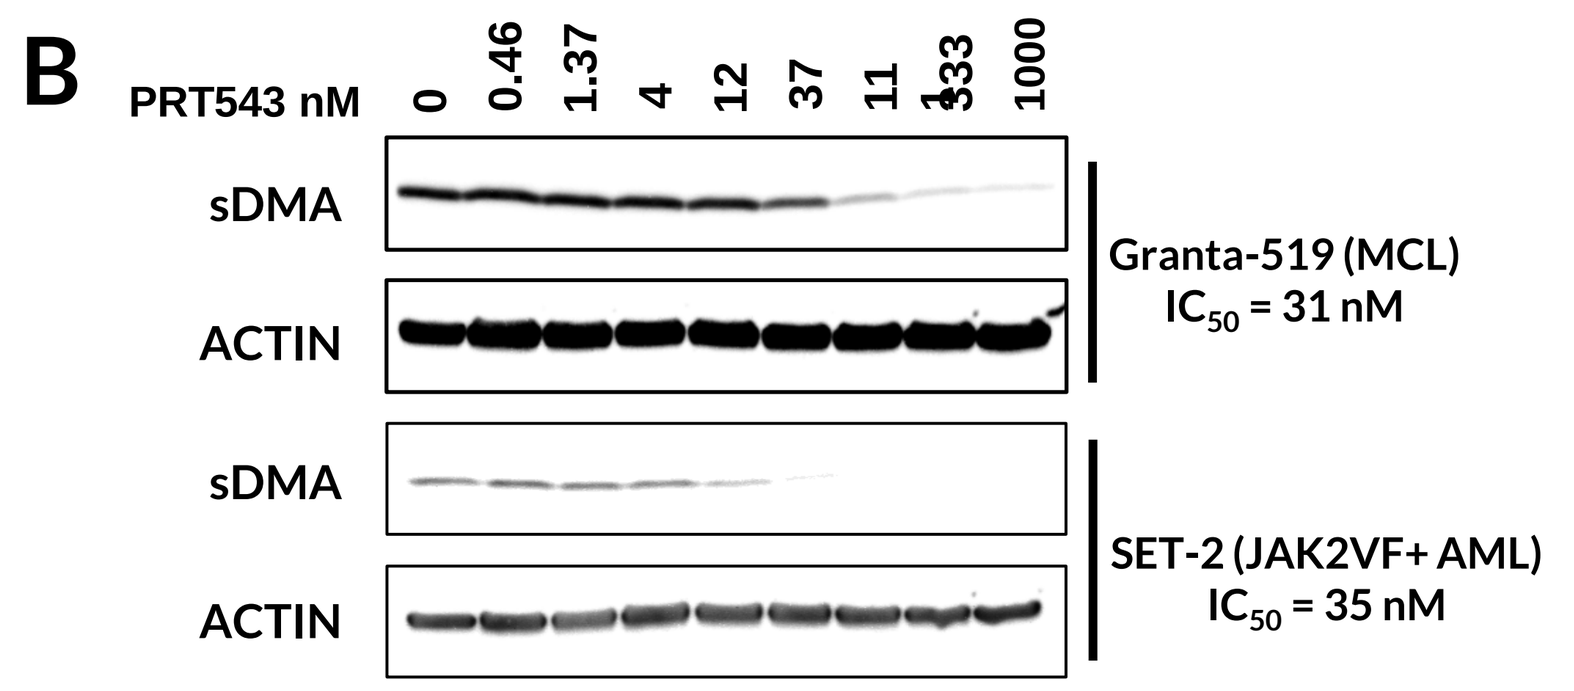
B
PRT543 nM
0
0.46
1.37
4
12
37
11
333
1000
sDMA
ACTIN
Granta-519 (MCL)
IC_{50} = 31 nM
sDMA
ACTIN
SET-2 (JAK2VF+ AML)
IC_{50} = 35 nM

Figure 2: A. Profile of anti-proliferative response to PRT543 in a panel of 85 cell lines following 10 days of treatment. Baseline corresponds to IC_{50} = 250 nM **B.** Western blot demonstrating concentration-dependent reduction of symmetrically demethylated SMD3, a known PRMT5 substrate, following 3 days of PRT543 treatment in indicated cell lines.

PRT543 DEMONSTRATES POTENT ANTI-TUMOR ACTIVITY IN MULTIPLE CELL LINE-DERIVED XENOGRAFT MODELS

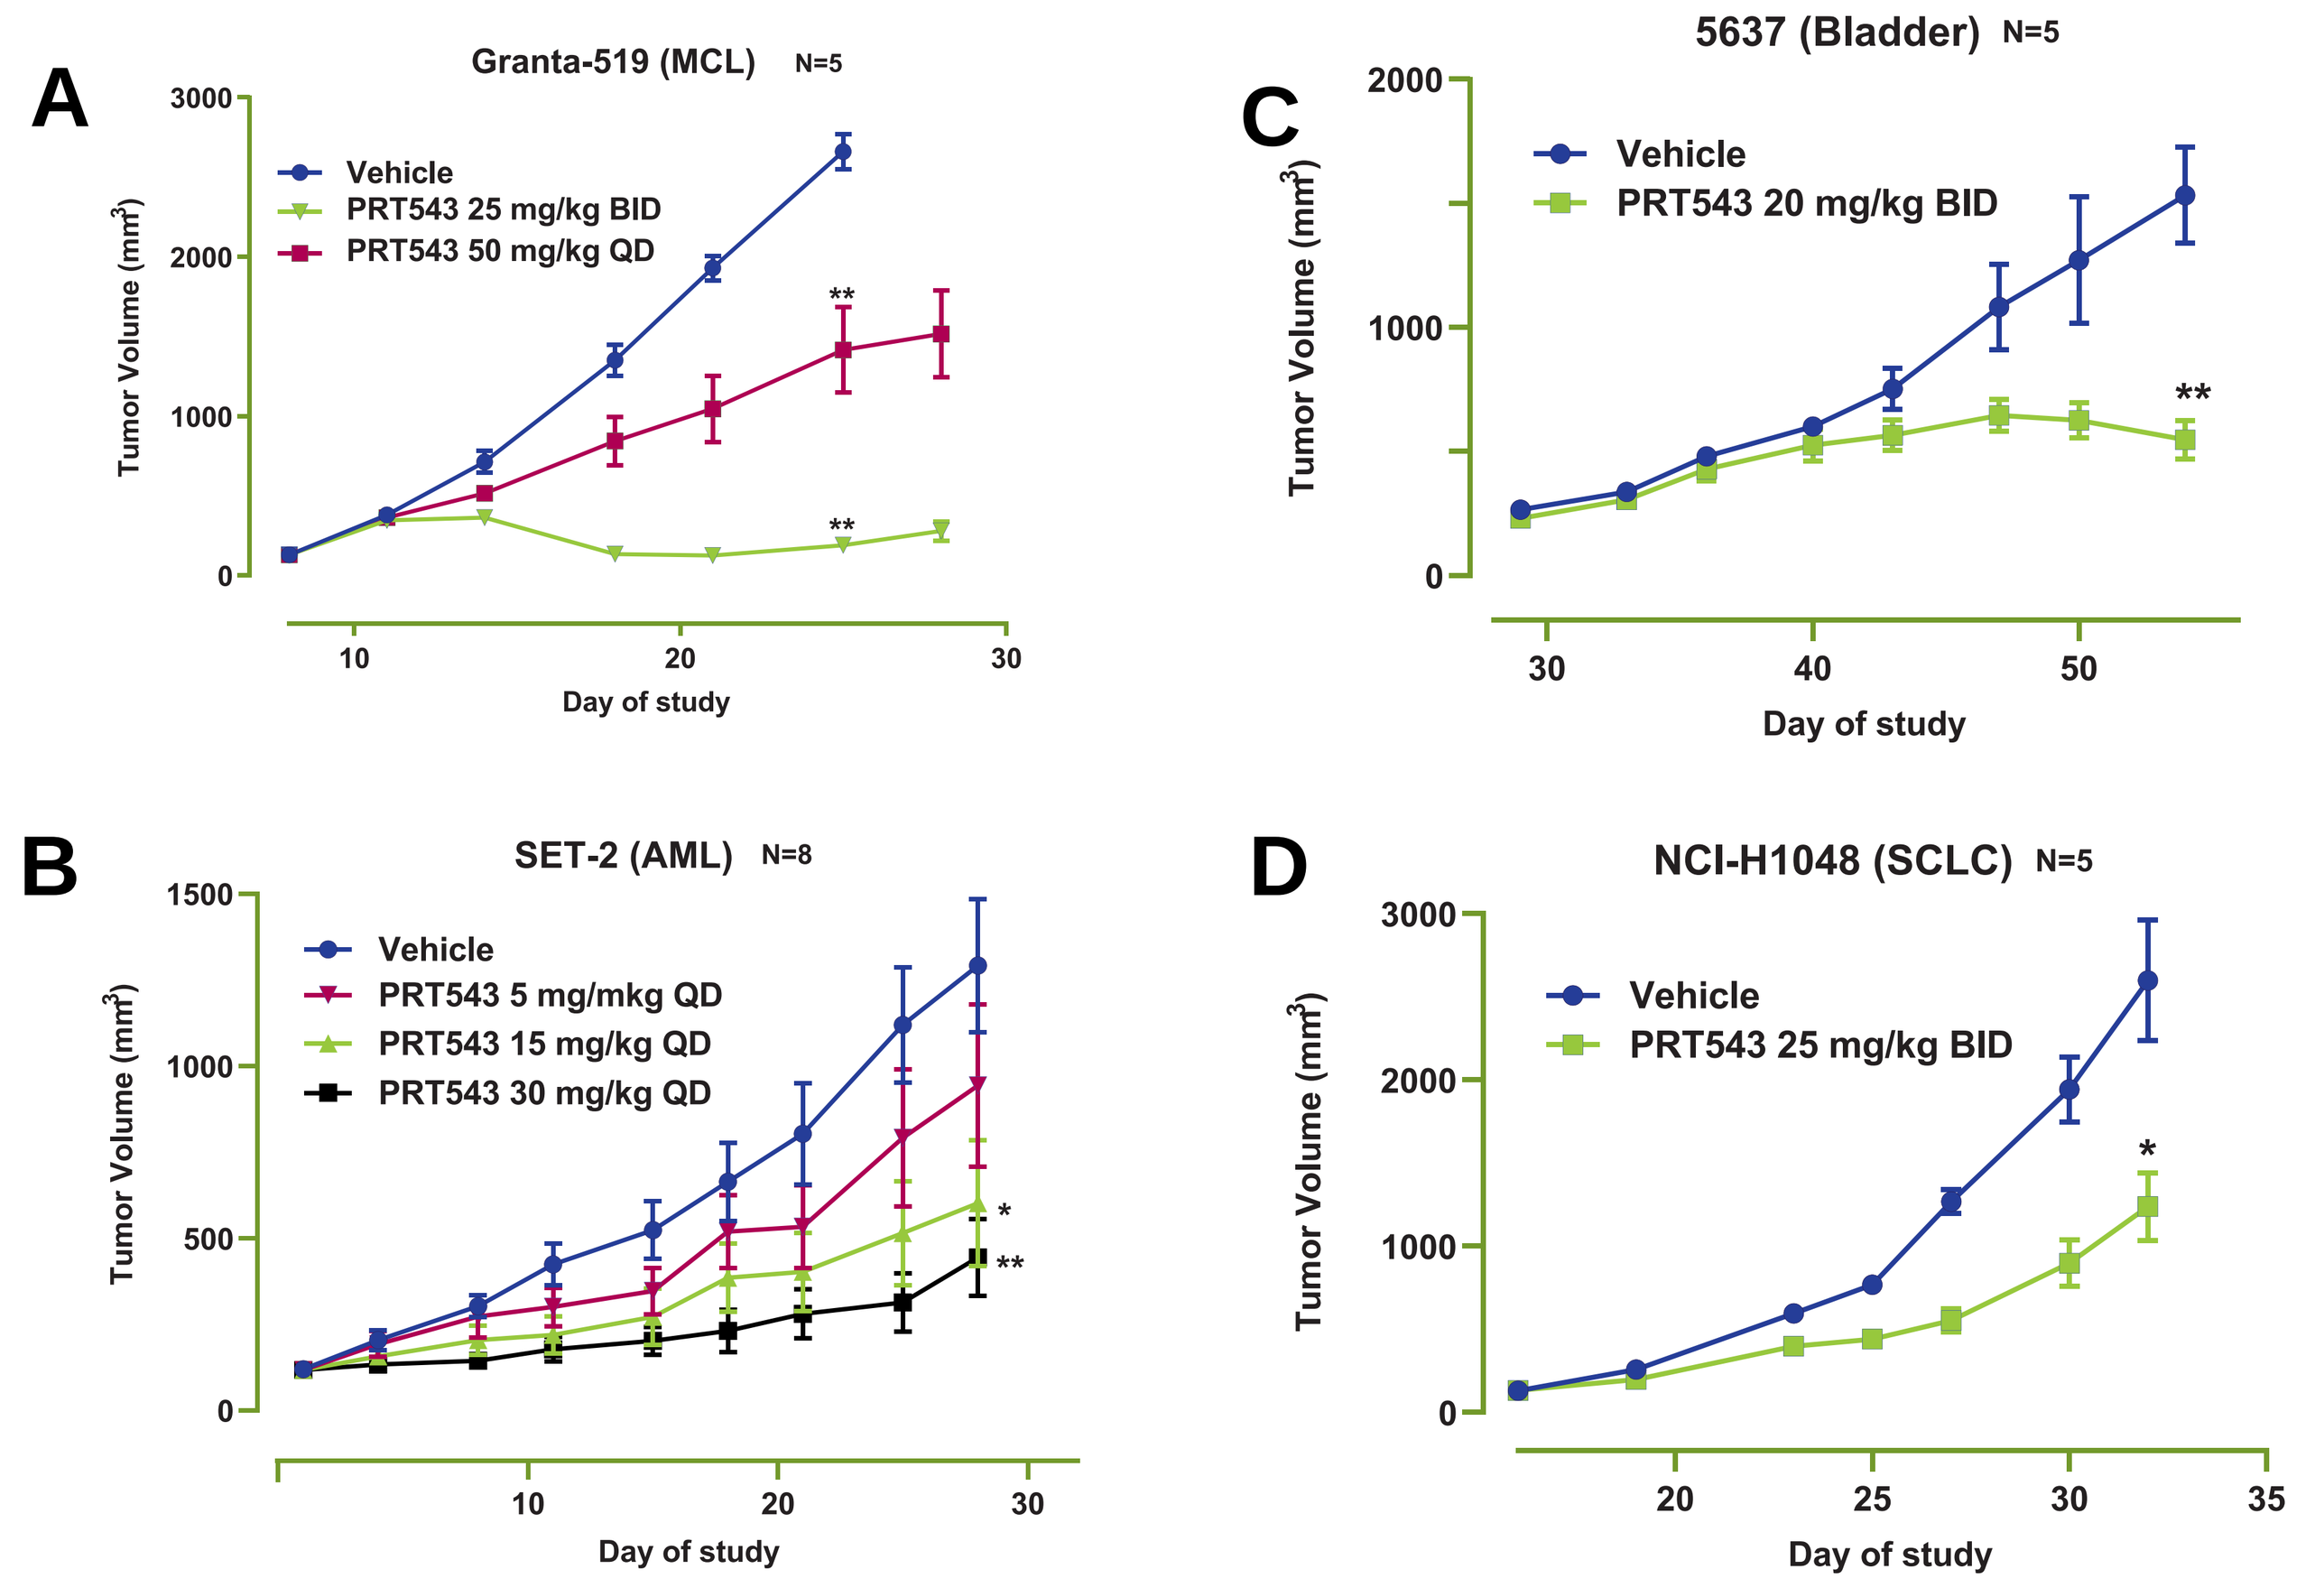
A
Granta-519 (MCL) N=5
Tumor Volume (mm^3)
Vehicle
PRT543 25 mg/kg BID
PRT543 50 mg/kg QD
Day of study
C
5637 (Bladder) N=5
Vehicle
PRT543 20 mg/kg BID
B
SET-2 (AML) N=8
Vehicle
PRT543 5 mg/mkg QD
PRT543 15 mg/kg QD
PRT543 30 mg/kg QD
D
NCI-H1048 (SCLC) N=5
Vehicle
PRT543 25 mg/kg BID

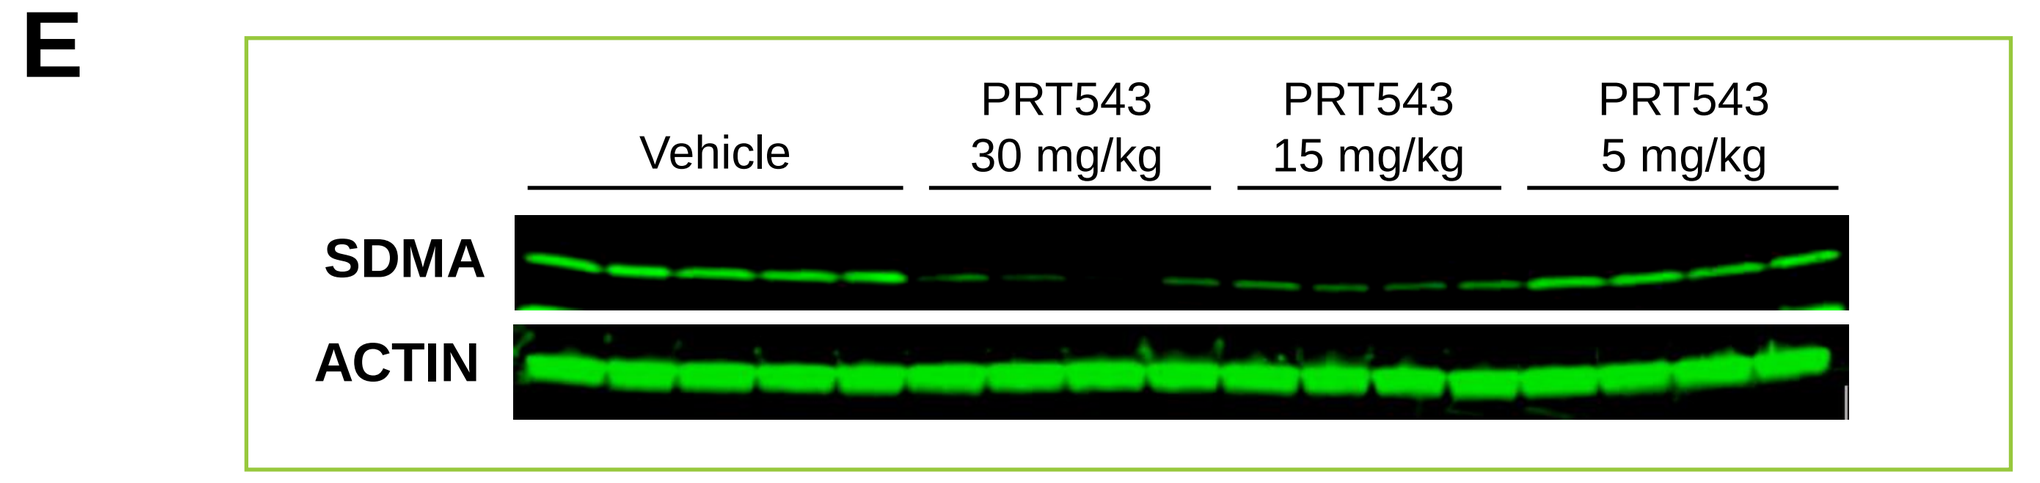
E
Vehicle
PRT543 30 mg/kg
PRT543 15 mg/kg
PRT543 5 mg/kg
SDMA
ACTIN

Figure 3: Oral administration of PRT543 leads to dose-dependent tumor growth inhibition in the **A**. Granta-519 MCL, **B**. SET-2 AML, **C**. 5637 bladder cancer, and **D**. NCI-H1048 SCLC xenograft model. Data represent mean ± SEM. **E**. Western blot showing sDMA reduction in SET-2 tumor tissue collected 4 hours after the last dose, at the end of a 28 day study. * *P* < 0.05, ** *P* < 0.01 vs. vehicle by Mann-Whitney U test.

PRT543 DEMONSTRATES SYNERGY IN COMBINATION WITH VENETOCLAX IN VITRO AND IN VIVO

A

[Venetoclax] nM (columns); [PRT543] nM (rows)

Granta-519 — ZIP score: **11**

[PRT543] nM	0	8	16	31	63	125	250	500
500	90	91	87	75	40	4	1	1
250	86	83	79	73	41	4	0	0
125	85	81	82	77	45	5	0	0
63	85	84	82	73	49	5	0	0
31	89	90	86	80	55	7	1	0
16	91	90	86	82	55	8	1	0
8	95	93	82	77	56	10	1	0
0	96	99	98	95	78	32	6	3

Z-138 — ZIP score: **9.6**

[PRT543] nM	0	8	16	31	63	125	250	500
333	77	75	73	55	3	-1	-1	0
167	80	81	74	51	9	-1	-1	-1
83	80	78	76	67	13	-1	-1	-1
42	77	86	83	76	31	-1	-1	-1
21	87	88	82	85	37	-1	-2	-1
10	87	86	91	84	54	-1	-2	-1
3	92	92	91	85	60	-1	-1	-1
0	102	99	99	98	80	0	-1	0

JVM-2 — ZIP score: **5.7**

[PRT543] nM	0	8	16	31	63	125	250	500
333	90	74	75	66	52	26	4	0
167	87	79	75	69	56	21	0	-2
83	83	65	70	63	49	21	1	-2
42	85	73	73	66	55	26	3	-2
21	86	82	71	65	53	29	5	0
10	96	88	74	76	59	32	5	0
3	99	95	83	74	56	29	5	1
0	106	93	90	74	64	33	8	4

MV4-11 — ZIP score: **7**

[PRT543] nM	0	8	16	31	63	125	250	500
500	25	22	21	14	2	1	1	1
250	51	54	45	35	2	-1	-1	-1
125	84	86	88	80	8	-1	-1	-1
63	91	93	95	96	26	-1	-1	-1
31	93	95	97	98	54	-1	-1	-1
16	96	97	99	99	60	0	-1	-1
8	96	94	97	99	64	0	-1	-1
0	96	100	101	101	84	1	0	1

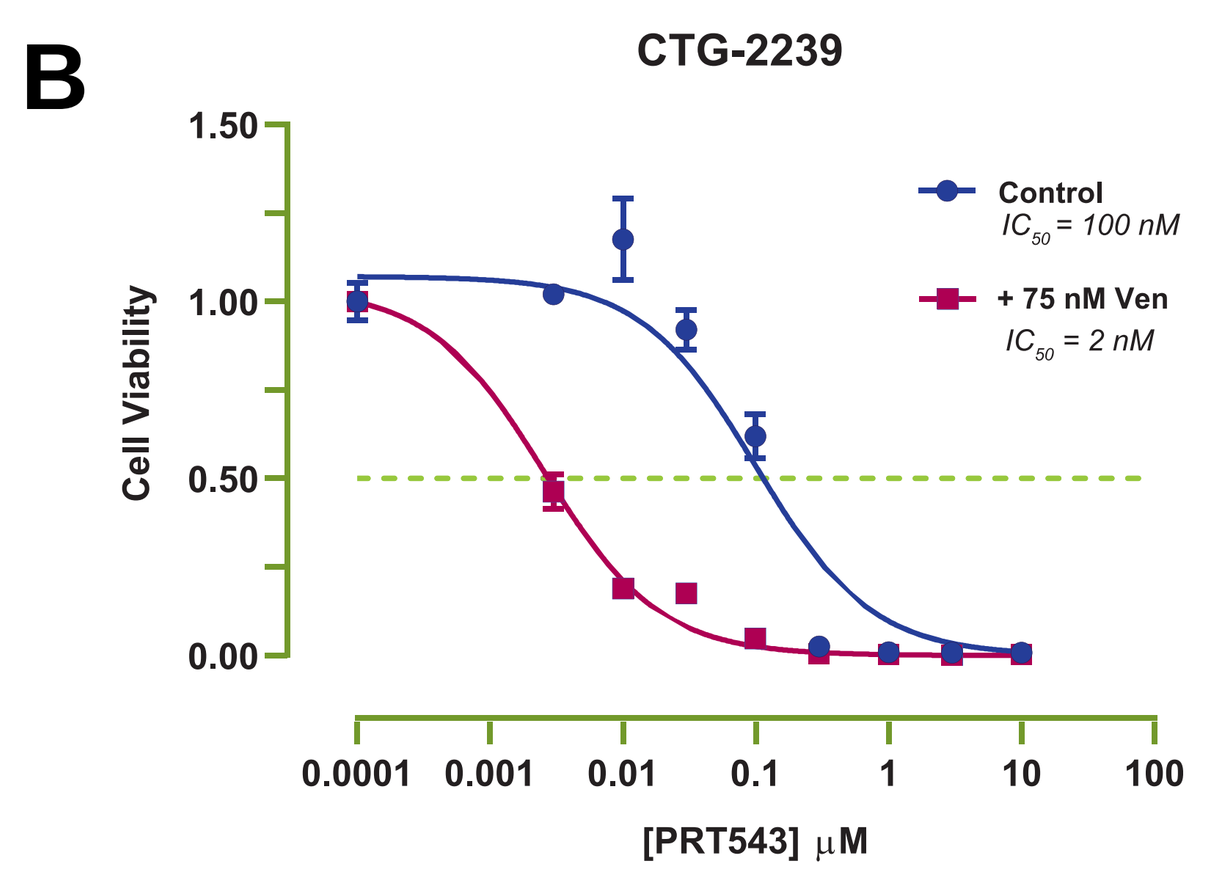
B
CTG-2239
Cell Viability
Control
IC_{50} = 100 nM
+ 75 nM Ven
IC_{50} = 2 nM
[PRT543] µM

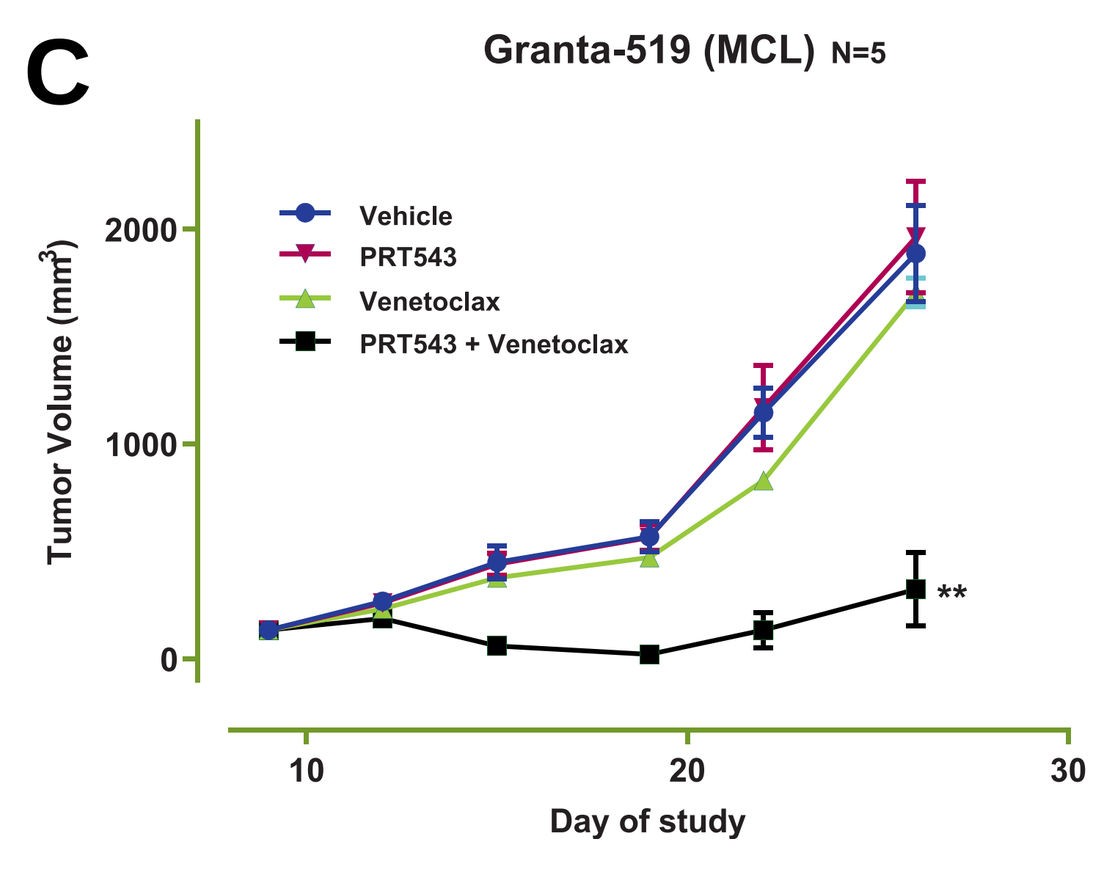
C
Granta-519 (MCL) N=5
Tumor Volume (mm^3)
Vehicle
PRT543
Venetoclax
PRT543 + Venetoclax
Day of study

Figure 4: A. Cell lines were treated with increasing concentrations of PRT543 for 10 days, with the addition of venetoclax at indicated concentrations for the last 2 days. **B**. Primary AML blasts isolated by leukapheresis were treated with increasing concentrations of PRT543, with and without 75 nM venetoclax for 10 days in a 2-D assay **C.** Oral administration of a combination of PRT543 and venetoclax results in significant tumor growth inhibition at doses that did not show efficacy as monotherapy for both agents in the Granta-519 CDX model. Data represent mean ± SEM. ** *P* < 0.01 vs. Vehicle by Mann-Whitney U test

PRT543 REDUCES MYELOPROLIFERATION IN PRECLINICAL MODELS OF MPN AND IS WELL-TOLERATED IN COMBINATION WITH RUXOLITINIB

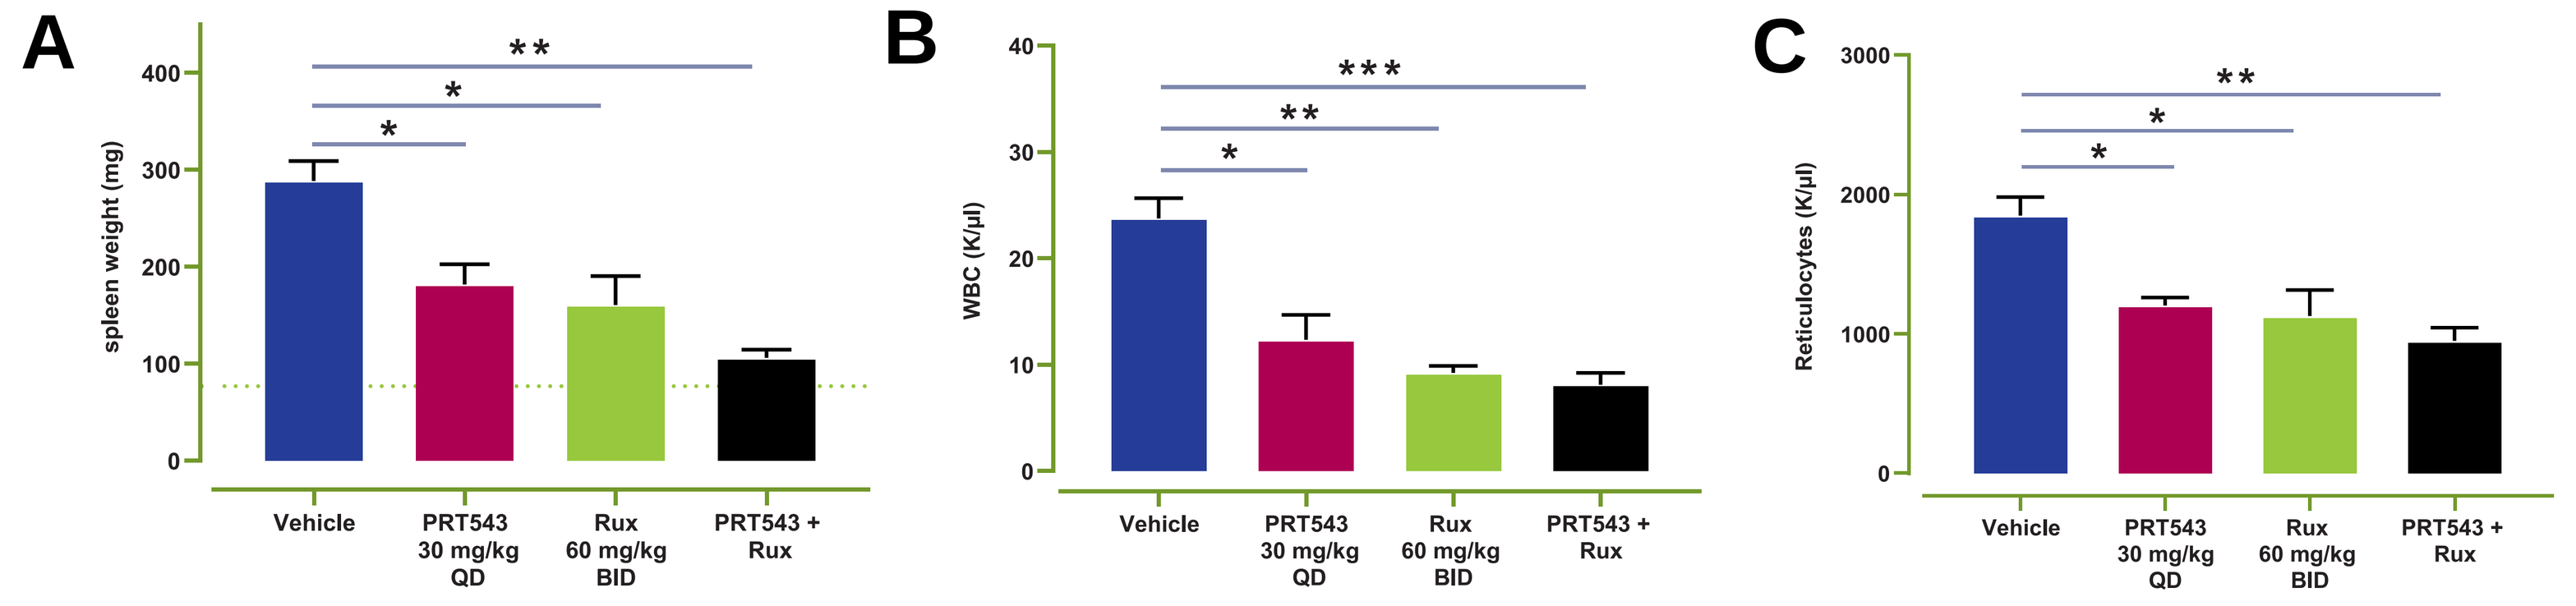
A
spleen weight (mg)
B
WBC (K/µl)
C
Reticulocytes (K/µl)
Vehicle
PRT543 30 mg/kg QD
Rux 60 mg/kg BID
PRT543 + Rux

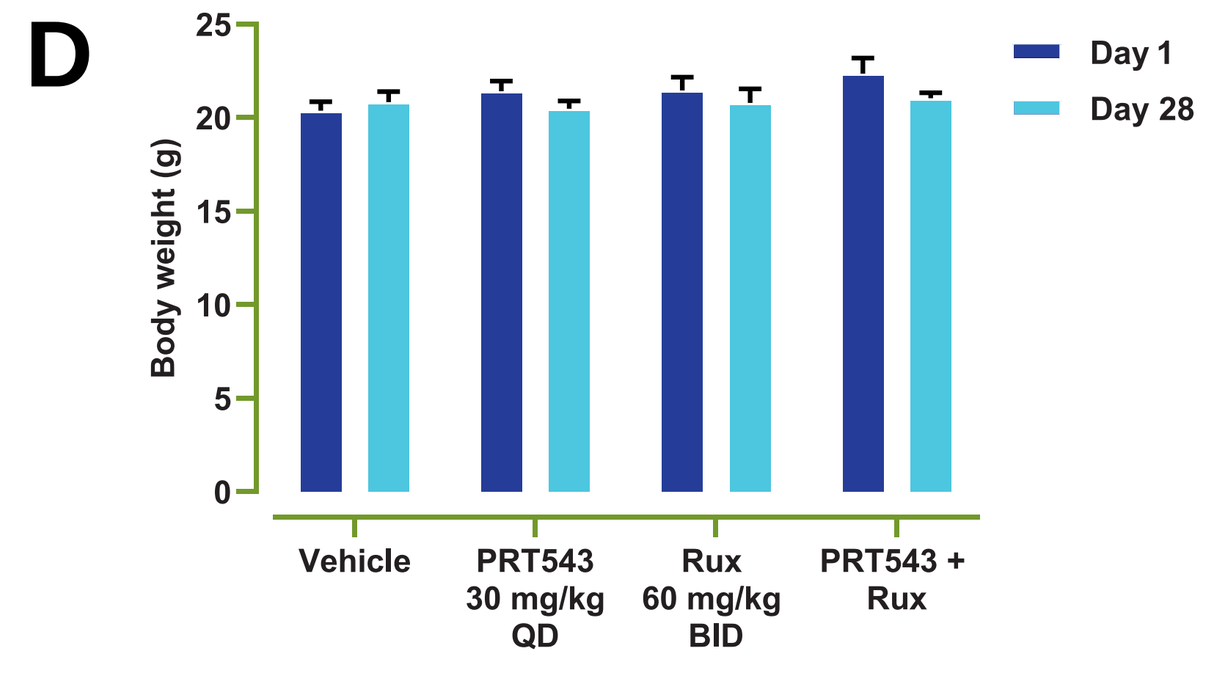
D
Body weight (g)
Day 1
Day 28
Vehicle
PRT543 30 mg/kg QD
Rux 60 mg/kg BID
PRT543 + Rux

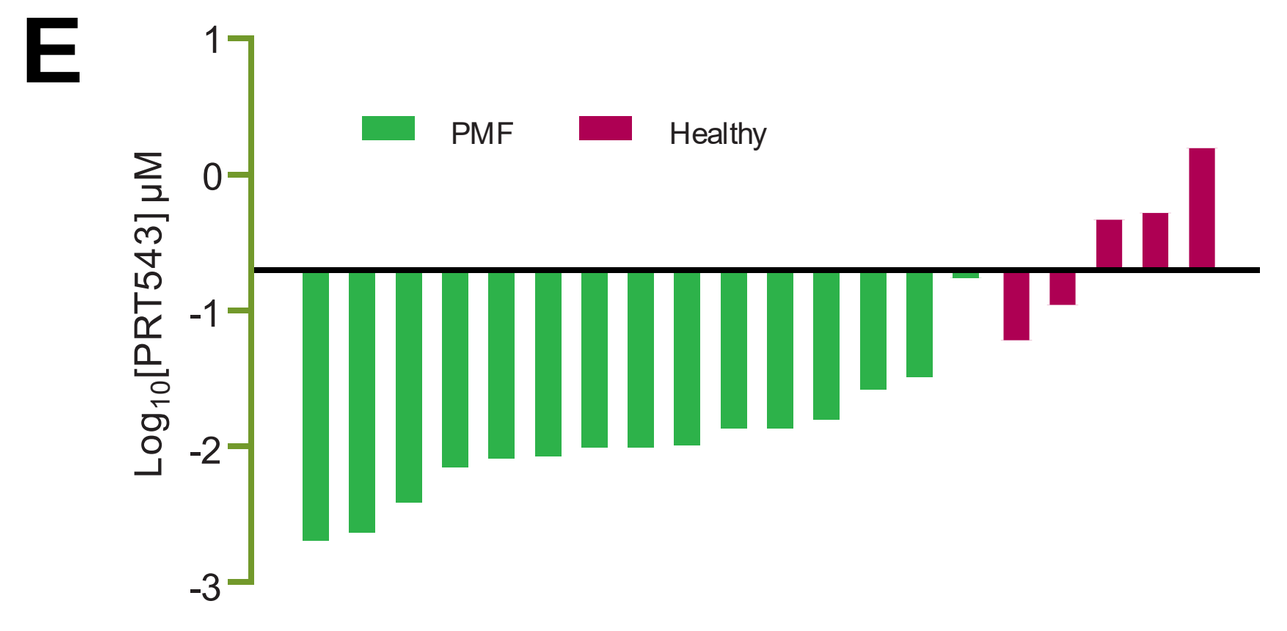
E
Log_{10}[PRT543] µM
PMF
Healthy

Figure 5: Oral administration of PRT543 as monotherapy and in combination with ruxolitinib led to significant decrease in **A.** spleen size, **B**. white blood cell counts, and **C**. reticulocytes in the JAK2VF bone marrow transplant model of polycythemia vera. Data represents mean ± SEM. Dotted line in **A** indicates mean spleen weight of WT transplanted mice. * *P* < 0.05, ** *P* < 0.01, *** *P* < 0.001 vs. vehicle by Mann-Whitney U test. **E**. Body weight changes over course of study indicate combination was well-tolerated. **F.** Primary BM mononuclear cells from PMF patients and healthy controls were treated ex vivo with increasing concentrations of PRT543 for 10 days and viability was assessed by an MTS assay. Baseline corresponds to IC_{50} = 200 nM. PMF: Primary myelofibrosis

PRT543 CAN OVERCOME RESISTANCE TO MULTIPLE TARGETED THERAPIES IN MYELOID MALIGNANCIES

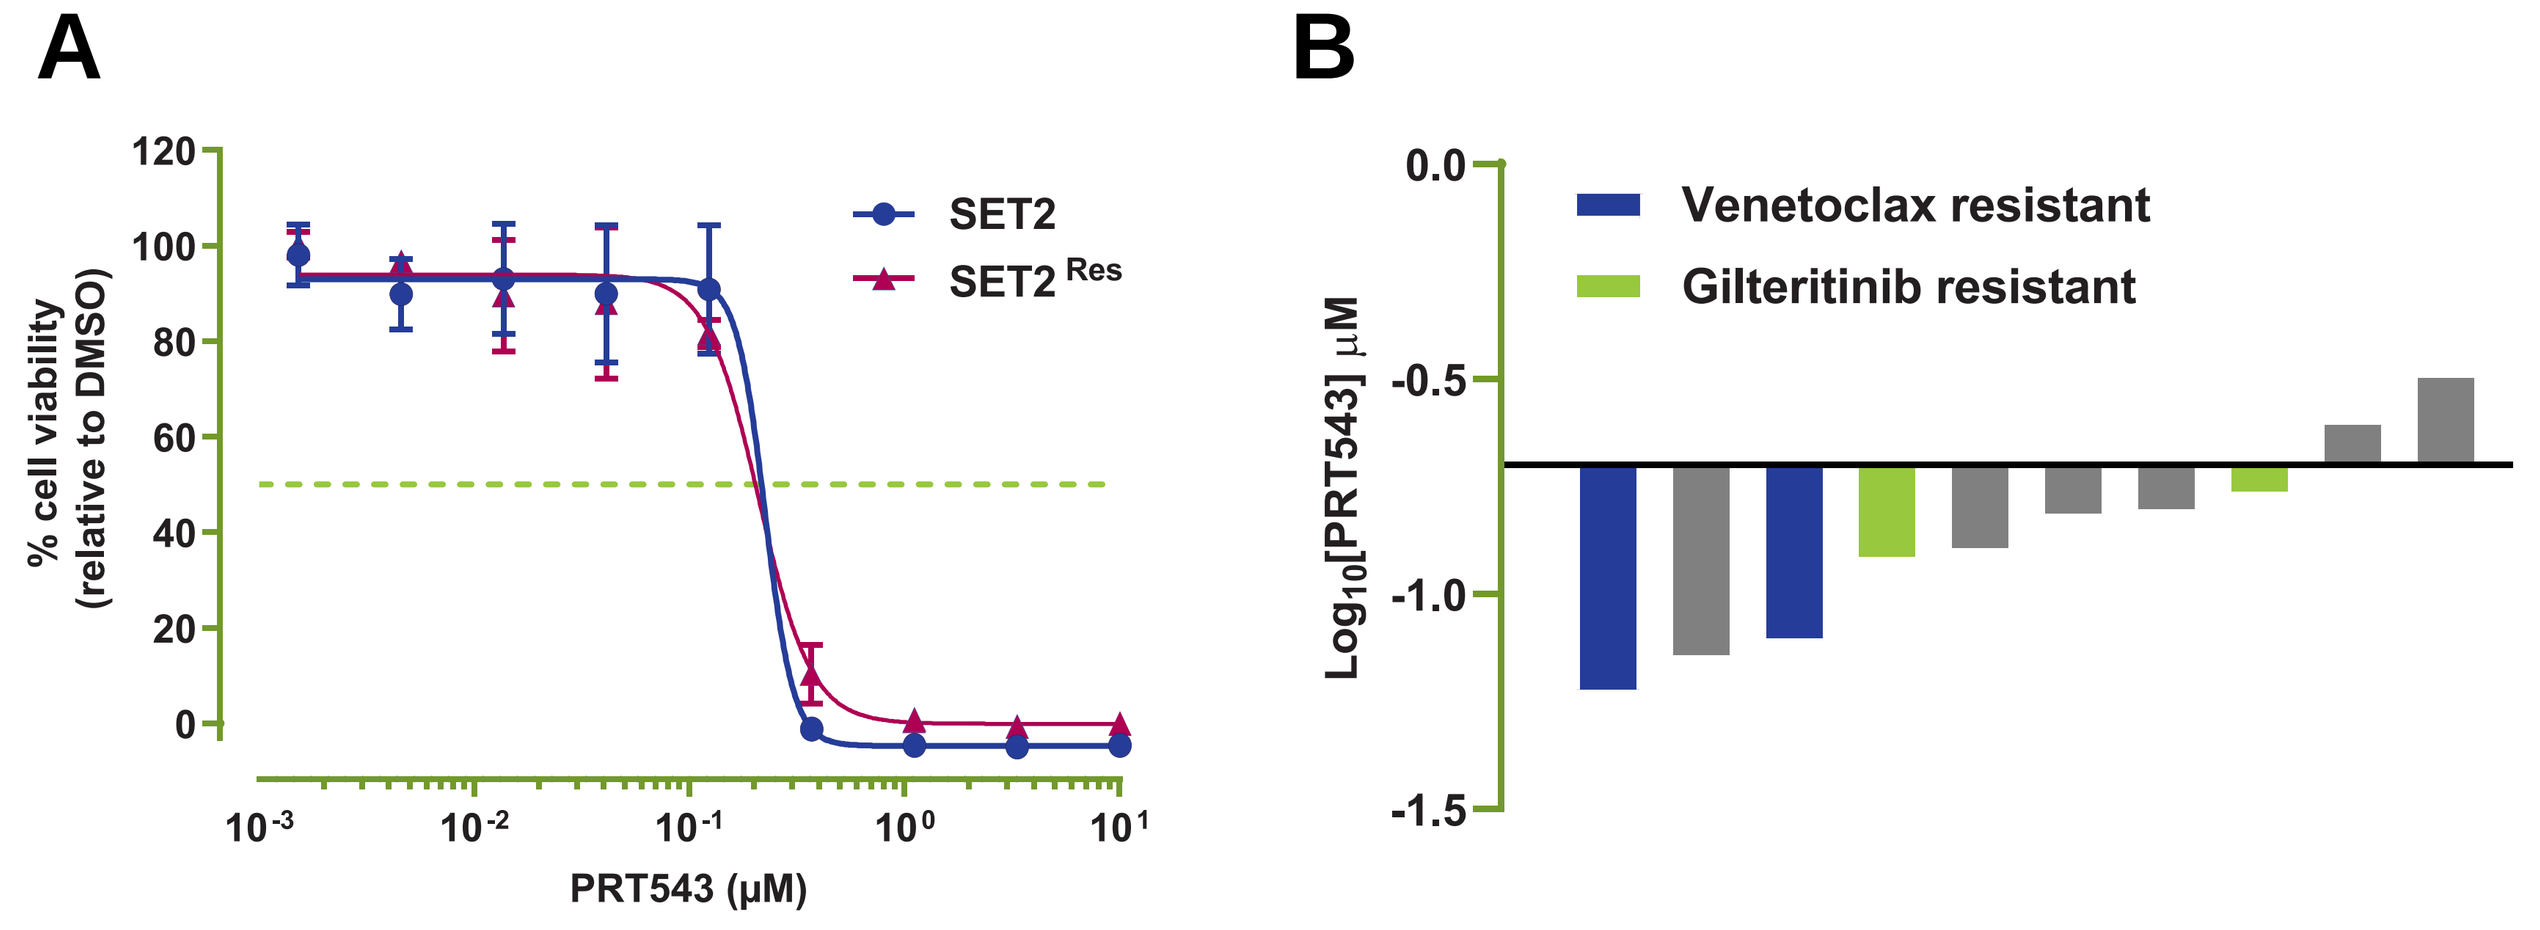
A
% cell viability (relative to DMSO)
SET2
SET2 Res
PRT543 (µM)
B
Log_{10}[PRT543] µM
Venetoclax resistant
Gilteritinib resistant

Figure 6: A. Ruxolitinib resistant SET-2 cells (SET-2[Res]) were generated by prolonged exposure to inhibitor. Parental and resistant SET-2 cells are equally sensitive to PRT543 treatment. **B**. Ex vivo PRT543 treatment demonstrated concentration-dependent inhibition of proliferation in a 2-D 10-day assay of primary AML blasts including FLT3 mutant samples that were resistant to gilteritinib (green bars) and samples that were resistant to venetoclax (blue bars). Baseline corresponds to IC_{50} = 200 nM

PRT543 DEMONSTRATES POTENT ACTIVITY IN SOLID TUMOR PDX MODELS

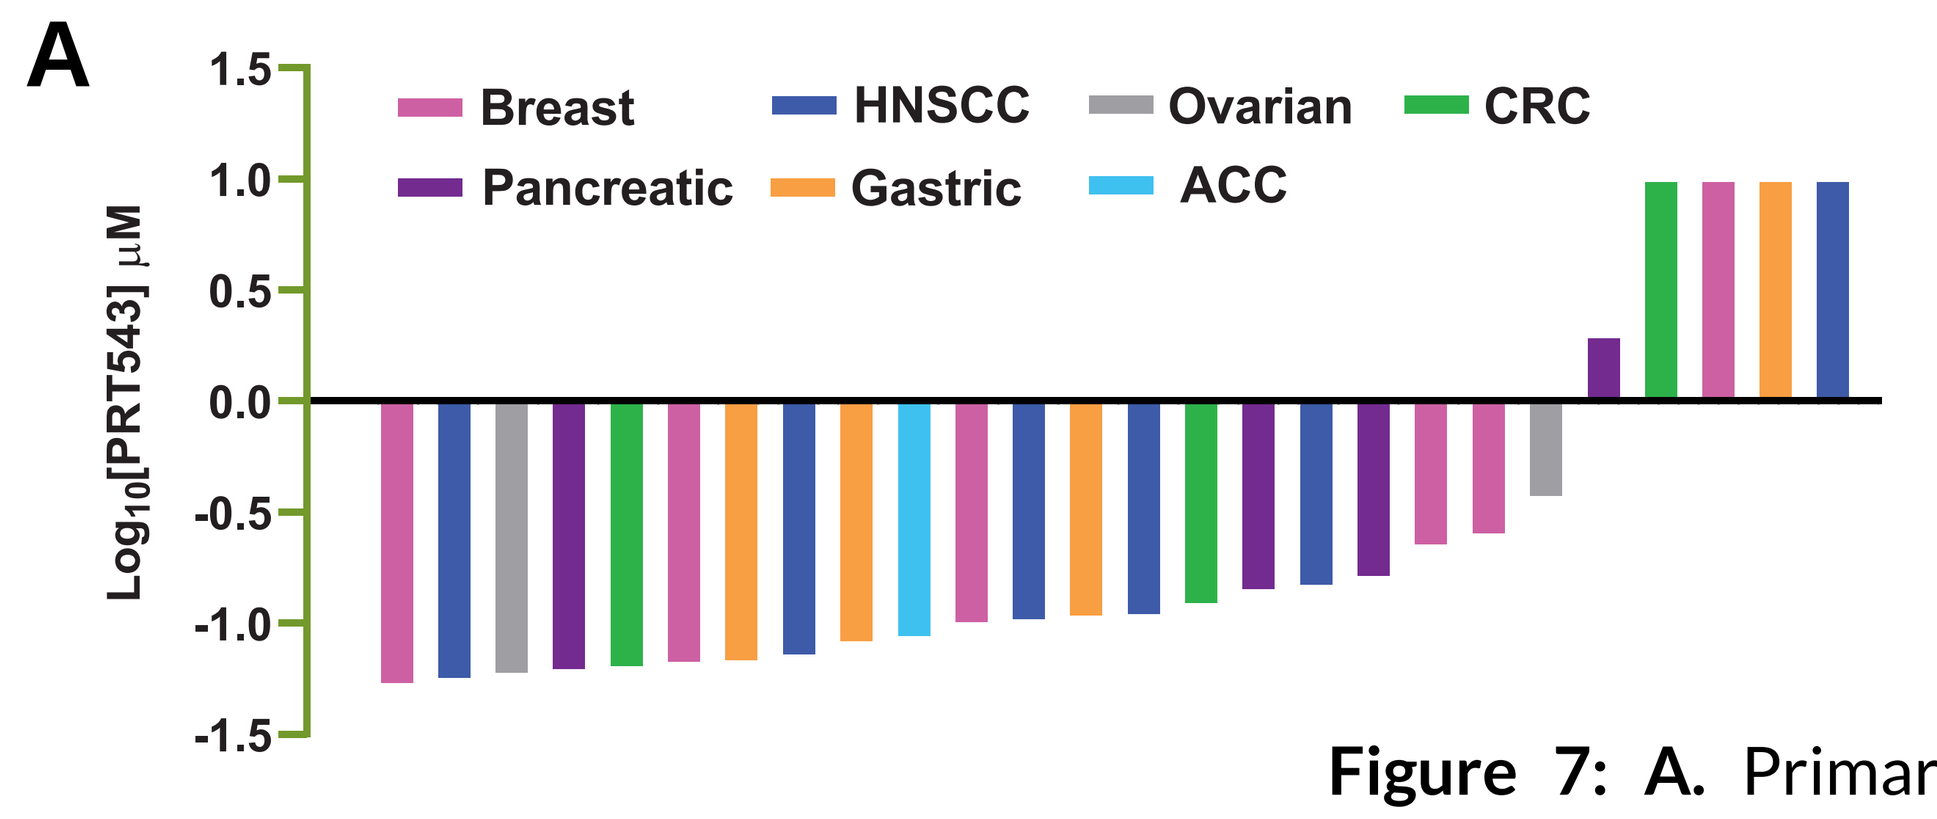
A
Log_{10}[PRT543] µM
Breast
Pancreatic
HNSCC
Gastric
Ovarian
ACC
CRC

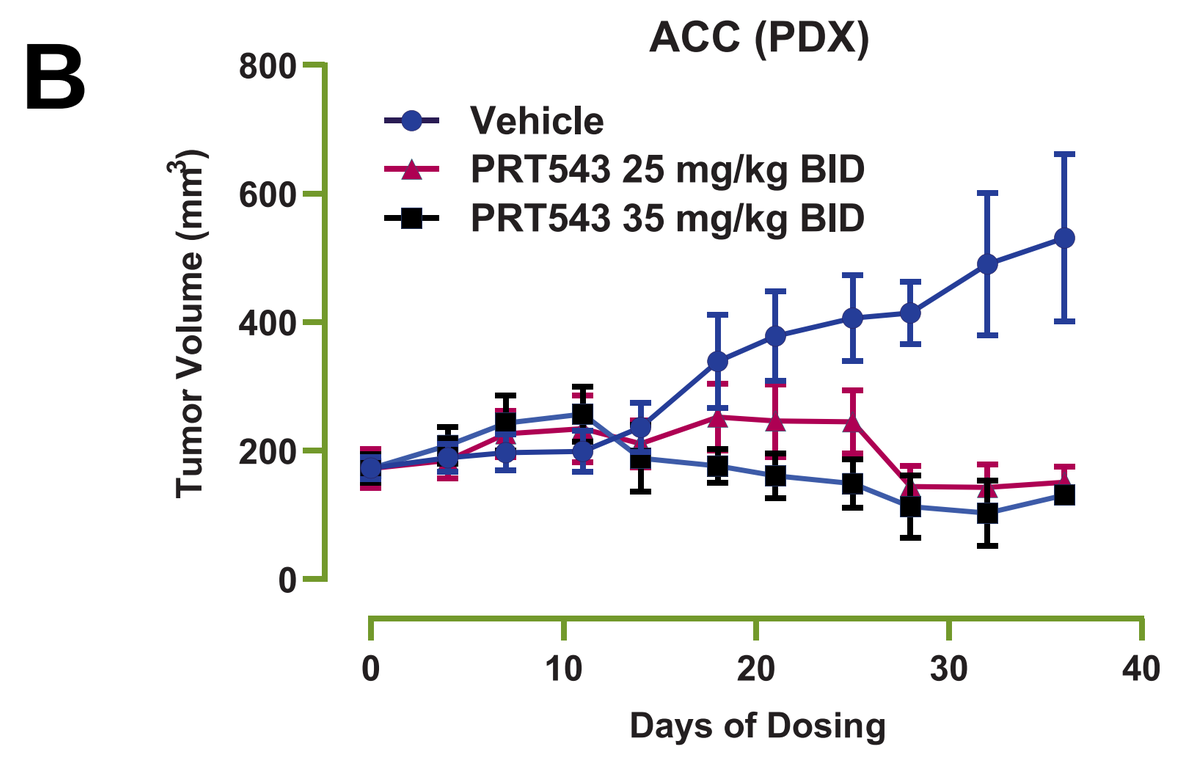
B
ACC (PDX)
Tumor Volume (mm^3)
Vehicle
PRT543 25 mg/kg BID
PRT543 35 mg/kg BID
Days of Dosing

Figure 7: A. Primary solid tumor PDX samples were treated with increasing concentrations of PRT543 ex vivo for 10 days in a 2-D assay and viability was assessed using a CellTiter Glo assay. Baseline corresponds to IC_{50} = 1 µM. **B.** Oral administration of PRT543 demonstrates potent anti-tumor efficacy in a PDX model of adenoid cystic carcinoma.

CONCLUSIONS

- **PRT543 is a potent and selective PRMT5 inhibitor.**
- **PRT543 demonstrates potent activity as monotherapy and in combination with standard of care therapies in solid and hematologic malignancies.**
- **PRT543 can overcome resistance to multiple targeted therapies, particularly in myeloid malignancies.**
- **PRT543 is currently under evaluation in a Phase I clinical trial in patients with advanced solid tumors and hematologic malignancies (NCT03886831).**